Eslicarbazepine acetate drug–drug interactions: characterization through a model-based population approach

W-20

Soujanya Sunkaraneni,[1] Elizabeth Ludwig,[2] David Blum,[1] Jill Fiedler-Kelly[2]

[1]Sunovion Pharmaceuticals Inc., Marlborough, MA, USA; [2]Cognigen Corporation (a wholly owned subsidiary of Simulations Plus, Inc.), Buffalo, NY, USA

INTRODUCTION

- Eslicarbazepine acetate (ESL) is a once-daily (QD) oral antiepileptic drug (AED), approved by the US Food and Drug Administration for the treatment of partial-onset seizures (POS) as monotherapy or adjunctive therapy. ESL is approved by the European Medicines Agency as adjunctive therapy of POS in adults.
- Following oral administration, ESL is rapidly and extensively metabolized to the active metabolite, eslicarbazepine, which accounts for approximately 95% of systemic drug exposure[1,2] (the minor metabolites, oxcarbazepine [OXC] and R-licarbazepine, account for <1% and ~5%, respectively).[3]
 - The maximum plasma concentration (C_{max}) of eslicarbazepine is reached approximately 3 hours post-dose, with steady-state being achieved after 4–5 days of QD dosing.[1,2]
 - Eslicarbazepine is mainly eliminated by renal excretion (unchanged or following glucuronidation).[2,4]
- The effect of concomitant administration of ESL on the pharmacokinetics (PK) of phenytoin (PHT), lamotrigine (LTG), topiramate (TPM), and carbamazepine (CBZ) was investigated in four Phase I, open-label, drug-drug interaction (DDI) studies in healthy volunteers.
- DDIs between ESL and other AEDs were further characterized using population PK models.
 - A population PK model was developed for eslicarbazepine using data from 11 Phase I studies and three Phase III studies of adjunctive ESL.
 - Population PK models for six other AEDs (CBZ, valproate [VAL], levetiracetam [LEV], phenobarbital [PB], PHT, and gabapentin [GBP]) were developed using data from the three Phase III studies of adjunctive ESL.
- The population PK models were used to characterize the effect of concomitant AED use on PK of eslicarbazepine, and the effect of concomitant ESL on the PK of the other AEDs.

OBJECTIVES

- Conduct a population model-based characterization of DDIs, to determine whether AEDs used concomitantly with ESL for the treatment of POS had clinically meaningful effects on eslicarbazepine PK, and whether ESL had clinically meaningful effects on the PK of the other AEDs.

METHODS

Phase I DDI studies

- Four open-label, Phase I studies (BIA-2093-119, -120, -121 and -129) were conducted to determine the effect of concomitant administration of ESL and LTG, TPM, PHT or CBZ, on the PK of eslicarbazepine and the other AEDs.
 - In each study, two groups of healthy volunteers (n = 16–20) took one of the other AEDs, alone and in combination with ESL, for 27 days (35 days for CBZ; see **Figure 1**)

