Population Pharmacokinetics of Dexmedetomidine (DEX) During Long-Term Continuous Infusion in Critically Ill Patients

J. Fiedler-Kelly[1], E. Ludwig[1], Q. Lu[1], D. Stalker[2]

[1]Cognigen Corporation, Buffalo, NY; [2]Hospira Incorporated, Lake Forest, IL

ABSTRACT

Background. Dexmedetomidine (DEX), a selective alpha$_2$-adrenoceptor agonist is approved for sedation. In this study, the population pharmacokinetics (PK) of DEX during long-term (> 24 hours) infusion was characterized and compared to previous results obtained after short-term infusion.

Methods. Data were obtained from 53 adults (62% male, 79% Caucasian, aged 18-87 years) enrolled in a Phase 4 study. DEX was administered as an optional loading dose (1 µg/kg), followed by a maintenance intravenous (IV) infusion that started at 0.84 µg/kg/h and was titrated to predefined sedation level. Plasma DEX concentrations (n = 291) were obtained at 2 and 4 hours after the infusion was started, within 15 minutes before the infusion was stopped, and 4 and 8 hours after the infusion was stopped. Interindividual variability (IIV) on clearance (Cl) and volume of distribution (Vd) and residual variability (RV) were estimated using exponential and log error models, respectively. Selected covariates were evaluated graphically.

Results. DEX PK was described by a 1-compartment model with first-order elimination. The mean parameter estimates (%SEM) were: 39.4 (9.7) L/h for Cl, and 152 (11.5) L for Vd. Estimated IIV for Cl and Vd were 63% and 68%, respectively, with RV approximately 59 %CV. DEX Cl was comparable to a previous estimate of 39.2 L/h demonstrated during short-term infusion in healthy subjects for less than 24 hours. Graphical covariate assessment indicated potential positive relationships between Cl and body weight, Vd and weight, as well as a possible relationship between DEX PK and age.

Conclusions. DEX PK after long-term infusion was consistent with DEX PK after short-term infusion.

INTRODUCTION

- The alpha$_2$ agonist dexmedetomidine was initially approved by the Food and Drug Administration for sedation in initially intubated and mechanically ventilated patients in an intensive care unit setting, with the recommendation that dexmedetomidine be administered by continuous infusion (0.2 µg/kg/h to 0.7 µg/kg/h) not exceeding 24 hours in duration.[1]
- Subsequent studies conducted in intensive care unit patients have assessed the use of dexmedetomidine infusions beyond 24 hours and up to 7 days.[2,3]
- Dexmedetomidine undergoes almost complete biotransformation with very little unchanged dexmedetomidine excreted in urine and feces.[1] Biotransformation involves both direct glucuronidation, as well as cytochrome P450 mediated metabolism.[1]
- Population pharmacokinetic (PK) parameter estimates for dexmedetomidine clearance (Cl), volume of distribution (Vd) at steady-state, $t_{1/2\alpha}$, and $t_{1/2\beta}$ were 39.2 L/h, 118.1 L, 5.9 minutes, and 2.4 hours, respectively, based on a 2-compartment model developed using pooled data from 7 studies of healthy volunteers. Dexmedetomidine concentrations were obtained during and after very brief infusions (1 - 10 minutes), as well as during and following discontinuation of 5- to 12-hour infusions in several studies.[4]

OBJECTIVES

- Compare dexmedetomidine mean population PK during long-term (> 24 hours) continuous intravenous (IV) infusion with dexmedetomidine PK during short-term infusion.
- Develop a structural model, including interindividual variability (IIV) and residual variability (RV), to describe the population PK of dexmedetomidine in patients requiring long-term, continuous IV infusion for sedation.
- Perform model validation using visual predictive check (VPC) and relative predictive performance methods.
- Conduct an exploratory graphical review of select covariate effects on dexmedetomidine PK.

METHODS

Study Design and Data

- Data were available from a randomized, double-blind, Phase 4 study of long-term, continuous infusions of dexmedetomidine versus midazolam for sedation in intensive care unit patients. Patients were initially intubated and mechanically ventilated, sedation was expected to be required during mechanical ventilation, and mechanical ventilation was expected to continue for at least 72 hours.
- Dexmedetomidine doses were an optional loading dose of up to 1.0 µg/kg infused over 10 minutes, followed by a maintenance infusion of 0.84 µg/kg/h initially, then titrated to maintain an adequate level of sedation for up to 30 days (dose limits of 0.2 µg/kg/h to 1.4 µg/kg/h).
- Blood was obtained for later measurement of dexmedetomidine concentrations at: 2 and 4 hours (± 30 minutes) after dexmedetomidine infusion initiation, within 15 minutes prior to discontinuation of the infusion, 4 and 8 (± 1 hour) hours after the infusion was discontinued.

Data Analysis

- Data preparation was performed using SAS®, Version 8.24, and the PK analysis was completed using NONMEM®, Version 6, Level 1.0. Model parameters were estimated using the first-order conditional estimation method with interaction.
- Structural model developed including estimation of between-patient variability in PK parameters and within-patient (residual) variability in drug concentrations.
- Goodness-of-fit of each NONMEM analysis was assessed by examination of scatterplots of predicted versus measured concentrations and versus weighted residuals, %SEM of the parameter estimates, and changes in the estimates of the IIV and RV.
- Patient-specific Bayesian estimates of exposure to dexmedetomidine and derived population parameters were generated based on this model.
- Model validation was performed using VPC procedure (1000 replicate datasets simulated based on estimates of fixed and random effects from the model including RV) and relative predictive performance (prediction errors and standardized mean prediction errors calculated).[5,6,7]

Covariate Analysis

- Graphical representations of individual estimates of PK parameters versus patient demographic factors and clinical laboratory indices were evaluated.
- Covariates assessed: gender, body weight, ethnicity, age, alanine aminotransferase (ALT), aspartate aminotransferase (AST), albumin, and estimated creatinine clearance (CrCL).[8]
- Using the base structural PK model, Bayesian estimates of parameters were generated for each individual patient. These Bayesian parameter estimates were plotted versus each of the patient covariates to identify potential relationships between unexplained variability in PK parameters and covariates.

RESULTS

Data

- There were 236 dexmedetomidine concentrations from 53 patients used in the analysis. Patients were 62% male; 79% Caucasian with median age of 60 years (range: 18 years to 87 years) and mean (SD) body weight of 94 (44.6) kg (range: 36.3 kg to 285 kg).
- Only 5 patients (9%) received a loading dose prior to the maintenance infusion. Thereafter, maintenance infusion rates were adjusted according to level of sedation (0 to 68 times per patient), with median total dexmedetomidine dose of 5573.33 µg (range: 1204 µg to 23,439.92 µg).

