P55

The steady-state pharmacokinetic profile across a range of patient body weight categories supports weight-based dosing for intravenous reslizumab

D. Jaworowicz[1], J. Fiedler-Kelly[1], L. Rabinovich-Guilatt[2] and M. Bond[2]

[1]Cognigen Corporation, a *SimulationsPlus* Company, Buffalo, NY, USA; [2]Teva Pharmaceutical Industries Ltd, Frazer, PA, USA

BACKGROUND AND AIM

- Reslizumab is an IgG4 kappa humanized monoclonal antibody that targets interleukin-5, leading to marked reductions in airway eosinophil levels, and is indicated as an 'add-on' maintenance treatment for adult patients (≥18 years of age) with severe asthma and eosinophilic phenotype.[1–3]
- The efficacy and safety of intravenous (IV) reslizumab (3.0mg/kg) in patients with severe asthma and elevated eosinophil levels has been shown in four large, placebo-controlled, phase 3 clinical trials (BREATH clinical program).[4–6]
- The dosage of intravenously administered reslizumab in these studies was based on patient body weight (weight-based dosing).[3]
- Our aim was to develop a population pharmacokinetic (PPK) model to characterize the disposition of reslizumab and to evaluate the influence of key covariates, including body weight, on steady-state reslizumab exposures in support of the recently US Food and Drug Administration-approved IV dosing regimen of reslizumab.

METHODS

- Data were pooled from eight reslizumab clinical trials (**Table 1**), which included healthy volunteers (n=130) or patients with asthma or nasal polyps (n=674).

TABLE 1. STUDY DESIGN CHARACTERISTICS

Phase (study number)	Dosing regimen (number of participants)	Medical condition	Median number (range) of PPK samples per participant
Phase 1 (I96-350)	Single dose: 0.03mg/kg (n=2) or 0.1mg/kg (n=4) IV bolus 0.3mg/kg (n=6) or 1.0mg/kg (n=12) IV infusion	Severe asthma	17.5 (14–20)
Phase 1 (P01942)	Single dose: 1.0mg/kg (n=8) or 3.0mg/kg (n=8) IV infusion	Recurrent nasal polyps after surgery or grades 3–4 nasal polyps in both nares	20 (16–22)
Phase 1 (C38072/1102)	5 doses, 28 (±2) days apart: 0.3mg/kg (n=18), 1.0mg/kg (n=20), 2.0mg/kg (n=20), or 3.0mg/kg (n=42) IV infusion	Healthy (Japanese and non-Japanese)	35 (7–35)
Phase 1 (C38072/1107)	Single dose: 220mg IV 20-minute infusion (n=30)	Healthy (Japanese and non-Japanese)	16 (9–16)
Phase 2 (P00290)	2 doses, 12 weeks apart: 1.0mg/kg (n=71) or 0.3mg/kg (n=74) IV infusion	Moderate to severe asthma maintained but not adequately controlled on inhaled corticosteroids	15 (2–18)
Phase 2 (Res-5-0010)	4 doses, 4 weeks (±7 days) apart: 3.0mg/kg IV infusion (n=51)	Poorly controlled asthma with eosinophilic airway inflammation	2 (1–2)
Phase 3 (3081)	4 doses, 4 weeks apart: 0.3mg/kg (n=102) or 3.0mg/kg (n=100) IV infusion	Eosinophilic asthma	8 (1–10)
Phase 3 (3082)	13 doses, 4 weeks (±7 days) apart: 3.0mg/kg (n=236) IV infusion	Eosinophilic asthma	10 (1–15)

IV: intravenous; PPK: population pharmacokinetic.

- A reslizumab PPK model was developed using nonlinear mixed effects modeling (NONMEM) based on 10,314 observed reslizumab serum concentrations collected in 804 subjects after single or multiple (ranging from a total of 2–13 doses taken 4 weeks or 12 weeks apart) IV administrations ranging from 0.03 to 3.0mg/kg.
- Covariate analysis to identify statistically significant predictors of PPK variability used a univariate forward selection – backward elimination procedure.
- The demographic and clinical covariates evaluated for their potential in explaining PPK variability were: age, body weight, body mass index, gender, race, renal function (based on Modification of Diet in Renal Disease estimated glomerular filtration rate [MDRD eGFR]), liver function (including total bilirubin, alanine aminotransferase (ALT), aspartate aminotransferase (AST), and the National Cancer Institute (NCI) Liver Dysfunction Group Classification), anti-drug antibody status, and concomitant medications (prednisone, montelukast, and classes for injectable corticosteroids or leukotriene antagonists).
- The clinical significance of the effects of baseline body weight, and other covariates of interest, on the PPK of reslizumab following 3.0mg/kg IV administration was assessed via simulations. Assuming a standard 4-week dosing interval, individual measures of reslizumab exposure at steady-state (area under the reslizumab serum concentration versus time curve [$AUC_{ss(0–4wk)}$], the average reslizumab serum concentration [$C_{av,ss(0–4wk)}$], the maximum reslizumab serum concentration [$C_{max,ss(0–4wk)}$], and the minimum reslizumab serum concentration [$C_{min,ss(0–4wk)}$]) were simulated using individual empiric Bayesian PK parameter estimates obtained from the PPK model in conjunction with each individual's covariate distribution.

RESULTS

PPK model

- The demographic and clinical characteristics of the PPK analysis population are summarized in **Table 2**.

