19

A POPULATION-BASED PK/PD ANALYSIS OF DASOTRALINE EFFICACY IN THE TREATMENT OF ADHD IN ADULTS

Seth C. Hopkins[1]; Soujanya Sunkaraneni[1]; Estela Skende[1]; Julie Passarell[2]; Jeremy Hing[2]; Antony Loebel[1]; Kenneth S. Koblan[1]

[1]Sunovion Pharmaceuticals Inc.; [2]Cognigen Corporation

ABSTRACT

Background: Dasotraline (SEP-225289) is a new chemical entity with a slow elimination half-life demonstrating dopamine (DAT) and norepinephrine (NET) transporter inhibition in clinical studies. Here we hypothesized dasotraline plasma concentrations, measured in an efficacy trial of adult ADHD, would be correlated with improvements in ADHD RS-IV across subjects, treatment groups, and visits.
Methods: Plasma concentrations of dasotraline from Phase I and Phase 2 studies were analyzed by population PK methodology. A 1-compartment population PK model with sequential zero-order followed by first-order absorption and dual (nonlinear and linear) elimination described dasotraline PK across a total of 395 subjects after single or multiple dose administrations ranging from 0.2 to 36 mg. Norepinephrine metabolite 3,4-dihydroxyphenylglycol (DHPG) concentrations from 220 subjects were modeled as a power function of the time-matched dasotraline concentrations as derived from the PK model. Population PK/PD model for ADHD RS-IV scores and dasotraline concentrations (Cav) was a sigmoid Emax time-course model.
Results: The population PK/PD model for ADHD RS-IV scores and dasotraline concentrations (Cav) was a sigmoid Emax time-course model. Steady-state plasma concentrations were attained by 2 weeks with a mean apparent half-life of 47 hours. Concentrations of the norepinephrine metabolite DHPG indicated central NET inhibition was achieved within the first days of dosing. An exposure-response relationship was found between increases in Cav, Emax and reductions in ADHD RS-IV.
Discussion: These results demonstrate a dose- and concentration-response relationship of pharmacological activity in ADHD, supporting the concept that maintaining constant, steady-state inhibition of both dopamine and norepinephrine transporters is a novel pharmacological approach to the management of ADHD symptoms.

Clinicaltrials.gov identifier: NCT01692782.

BACKGROUND

- ADHD (Attention-Deficit/Hyperactivity Disorder) is a neurodevelopmental disorder characterized by symptoms of inattention, hyperactivity and impulsivity associated with clinically significant impairment in functioning
- Dopamine and norepinephrine are associated with the pathophysiology of ADHD, and drugs that facilitate synaptic concentrations of dopamine and norepinephrine are clinically useful in the pharmacological management of ADHD symptoms
- Dasotraline [(1R,4S)-4-(3,4-Dichlorophenyl)-1,2,3,4-tetrahydronaphthalen-1-amine] is a novel compound in clinical development for the treatment of ADHD
- Dasotraline is a potent inhibitor of human DA transporters (DAT; dopamine uptake IC_{50} 3 nM) and NE transporters (NET; norepinephrine uptake IC_{50} 4 nM), and a weaker inhibitor of human serotonin transporters (SERT; serotonin uptake IC_{50} 15 nM) (Sunovion data on file)
- The dasotraline pharmacokinetic profile of slow absorption/elimination is unique among current stimulant and nonstimulant medications indicated for ADHD, and can support relatively stable plasma concentrations over a 24-hour daily dosing interval
- A Phase 2 clinical trial with dasotraline demonstrated statistically and clinically meaningful effects in adults with ADHD

OBJECTIVE

- The objective of these pharmacokinetic (PK) and pharmacodynamics (PD) analyses was to characterize the time-course and exposure-response relationships between dasotraline, the norepinephrine (NE) metabolite 3,4-dihydroxyphenylglycol (DHPG), and improvement in severity of ADHD symptoms as measured by the ADHD RS-IV

METHODS

Population PK/PD

- A dasotraline population PK model was developed based on 4,570 dasotraline measurements in 395 subjects after single or multiple administrations of dasotraline in doses ranging from 0.2 to 36 mg
- A total of 759 DHPG measurements from 220 subjects treated with dasotraline in Study 201 were included in the PK/PD DHPG analysis

Pharmacodynamic Responses

- Data from a Phase 2 study (NCT01692782)[1] were used for PK/PD analyses of plasma DHPG concentrations and ADHD RS-IV total scores (assessed at each study visit)
- A total of 1847 measurements from 330 patients were included in the PK/PD analysis of the effect of dasotraline plasma concentrations on change in ADHD RS-IV total scores
- The relationship between ADHD RS-IV responses and dasotraline concentrations was a sigmoid Emax time-course model with concentration as a linear function on Emax
- Concentrations of the norepinephrine metabolite, DHPG, obtained from 220 subjects were modeled as a power function of the time-matched dasotraline concentrations as derived from the PK model

Exposure Measurements

- Dasotraline and DHPG concentrations were determined using HPLC with MS/MS detection
- For dasotraline measurements in the 201 study, the inter-assay coefficient of variation for quality control samples ranged from 2.0-2.7%; lower limit of quantitation (LLOQ) was 10.0 pg/mL
- For DHPG measurements in the 201 study, the inter-assay coefficient of variation for quality control samples ranged from 4.6-7.1%; lower limit of quantitation (LLOQ) was 200 pg/mL
- The population PK model for dasotraline was used to generate empiric Bayesian PK parameters for each individual

RESULTS

Pharmacokinetics of Dasotraline

Figure 1. Slow Absorption and Elimination of Dasotraline in Adult Subjects Requires Daily Doses to Steady State