Population Pharmacokinetic Model Development

- Both 1- and 2-compartment models with first-order elimination were evaluated.
- The concentration-time profile based on the typical-value parameter estimates from 1-compartment model fit for a dose of 5573 µg administered over 86 hours is overlaid on the raw concentration data in Figure 1.
- A 2-compartment model also provided a reasonable fit to the data. The estimated α- and β-half-lives were 1.35 hours and 4.9 hours. This α-half-life estimate is much higher than reported previously (5.9 minutes),[4] suggesting that there was insufficient information to characterize the initial distribution phase for dexmedetomidine since the first sample after infusion discontinuation was obtained at 4 hours.
- Figure 1 (inset) also provides a comparison of fit of 1- and 2-compartment models and shows that the differences between the predicted curves are quite small and apparent only at 4 hours after the end of the infusion.

Figure 1. Typical Concentration-Versus-Time Profile for the Fit of a 1- and 2-Compartment Model Overlaid on Observed Concentration Data

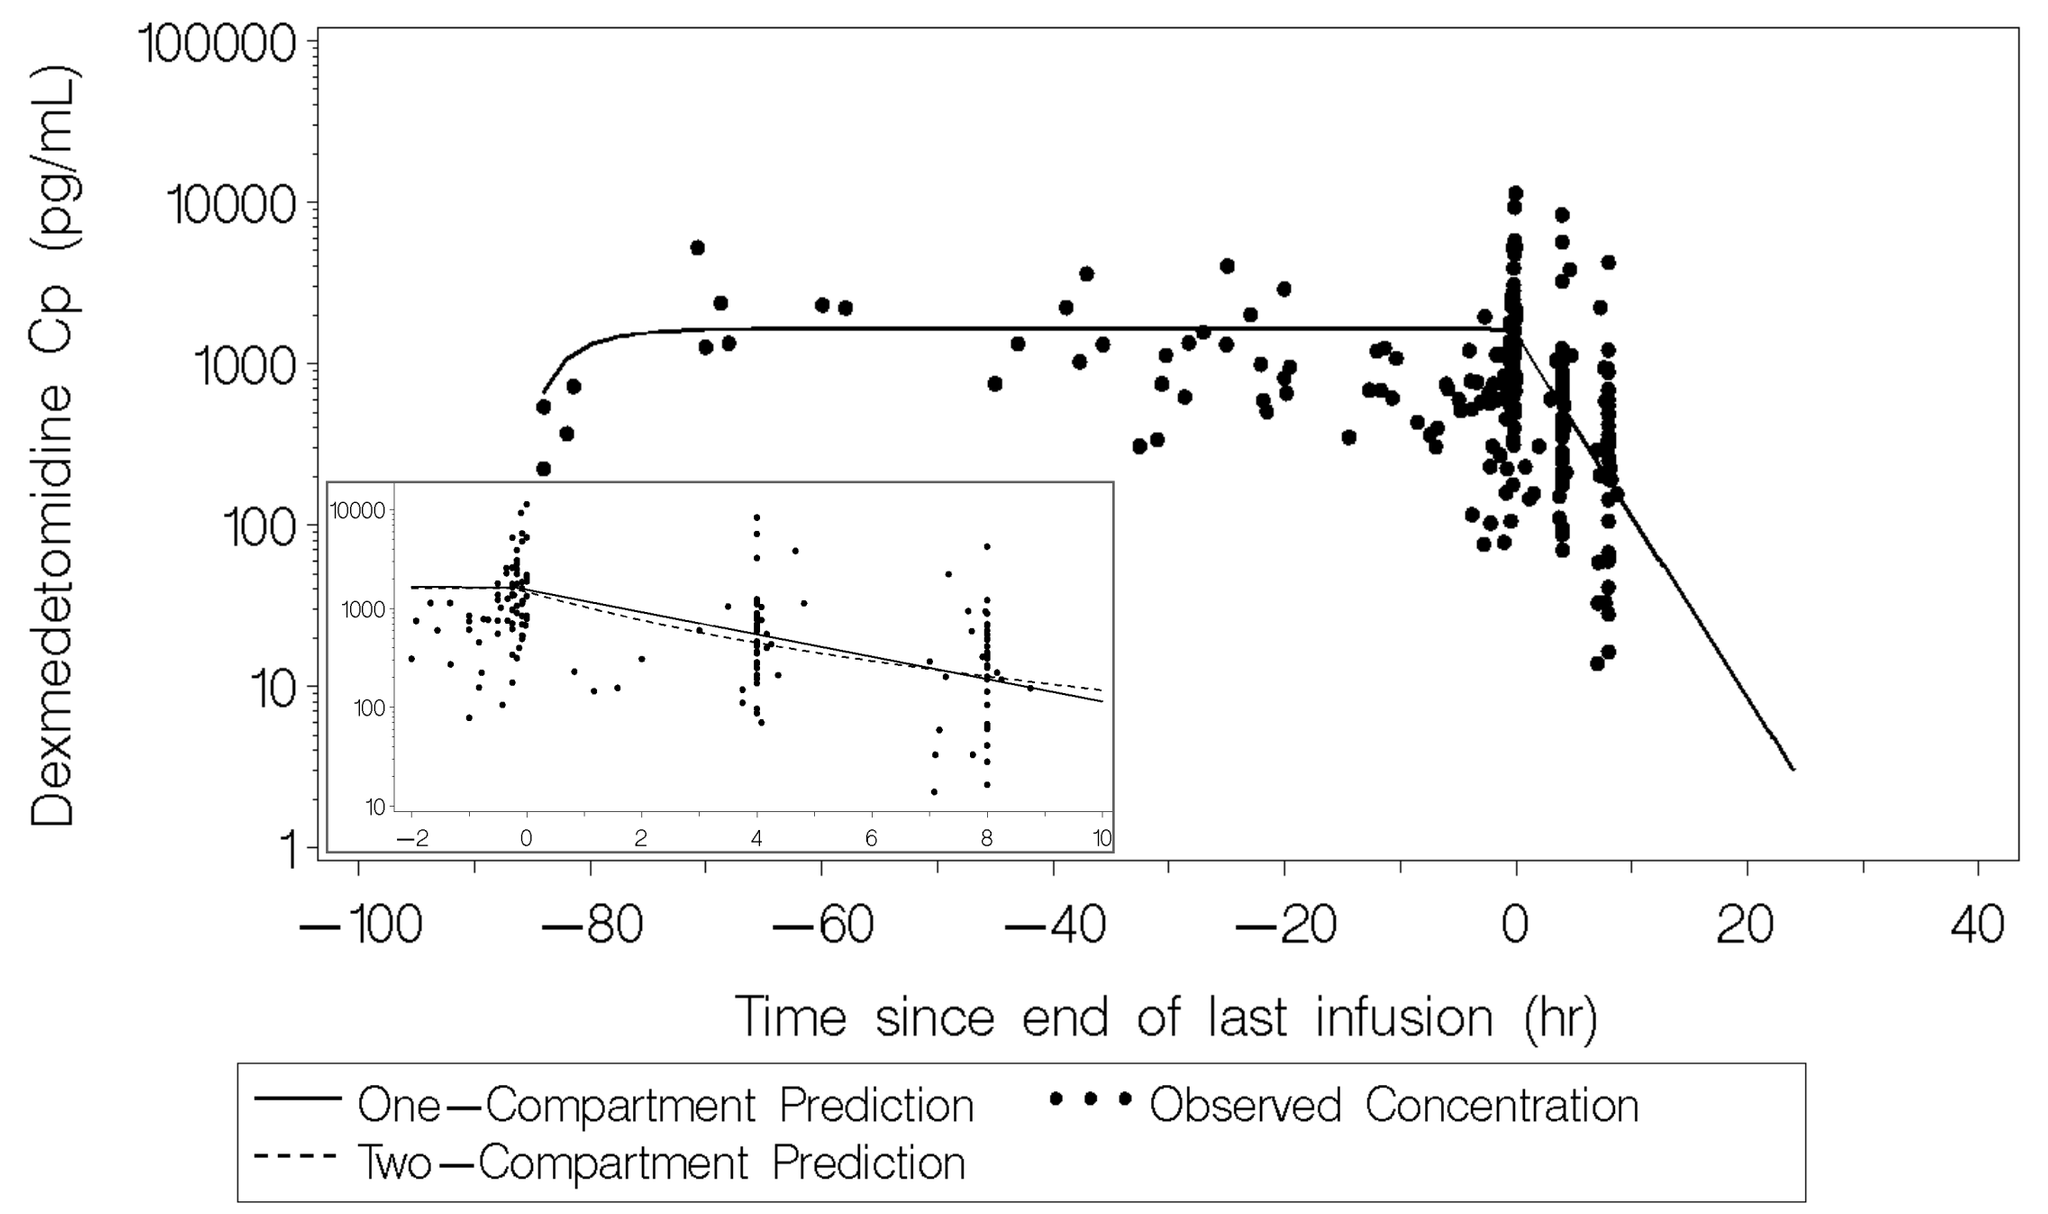
Dexmedetomidine Cp (pg/mL)
Time since end of last infusion (hr)
One−Compartment Prediction
Two−Compartment Prediction
Observed Concentration