Figure 1. Design of the Phase I DDI studies

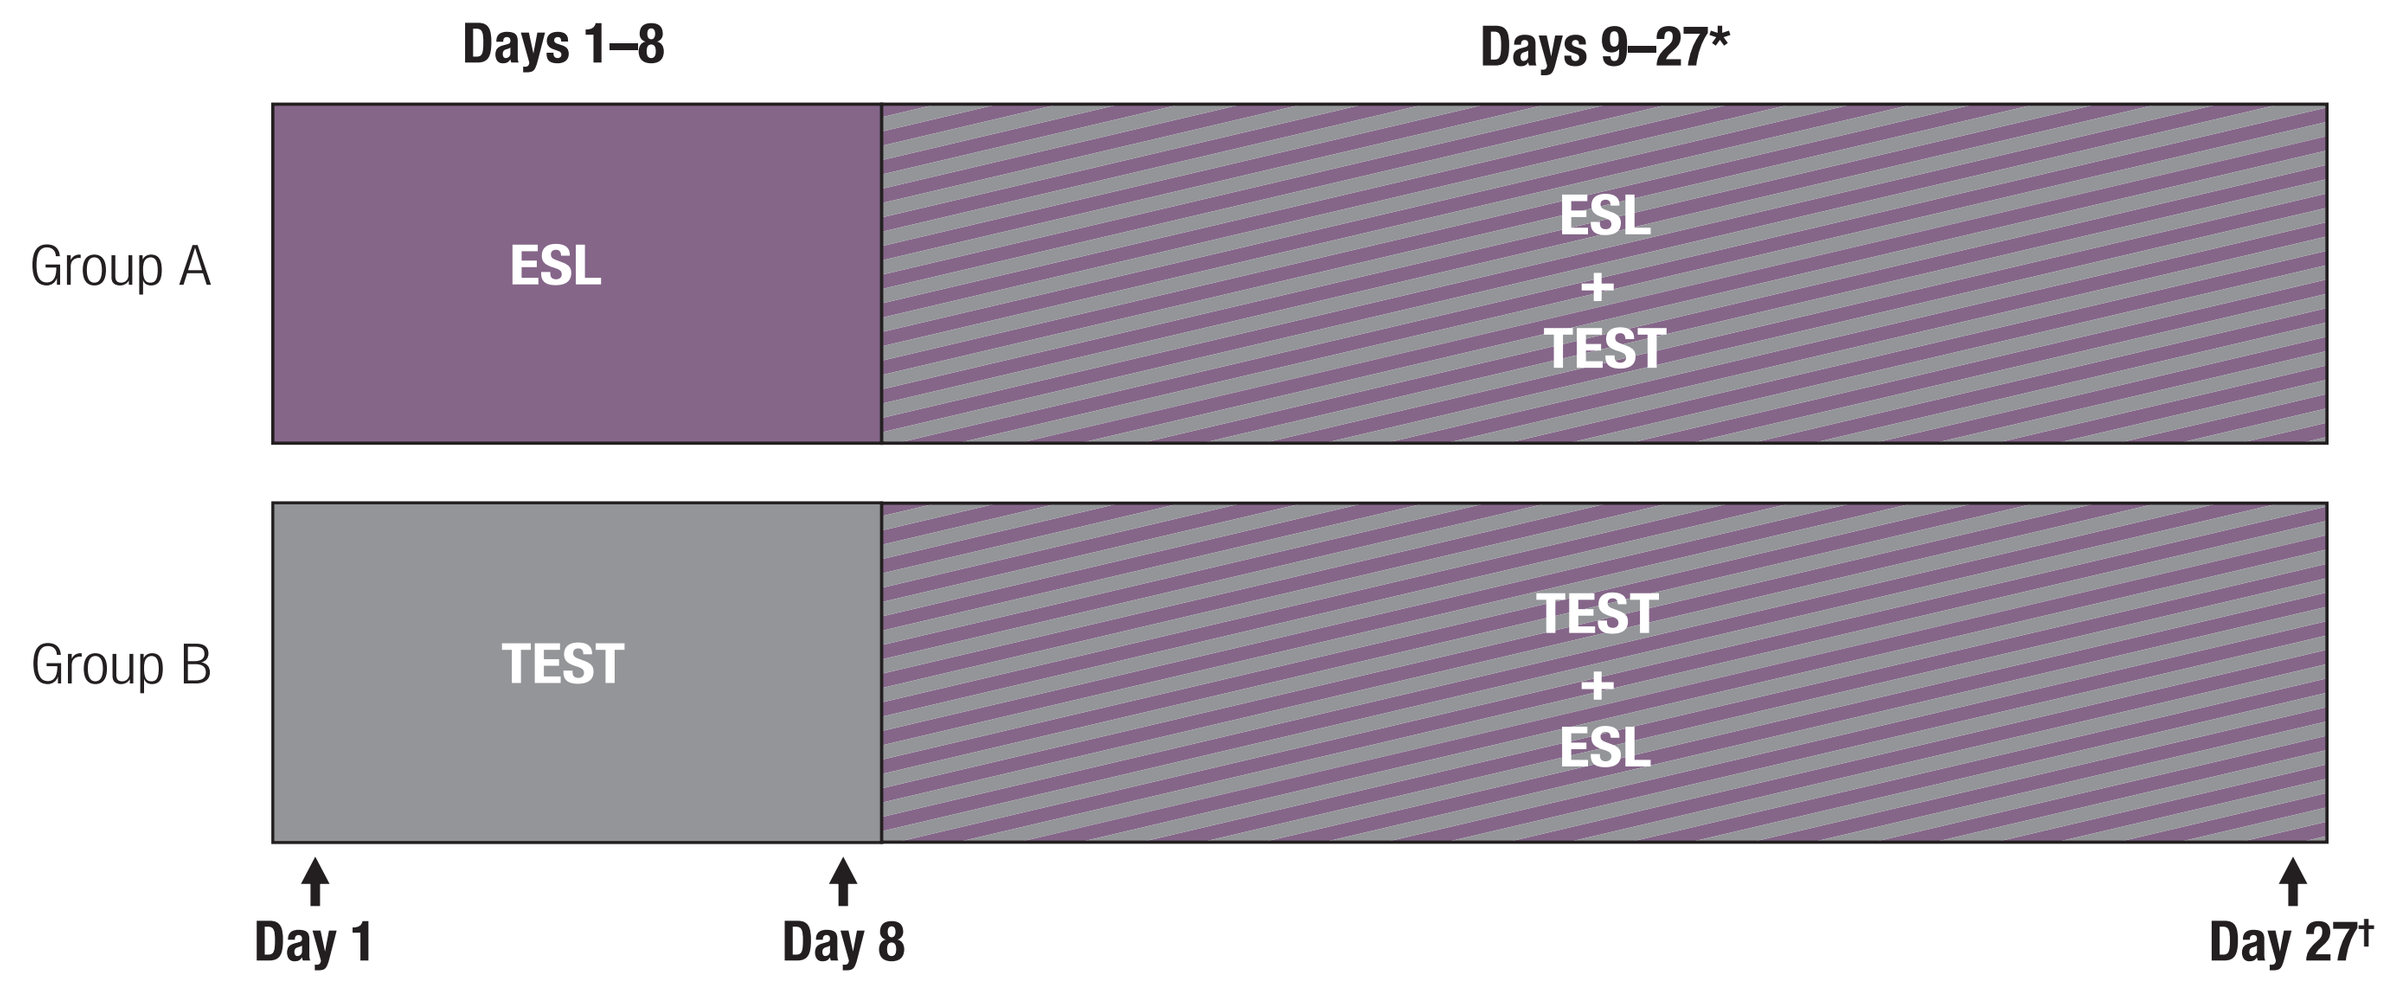
Days 1–8
Days 9–27*
Group A
ESL
ESL + TEST
Group B
TEST
TEST + ESL
Day 1
Day 8
Day 27†

ESL: 600 mg QD for 2 days, then 800 mg QD (initial dose 800 mg in CBZ study)
TEST AEDs: dosing
LTG: 150 mg QD
TPM: 200 mg QD
PHT: 300 mg QD
CBZ: 400 mg BID
↑ Blood sample taken for PK analysis.
*CBZ study: Days 8–35.
†Day 35.
AED: antiepileptic drug; BID: twice-daily; CBZ: carbamazepine; ESL: eslicarbazepine acetate; LTG: lamotrigine; PHT: phenytoin PK: pharmacokinetic; QD: once-daily; TPM: topiramate.

- Interactions were assessed using a random analysis of variance (ANOVA) model.
 - If the 90% confidence interval (CI) for the exponential of the difference in the ln-transformed parameter of the area under the plasma concentration–time curve during 24 hours (AUC_{0-24}) between Day 8 and the last day of treatment, was between 80% and 125% in both Group 1 and Group 2, then it could be concluded that no interaction had occurred.

