PIII-10

Application of Real-Time Data Assembly (RTDA) to a Pivotal Phase III Pediatric Trial: A Proactive Approach to Population Pharmacokinetic/Pharmacodynamic (PK/PD) Dataset Creation

CM Rubino,[1] ME McPhee,[1] M Vo,[1] GL Jungbluth[2]

[1]Cognigen Corporation, Buffalo, NY and [2]Pharmacia Corporation, Kalamazoo , MI

ABSTRACT

Purpose.
To implement an RTDA process, similar to that described in the FDA Guidance for Industry: Population Pharmacokinetics, during a Phase III trial of linezolid (IV to oral) in pediatric patients which utilized an every 8 hour dosing regimen.

Methods.
During study enrollment, data was transmitted monthly; data merges were performed to generate population PK-specific queries; queries were communicated for resolution. Descriptive figures and tables were provided to sponsor for internal discussions. The impact of the new dosing regimen on linezolid plasma concentrations was evaluated on a monthly basis as well. As it was not known at the start of the study how many patients would have PK samples drawn during administration of the oral suspension, this was monitored throughout the study in order to prospectively plan the analysis.

Results.
Approximately 50 figures and queries were generated each month and over 100 data issues were resolved proactively. The complicated process for merging of concentration data to CRF data was resolved prior to data lock, saving several weeks of dataset creation time.

Conclusions.
Implementing RTDA improved data quality, reduced data exclusions, and facilitated rapid dataset creation upon data lock. This process allowed early confirmation of the appropriateness of the new dosing regimen. Dataset creation issues were proactively resolved and the analysis was prospectively designed, allowing the population PK/PD analysis to be included in the FDA submission.

INTRODUCTION

The goal of drug development is to obtain regulatory approval to market a safe and effective medication, with optimal competitive advantage and commercial potential. Regulatory approval should be secured with the most efficient and productive expenditure of resources and in the shortest timeframe possible. Knowledge accumulated during drug development needs to be readily available for global development teams to make strategic program adjustments and take advantage of the knowledge gained while studies are ongoing. A systems approach must be used to accelerate the availability of knowledge and development programs must be flexible to implement necessary changes.

Real-time data assembly (RTDA) is a rational strategy to facilitate the inclusion of important pharmacokinetic analyses into development programs. RTDA optimizes drug development by generating and integrating pharmacokinetic knowledge early in the program thereby permitting timely access to important analyses. Ultimately this knowledge-based decision making strategy improves the likelihood of drug approval by producing a well-characterized drug with a better chance of success in the market place.

In this case, pediatric studies of linezolid (LZD) had suggested that a TID dosing regimen might be more appropriate for children than the approved adult BID regimen. For this reason, the pivotal Phase III trial in children less than 11 years of age (Study 82) with resistant infections used a TID regimen. Due to the regimen change, which occurred relatively late in the pediatric development, as well as the need to be as efficient as possible in analyzing the PK/PD data, a real-time data assembly (RTDA) approach was utilized for this trial. It was hoped that this would accomplish two goals: 1) allow early confirmation of the new dosage regimen, and 2) to allow for rapid and efficient data assembly to help meet the rigorous timelines for the pediatric submission.

Quote from the FDA Guidance for Industry, Population Pharmacokinetics:

"Real-time data assembly prevents the problems that generally arise when population PK data are stored until the end of a clinical trial. Real-time data assembly permits an ongoing evaluation of site compliance with the study protocol and creates the opportunity to correct violations of study procedures and policy (32). Evaluation of pharmacokinetic data can provide the safety data monitoring board with insight into drug exposure safety evaluations and drug-drug interactions. Real-time data assembly creates the opportunity for editing the concentration-time data, drug dosing history, and covariates data in a timely manner to meet the pharmacokinetic objectives of a clinical trial (33) and to facilitate the model building process. It also allows practical analysis and development of software protocols for the final analysis, thereby saving much time in data analysis. If real-time data analysis will be implemented for an add-on population PK study, adequate policies and procedures should be in place for study blind maintenance (29)."