Concentrations measured prior to 86 hours before the end of the infusion deleted for graphing and visualization purpose.

- Similarity in concentration-time profiles justifies the selection of a 1-compartment model as the more appropriate (and parsimonious) description of dexmedetomidine PK following long-term infusion.
- Final parameter estimates for the 1-compartment model are shown in Table 1. Clearance was estimated at 39.4 L/h, Vd at 152 L, and the half-life was estimated as 2.67 hours. All parameters estimated with reasonable precision (%SEMs < 35%). These estimates for Cl and β-half-life were consistent with previous Cl (39.2 L/h), and β-half-life (2.4 hours) estimates from dexmedetomidine infusions of < 24 hours in duration.[4] The estimate of Vd from the previous analysis was somewhat lower (118.1 L).
- Interindividual variability was estimated for Cl and Vd using an exponential error model, and RV was estimated using a log error model.
- Diagnostic goodness-of-fit plots for the 1-compartment model as shown in Figure 2.

Table 1. Parameter Estimates and Standard Errors for 1-Compartment Model

Parameter	Typical Value Estimate	%SEM
Clearance (L/hr)	39.4	9.7
Volume of distribution (L)	152	11.5
IIV on clearance (%CV)[a]	63.2%	30.3
IIV on volume of distribution (%CV)	67.5%	29.2
Cov (IIV on CL, IIV on V_c)[a,b]	0.317	34.1
Residual Variability (SD in log-concentration units)	0.587	9.1
Minimum Value of the Objective Function = 145.978		

Abbreviations: Cov, covariance; hr, hour; IIV, interindividual variability, SD, standard deviation.

[a] The associated correlation coefficient for IIV on CL and V_c is 0.552 (r^2).

[b] IIV on CL and cov (IIV on CL, IIV on Vc) correlated with r^2 = 0.848, indicating that independent estimates of these parameters were not obtained.

Fiedler-Kelly J, Ludwig E, Lu Q, Stalker D. Population pharmacokinetics of dexmedetomidine (DEX) during long-term continuous infusion in critically ill patients. Poster presented at: American Society for Clinical Pharmacology and Therapeutics Annual Meeting (ASCPT); March 17-20, 2010; Atlanta, GA.

For additional information, please contact
Jill Fiedler-Kelly, Vice President, Pharmacometric Services and CSO
Cognigen Corporation
395 South Youngs Road, Buffalo, NY 14221
(716) 633-3463, ext. 228 or jill.fiedler-kelly@cognigencorp.com

Figure 2. Goodness-of-Fit Diagnostic Plots for Fit of the Final 1-Compartment Model