Population PK models for eslicarbazepine and the other AEDs

- Population PK models were developed for eslicarbazepine, CBZ, VAL, LEV, PB, PHT and GBP, in order to estimate the apparent oral clearance (CL/F) of each drug, and to evaluate the effect of covariates on the variability in CL/F, including (for the other AEDs) the effect of concomitant ESL.
 - Models were not developed for LTG or TPM, because in the Phase I studies, ESL had no clinically meaningful effects on the PK of these AEDs.
 - The models for CBZ, VAL, LEV, PB, PHT, and GBP were developed using data from three Phase III studies of adjunctive ESL in patients with refractory POS (BIA-2093-301, -302, and -304).
 - The design of the three studies is shown in **Figure 2**. In each study, after an 8-week baseline period, eligible patients were randomized to receive adjunctive ESL or placebo for 14 weeks, while continuing to take the AEDs that they had been taking during baseline.
 - Data from the double-blind phases of the studies and the 1-year open-label extension of study -301 were available for analysis.
 - The population PK model for eslicarbazepine was developed using data from the three Phase III studies of adjunctive ESL, plus 11 densely sampled Phase I studies with comparable conditions to the Phase III studies, i.e., multiple doses of ESL 400–1200 mg given QD, with PK sampling in the 24-hour period following dosing (BIA-2093-105, -110, -111, -115, -116, -119, -120, -121, -127, -129, and -150).