TABLE 2. DEMOGRAPHIC AND CLINICAL CHARACTERISTICS OF THE PPK ANALYSIS POPULATION

Subject characteristic	Summary statistics	
Continuous covariates		
Age (years)	Mean (SD)	43 (14)
	Median [Min–Max]	44 [12–77]
Weight (kg)	Mean (SD)	75.8 (17.9)
	Median [Min–Max]	73.0 [33.9–156.0]
Body mass index (kg/m²)	Mean (SD)	27.1 (6.1)
	Median [Min–Max]	26.2 [15.3–53.7]
MDRD eGFR (mL/min/1.73m²)	Mean (SD)	85.73 (21.77)
	Median [Min–Max]	83.63 [27.1–227.2]
Categorical covariates		
Gender, n (%)	Male	373 (46.4)
	Female	431 (53.6)
Race, n (%)	White	626 (77.9)
	Black	37 (4.6)
	Asian	56 (7.0)
	Japanese	50 (6.2)
	Other	35 (4.4)
Classification of renal function (MDRD eGFR), n (%)	Normal or high	294 (36.6)
	Mildly decreased	446 (55.5)
	Mildly to moderately decreased	60 (7.5)
	Moderately to severely decreased	3 (0.4)
	Severely decreased	1 (0.1)
Total bilirubin category, n (%)	Normal	785 (97.6)
	Grade 1	16 (2.0)
	Grade 2	3 (0.4)
AST category, n (%)	Normal	784 (97.5)
	Grade 1	20 (2.5)
ALT category, n (%)	Normal	757 (94.2)
	Grade 1	47 (5.8)
NCI Liver Dysfunction Group Classification, n (%)	Normal	766 (95.3)
	Mild	35 (4.4)
	Moderate	3 (0.4)
Anti-drug antibody status, n (%)	Negative	725 (90.2)
	Positive	42 (5.2)
	Borderline	13 (1.6)
	Missing	24 (3.0)
Concomitant medications (time-varying)		
Corticosteroids, n (%)	Never	661 (82.2)
	Ever	143 (17.8)
Leukotriene antagonists, n (%)	Never	671 (83.5)
	Ever	133 (16.5)
Prednisone, n (%)	Never	714 (88.8)
	Ever	90 (11.2)
Montelukast, n (%)	Never	678 (84.3)
	Ever	126 (15.7)

ALT: alanine aminotransferase; AST: aspartate aminotransferase; eGFR: estimated glomerular filtration rate; MDRD: Modification of Diet in Renal Disease; NCI: National Cancer Institute; SD: standard deviation.

- The reslizumab serum concentration-time course following IV administration was accurately described by a two-compartment PK model with zero order input and first-order elimination (**Table 3**).
- Inter-individual variability (IIV) was estimated for clearance (CL), central volume of distribution (V_c), distributional clearance (Q), and peripheral volume of distribution (V_p) using an exponential error model. The residual variability (RV) was estimated using separate log error models applied to full-profile PK data, phase 2 sparse data, and phase 3 sparse data.
- Final parameter estimates for the final PPK model are presented in **Table 3**. All parameters were estimated with good precision (≤9.28% standard error of mean [SEM] for fixed effects and ≤39.6% SEM for random effects).
- Diagnostic delta plots (individual Bayesian parameter estimate minus the population typical value of the parameter) of CL and V_c versus body weight (**Figure 1**) support a strong dependence of both PK parameters on body weight.

TABLE 3. PARAMETER ESTIMATES AND STANDARD ERRORS FROM THE RESLIZUMAB FINAL PPK MODEL

Parameter	Final parameter estimate		IIV/RV	
	Typical value	%SEM	Magnitude	%SEM
CL, mL/h	7.16	1.36	33.3%CV	6.54
Power for body weight on CL	0.561	8.15		
V_c, mL	3130	1.18	26.0%CV	16.0
Power for body weight on V_c	0.606	9.28		
Q, mL/h	10.0	7.51	97.2 %CV	13.2
V_p, mL	2050	3.35	54.8 %CV	10.1
Covariance (IIV in CL, IIV in V_c)	0.0410	14.8	NA	NA
Covariance (IIV in V_p, IIV in V_c)	0.0332	22.4		
Covariance (IIV in V_p, IIV in CL)	0.129	8.83		
Covariance (IIV in Q, IIV in CL)	0.138	16.9		
Covariance (IIV in Q, IIV in V_p)	0.382	14.6		
RV (log scale, full-profile)	0.0398	5.70	0.199 SD	NA
RV (log scale, phase 2 sparse)	0.337	39.6	0.581 SD	
RV (log scale, phase 3)	0.107	9.15	0.327 SD	
Minimum value of the objective function = –14614.946				

The calculated correlation coefficients (r^2) of the off-diagonal omegas were as follows: 0.225 for covariance (IIV in CL, IIV in V_c), 0.0545 for covariance (IIV in V_p, IIV in V_c), 0.500 for covariance (IIV in V_p, IIV in CL), 0.182 for covariance (IIV in Q, IIV in CL), 0.515 for covariance (IIV in Q, IIV in V_p).
CL: clearance; %CV: coefficient of variation expressed as a percentage; IIV: inter-individual variability; NA: not applicable; PPK: population pharmacokinetic; Q: distributional clearance; RV: residual variability; SD: standard deviation; %SEM: standard error of the mean expressed as a percentage; V_c: central volume of distribution; V_p: peripheral volume of distribution.

FIGURE 1. DELTA PLOTS OF CL AND V_C VERSUS BODY WEIGHT IN THE BASE PPK MODEL PRIOR TO INCORPORATING THE EFFECTS OF BODY WEIGHT ON THESE PK PARAMETERS