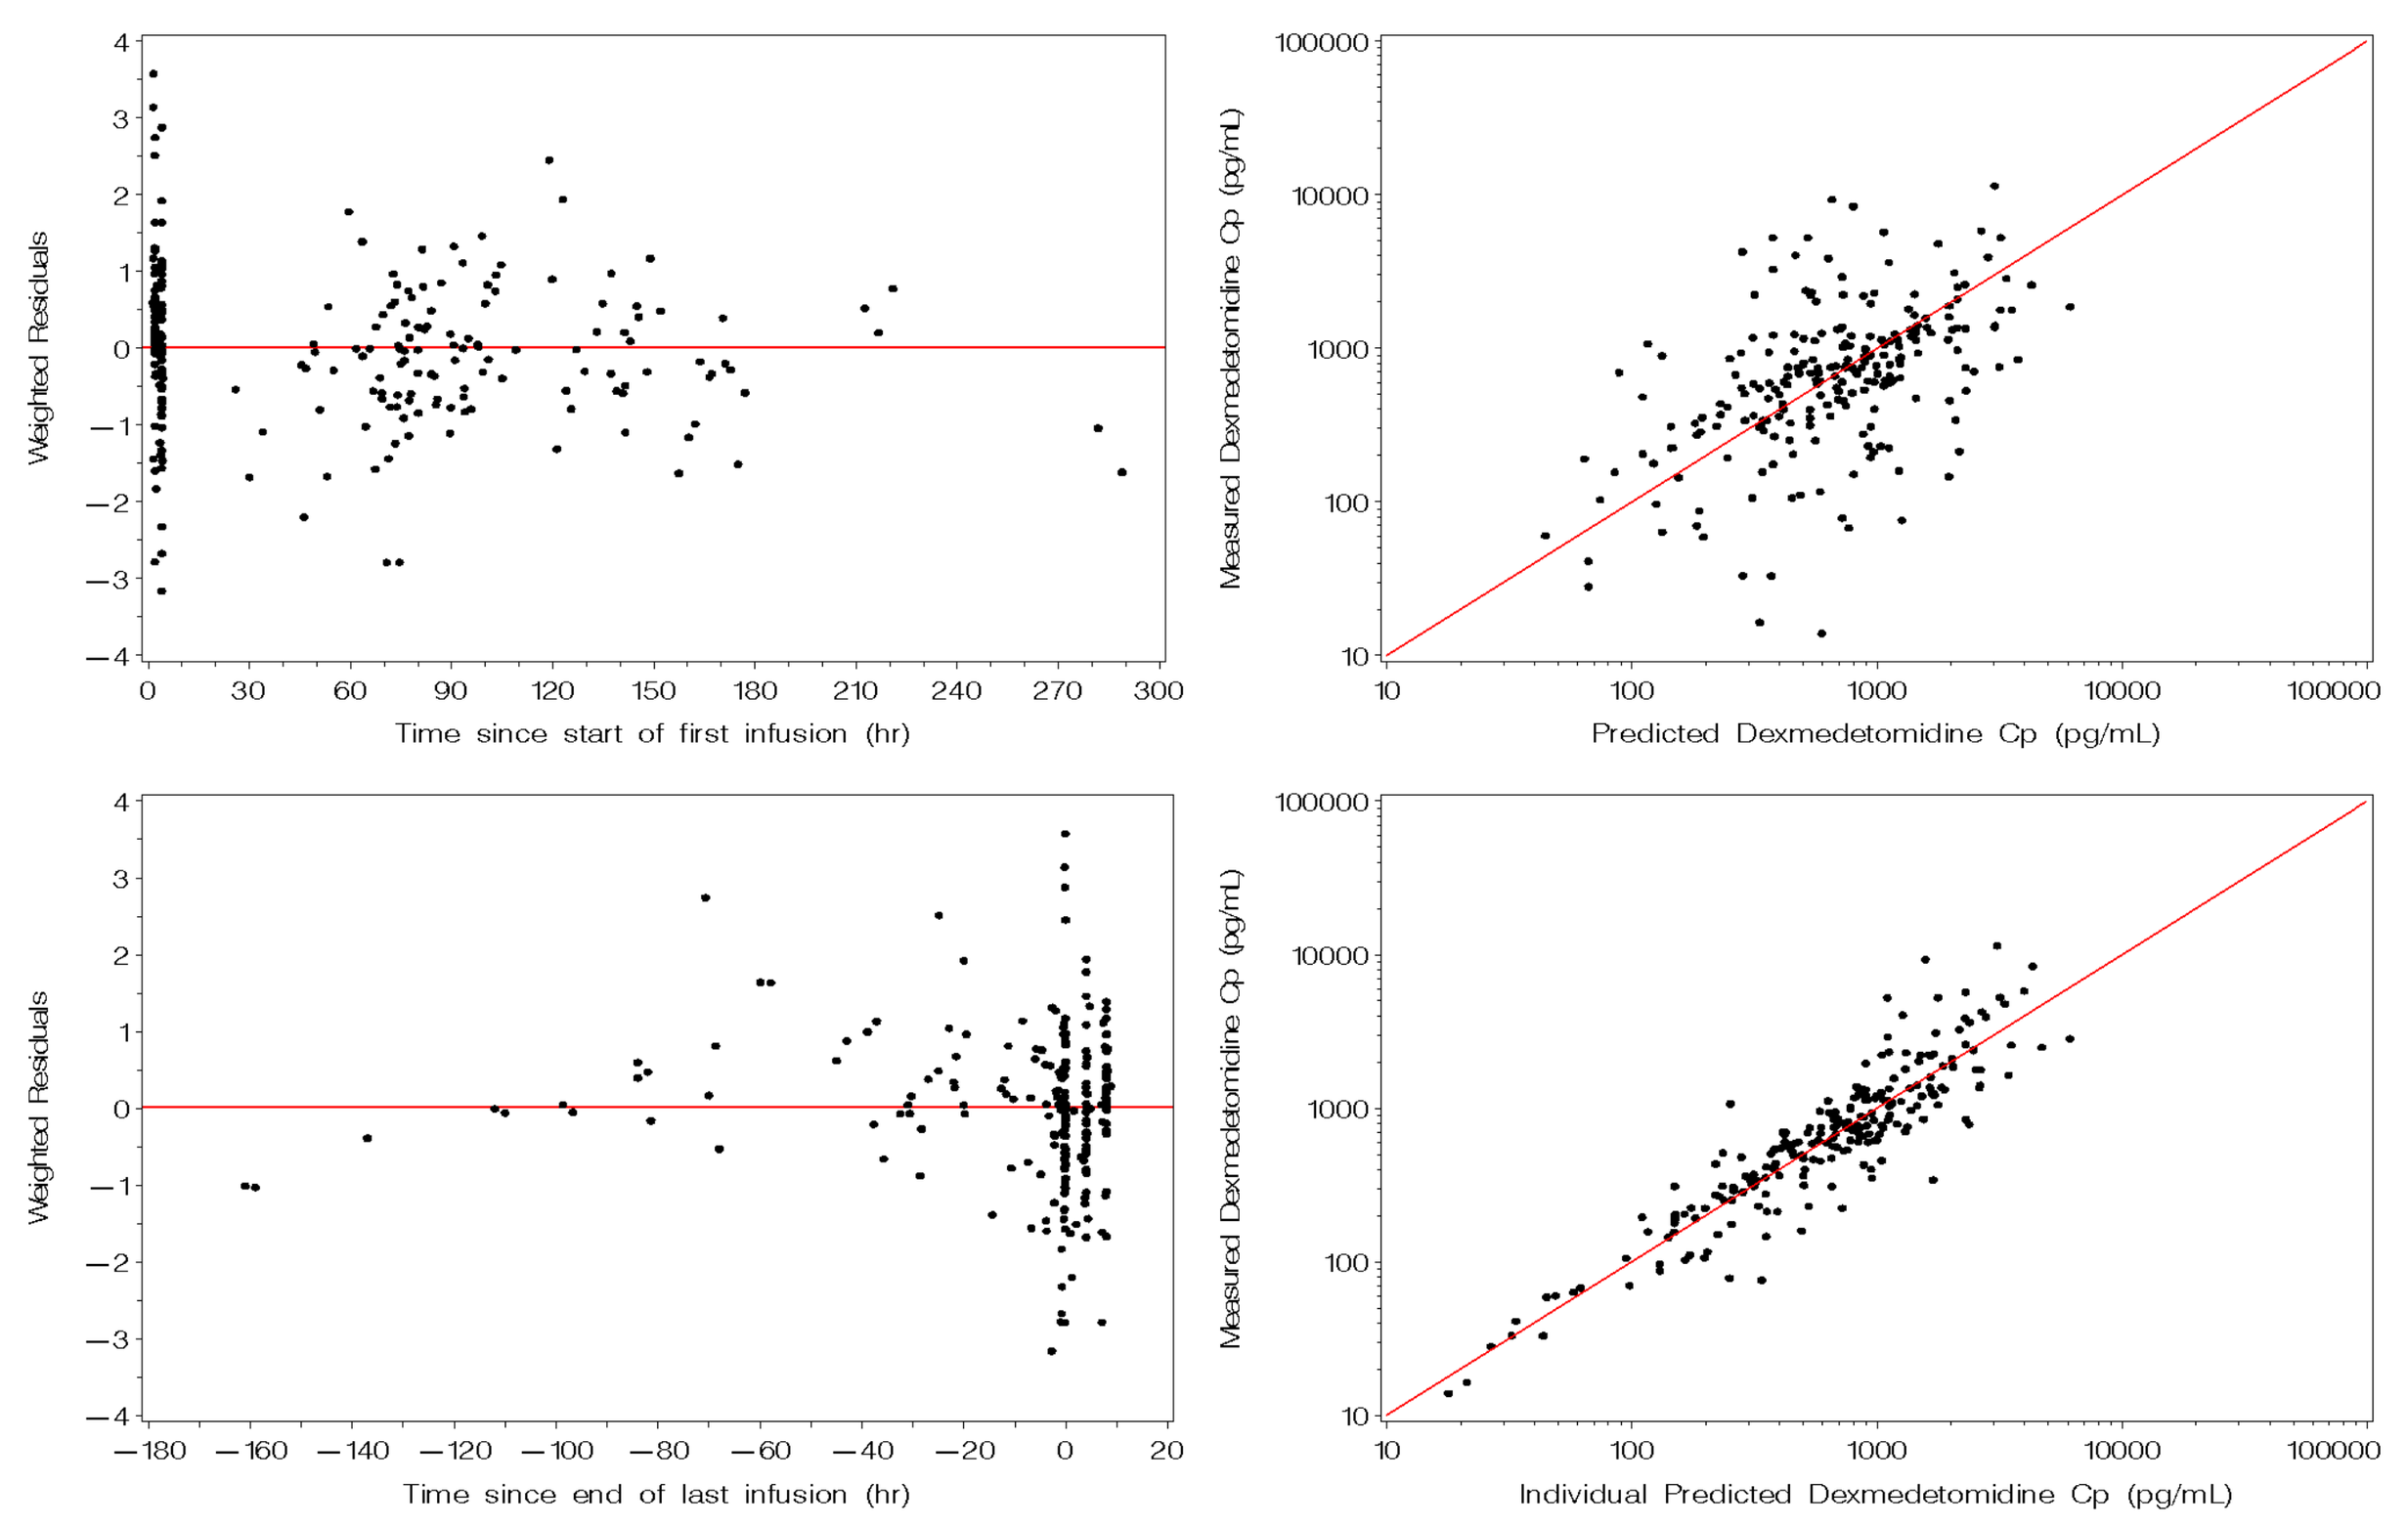
Weighted Residuals
Time since start of first infusion (hr)
Measured Dexmedetomidine Cp (pg/mL)
Predicted Dexmedetomidine Cp (pg/mL)
Time since end of last infusion (hr)
Individual Predicted Dexmedetomidine Cp (pg/mL)