Figure 2. Design of the Phase III studies

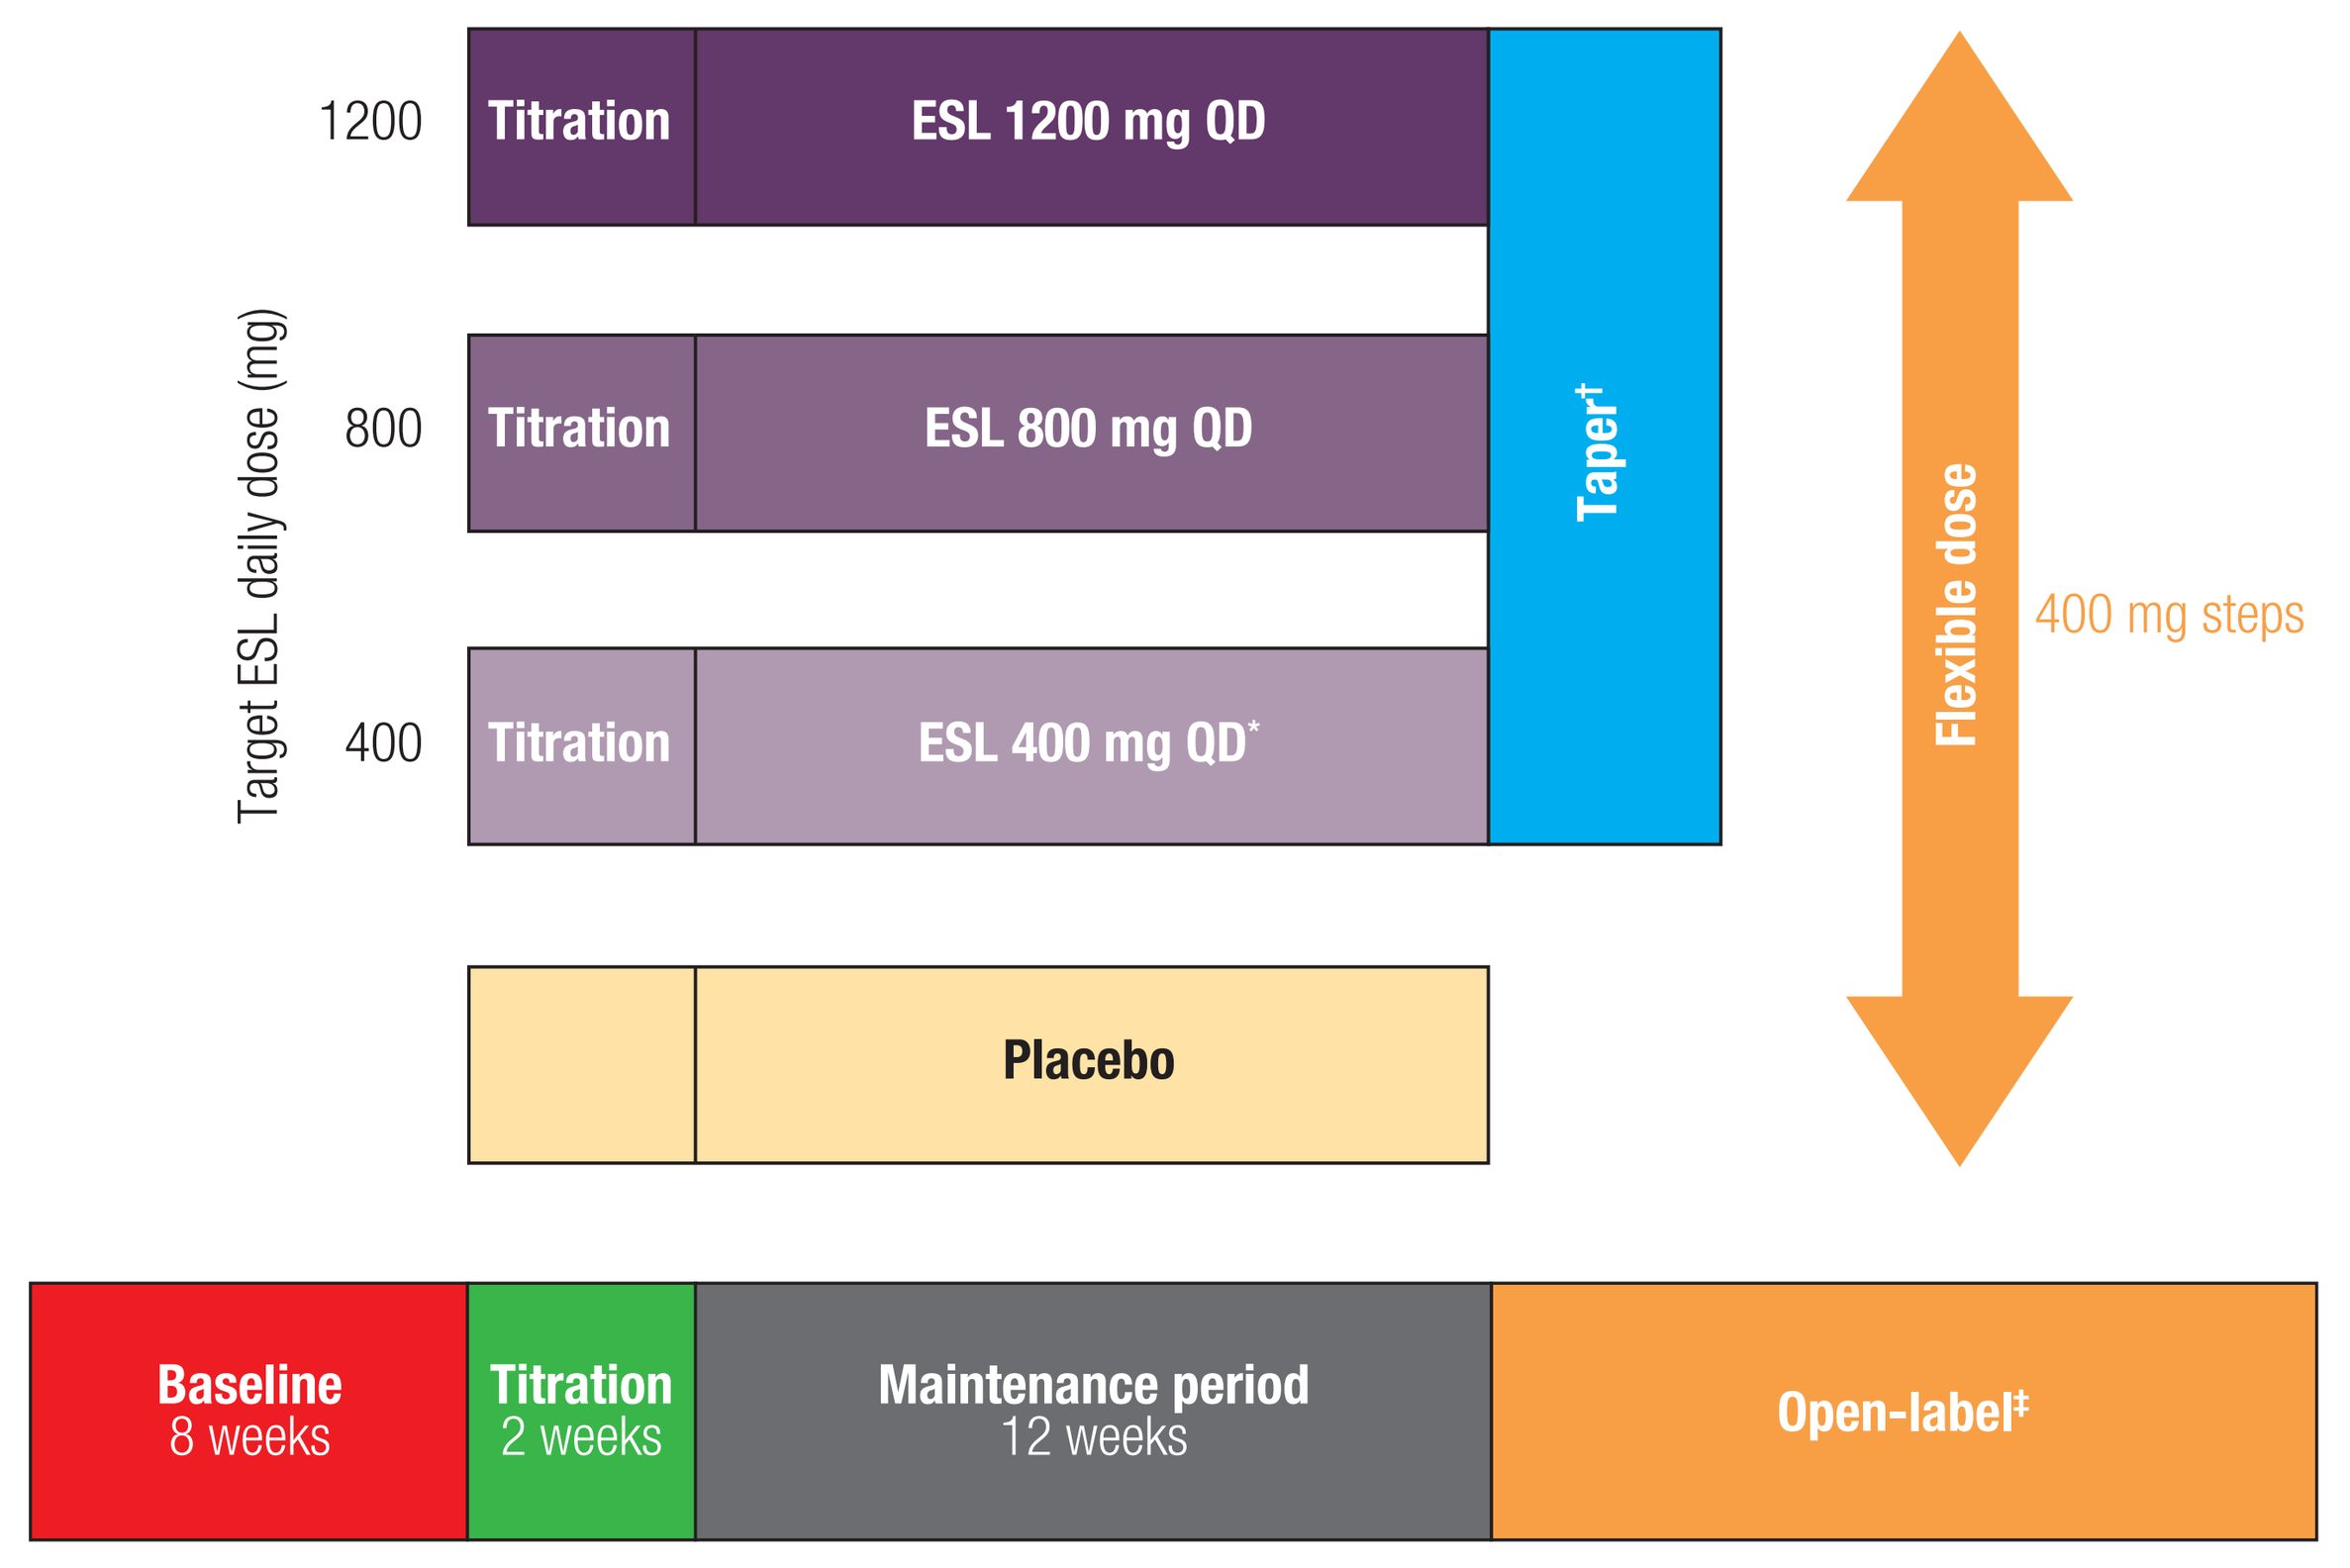
Target ESL daily dose (mg)
1200
800
400
Titration
ESL 1200 mg QD
Titration
ESL 800 mg QD
Titration
ESL 400 mg QD*
Taper†
Placebo
Flexible dose
400 mg steps
Baseline
8 weeks
Titration
2 weeks
Maintenance period
12 weeks
Open-label‡

