Population Pharmacokinetics of Paliperidone ER in Healthy Subjects and Patients With Schizophrenia

B. Cirincione,[1] M. Redman,[1] J. Fiedler-Kelly,[1] E. Ludwig,[1] A. Vermeulen[2]

[1]Cognigen Corporation, Buffalo, NY and [2]Johnson & Johnson Pharmaceutical Research and Development, Beerse, Belgium

ABSTRACT

Purpose. To develop a population pharmacokinetic (PPK) model for paliperidone extended-release (ER) and to evaluate the influence of selected covariates.

Methods. Paliperidone was administered once daily as paliperidone ER 3 - 15 mg (n=1368) or 1-mg intravenous dose (n=20). PPK analysis was based on nine Phase 1 and four Phase 3 trials. Model development included five Phase 1 studies (n=206), model refinement and covariate analysis included three additional Phase 1 studies and two Phase 3 studies (n=692), and validation included one Phase 1 and two Phase 3 studies (n=470).

Results. Data were described with a linear, two-compartment model with zero-order input (estimated duration 23.9h) and first-order absorption (estimated lag time 0.668h). Apparent total paliperidone clearance and steady-state volume of distribution were estimated to be 13.8L/h and 487L, respectively. Lean body mass (LBM) and CrCL were significant predictors of paliperidone clearance. In subjects with normal renal function, predicted paliperidone AUC (paliperidone ER 6 mg) decreased from 482 to 410 (ng•hr/mL) as LBM increased from 42.8 to 67.5kg. For any given LBM, however, a change from normal renal function to mild impairment was predicted to increase paliperidone exposure by approximately 20%.

Conclusions. A two-compartment model with zero-order input, first-order absorption, and first-order elimination best described the PPK for paliperidone ER.

INTRODUCTION

- Paliperidone (Invega™), an atypical antipsychotic that differs from risperidone by the addition of a hydroxyl group is delivered via proprietary extended-release technology (OROS®) designed to minimize peak to trough concentration fluctuations.
- The Food and Drug Administration approved paliperidone extended-release (ER) tablets for the once-daily treatment of schizophrenia in December 2006.
- Data from three 6-week trials of nonelderly patients (N = 1665; mean age, 37 years) indicated that the use of paliperidone ER at any dose (3, 6, 9, 12, or 15mg/day) was significantly more effective than placebo for improving symptoms of schizophrenia.
- Among the commonly reported adverse events associated with paliperidone ER treatment were restlessness, extrapyramidal symptoms (i.e., movement disorders), tachycardia, and somnolence.
- Dose-proportional increases in the pharmacokinetic (PK) parameters of paliperidone ER are observed over the dose range of 3 - 15 mg. The elimination $t_{1/2}$ is about 23 to 30 hours. Renal excretion is the primary route of paliperidone elimination.

OBJECTIVES

The PPK analysis program was comprised of three main stages: model development, model refinement, and model evaluation. The primary objectives of these analyses were to:

- develop a structural PPK model for paliperidone based on data obtained after oral ER and intravenous (IV) administration in five Phase 1 trials;
- refine the structural PK model using data from three additional Phase 1 trials and two Phase 3 trials;
- evaluate the influence of subject demographic factors and selected laboratory indices on paliperidone pharmacokinetics; and
- perform external model evaluation using data from one Phase 1 and two Phase 3 trials.

METHODS

Study Design and Data

- Data from nine Phase 1 studies and four Phase 3 studies were used for the analyses.
- Doses of paliperidone used in these studies ranged from 1 mg of an immediate-release (IR) solution for intravenous infusion to 3 - 15 mg of oral paliperidone ER tablets daily.
- Covariate analysis of subject demographic factors and selected laboratory parameters was completed during model refinement and after model evaluation.
- 21,183 paliperidone plasma concentrations were available from 1368 subjects from all stages of the analysis.