Model Validation

- In Figure 3, the median of the observed data and the 50th percentile of the simulated datasets are shown to correspond well across the time period displayed, with the exception of the time point between the end of the infusion and the 4-hour post-end-of-infusion sample due to the extremely small number of samples collected at around this time.
- The 5th and 95th percentiles of simulated data fall slightly outside corresponding percentiles of observed data, especially at 8 hours post-end-of-infusion, indicating slight over-estimation of variability with the model.

Figure 3. Percentiles of the Observed and Simulated Data for the Final 1-Compartment Model

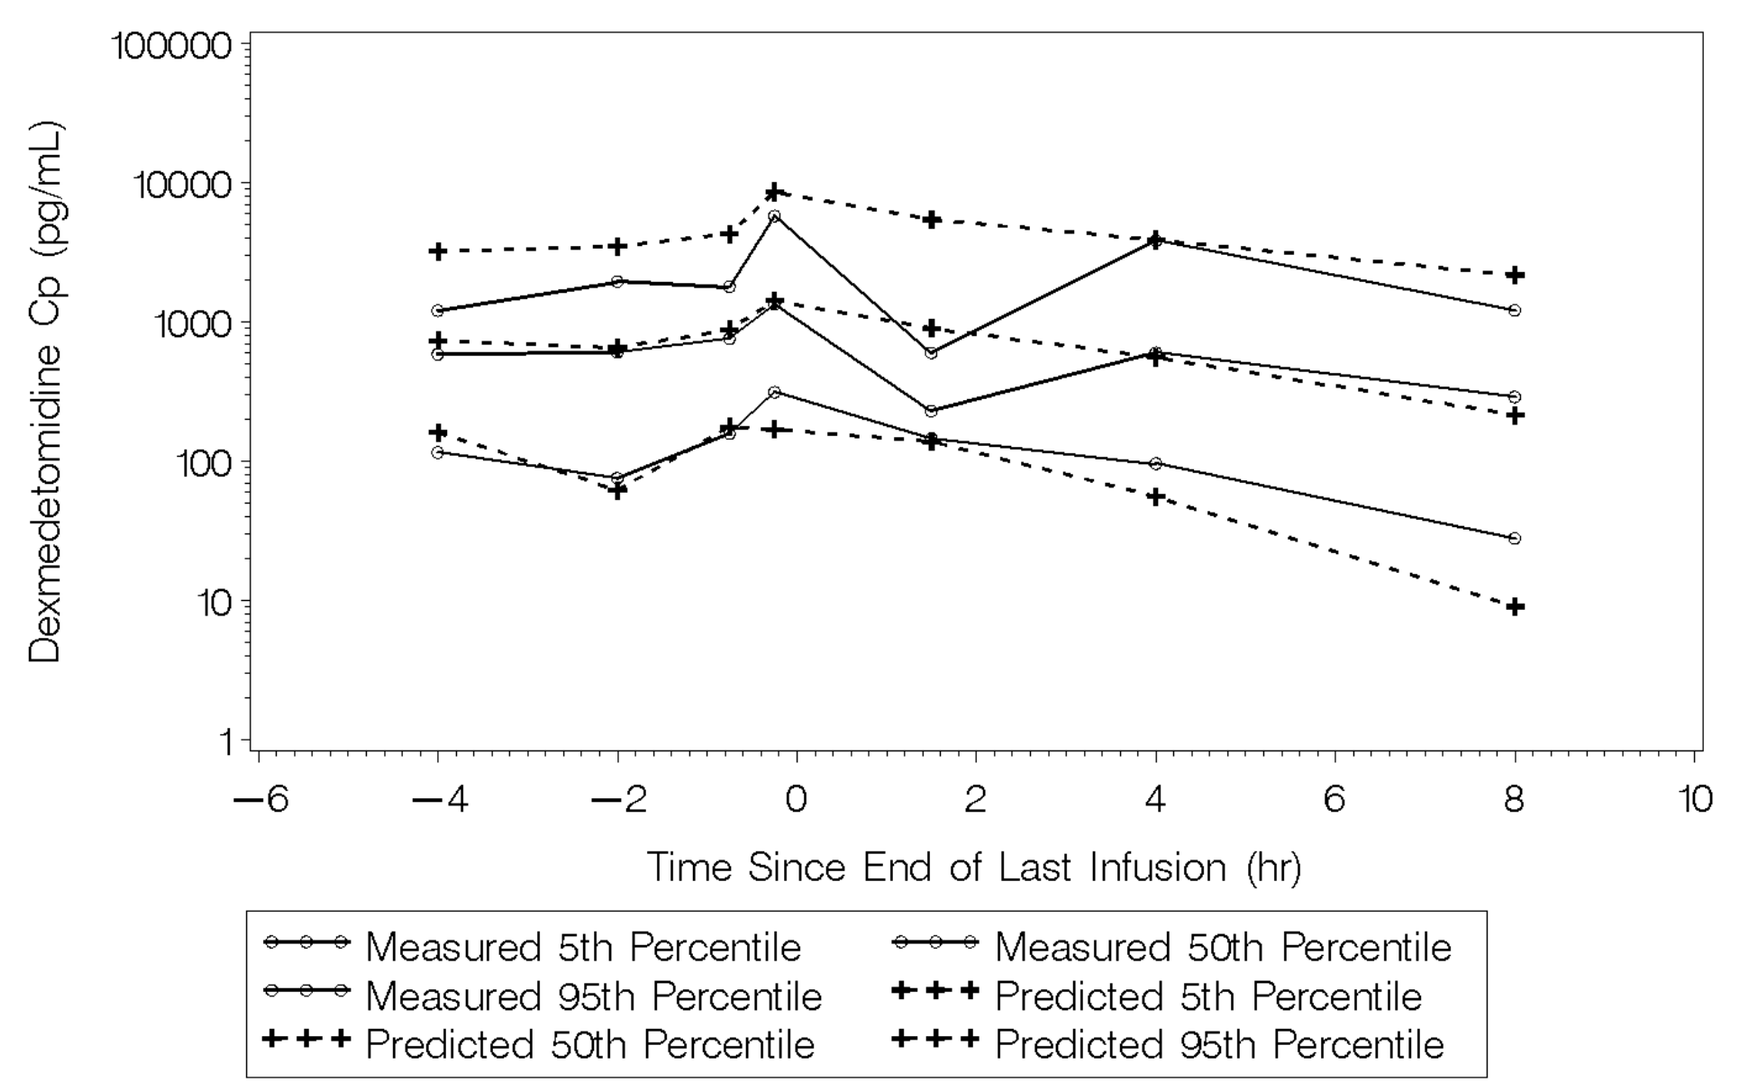
Dexmedetomidine Cp (pg/mL)
Time Since End of Last Infusion (hr)
Measured 5th Percentile
Measured 50th Percentile
Measured 95th Percentile
Predicted 5th Percentile
Predicted 50th Percentile
Predicted 95th Percentile