METHODS

Study Design

- This was a Phase III, randomized (2:1 LZD to vancomycin), open-label, comparator-control, multicenter, PK/PD study.
- Patients aged from birth to 11 years including term and preterm infants with suspected or proven resistant Gram-positive bacterial infections were enrolled in the trial.
- Initially, all LZD patients were given IV LZD over 30-120 minutes infusion. After at least three days of IV dosing under the investigator's discretion, the patients were allowed to be switched to PO LZD. Patients with documented vancomycin-resistant Enterococci (on or before Day 3) who had been randomized to vancomycin were also allowed to switch to LZD.
- Planned duration of therapy was to be at least 10 days with a maximum of 28 days.
- One plasma sample per day for the determination of LZD concentrations was drawn on Days 3, 10, 17, and 24 depending on the duration of LZD therapy.

Real-Time Data Assembly Defined

- RTDA is a prospective process that initiates data assembly and analysis while studies are ongoing. It is a structured quality assurance program for the rapid retrieval, clean up, assembly, and analysis of data during the conduct of a clinical trial.
- Data transfers can be scheduled periodically (middle and end of study), monthly, or even daily when continuously monitoring for safety.
- RTDA yields a comprehensive database for analysis of drug-drug interactions, lab data, and adverse events and can provide prompt feedback of drug exposure estimates for dose adjustments during the trial.
- As the warehouse is built, relational evaluations take place to seek out missing, invalid, or out of range data points, and sample / dosing date times that are not correct. Concentration results that are improbable in relation to randomization or time since last dose are evaluated as are missing, invalid, or out of range covariates.
- PERSPECTIVE Hypertext Data Analysis Mapping is used as a communication tool to allow for rapid dissemination of knowledge to the entire project team, including CRO and Sponsor personnel, located at many sites.
- RTDA offers a strategy for data scrubbing and analysis before the end of a trial. Results are available for crucial program decisions, facilitating the review process and allotting additional time to prepare a fully integrated regulatory submission.

Figure 1: Schematic Describing the RTDA Process for the Pivotal Phase III Trial of LZD in Pediatric Patients