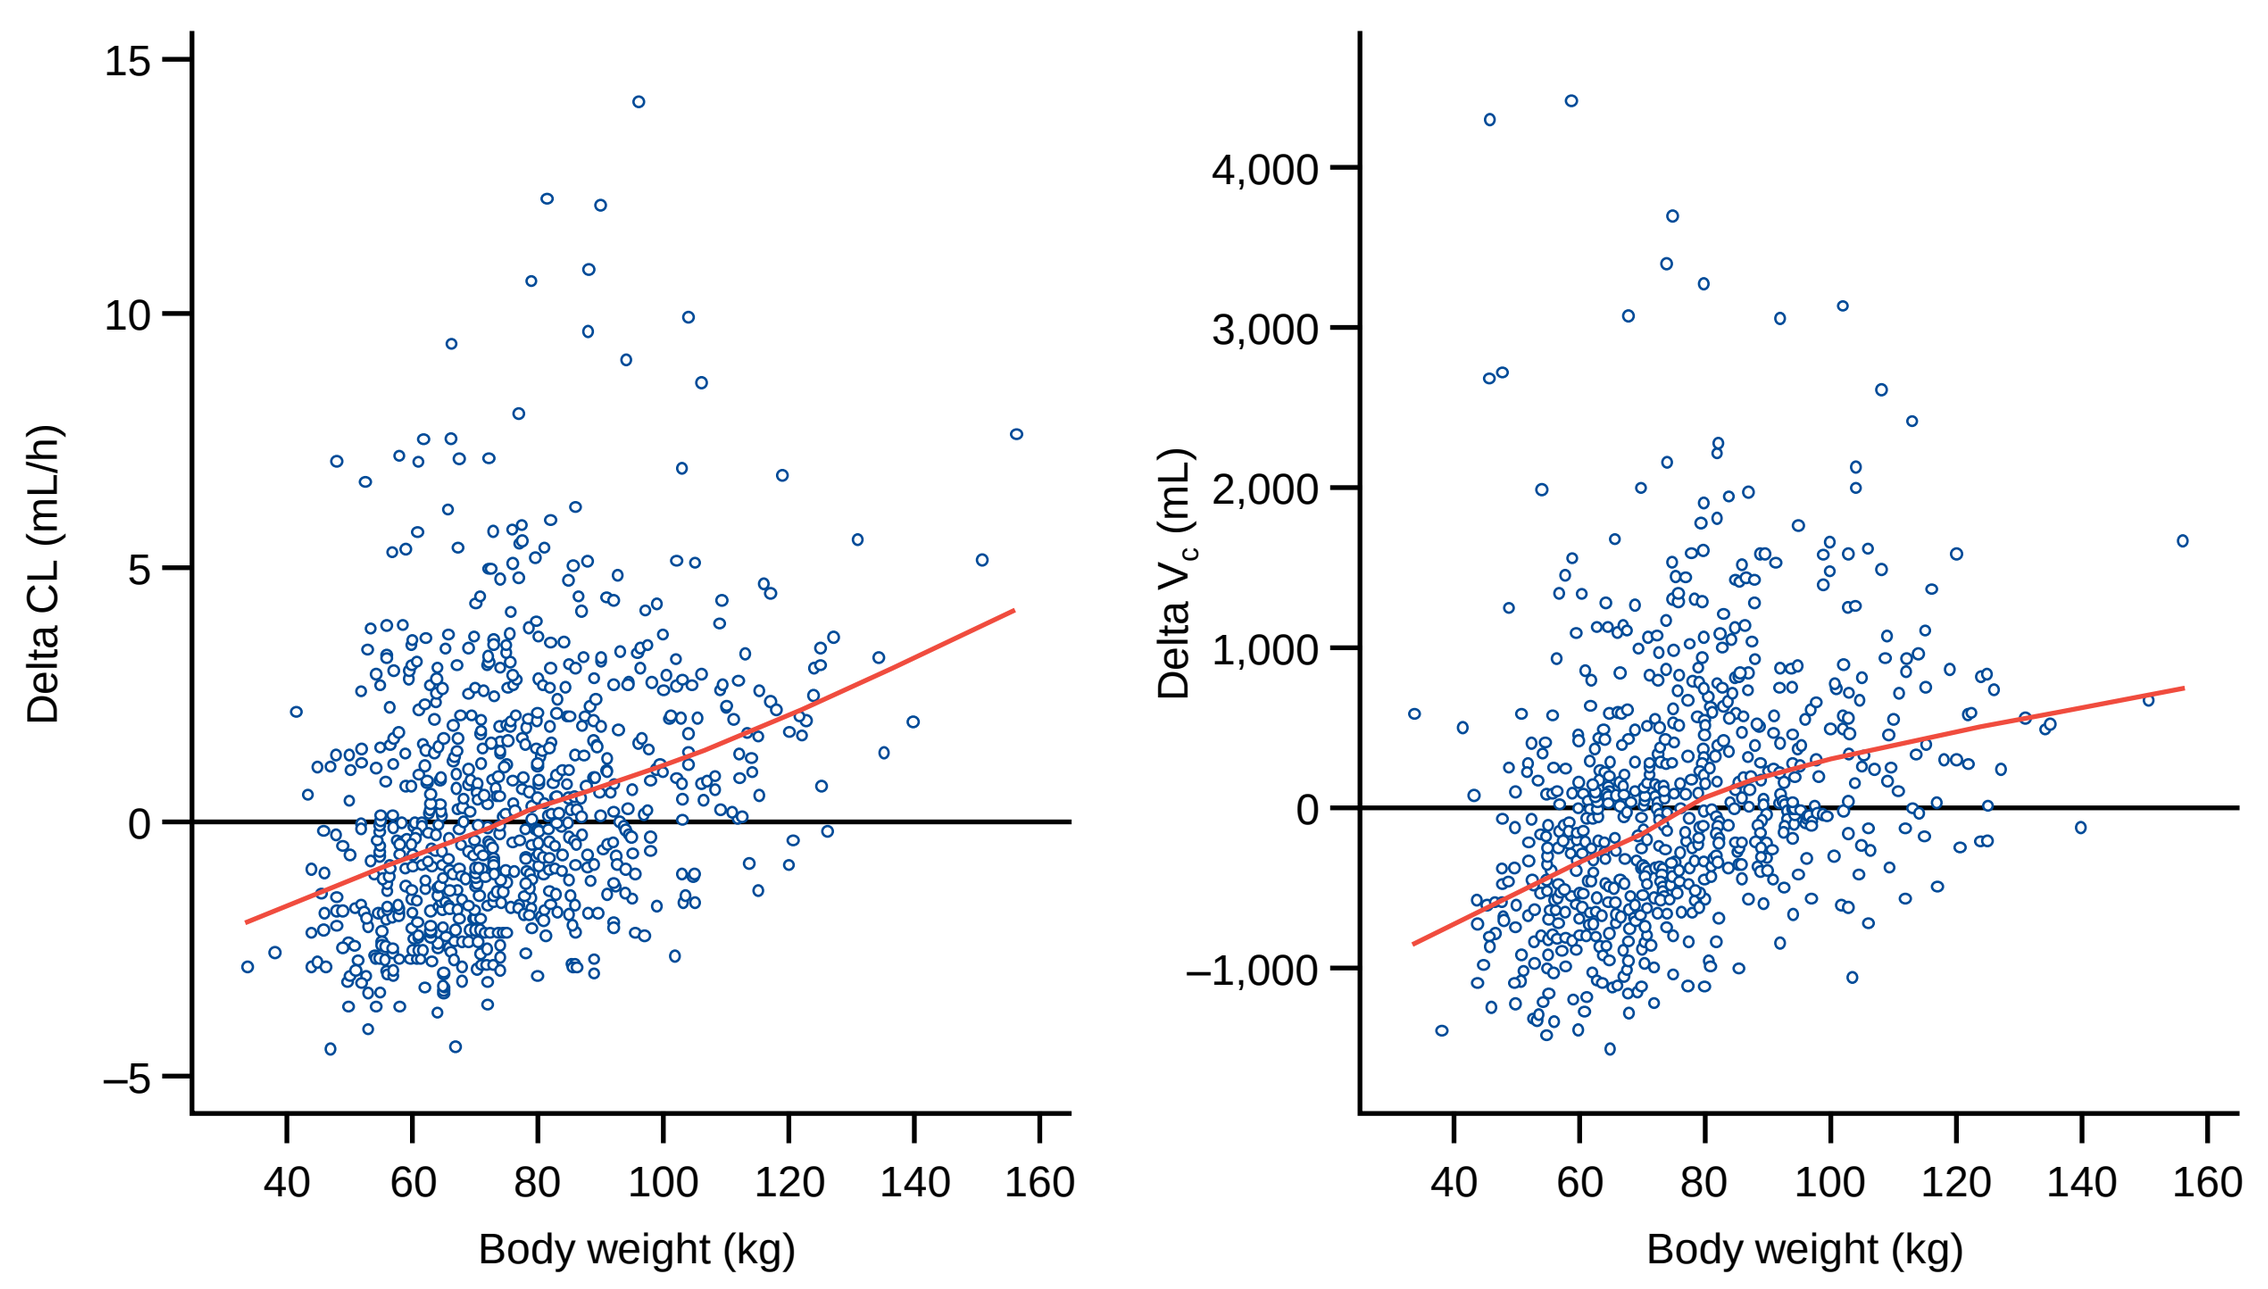

Delta CL (mL/h)
Body weight (kg)
Delta V_c (mL)
Body weight (kg)

Delta parameter values are calculated as the subject-specific Bayesian PK parameter estimate minus the typical value of that PK parameter. CL: clearance; PK: pharmacokinetic; V_c: central volume of distribution.

- Covariate analysis identified body weight as a statistically significant (p<0.001) predictor of both CL and V_c (**Table 3**), with typical CL and V_c parameter values predicted to increase less than proportionally with increasing body weight, according to the power function equations provided below.

$$\tilde{CL}_i = 7.16 \times \left(\frac{WTKG_i}{73}\right)^{0.561}$$

$$\tilde{V}_{ci} = 3130 \times \left(\frac{WTKG_i}{73}\right)^{0.606}$$

Where:
$\tilde{CL}_i$ is the typical value of CL (mL/h) in the i^{th} subject;
$\tilde{V}_{ci}$ is the typical value of central volume of distribution (mL) in the i^{th} subject; and
$WTKG_i$ is the body weight (kg) in the i^{th} subject.

- The effect of body weight on CL and V_c was independent of other measured covariates. No other covariates, including ADAs or concomitant medications, were found to be significant descriptors of variability in reslizumab PK.
- Model diagnostics (including standard goodness-of-fit plots and visual predictive checks) demonstrated that the model adequately characterized the PK data across the analysis population.
- **Figure 2** displays plots of the typical model predicted concentration-time curves for hypothetical patients with body weights representing the 5th, 50th, and 95th percentiles of the observed weight distribution in the analysis population overlaid on the observed data for 3.0mg/kg reslizumab following multiple dosing, demonstrating how the model accurately captures the central tendency and shape of the PK profiles.

FIGURE 2. TYPICAL VALUE MODEL PREDICTED PROFILES BY PERCENTILES OF BODY WEIGHT OVERLAID ON OBSERVED RESLIZUMAB CONCENTRATIONS VERSUS TIME SINCE PREVIOUS DOSE FOLLOWING MULTIPLE DOSING