PK Model Development Procedure for Each Analysis Stage

Prior to model development and refinement, datasets were randomly split into index (70%) and evaluation (30%) datasets. The following steps were taken in the analysis:

1. Exploratory analysis on combined index and evaluation datasets
2. Structural model development on index dataset
3. Subject covariate analyses on index dataset
4. Optimization of the random effect matrices on index dataset
5. Internal model evaluation on evaluation dataset
6. Estimation of the model parameters using the combined index and evaluation datasets
7. External model evaluation

Covariate Analysis

- Subject demographics: age, sex, race, body weight, LBM,[1] and other derived body size variables
- Laboratory parameters: aspartate transaminase, alanine transaminase, lactate dehydrogenase, alkaline phosphatase, total bilirubin, gamma glutamyl transferase, serum creatinine, creatinine clearance,[2] total protein, and albumin
- Other: paliperidone formulation, smoking history, health status, country, study, and predicted CYP2D6 phenotype (graphical evaluation only for final PPK model as data available in only 50% of subjects)

Statistical Analysis

- All analyses were performed using NONMEM®, Version 5 Level 1.1.
- Statistical significance: univariate analyses: α=0.05; backward elimination: α=0.001
- Goodness-of-fit of each NONMEM® analysis was assessed by examination of:
 - scatterplots of predicted versus measured concentrations and versus weighted residuals;
 - % SEM of the parameter estimates; and
 - changes in the estimates of the interindividual and residual variability.

RESULTS

Figure 1: **Paliperidone Concentrations Versus Time Since Last Dose from All Stages**

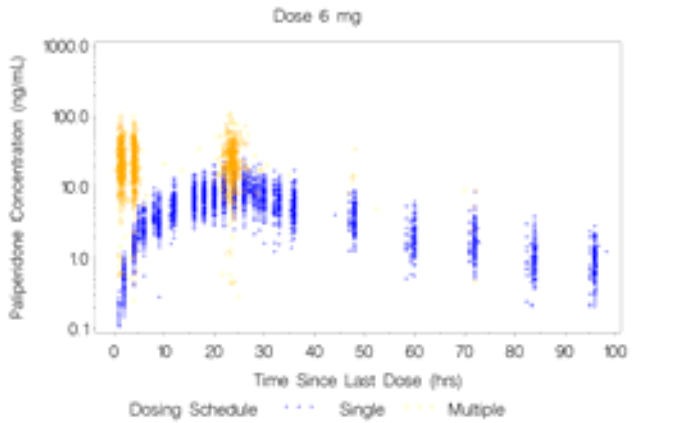

Dose 6 mg
Paliperidone Concentration (ng/mL)
1000.0
100.0
10.0
1.0
0.1
0
10
20
30
40
50
60
70
80
90
100
Time Since Last Dose (hrs)
Dosing Schedule
Single
Multiple

Subject Characteristics

- 65% male, mean age was 40 yrs (range 18 - 85)
- Mean (SD) LBM was 55 (range 17.2 - 87.4)
- 77% were patients diagnosed with schizophrenia or schizoaffective disorder
- 51% of subjects had CYP2D6 genotype-derived phenotype, 88% were extensive metabolizers
- A range of renal function was represented within the subjects (10.2, 150.0)

PK Model Development Stage

- Evaluation of pooled paliperidone IV and oral ER data
 - A linear three-compartment model with consecutive zero-order (into the depot compartment) and first-order absorption (from depot compartment to the central compartment), and first-order elimination
 - Influence of gender on bioavailability (0.442 F and 0.301 M) and study on absorption lag time were identified as significant

PK Model Refinement Stage

- Evaluation of paliperidone ER data
 - Percentage of data from the IV formulation was not large enough to support the estimation of a three-compartment model
- A linear, two-compartment model with consecutive zero- and first-order absorption and first-order elimination from the central compartment
- LBM and renal impairment status were significant predictors of apparent oral clearance.

PK Model Evaluation and Refinement of the Covariate Model

- A two-compartment model was used to generate population and empirical Bayesian (individual) predictions for all concentrations in the model evaluation dataset.
- Diagnostic plots show that the PK model described the model evaluation data reasonably well.