*Studies 301 and 302 only. †Taper: Study 301: 2-week down-titration; Study 302: abrupt discontinuation; Study 304: ESL 800 mg QD.
‡ESL 800 mg QD for the first month, followed by flexible dosing. ESL: eslicarbazepine acetate; QD: once-daily.

Evaluation of the effect of concomitant AED use on PK of eslicarbazepine and other AEDs

- The population PK models were used to evaluate the effects of concomitant AED use on eslicarbazepine PK, and the effects of concomitant ESL use on the PK of the other AEDs.
 - Structural PK models included estimation of fixed effects (clearance, distribution volume) and random effects (between-patient variability in PK parameters, and within-patient variability in drug concentrations).
 - Covariate analyses were performed to evaluate effects of ESL on exposure of concomitantly used AEDs, and effects of concomitantly used AEDs on eslicarbazepine exposure.
 - Effects were evaluated sequentially (presence/absence) and, if significant, according to drug dose and/or plasma concentration.
 - Covariates were evaluated using forward selection ($\alpha = 0.01$) and backward elimination ($\alpha = 0.001$).

RESULTS

Phase I DDI interaction studies

- Using a bioequivalence approach, analysis of data from the four Phase I DDI studies indicated that concomitant administration of ESL had some effects on measures of exposure for specific AEDs.
 - LTG, TPM, and CBZ exposure was reduced during concomitant administration of ESL, by 14%, 18% and 10% respectively, whereas PHT exposure was increased by 35%.
- In the same subjects, concurrent administration of LTG, TPM, CBZ, and PHT reduced eslicarbazepine exposure by 4%, 7%, 31.9%, and 31–33%, respectively.

Population PK analyses: impact of concomitant administration on PK parameters for eslicarbazepine and other AEDs

- The analysis populations (adult patients with evaluable PK data) were as follows: eslicarbazepine, n = 1039; CBZ, n = 628; VAL, n = 262; LEV, n = 232; PB, n = 115; PHT, n = 106; GBP, n = 29.

Eslicarbazepine

- Eslicarbazepine PK during QD oral dosing with ESL was described by a one-compartment model with first-order absorption and linear elimination.
 - In the absence of concomitant AEDs, population mean PK parameter estimates for eslicarbazepine were: apparent clearance (CL/F) = 2.43 L/h; volume of distribution (V/F) = 61.3 L.
 - Eslicarbazepine exposure (AUC at steady-state; AUC_{ss}) was 33.8% lower when ESL was used together with concomitant enzyme-inducing AEDs (EIAEDs, i.e., PB, PHT and primidone [PD]) compared with eslicarbazepine exposure when ESL was used alone.
 - Concomitant administration of CBZ led to a reduction in eslicarbazepine AUC_{ss} of 25.1–46.8% versus ESL used alone (**Figure 3**). CBZ 400 mg twice daily led to a median reduction in eslicarbazepine AUC_{ss} of 30.8%.
 - Concomitant use of VAL, LTG, and LEV had no appreciable impact on eslicarbazepine PK parameters.
- **Figure 3** summarizes the predicted effects of concomitant AED use on eslicarbazepine exposure (based on the results of the Phase I DDI studies and the population PK analyses).