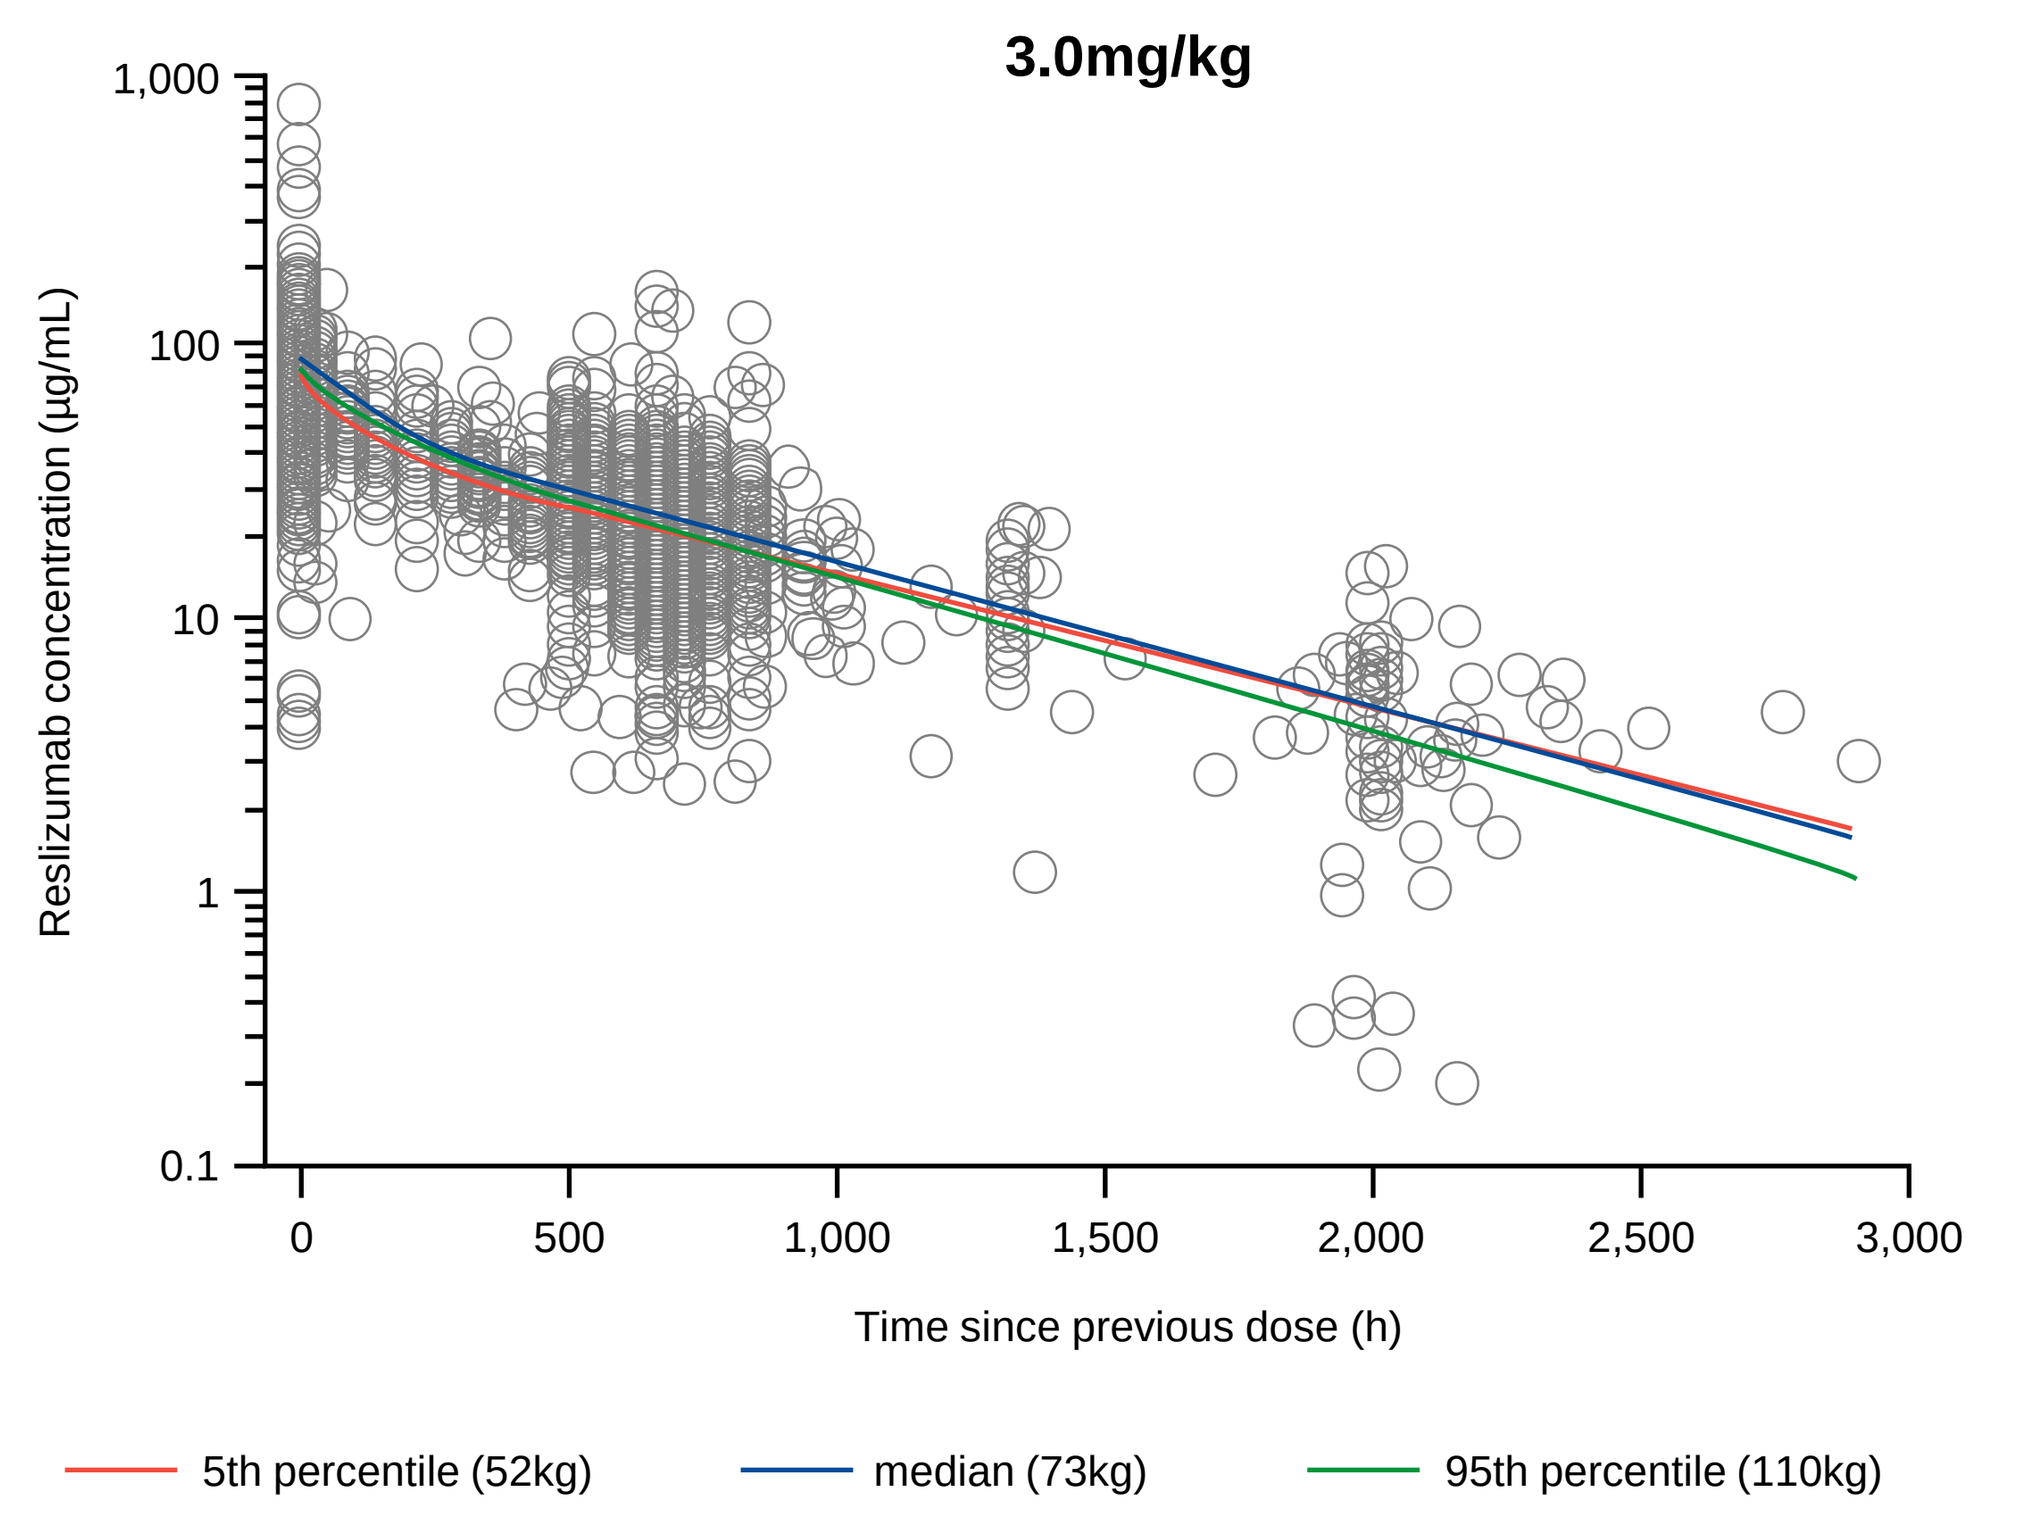

3.0mg/kg
Reslizumab concentration (µg/mL)
Time since previous dose (h)
5th percentile (52kg)
median (73kg)
95th percentile (110kg)

Steady-state exposure assessments

- **Figure 3** depicts the simulated reslizumab steady-state exposures normalized for 3.0mg/kg dosing, stratified by quartiles of the observed weight distribution in the analysis population:
 - 33.9–63kg, >63–73kg, >73–85.5kg, and >85.5–156kg.
- Overall, the steady-state exposures of reslizumab following 3.0mg/kg IV administration were comparable across the different body weight groups, with only relatively minor numerical differences in mean exposures between the lowest and highest body weight quartiles.
 - Mean $AUC_{ss(0–4wk)}$ values ranged from 27.2mg·h/mL to 33.1mg·h/mL and $C_{av,ss(0–4wk)}$ values ranged from 40.4µg/mL to 49.3µg/mL between the lowest (33.9–63kg) to highest (>85.5–156kg) body weight groups.

FIGURE 3. BOXPLOTS OF MODEL-PREDICTED $AUC_{ss(0–4wk)}$, $C_{av,ss(0–4wk)}$, $C_{max,ss(0–4wk)}$, and $C_{min,ss(0–4wk)}$, NORMALIZED TO A 3.0MG/KG DOSE, BY BODY WEIGHT FOLLOWING IV DOSING EVERY 4 WEEKS