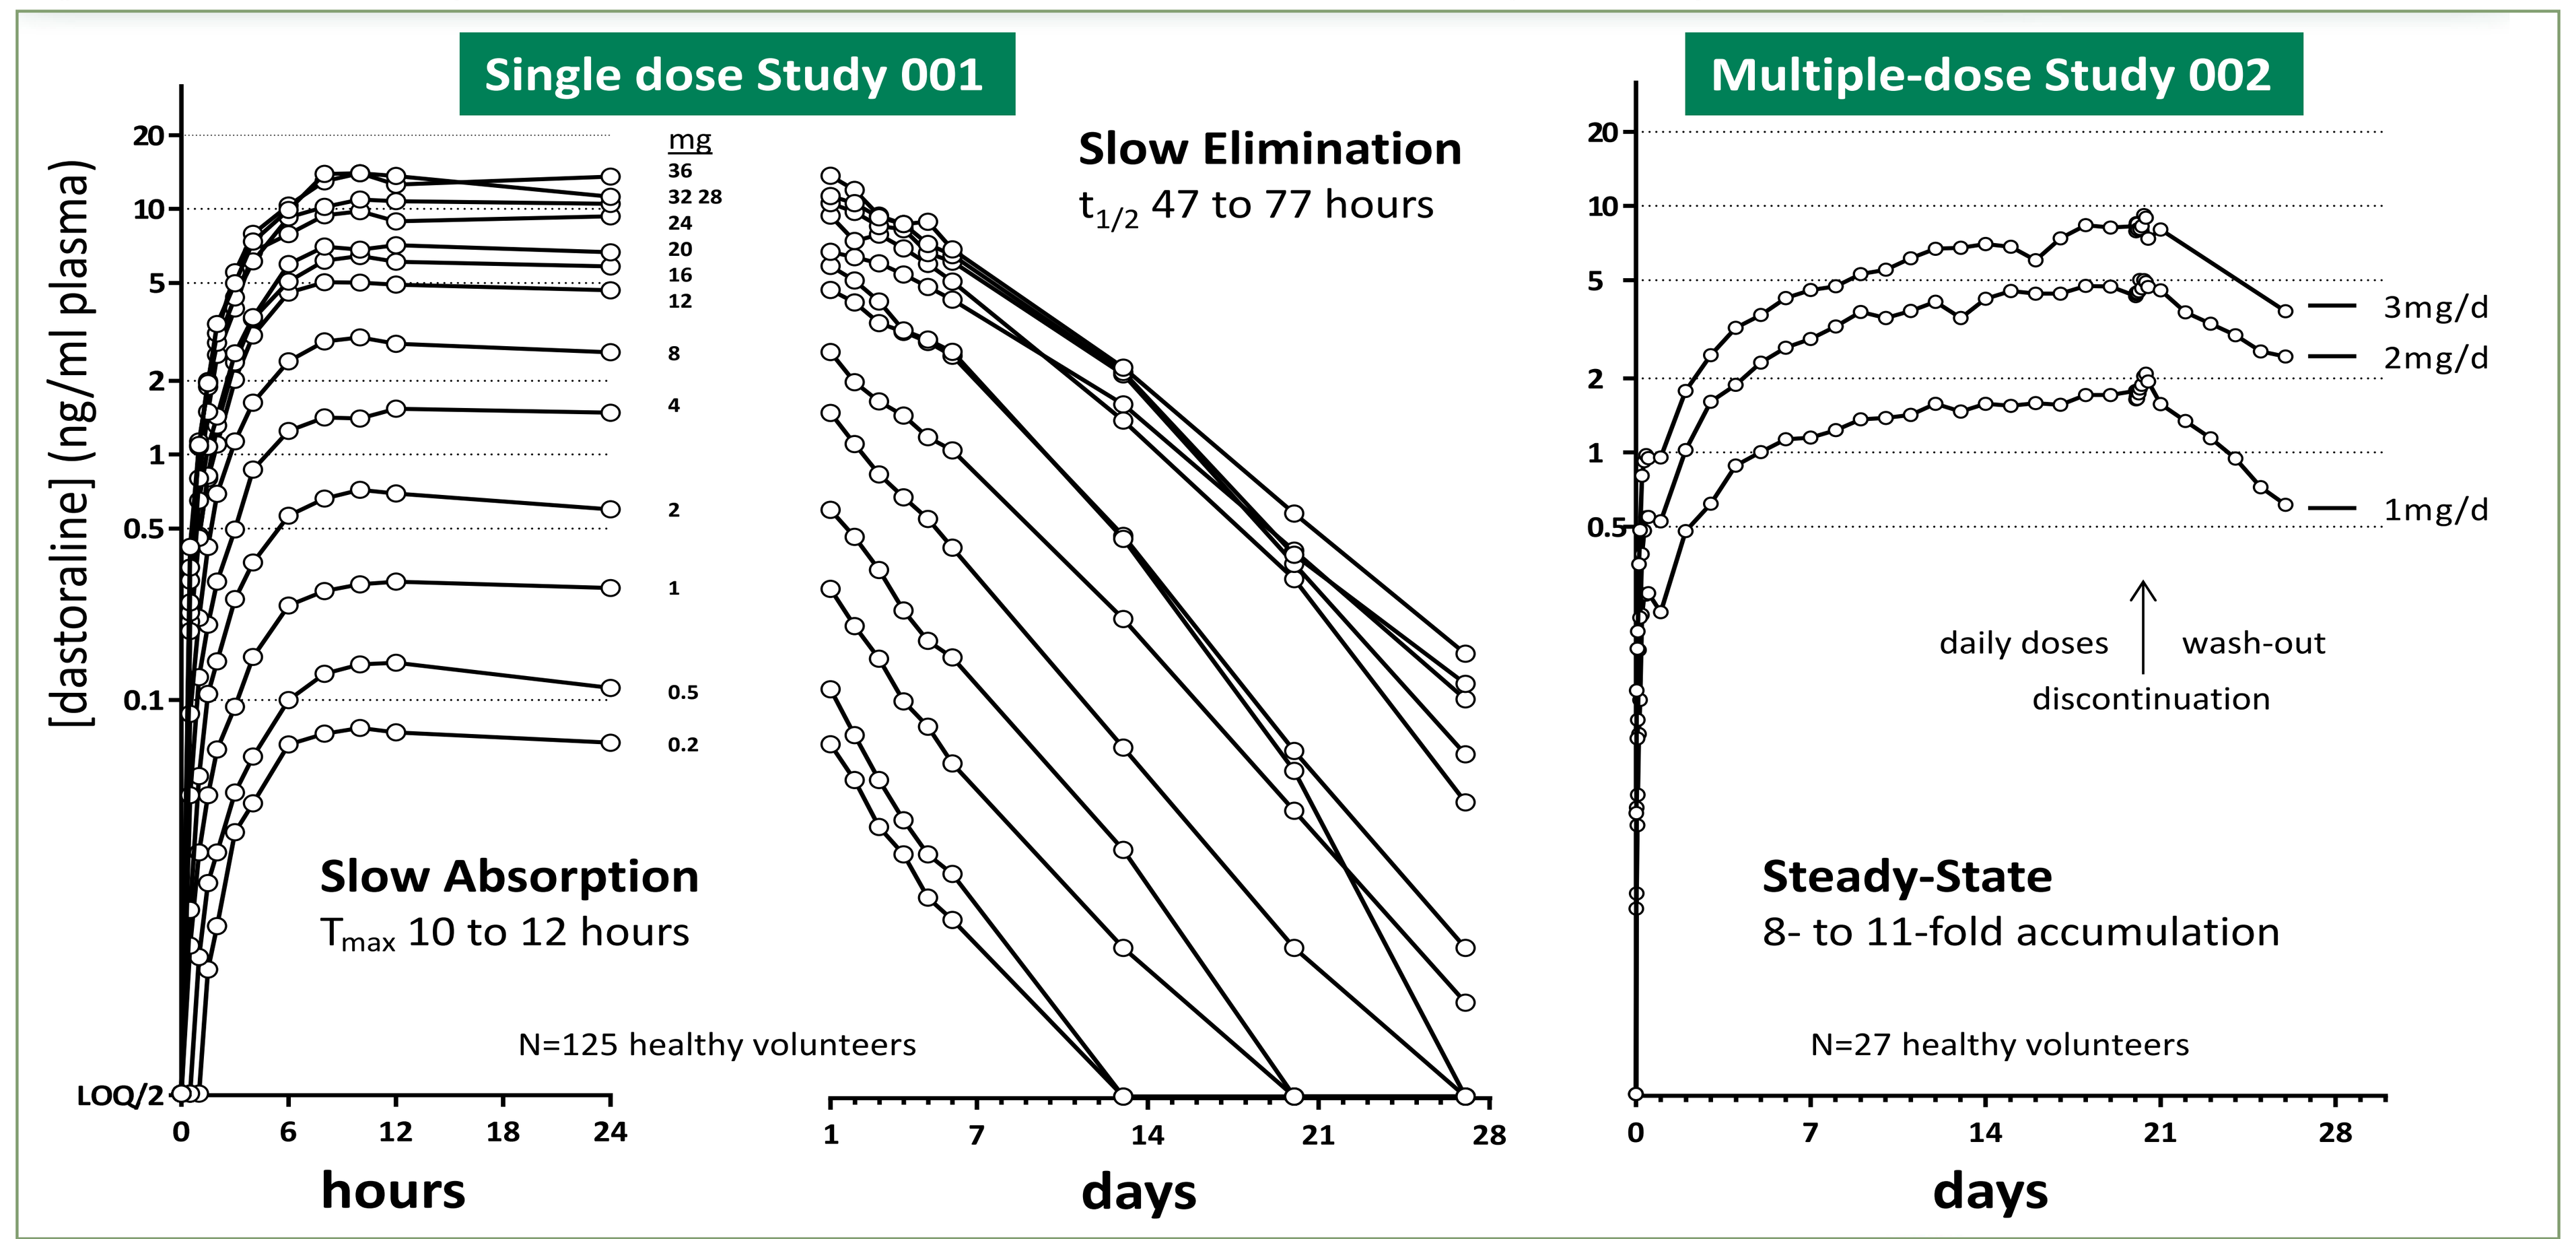

Single dose Study 001
Multiple-dose Study 002
[dastoraline] (ng/ml plasma)
Slow Absorption
T_{max} 10 to 12 hours
Slow Elimination
$t_{1/2}$ 47 to 77 hours
N=125 healthy volunteers
hours
days
Steady-State
8- to 11-fold accumulation
daily doses
wash-out
discontinuation
3mg/d
2mg/d
1mg/d
N=27 healthy volunteers
days

- Mean dasotraline plasma concentrations (ng/mL) are presented on a logarithmic scale:
- Left Panel: following single doses from 0.2 mg to 36 mg, N=9 subjects (N=8 for 0.5 mg)
- Right Panel: following multiple doses for 21 days followed by 7 day washout (N=9, 8, 7 for 1, 2, 3 mg/d, respectively)

Population PK Model

Figure 2. Population PK Model: Dasotraline Pharmacokinetics Are Well-described by a 1-compartment Model