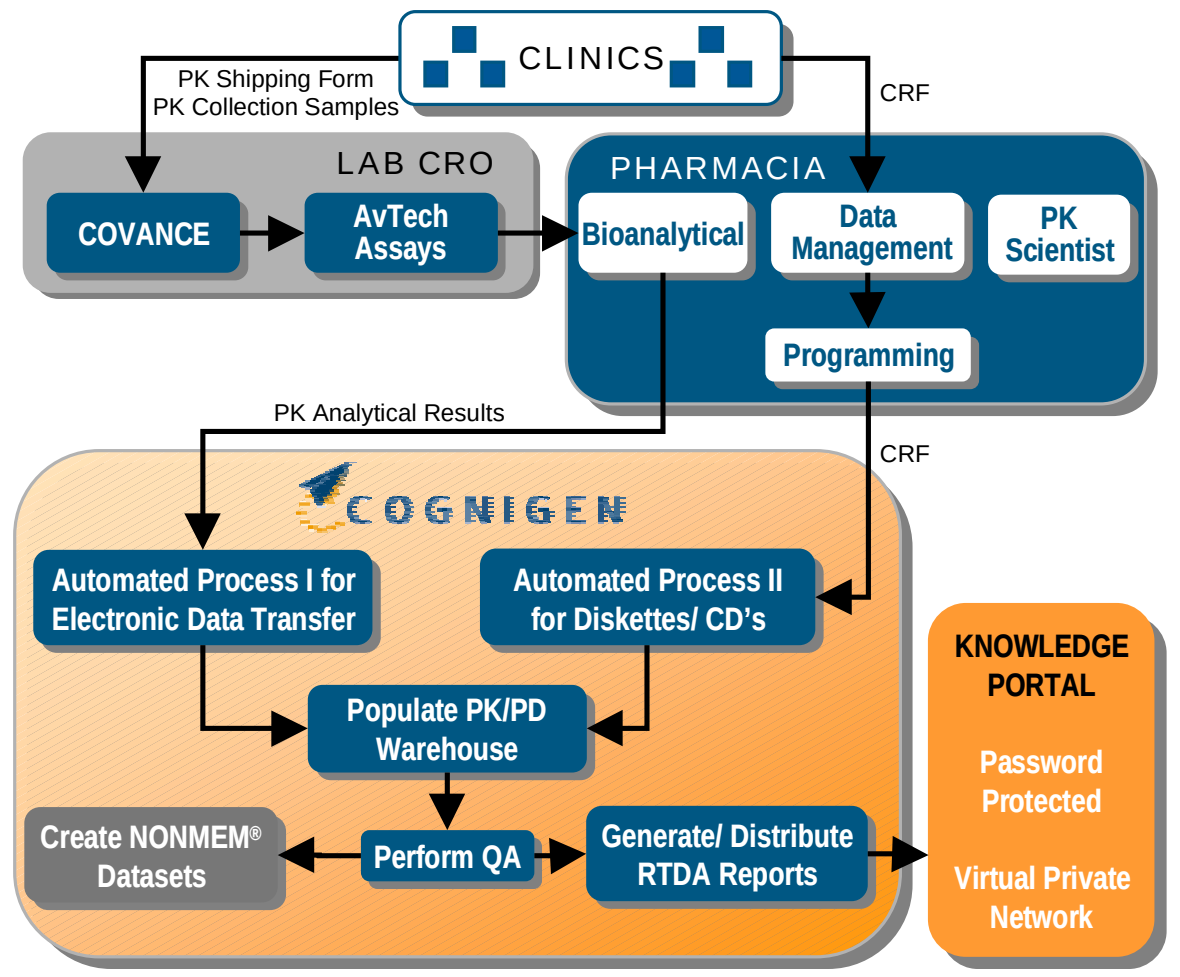

Pediatrics Linezolid Study 0082
CLINICS
PK Shipping Form
PK Collection Samples
CRF
LAB CRO
COVANCE
AvTech Assays
PHARMACIA
Bioanalytical
Data Management
PK Scientist
Programming
PK Analytical Results
CRF
COGNIGEN
Automated Process I for Electronic Data Transfer
Automated Process II for Diskettes/ CD's
Populate PK/PD Warehouse
Create NONMEM® Datasets
Perform QA
Generate/ Distribute RTDA Reports
KNOWLEDGE PORTAL
Password Protected
Virtual Private Network

RTDA Process for Present Study

1. While the trial was ongoing, drug dosing, concentration sampling, and patient demographic data was transmitted to Cognigen on a monthly basis.
2. Cognigen performed the necessary data merges in order to generate population PK-specific queries regarding questionable data, e.g., samples with inordinately long time since last dose or PK-specific protocol violations.
3. These queries were then communicated back to Pharmacia on a monthly basis for resolution.
4. Basic figures and tables were created describing the patient demographics, numbers of samples per patient, number of patients receiving PO therapy, LZD concentration vs time since last dose, etc.
5. After database lock, the final data was transmitted to Cognigen for creation of the final NONMEM® datasets.

RESULTS

Real-Time Data Exploration

Figure 2: Scatterplot of LZD Concentration versus Time Since Last Dose, Stratified by Route of Administration of Preceding Two Doses