Figure 3. Potential effects of concomitant AED use on eslicarbazepine exposure*

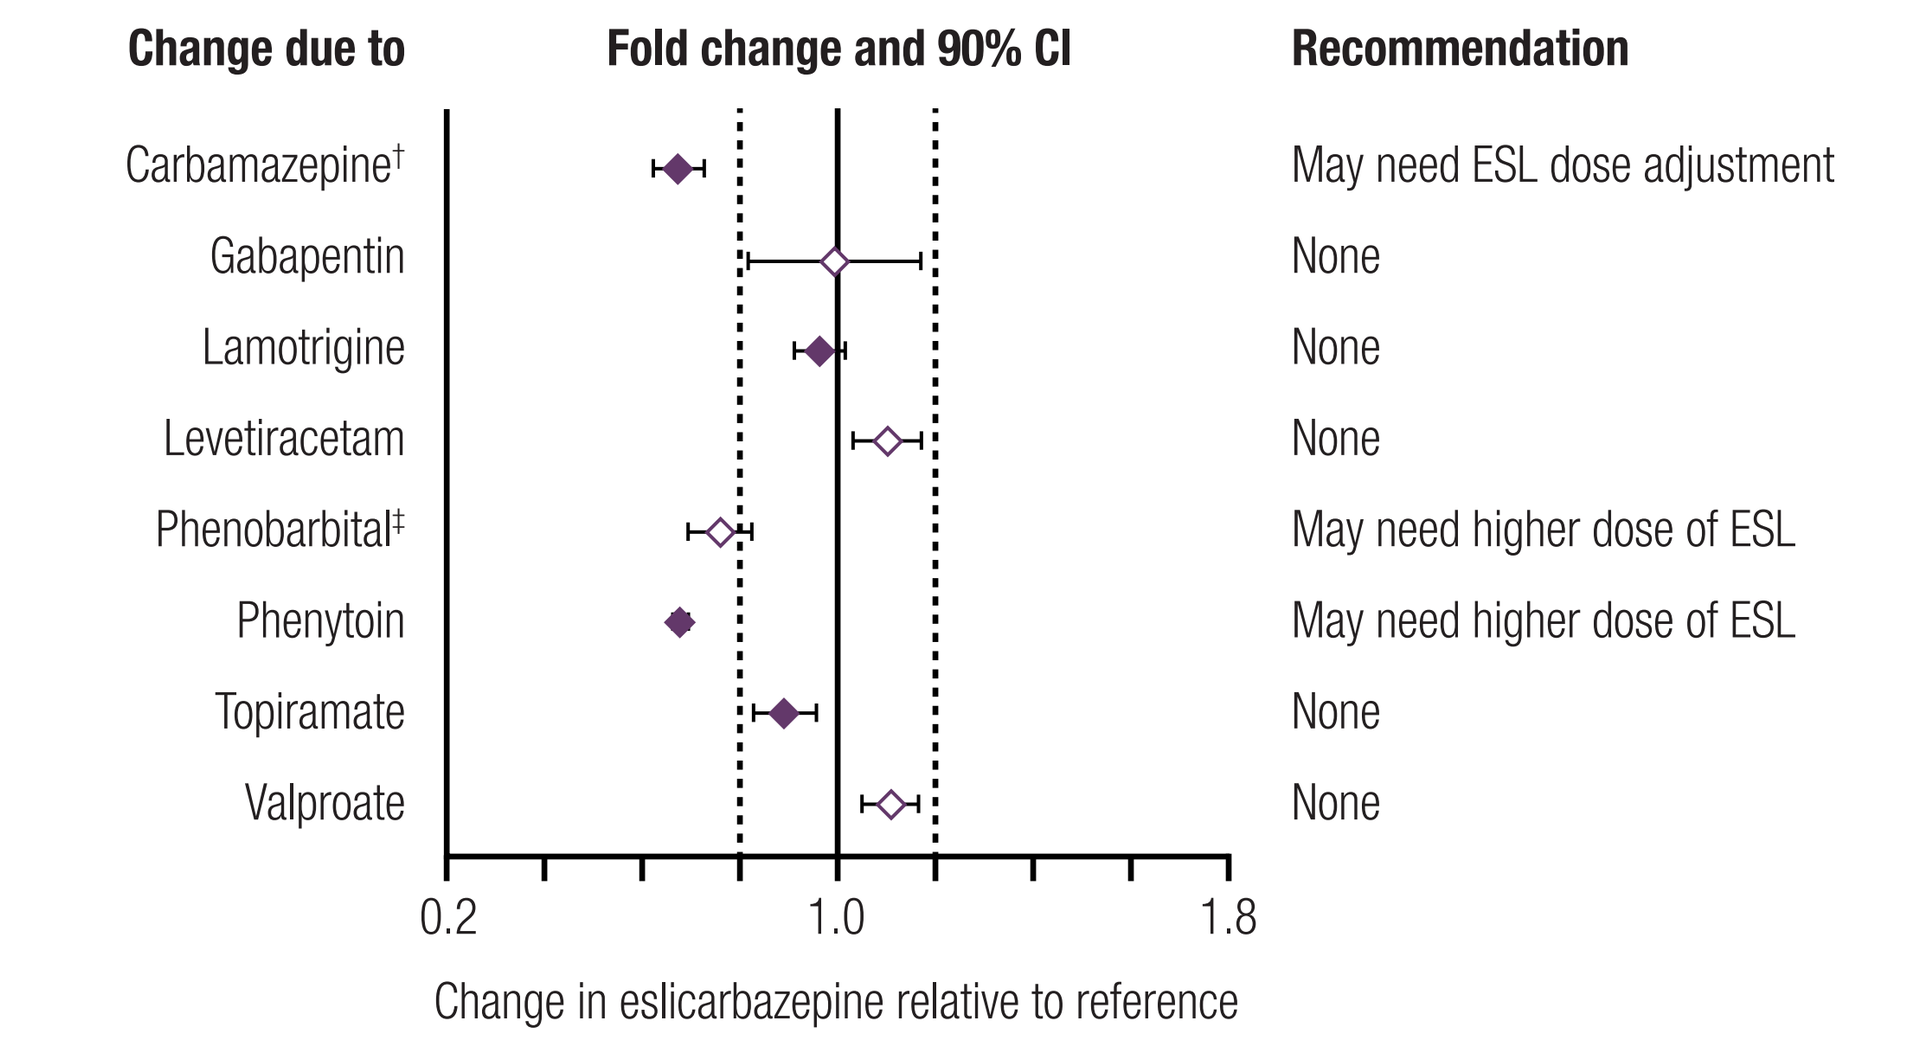
Change due to
Fold change and 90% CI
Recommendation
Carbamazepine†
May need ESL dose adjustment
Gabapentin
None
Lamotrigine
None
Levetiracetam
None
Phenobarbital‡
May need higher dose of ESL
Phenytoin
May need higher dose of ESL
Topiramate
None
Valproate
None
0.2
1.0
1.8
Change in eslicarbazepine relative to reference

*Based on AUC.
†Based on population PK data from both the Phase III and Phase I studies.
‡Phenobarbital and/or phenobarbital-like AEDs (e.g., PD).
Open markers: population PK data from Phase III study; solid markers: Phase I study.
AEDs: antiepileptic drugs; AUC: area under the plasma concentration–time curve; CI: confidence interval
ESL: eslicarbazepine acetate; PD: primidone; PK: pharmacokinetic.