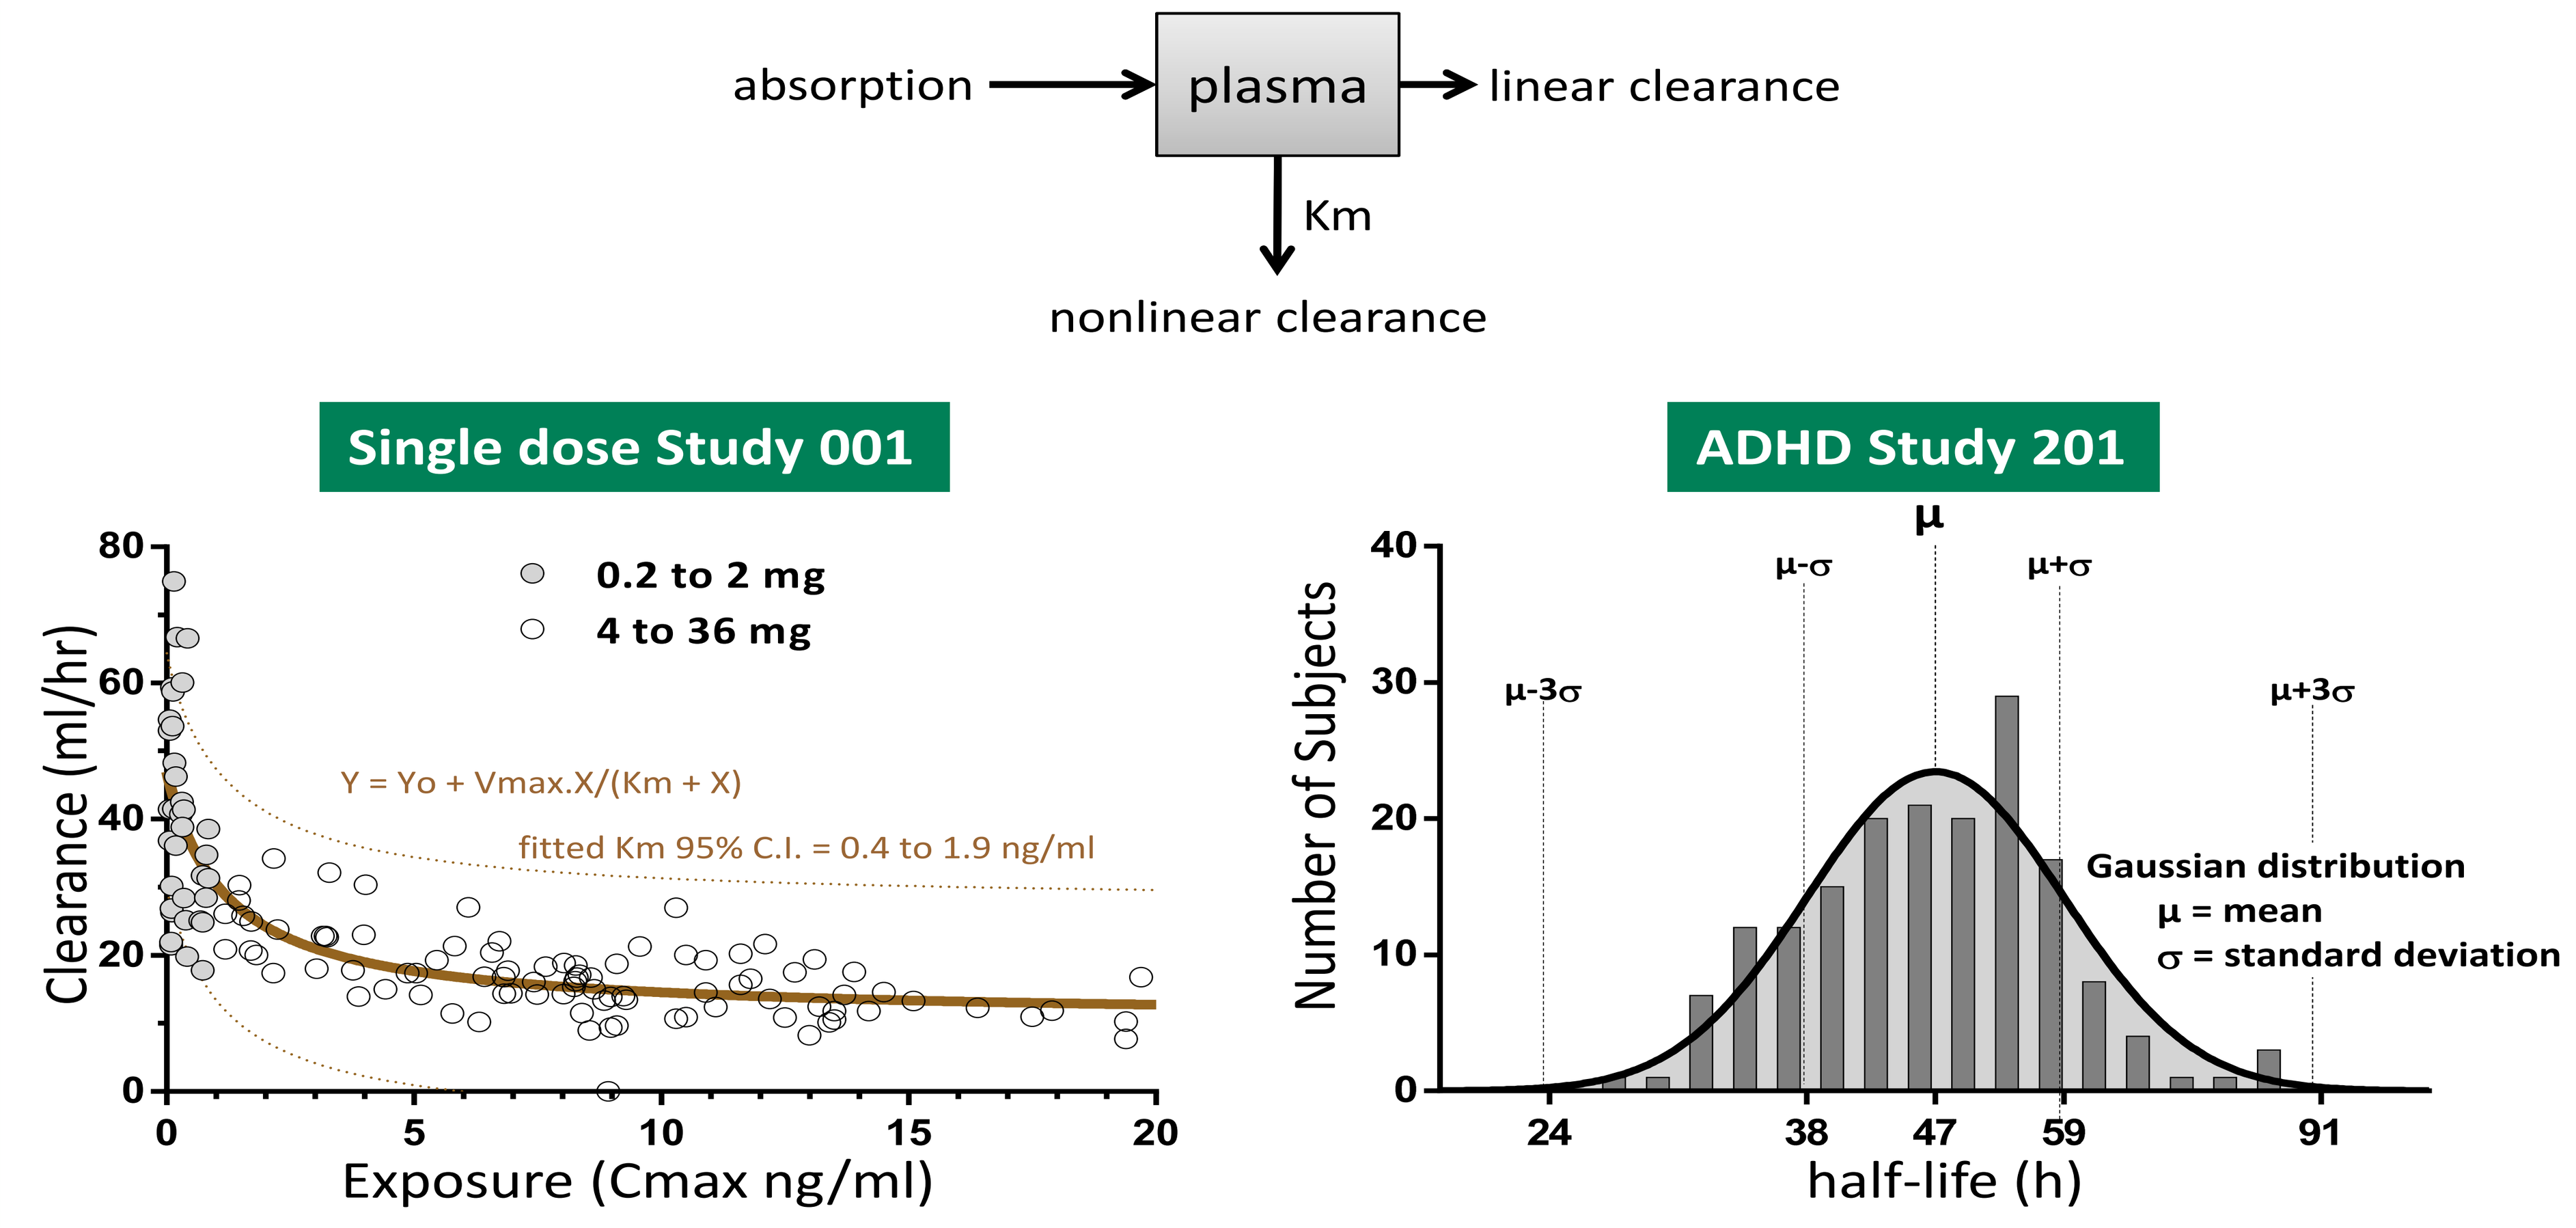

absorption
plasma
linear clearance
Km
nonlinear clearance
Single dose Study 001
0.2 to 2 mg
4 to 36 mg
Y = Yo + Vmax.X/(Km + X)
fitted Km 95% C.I. = 0.4 to 1.9 ng/ml
Clearance (ml/hr)
Exposure (Cmax ng/ml)
ADHD Study 201
Gaussian distribution
μ = mean
σ = standard deviation
Number of Subjects
half-life (h)

- Schematic: Dasotraline pharmacokinetics was described by a 1-compartment model with dual (nonlinear and linear) elimination
- Left Panel: In a single dose study of dasotraline, the relationship between clearance and exposure (Cmax) was found to be nonlinear. The nonlinear component saturated with apparent half-maximal (Km) of 0.4 to 1.9 ng/ml (95% CI from curve fitting), consistent with dual (linear and nonlinear) elimination
- Right Panel: Distribution of half-life values during washout, following 4 weeks of daily dosing, were obtained by simulations of individuals in Study 201 with the population PK model
- As concentrations of dasotraline increased above 3 ng/mL, the nonlinear component contributed less to the total elimination
- The following variables were not found to be associated with variability in the population PK model: age, total bilirubin, alanine aminotransferase (ALT), gender, race, or ethnicity

Figure 3. PK/PD Models: Relationship Between Dasotraline and DHPG Values; and ADHD RS-IV Values