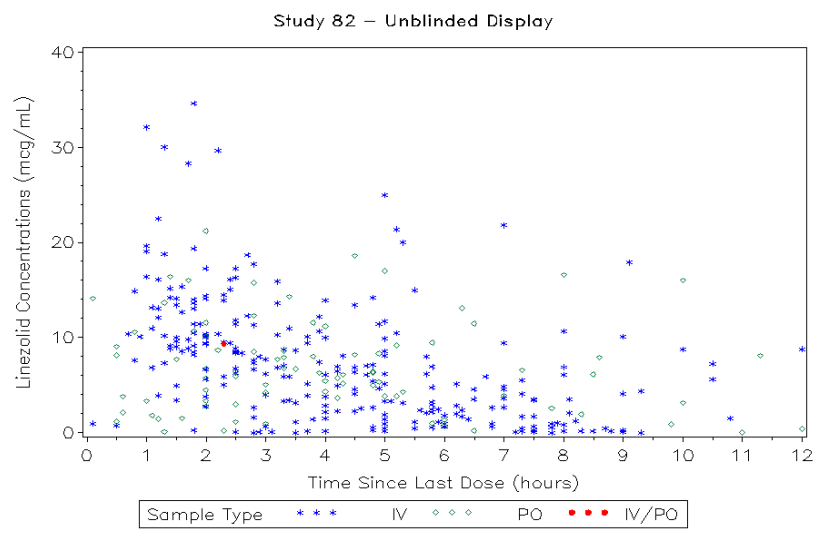

Study 82 – Unblinded Display
Linezolid Concentrations (mcg/mL)
Time Since Last Dose (hours)
Sample Type
IV
PO
IV/PO

Figure 3: Scatterplot of LZD Concentration versus Time Since Last Dose Used to Identify Potential Outliers for Further Follow-Up

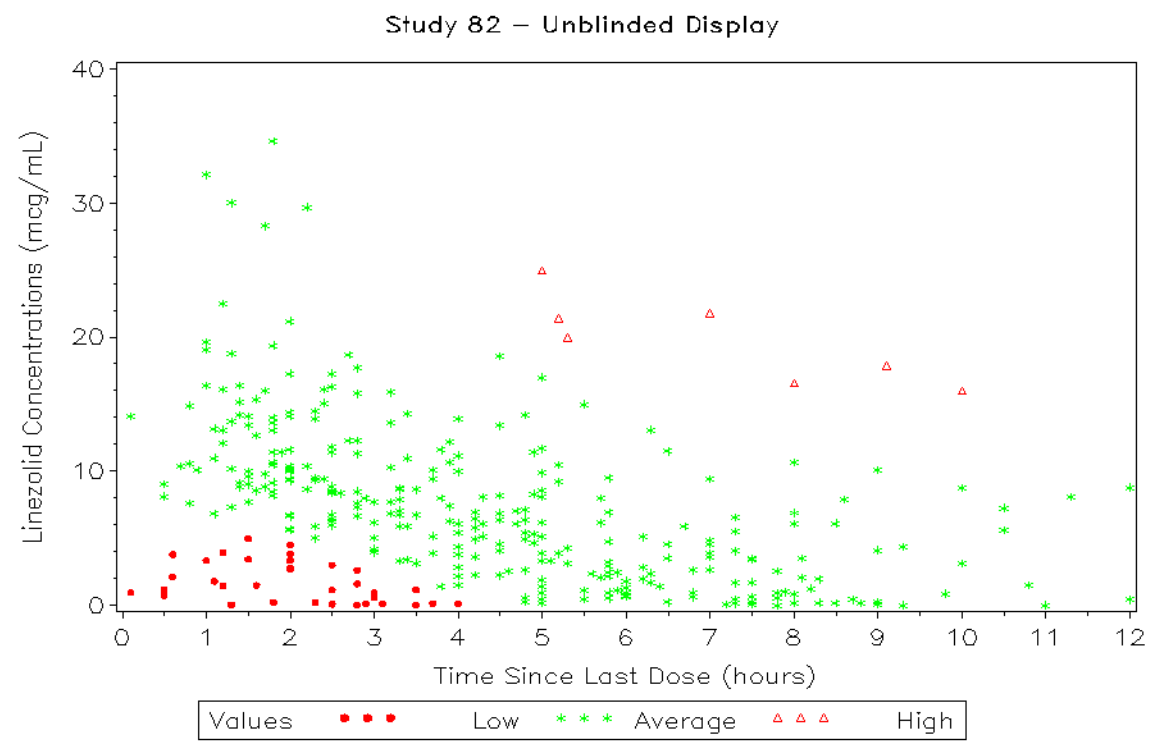

Study 82 – Unblinded Display
Linezolid Concentrations (mcg/mL)
Time Since Last Dose (hours)
Values
Low
Average
High

Figure 4: Frequency Distribution Histogram of Number of Samples per Patient, Stratified by Route of Administration