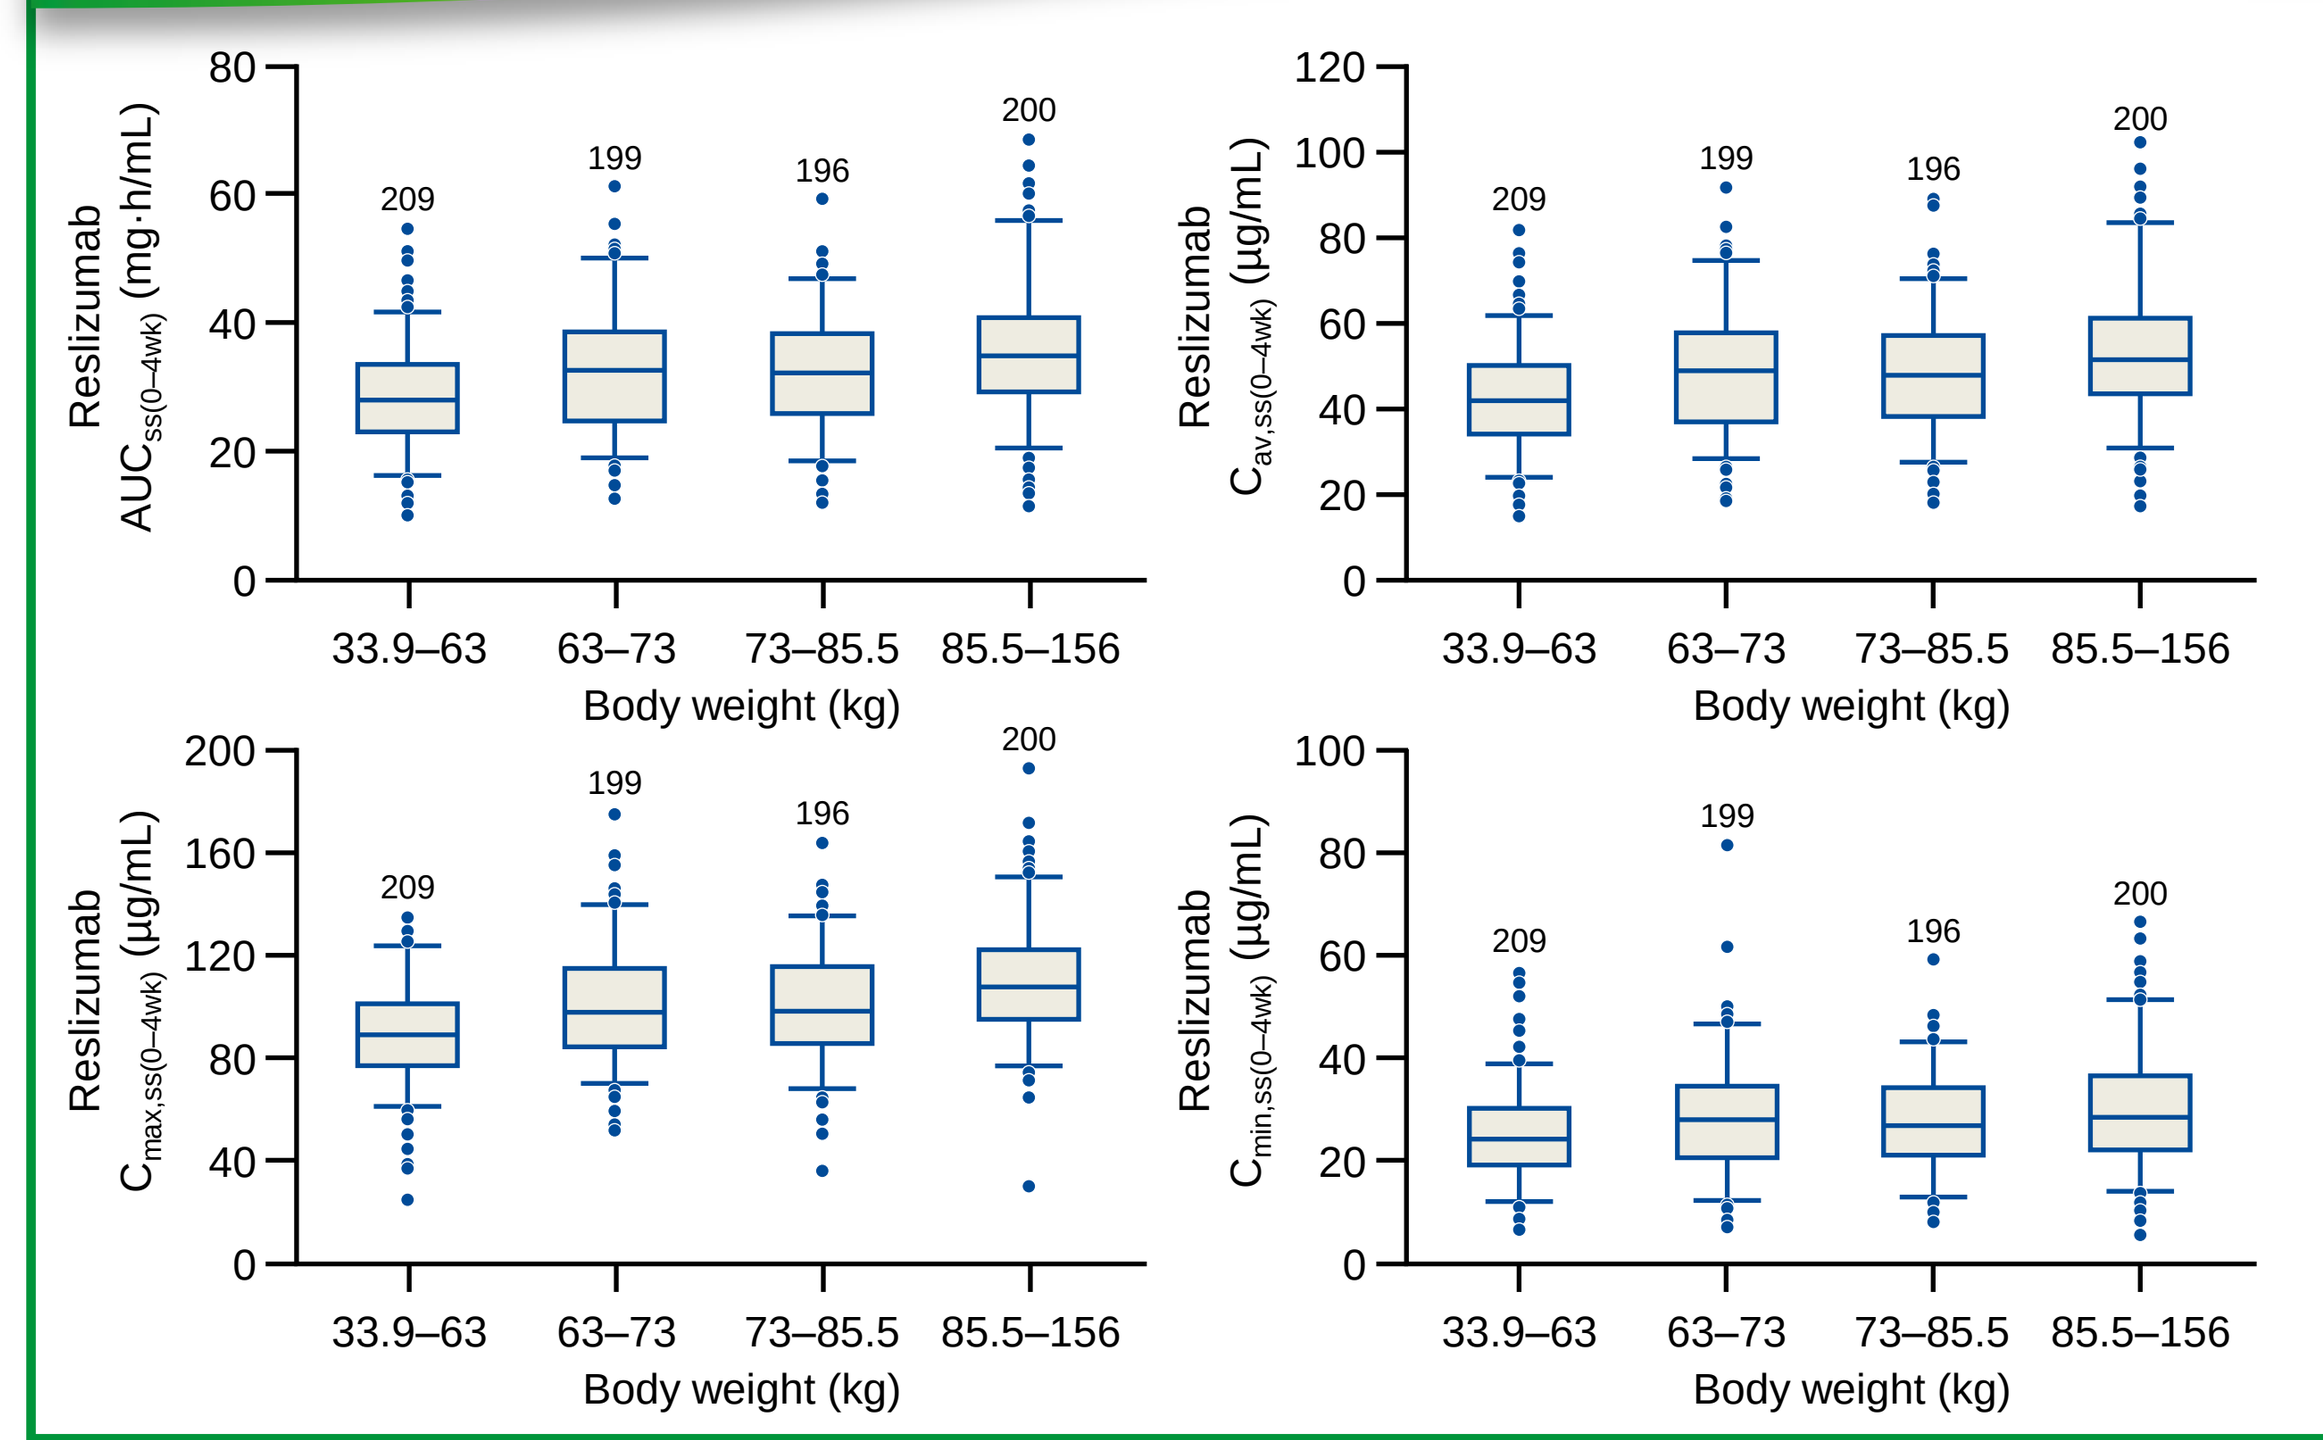

Reslizumab $AUC_{ss(0–4wk)}$ (mg·h/mL)
Reslizumab $C_{av,ss(0–4wk)}$ (µg/mL)
Reslizumab $C_{max,ss(0–4wk)}$ (µg/mL)
Reslizumab $C_{min,ss(0–4wk)}$ (µg/mL)
33.9–63
63–73
73–85.5
85.5–156
Body weight (kg)
209
199
196
200

Boxes are 25th, 50th and 75th percentiles; whiskers are 5th to 95th percentiles; asterisks show data points outside this range; the number of subjects is above each box.
$AUC_{ss(0–4wk)}$: area under the reslizumab serum concentration versus time curve; $C_{av,ss(0–4wk)}$: average reslizumab serum concentration; $C_{max,ss(0–4wk)}$: maximum reslizumab serum concentration; $C_{min,ss(0–4wk)}$: minimum reslizumab serum concentration

CONCLUSIONS

- **A two-compartment PK model with zero order input and first-order elimination adequately describes the PPK of reslizumab following IV administration.**
- **The PPK analysis identified an increase in both CL and V_c with increasing body weight, suggesting that a fixed IV dose administered across a range of body weights would result in a relative decrease in reslizumab exposures with increasing body weight.**
- **The simulated steady-state exposures of reslizumab following a 3.0mg/kg once-monthly IV dosing regimen were numerically comparable across a greater than four-fold range of body weights.**