- A summary of the bias and precision measures for the 1-compartment model are shown in Table 2. Non-parametric statistical assessment of the median prediction error indicated a lack of bias in the fit.
- Only a small number (roughly 10%) of concentrations were substantially under- and over-predicted, however, no consistent patterns were identified with respect to the timing of these samples relative to dosing.

Table 2. Summary Measures of Bias and Precision for the 1-Compartment Model

Statistic	Percentile	1-Compartment Model
Prediction Error	50th	-18.4667[a]
	5th, 95th	-1616.98, 2884.28
Percent Prediction Error	50th	-3.7071
	5th, 95th	-431.70, 83.59
Percent Individual Prediction Error	50th	3.4877
	5th, 95th	-128.74, 51.86
Absolute Percent Prediction Error	50th	47.8163
	5th, 95th	3.71, 431.70
Absolute Percent Individual Prediction Error	50th	27.2776
	5th, 95th	1.75, 128.74

[a] Non-parametric sign test p-value = 0.3549 indicating no statistically significant difference from 0.

- The standardized mean prediction error (95% confidence interval) of -0.0900 (-1.90306, 1.48112) was not statistically different from the expected value of 0, indicating a lack of model bias.
- The standard deviation of the standardized mean prediction error (95% confidence interval) of 1.0127 (0.71639, 1.28225) was not statistically different from the expected value of 1, indicating that the model accurately describes between- and within-patient variability in observed concentrations.

Covariates

- Variability in patient demographics and baseline lab values, as well as variable dose administration (total dose amount, overall length of infusion, infusion rate changes) likely contributed to the relatively high degree of unexplained IIV in dexmedetomidine Cl and Vd, and high RV.
- Relationships between patient covariates (weight, age, CrCL) and DEX PK likely explain some inter-patient differences.
- An apparent positive relationship between Cl and body weight is evident in Figure 4, with increasing dexmedetomidine Cl as body weight increases. A similar pattern can be observed between Vd and body weight in Figure 5.
- No relationship between dexmedetomidine Cl and estimated CrCL (capped at 160 mL/min) was apparent as shown in Figure 6.