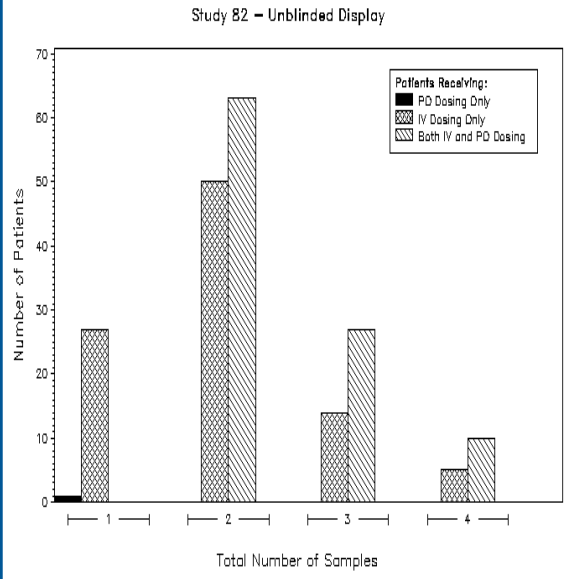

Study 82 – Unblinded Display
Patients Receiving:
PO Dosing Only
IV Dosing Only
Both IV and PO Dosing
Number of Patients
Total Number of Samples

Figure 5: Frequency Distribution Histogram of Patient Age, Stratified by Route of Administration

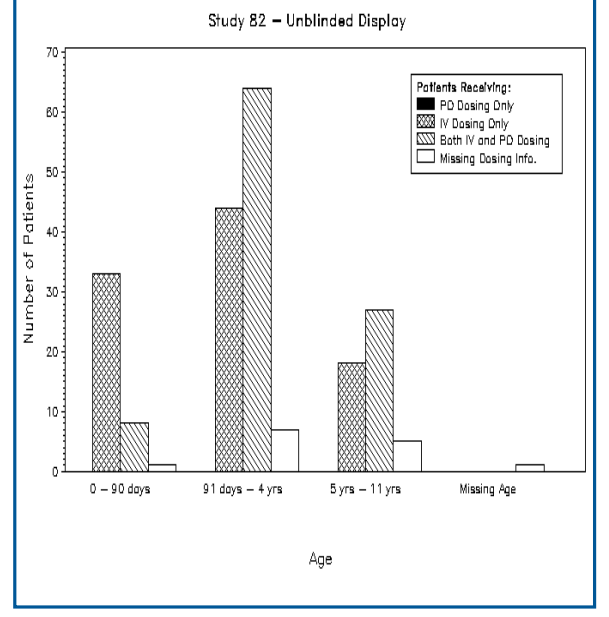

Study 82 – Unblinded Display
Patients Receiving:
PO Dosing Only
IV Dosing Only
Both IV and PO Dosing
Missing Dosing Info.
Number of Patients
0 – 90 days
91 days – 4 yrs
5 yrs – 11 yrs
Missing Age
Age

Queries Generated

Category	Description	Number of Listings*
Critical Data Errors	Errors requiring follow-up; Unlikely that Sponsor's data cleansing process will rectify	6-8
Time Since Last Dose Listings	Potential errors associated with the calculation of time since last dose	4-6
PK Sample Data Listings	Potential errors associated with the PK sample date/time	9-12
Dosing Data Listings	Potential errors associated with the dosing date/time	10-12
Demographic Data Listings	Potential errors related to demographic data	3-8