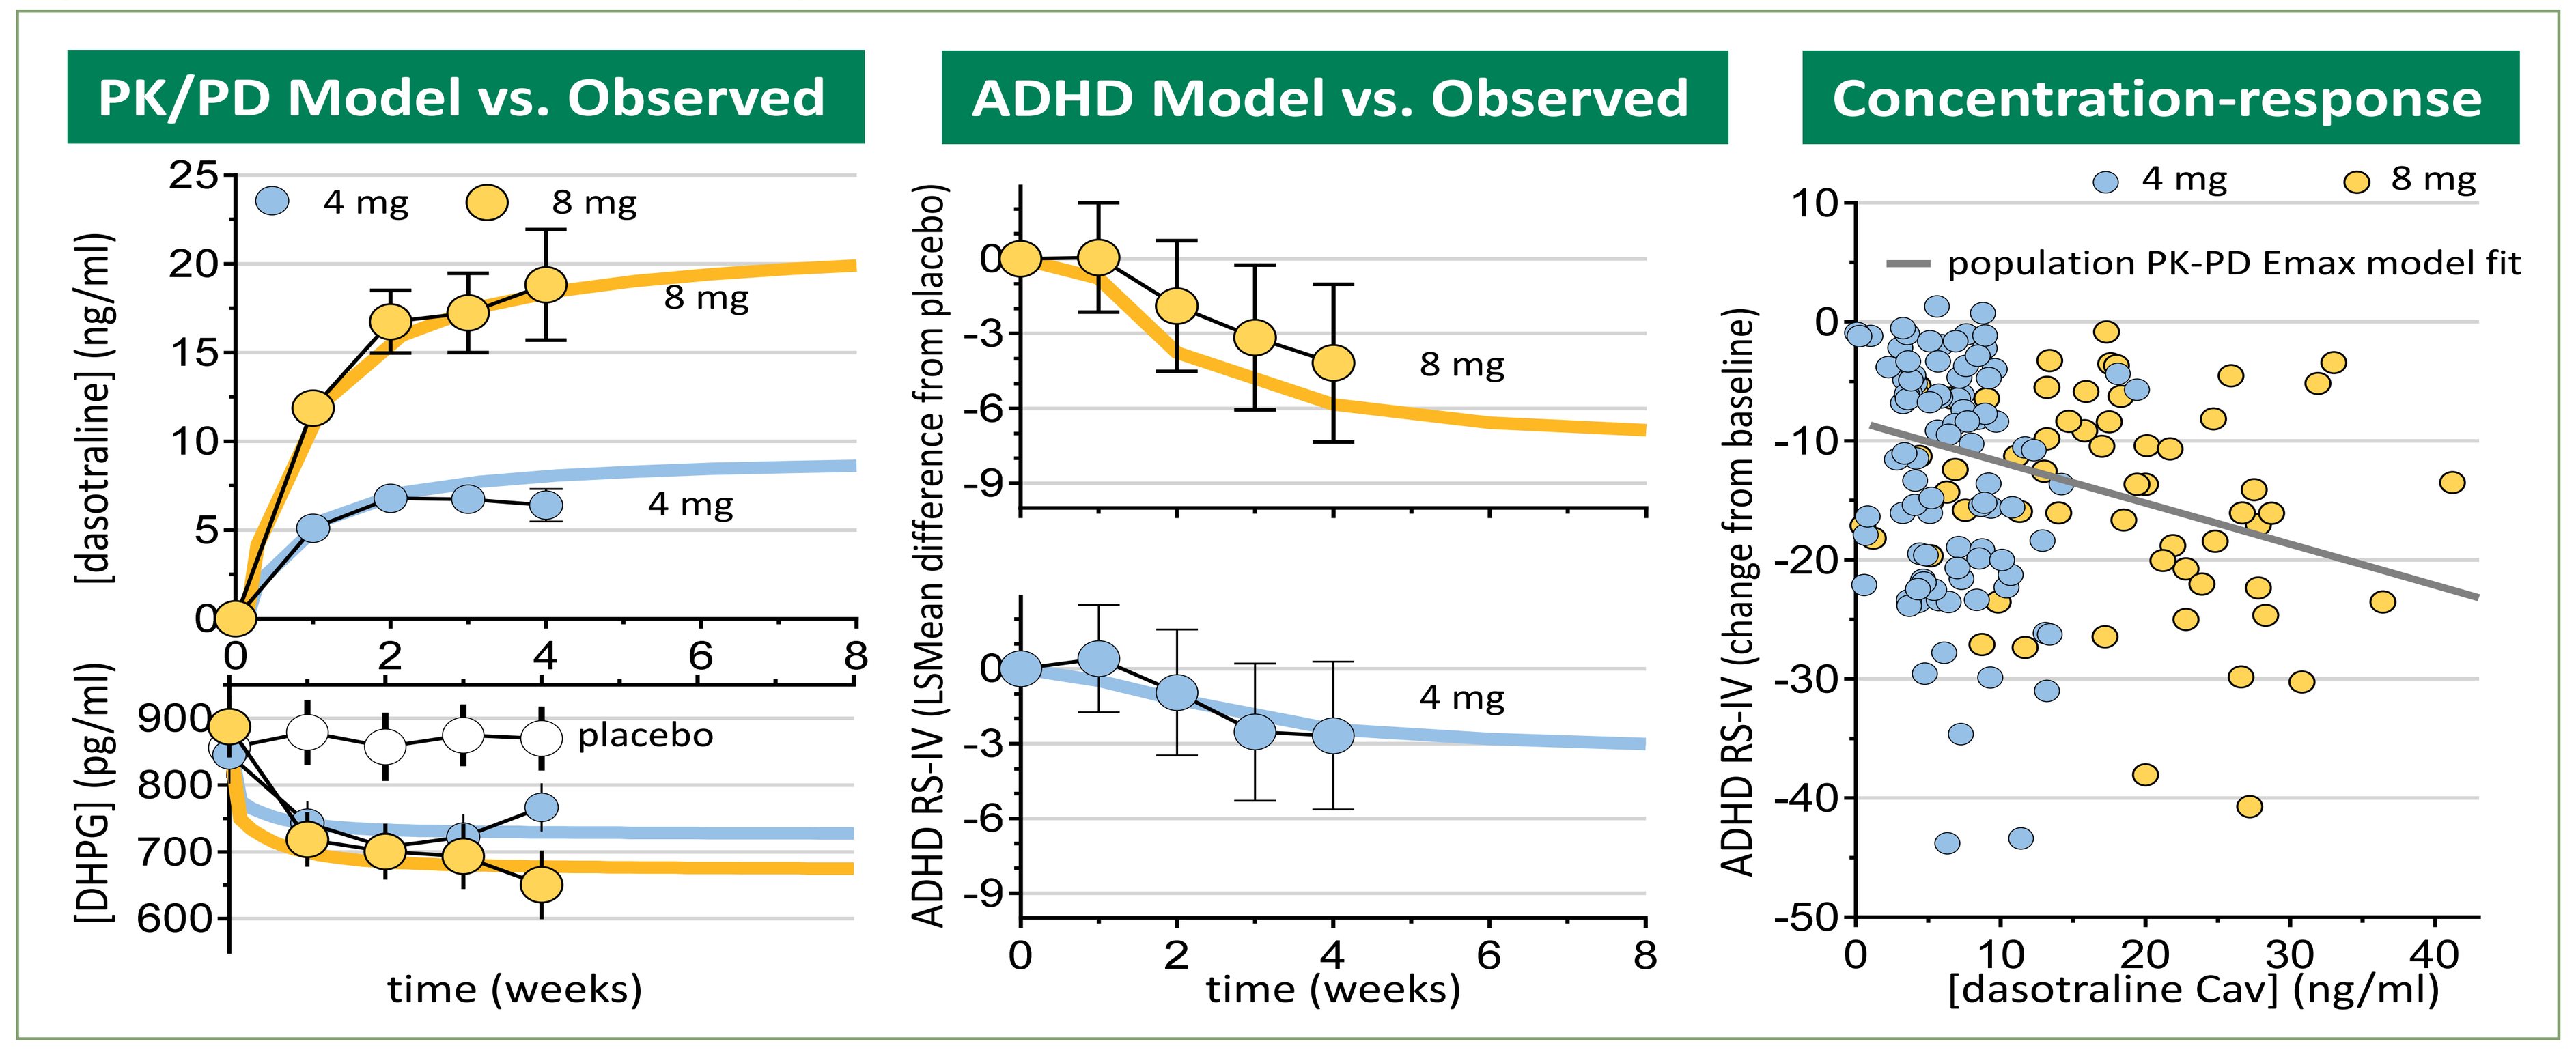

PK/PD Model vs. Observed
ADHD Model vs. Observed
Concentration-response
4 mg
8 mg
[dasotraline] (ng/ml)
[DHPG] (pg/ml)
placebo
time (weeks)
ADHD RS-IV (LSMean difference from placebo)
population PK-PD Emax model fit
ADHD RS-IV (change from baseline)
[dasotraline Cav] (ng/ml)

PK/PD Model vs. Observed

- Levels of DHPG decreased as plasma concentrations of dasotraline increased, indicating that treatment with dasotraline was associated with NET inhibition
- In an analysis of the influence of covariate effects on reduction in DHPG levels, the following variables had no effect: age, baseline weight or BMI, baseline DHPG, gender, race, or ethnicity