- **This modeling and simulation analysis provides support for the appropriateness of weight-based dosing of 3.0mg/kg IV reslizumab in order to achieve similar steady-state exposures across a wide range of body weights.**

References

1. Egan RW, et al. Arzneimittelforschung 1999;49:779–90.
2. Kips JC. Am J Respir Crit Care Med 2003;167:1655–59.
3. Reslizumab prescribing information: http://www.cinqair.com/.
4. Castro M, et al, Lancet Resp Med 2015;3:355–66.
5. Bjermer L, et al, CHEST 2016; Apr 4. doi: 10.1016/j.chest.2016.03.032. [Epub ahead of print].
6. Corren J, et al, CHEST 2016; Mar 24. doi: 10.1016/j.chest.2016.03.018. [Epub ahead of print].

Acknowledgements

The authors would like to thank Donna Cox for her critical review of this poster. Editorial support provided by GeoMed, an Ashfield company, part of UDG Healthcare plc, was funded by Teva Branded Pharmaceutical Products R & D, Inc. (Frazer, PA). Teva provided a full review of the poster.
This study was sponsored by Teva Branded Pharmaceutical Products R & D, Inc.

Copies of this poster obtained through Quick Response Code are for personal use only and may not be reproduced without permission from ATS and the author of this poster.

Presented at the AMERICAN THORACIC SOCIETY (ATS) INTERNATIONAL CONGRESS, San Francisco, CA, USA, May 13–18, 2016.