Other AEDs

- Population PK models for the other AEDs identified the following covariates as statistically significant predictors of CL/F for each AED.
 - CBZ: CBZ dose; total bilirubin; ESL dose.
 - VAL: VAL dose; CBZ dose (no effect of ESL).
 - LEV: creatinine clearance (CrCL); CBZ dose; LEV dose (no effect of ESL).
 - PB: PB dose (no effect of ESL).
 - PHT: no covariates were statistically significant predictors of CL/F.
 - GBP: CrCL (no effect of ESL).
- Based on the covariate analysis of the effect of ESL on concurrent AEDs, the potential interactions between ESL and AEDs are shown in **Figure 4**.

Figure 4. Potential effects of concomitant ESL use on AED exposure*

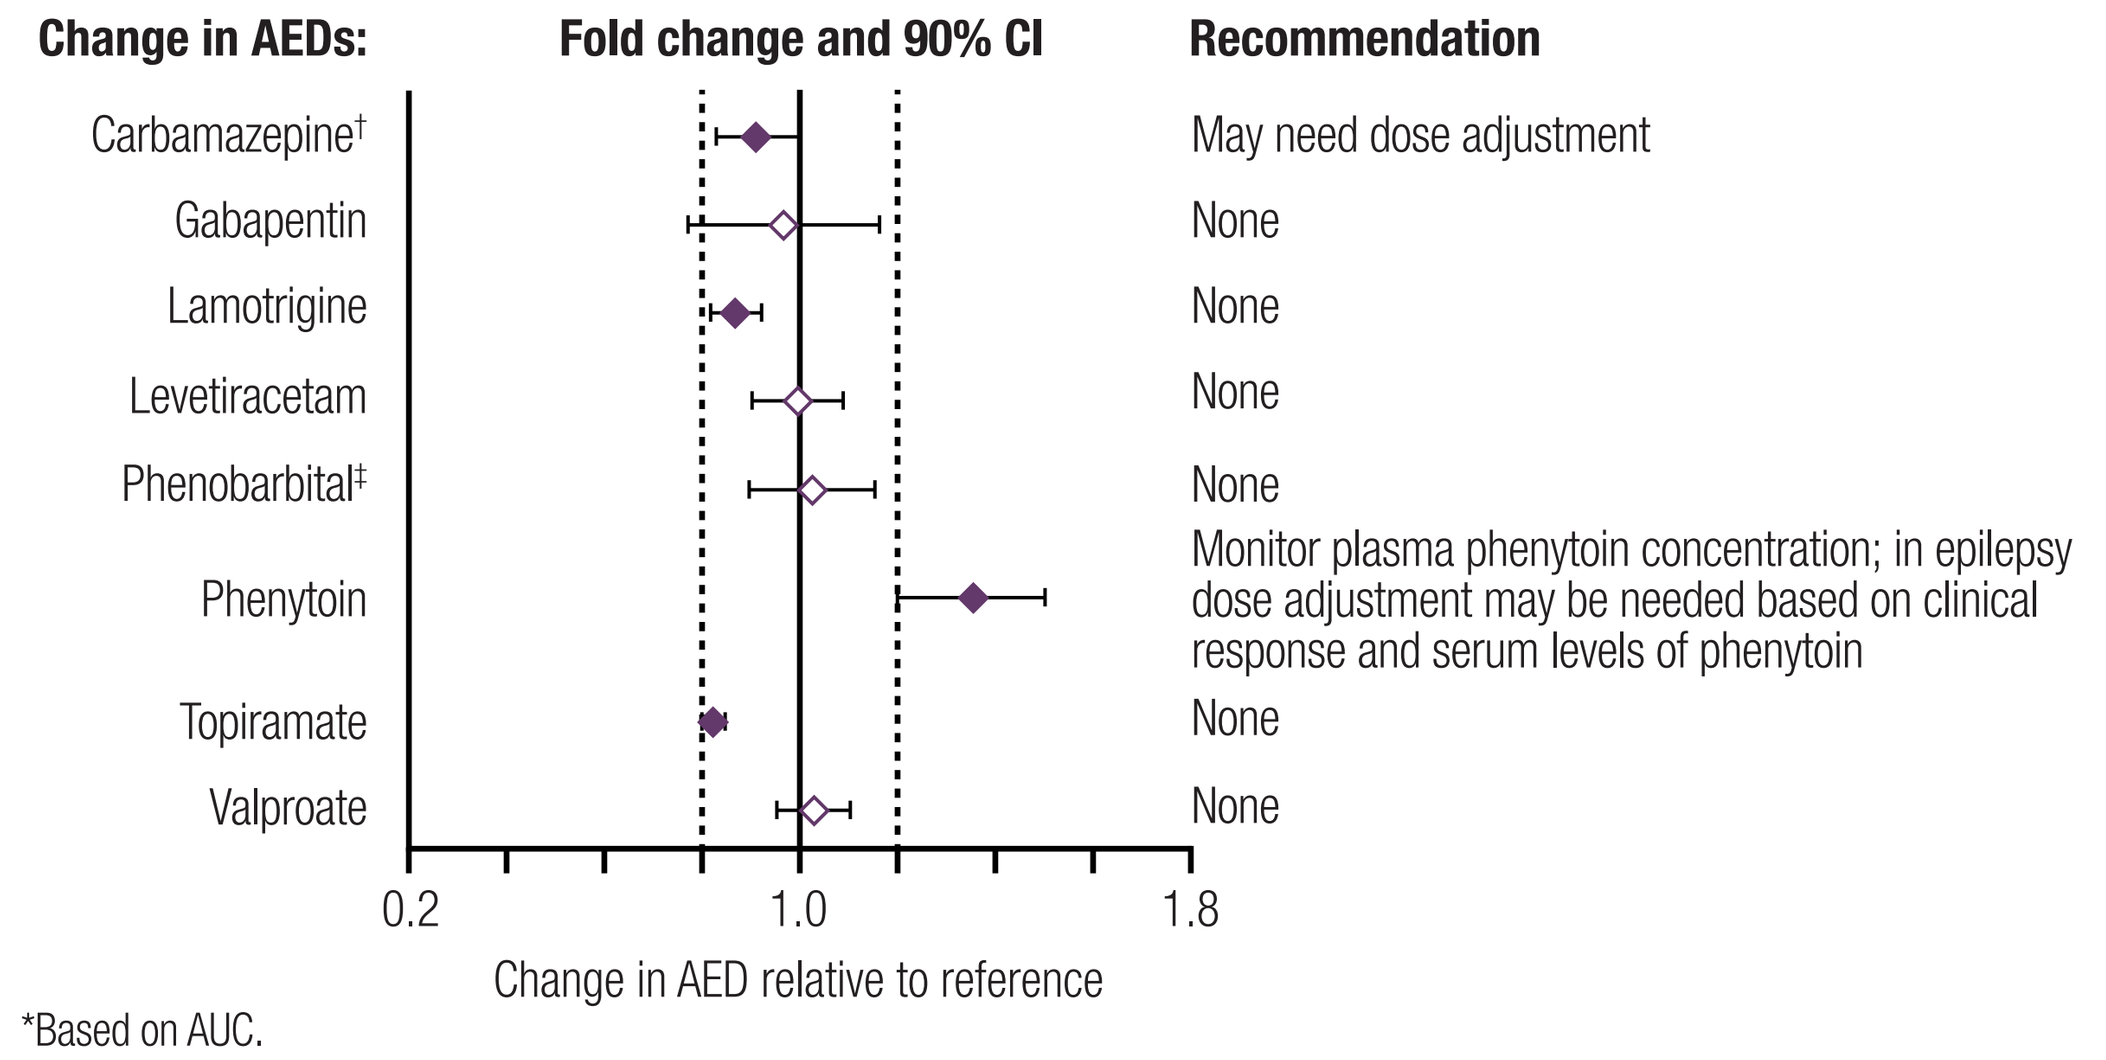
Change in AEDs:
Fold change and 90% CI
Recommendation
Carbamazepine†
May need dose adjustment
Gabapentin
None
Lamotrigine
None
Levetiracetam
None
Phenobarbital‡
None
Phenytoin
Monitor plasma phenytoin concentration; in epilepsy dose adjustment may be needed based on clinical response and serum levels of phenytoin
Topiramate
None
Valproate
None
0.2
1.0
1.8
Change in AED relative to reference