Figure 4. Individual Estimates of Dexmedetomidine Clearance Versus Body Weight

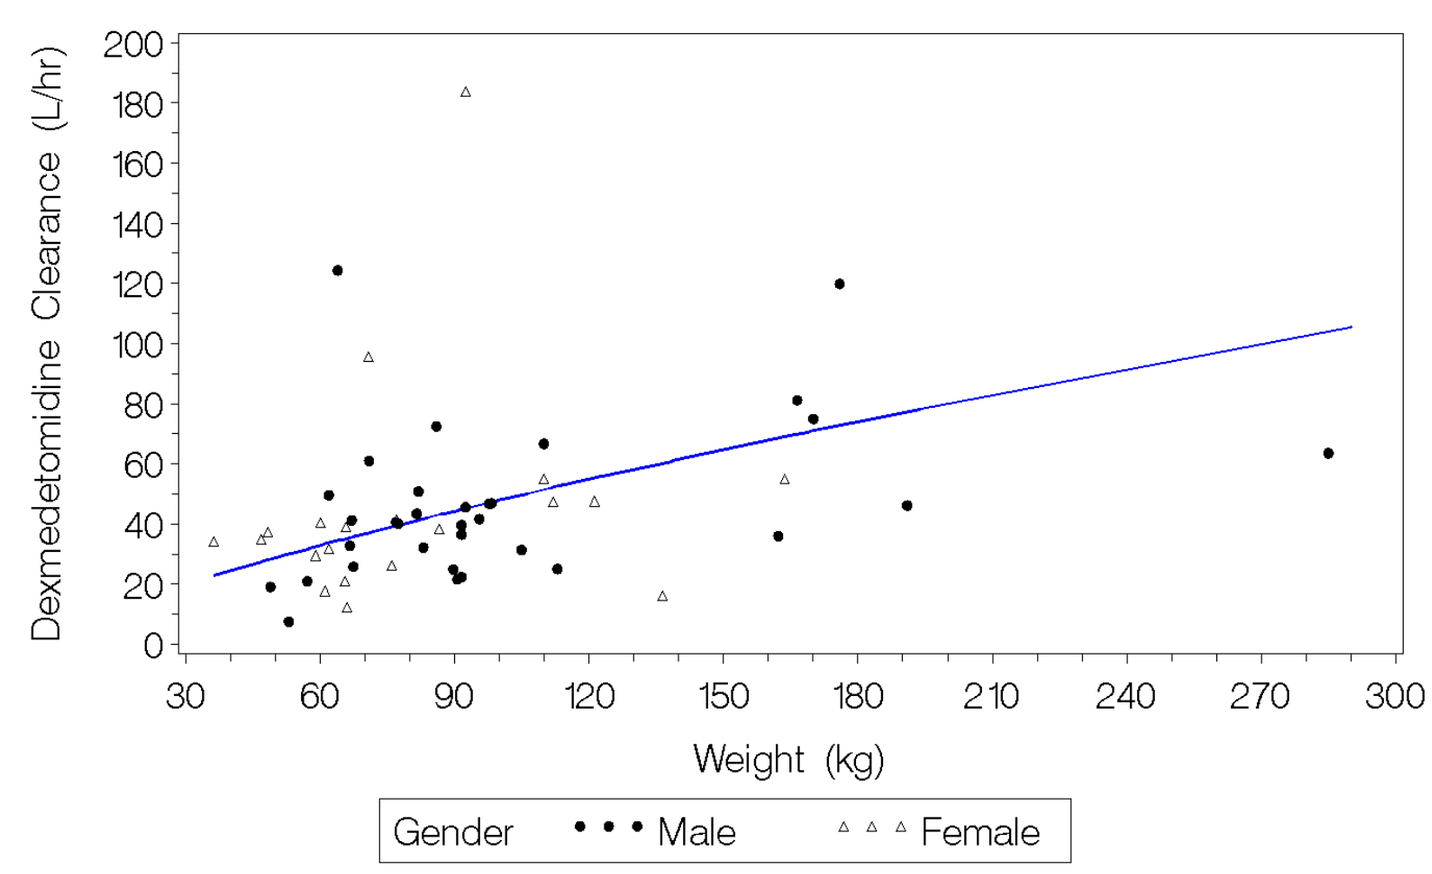
Dexmedetomidine Clearance (L/hr)
Weight (kg)
Gender
Male
Female

For visualization purposes, an allometric function is overlaid, where CL = 1.48*weight^0.75.

Figure 5. Individual Estimates of Dexmedetomidine Volume of Distribution Versus Body Weight

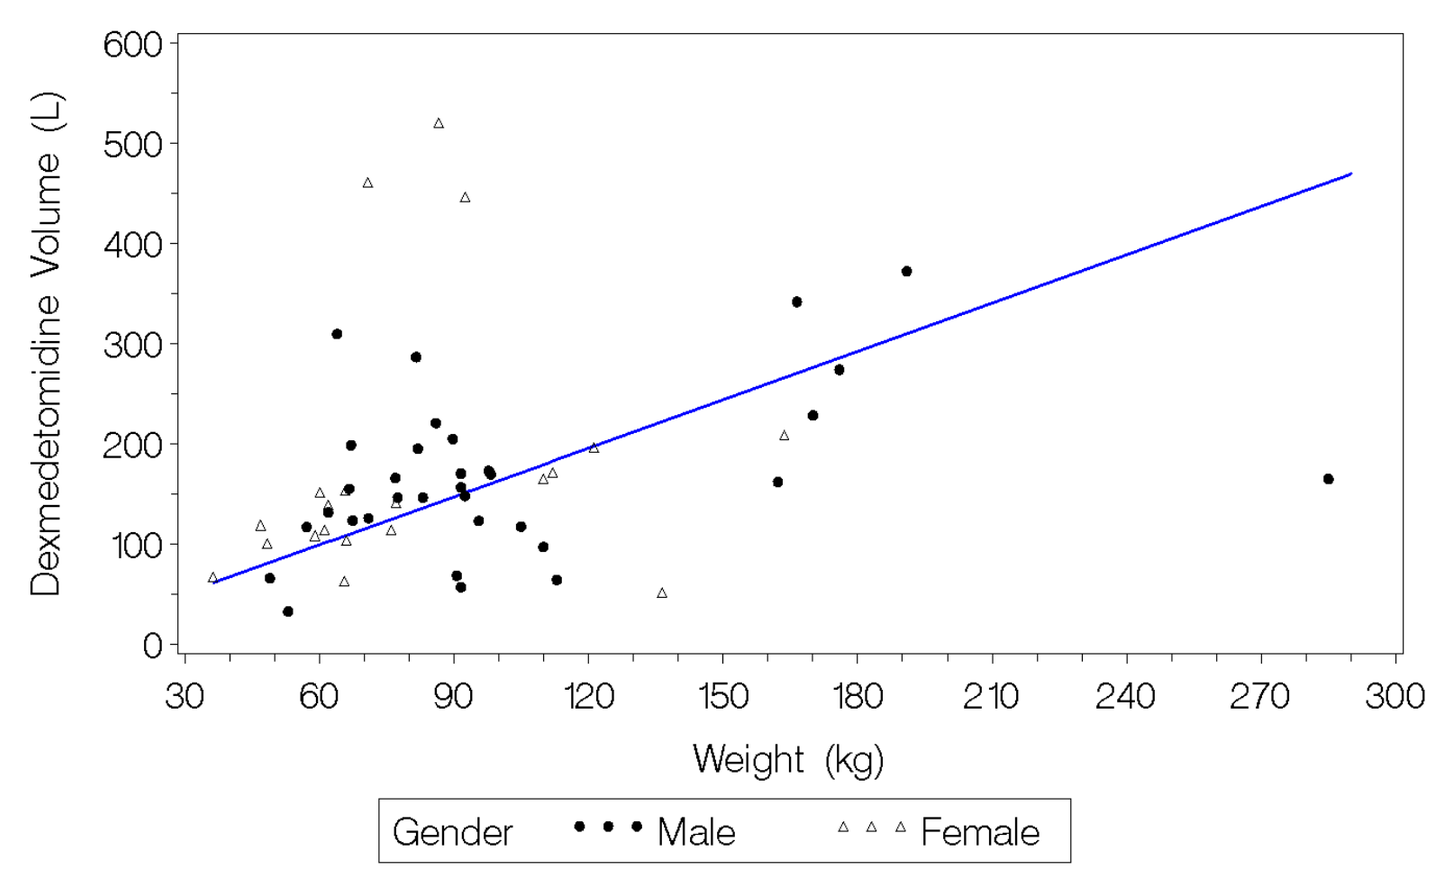
Dexmedetomidine Volume (L)
Weight (kg)
Gender
Male
Female

For visualization purposes, an allometric function is overlaid, where V = 1.61*weight.