Figure 2: **Measured Versus Individual Predicted Paliperidone Concentrations for the External Model Evaluation**

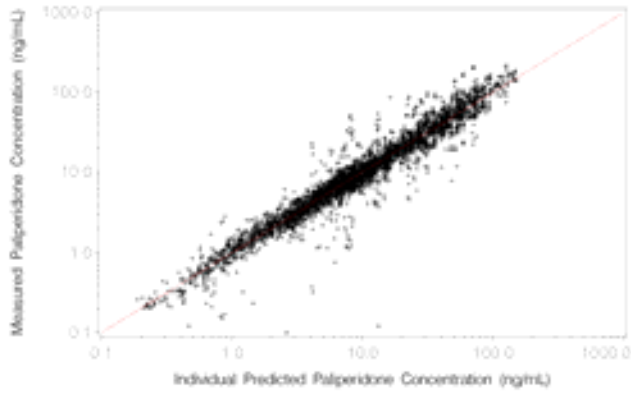

Measured Paliperidone Concentration (ng/mL)
1000.0
100.0
10.0
1.0
0.1
1.0
10.0
100.0
1000.0
Individual Predicted Paliperidone Concentration (ng/mL)

Final PK Model

- Pooled data from all stages.
- A linear, two-compartment model with consecutive zero- and first-order absorption and first-order elimination from the central compartment was used to describe the plasma concentrations of paliperidone ER.

Figure 3: **Two-Compartment Disposition Model**

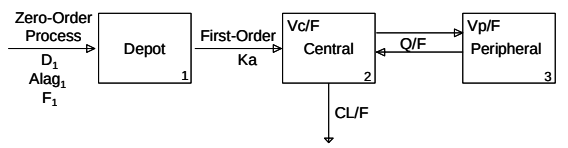

Zero-Order Process
D1
Alag1
F1
Depot
1
First-Order
Ka
Vc/F
Central
2
Q/F
Vp/F
Peripheral
3
CL/F

- The duration of the zero-order input (the rate-limiting step in the absorption process) was estimated to be 23.9 hr, in line with the expected T_{max} of about one day, while the absorption rate constant was 0.565 1/hr, with IIV and IOV of 49.0 and 80.5%, respectively.

Table 1: **Parameter Estimates and Standard Errors from the Final Population PK Model for Paliperidone ER**

	Population Mean[a]		Interindividual Variability (%CV)		Interoccasion Variability (%CV)	
Parameter	**Estimate**	**RSE%**	**Estimate**	**RSE%**	**Estimate**	**RSE%**
CL/F (L/hr) Intercept[b]	8.02	14.0			NA	NA
Power on CL/F for LBM[b]	0.636	33.2			NA	NA
Linear Slope on CL/F for CrCL[b]	0.0512	17.9	52.25	16.0	NA	NA
Vc/F (L)	260	10.2			NA	NA
Q/F (L/hr)	34.5	18.6	132.66	118.2	NA	NA
Vp/F (L)	227	8.1	57.79	46.1	NA	NA
Ka (1/hr)	0.565	10.5	48.99	38.2	80.50	21.1
D_1 (hr)	23.9	0.5	NA	NA	NA	NA
$Alag_1$ (hr)	0.668	4.8	63.01	22.9	NA	NA
F_1	NE	NA	NA	NA	50.79	10.1
RV ($\log_e$ SD) (Phase 1)	0.18	3.6	NA	NA	NA	NA
RV ($\log_e$ SD) (Phase 3)	0.22	27.7	NA	NA	NA	NA
Minimum Value of the Objective Function = -33555.188						

[a] Correlated parameter estimates (|r| > 0.9):

Correlated Parameters	Coefficient of Determination (r)	Correlation Coefficient (r^2)
CL (L/hr), CrCL Linear for CL	-0.957	0.916
Vc, IIV Q	0.904	0.812