*Note that each listing contained from one to > 50 queries

Data Queries
Screen Shot of Page from the RTDA Knowledge Portal

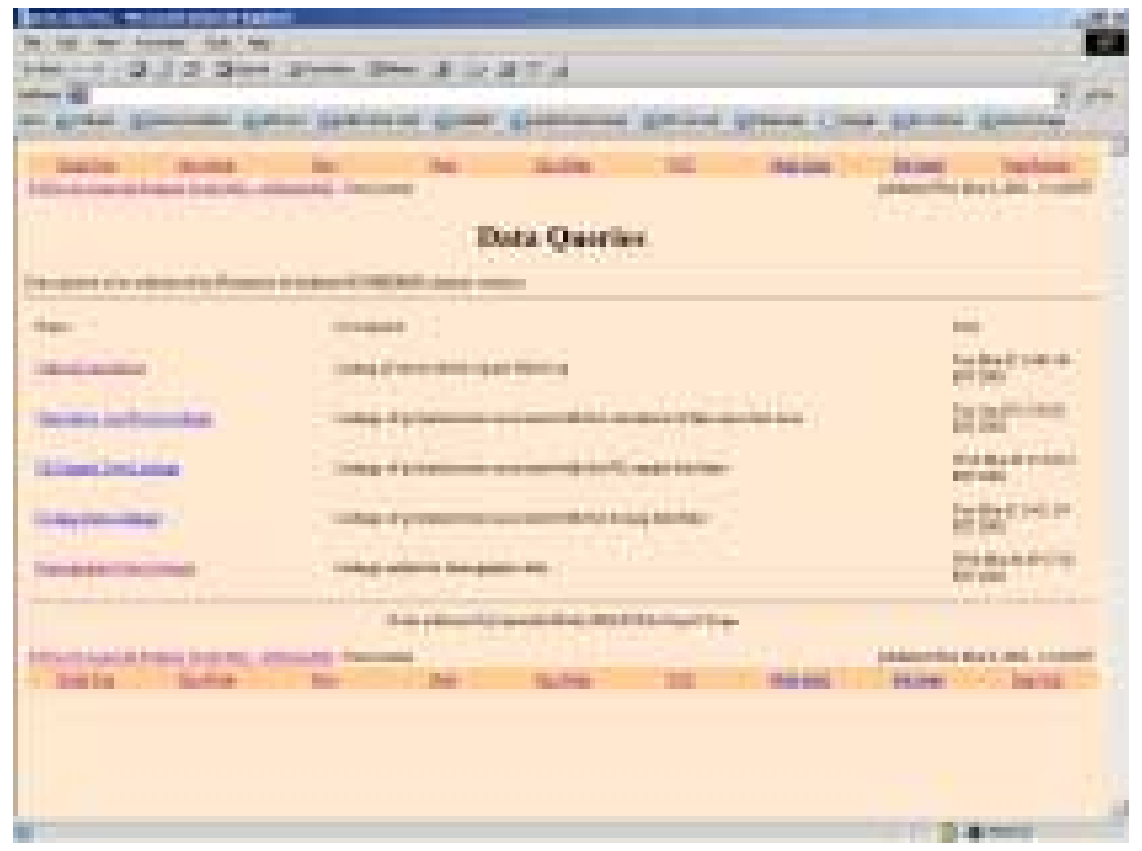

Data Queries

Benefits of Implementing RTDA

Quality	Over 100 PK-related queries were proactively resolved while the trial was ongoing. As a conservative estimate, if these queries had gone unresolved, at least 20% of the data would have been unusable from a PK/PD standpoint.
Efficiency	As soon as the first batch of concentration data was received, a potential issue was identified involving the process of merging the concentration results with the sampling time information. This issue was resolved proactively by identifying additional variables which could be used to accurately merge the data. If this had not been identified prior to database lock, this would have delayed dataset creation by at least three weeks.
Safety	Real-time data exploration allowed Pharmacia scientists to examine the plasma concentrations as the trial was ongoing. This was identified early on as a critical objective since this trial used a new dosing regimen in a pediatric population.

CONCLUSIONS

- Dataset creation issues were proactively resolved and the analysis was prospectively designed, allowing the population PK/PD analysis to be included in the FDA submission.
- Implementing RTDA improved data quality, reduced data exclusions, and facilitated rapid dataset creation upon data lock.
- This process allowed early confirmation of the appropriateness of the new dosing regimen through real-time data exploration. This was critical due to the potential vulnerability of this pediatric study population.

ACKNOWLEDGEMENTS

The authors would like to acknowledge the efforts of several individuals who were instrumental in the success of this project: Monika Sokolowski of Cognigen Corporation and Joann Thompson, Jill Bokros, Barb Padbury, and Monica Elfring of Pharmacia Corporation.