*Based on AUC.
†Based on both the population PK from the Phase III studies, and the Phase I study.
‡Phenobarbital and/or phenobarbital-like AEDs (e.g., PD).
Open markers: population PK from the Phase III studies; solid markers: Phase I study.
AEDs: antiepileptic drugs; AUC: area under the plasma concentration–time curve; CI: confidence interval; ESL: eslicarbazepine acetate
PD: primidone; PK: pharmacokinetic.

- Use of concomitant ESL had a notable impact on CBZ PK parameters.
 - In patients with a total bilirubin concentration of 0.29 mg/dL (the population median) taking 1000 mg CBZ daily, co-administration of ESL 400, 800 and 1200 mg QD is predicted to lead to reductions in CBZ exposure (C_{av-ss}) of 3.5%, 7.0% and 10.0% respectively. However, these reductions in exposure are not likely to be clinically relevant.
- Co-administration of ESL had no effect on exposure to VAL, LEV, PB, PHT or GBP (**Figure 4**).
 - The lack of effect on PHT exposure contrasts with the results of the Phase I study 2093-121, in which co-administration of ESL was found to increase PHT exposure (**Figure 4**).
- **Figure 4** summarizes the predicted effects of concomitant ESL use on exposure for the other AEDs (based on the results of the Phase I DDI studies and the population PK analyses).

CONCLUSIONS

- Concomitant administration of CBZ and EIAEDs (PB, PHT and PD) with ESL led to potentially clinically meaningful reductions in eslicarbazepine exposure.
 - Higher doses of ESL may be required when EIAEDs are co-administered.
 - ESL dose adjustment may be warranted during concomitant use of CBZ, based on efficacy and tolerability.
- Co-administration of ESL with CBZ led to a reduction in CBZ exposure, which may require dose adjustment for CBZ.
- Altered PHT exposure during concomitant ESL administration suggests that monitoring plasma PHT concentrations may be warranted; dose adjustments for PHT may be needed, based on clinical response and serum phenytoin levels.
- Based on the Phase I studies and population PK analyses, concomitant administration of ESL had no effect on PK parameters for LEV, GBP, LTG, TPM, or VAL, and therefore, no dose adjustments are required when using ESL with these AEDs.

REFERENCES

1. Almeida L, et al. J Clin Pharmacol 2005;45:1062–6.
2. Bialer M, Soares-da-Silva P. Epilepsia 2012;53:935–46.
3. Aptiom® (eslicarbazepine acetate) Prescribing Information. Sunovion Pharmaceuticals Inc., 2013. Available at: http://www.aptiom.com/Aptiom-Prescribing-Information.pdf. Accessed September 5, 2015.
4. Maia J, et al. Int J Clin Pharmacol Ther 2008;46:119–30.

DISCLOSURES

These data were previously presented in part at the American Academy of Neurology Annual Meeting, 2011, Honolulu, Hawaii (Phase III data from two studies) and at the 67th Annual American Epilepsy Society Meeting, 2013, Washington, DC, USA.

ACKNOWLEDGMENTS

Medical writing support was funded by Sunovion Pharmaceuticals Inc., and was provided by Michael Simpson of FireKite, an Ashfield company, part of UDG Healthcare plc.

6th American Conference on Pharmacometrics, October 4–7, 2015, Crystal City, VA, USA

57380F ACoP Sunkaraneni Poster 42x66inches_v1e.indd 1
17/09/2015 11:46