[b] $$CL/F = 8.02 \cdot \left(\frac{LBM}{58.4}\right)^{0.636} + 0.0512 \cdot CrCL$$

NE = not estimated (set to 1 for all subjects)
NA = not applicable

Figure 4: **Goodness-of-Fit Plots for the Final Population PK Model for Paliperidone ER**

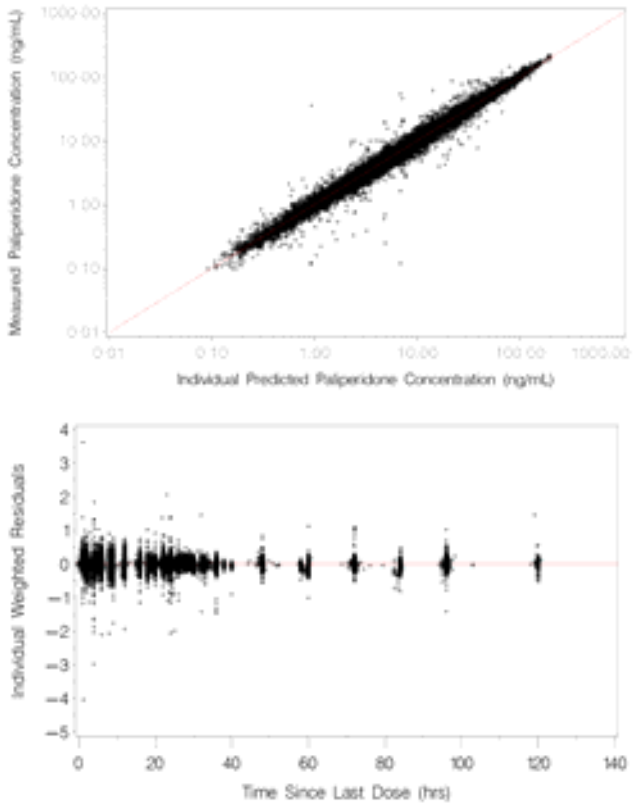

Measured Paliperidone Concentration (ng/mL)
1000.00
100.00
10.00
1.00
0.10
0.01
0.01
0.10
1.00
10.00
100.00
1000.00
Individual Predicted Paliperidone Concentration (ng/mL)
Individual Weighted Residuals
4
3
2
1
0
−1
−2
−3
−4
−5
0
20
40
60
80
100
120
140
Time Since Last Dose (hrs)

For graphing purposes, three concentrations, collected from one subject, associated with a time since last dose value >130 were removed.

- A single clearance parameter described elimination of paliperidone by all renal and non-renal pathways. The renal component of the total apparent clearance is approximated by 0.0512•CrCL, amounting to 5.8 L/hr for a subject with a normal CrCL of 114 mL/min. Non-renal clearance is approximated by $8.02 \cdot (LBM/58.4)^{0.636}$, and is 8.0 L/hr for a subject with a LBM of 58.4 kg.
- The total apparent clearance is 13.8 L/hr (the sum of 5.8 and 8.0 L/hr), consistent with previous values based on an absolute oral bioavailability of about 28%.
- In the population receiving 6-mg paliperidone ER tablets at steady-state with normal renal function (CrCL=114.4 mL/min), as LBM increases from 42.8 to 67.5, paliperidone $AUC_{SS(0-\tau)}$ decreases from 482 to 410 (ng•hr/mL). In addition, for any given LBM level, as renal impairment levels decrease from normal (CrCL=114.4) to mild (CrCL=72.7), an approximate 20% increase in mean exposure is predicted.