ADHD RS-IV Change Model vs. Observed

- In Study 201, a randomized, double-blind, placebo controlled proof-of-concept study in adults with ADHD, LS mean improvement at Week 4 in ADHD RS-IV total score were significantly greater for dasotraline 8 mg/d vs placebo (-13.9 vs -9.7; P=0.019), and non-significantly greater for 4 mg/d (-12.4; P=0.076)
- Reductions in ADHD symptoms observed in ADHD Study 201 (circles show LS mean [+95% C.I] vs placebo) compared to population PK/PD model predictions (lines)
- In an analysis of the influence of covariate effects on reduction in ADHD RS-IV total scores, the following variables had no statistically significant effect: age, baseline weight, baseline BMI, baseline ADHD RS-IV total score, baseline DHPG, baseline insomnia severity index, gender, race, and ethnicity

Figure 4. Effect Sizes of Dasotraline Are Comparable to Methylphenidate in Adults With ADHD

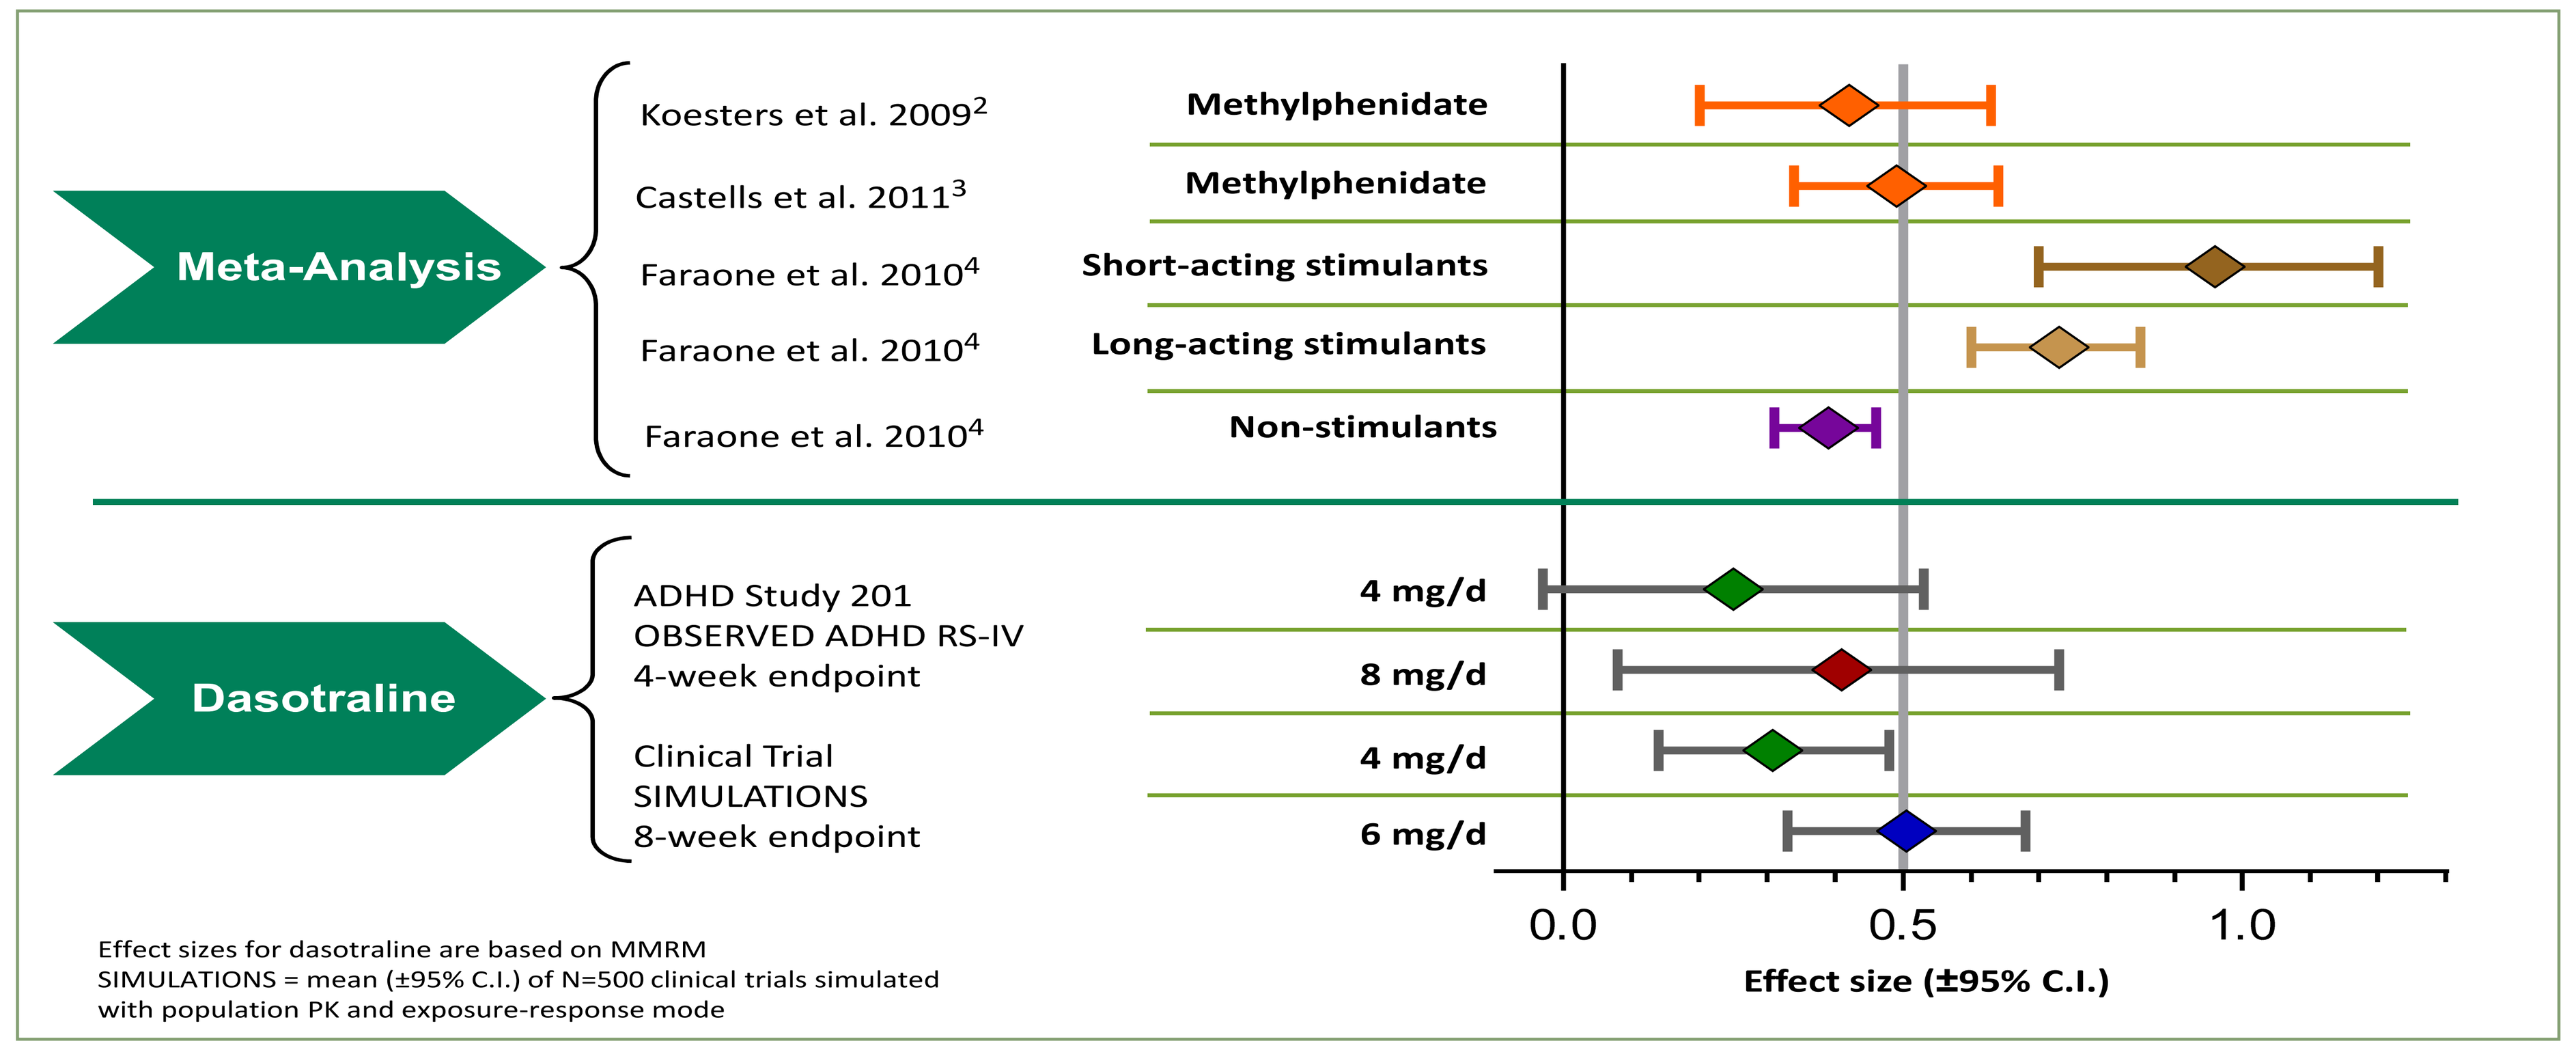

Meta-Analysis
Koesters et al. 2009[2]
Castells et al. 2011[3]
Faraone et al. 2010[4]
Faraone et al. 2010[4]
Faraone et al. 2010[4]
Methylphenidate
Methylphenidate
Short-acting stimulants
Long-acting stimulants
Non-stimulants
Dasotraline
ADHD Study 201
OBSERVED ADHD RS-IV
4-week endpoint
Clinical Trial
SIMULATIONS
8-week endpoint
4 mg/d
8 mg/d
4 mg/d
6 mg/d
Effect sizes for dasotraline are based on MMRM
SIMULATIONS = mean (±95% C.I.) of N=500 clinical trials simulated with population PK and exposure-response mode
Effect size (±95% C.I.)

DISCUSSION

- Individual measures of dasotraline exposure were well-described by the dasotraline population PK model
- Models of time-course and exposure-response described correlations between dasotraline exposure and pharmacodynamic outcomes (reduction in ADHD symptoms); and inhibition of norepinephrine reuptake (decreases in plasma DHPG concentrations)
- The pharmacokinetic profile of dasotraline, with slow absorption/elimination, is unique among ADHD treatments