Figure 6. Individual Estimates of Dexmedetomidine Clearance Versus Renal Function

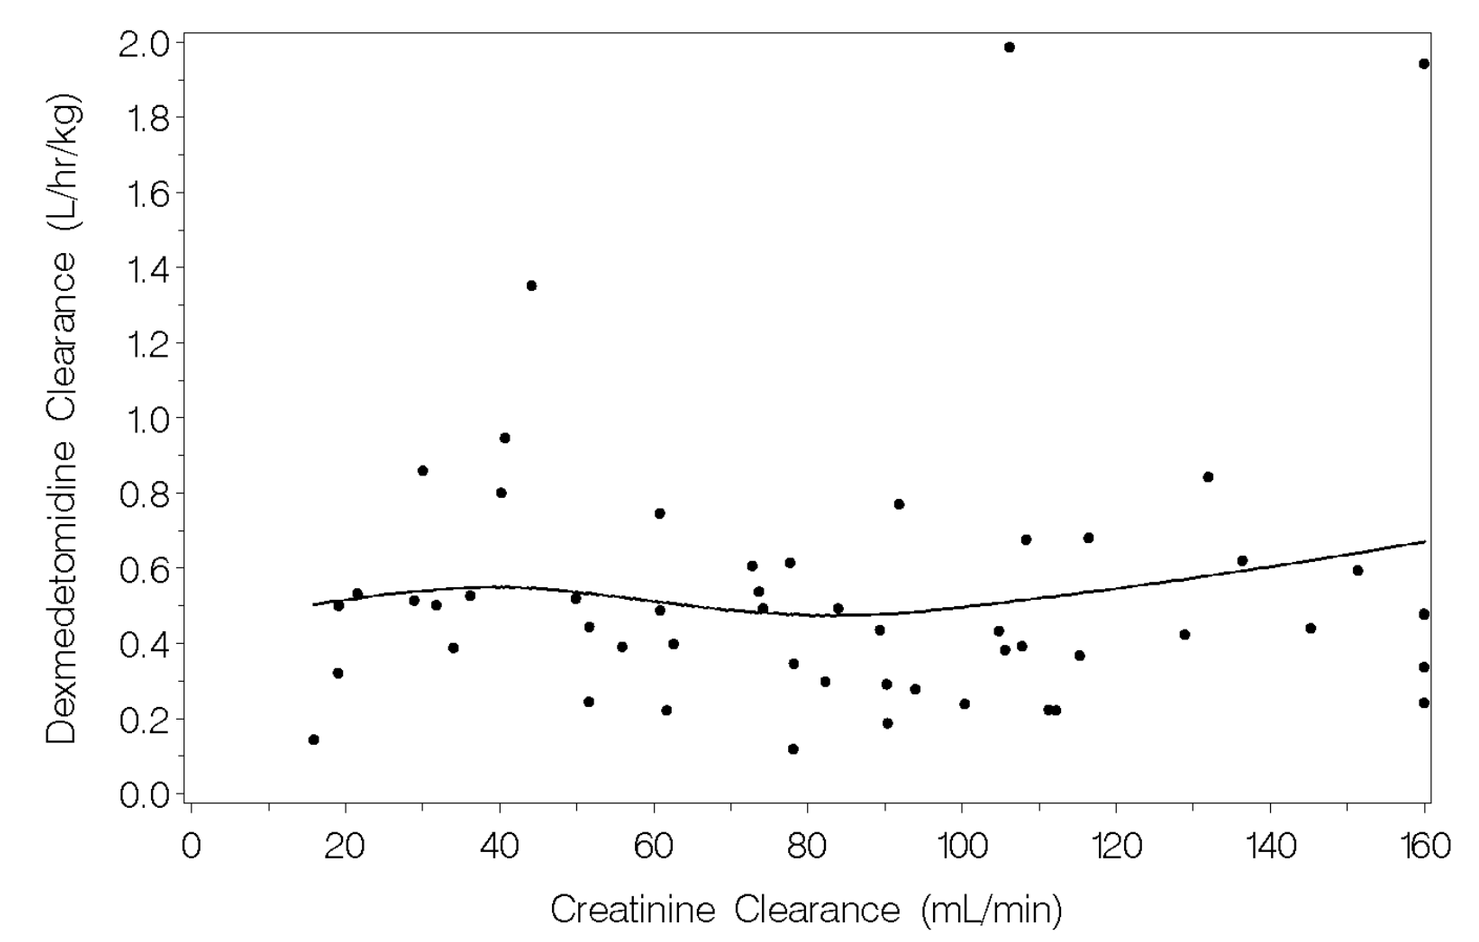
Dexmedetomidine Clearance (L/hr/kg)
Creatinine Clearance (mL/min)

The line represents a smoothing spline fit to the data.

CONCLUSIONS

- A 1-compartment model with linear elimination describes the sparsely sampled dexmedetomidine concentration-time data obtained from long-term infusions in the intensive care unit setting.
- The PK parameter estimates for dexmedetomidine clearance and elimination half-life are similar in ICU patients receiving long-term infusions as to previous estimates obtained in pooled analysis of short-term infusion studies in healthy subjects.
- The model validation results demonstrate adequate model fit for the 1-compartment model, given the variability in the observed data and the blood sampling strategy for dexmedetomidine concentrations employed in this Phase 4 study.
- Based on graphical evaluation, dexmedetomidine clearance and volume of distribution appear to increase with increasing body weight.
- Dexmedetomidine clearance does not appear to be influenced by renal function.

REFERENCES

1. Abbott Laboratories. Precedex (Dexmedetomidine Hydrochloride Injection) [package insert]. 2008.
2. Venn RM, Newman PJ, Grounds RM. A phase II study to evaluate the efficacy of dexmedetomidine for sedation in the medical intensive care unit. *Intensive Care Med*. 2003;29:201-207.
3. Hospira, Inc. *Investigator's Brochure for Dexmedetomidine HCl Injection*. IND Number 32,934. Lake Forest, IL: Hospira, Inc.; May 15, 2007.
4. Abbott Laboratories. *Dexmedetomidine as an Anesthetic Adjunct in Coronary Artery Bypass Grafting*. Drug Metabolism Report No. 24. Abbott-85499.
5. Wen YH, et al. Pharmacokinetic modeling in toxicology: a critical perspective. *Environ Rev*. 1999;7:1-18.
6. Guidance for Industry on Population Pharmacokinetics; Food and Drug Administration, HHS. *Fed Regist*. 1999;64:6663-6664.
7. Vozeh S, Maitre PO, Stanski DR. Evaluation of population (NONMEM) pharmacokinetic parameter estimates. *J Pharmacokinet Biopharm*. 1990;18:161-173.
8. Cockcroft DW, Gault MH. Prediction of creatinine clearance from serum creatinine. *Nephron*. 1976;16:31-41.