Figure 5: **Influence of Creatinine Clearance and Lean Body Mass on the Population Mean Predicted Apparent Oral Clearance of Paliperidone from the Final Model**

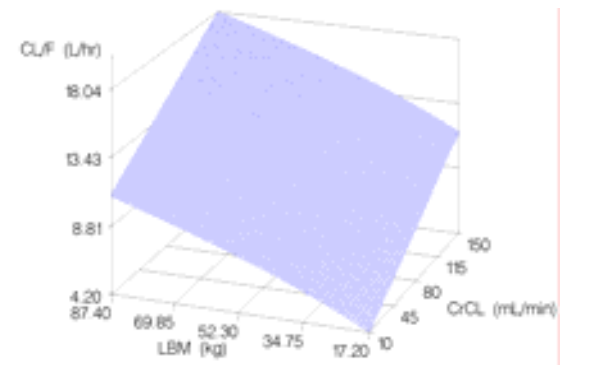

CL/F (L/hr)
18.04
13.43
8.81
4.20
87.40
69.85
52.30
34.75
17.20
LBM (kg)
10
45
80
115
150
CrCL (mL/min)

Evaluation of CYP2D6 Phenotype

- No substantial differences were observed in the apparent oral clearances between predicted CYP2D6 phenotypes.

Figure 6: **Boxplots of Bayesian CL Versus Predicted Phenotype from All Stages**

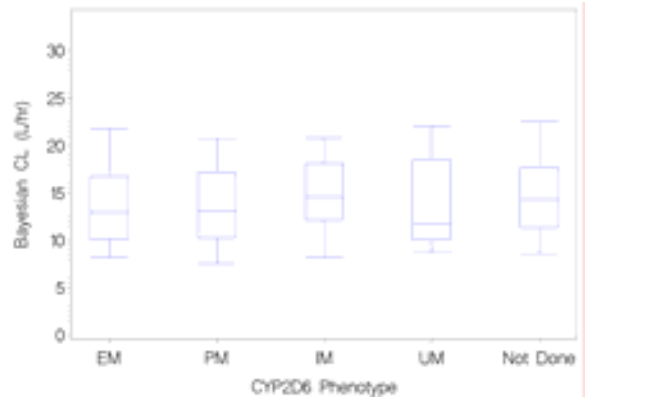

Bayesian CL (L/hr)
30
25
20
15
10
5
0
EM
PM
IM
UM
Not Done
CYP2D6 Phenotype

The box represents the 25th, 50th, and 75th percentiles. The whiskers extend from the 10th to the 90th percentiles of the data.
EM=Extensive Metabolizer
PM=Poor Metabolizer
IM=Intermediate Metabolizer
UM=Ultrarapid Metabolizer

CONCLUSIONS

- Population PK model development was integrated throughout the clinical development program of paliperidone ER.
- A two-compartment model with consecutive zero- and first-order absorption with a lag-time, and first-order elimination from the central compartment best described the paliperidone concentration-time data from healthy subjects and patients with schizophrenia.
- Large variabilities in absorption parameters were observed, consistent with the release of paliperidone from the ER formulation over a prolonged period of time (T_{max} of one day) and throughout the gastrointestinal tract.
- Lean body mass and creatinine clearance were shown to significantly influence the apparent oral clearance of paliperidone, however, only the magnitude of the renal function effect was substantial enough to require dose adjustment based on this factor.
- None of the other covariates tested contributed to the interindividual variability in any of the PK parameters to a statistically significant extent.
- No visual differences in paliperidone exposure between predicted phenotypes were observed; therefore, adjustment of the paliperidone ER dose based on predicted CYP2D6 phenotype is not warranted.

REFERENCES

1. Minto CF, Schnider TW, Egan TD, Youngs E, Lemmens HJ, Gambus PL, Billard V, Hoke JF, Moore KH, Hermann DJ, Muir KT, Mandema JW, Shafer SL. Influence of age and gender on the pharmacokinetics and pharmacodynamics of remifentanil. I. Model development. Anesthesiology, 1997 Jan;86(1):10-23.
2. Cockcroft DW, Gault MH. Prediction of creatinine clearance from serum creatinine. Nephron; 1976;16:31-41.

Presented at:
American Society for Clinical Pharmacology and Therapeutics (ASCPT)
Annual Meeting
Anaheim, California
March 21-24, 2007