- After a 10-12 hour absorption phase is complete, plasma concentrations of dasotraline remain relatively stable over a 24-hour dosing interval
- Plasma concentrations of dasotraline accumulate slowly over the approximately 2 weeks of daily doses to establish stead-state levels
- The population mean estimate of dasotraline half-lives in a virtual population of ADHD subjects administered dasotraline 4 or 8 mg/d for 4 weeks was 47 hours, indicating 10 days is an appropriate clinical estimate of the 5 half-lives for washout or establishing steady-state
- The population distribution of dasotraline half-lives was relatively narrow, with 1 standard deviation below and above the mean 47 hours estimated at 38 hours to 59 hours, respectively

CONCLUSIONS

- These results describe quantitatively the exposure-response relationships of dasotraline and its pharmacological activity in ADHD
- The PK and PK/PD models described here were developed to support the further clinical development of dasotraline in ADHD
- The novel PK/PD profile of dasotraline in ADHD supports the concept that maintaining constant, steady-state inhibition of both dopamine and norepinephrine transporters is a novel pharmacological approach to the management of ADHD symptoms. The results of the initial proof-of-concept study in ADHD (Study 201) indicated that effect sizes were comparable to methylphenidate

REFERENCES

1. Koblan KS et al. Poster presentation, American College of Neuropsychopharmacology annual meeting, Phoenix, AZ, Dec 7-11, 2014.
2. Koesters M et al. *J Psychopharmacol.* 2009;23:733-44.
3. Castells X et al. *CNS Drugs.* 2011;25:157-69.
4. Faraone SV, Glatt SJ. *J Clin Psychiatry.* 2010;71:754-63.

DISCLOSURES

Drs. Hopkins, Sunkaraneni, Skende, Loebel, and Koblan are employees of Sunovion Pharmaceuticals Inc.
Drs. Passarell and Hing are employees of Cognigen Corp., which performed the PK/PD analyses under a contract with Sunovion.

Sponsored by Sunovion Pharmaceuticals Inc.

American Professional Society of ADHD and Related Disorders, Jan 16-18, 2015, Washington, D.C.