Suppressing TGF-β activation to reduce lung fibrosis:
In search for an efficacious dose regimen for alpha V integrin inhibitors

Sergey Ermakov[a,†], Grant Generaux[a], Fulya Akpinar Singh[b,‡], Ankit Chandra[b], Scott Q Siler[a]

[a]DILIsym Services, Inc., a Simulations Plus company, Research Triangle Park, NC; [b]Bristol Myers Squibb, Princeton, NJ; Current affiliation: [†]Daiichi Sankyo, Basking Ridge, NJ; [‡]Genmab, Princeton, NJ;

ABSTRACT

Objectives: *Elevated levels of TGF-β and specifically its activated form is regarded as one of the disease drivers in patients with idiopathic pulmonary fibrosis (IPF). TGF-β can bind to the extracellular matrix and be subsequently liberated and activated via interaction with αV integrins present on the surface of epithelial and myofibroblast cells in the lungs. Blocking αV integrin molecules with a drug prevents TGF-β liberation and activation, however this may drive concentration of the ECM-bound latent TGF-β higher, thus reducing the drug efficacy. The goal of this study is to model the underlying biology and to identify optimal αV integrin inhibitor regimens that effectively suppress TGF-β activation.*

Methods: *We developed a computer model describing the dynamics of the TGF-β latent and active forms in the lung tissue that enables the stoichiometric calculation of the inhibitor and substrate binding to αV integrin. The model includes the following features:*

- *Latent TGF-β is produced by resident cells within the lungs and released into the extracellular fluid (ECF) where a portion can be incorporated into the extracellular matrix (ECM).*
- *TGF-β is released back to ECF during ongoing ECM turnover.*
- *Exchange between the tissue and the circulating TGF-β pools is included.*
- *Activated TGF-β is released upon interaction with αV integrins.*
- *When the drug is present it competes with TGF-β for αV integrin binding sites and inhibits TGF-β liberation and activation.*
- *The drug+αV integrin complex can be internalized and degraded inside the cell.*

The above mechanisms are modelled by the set of ODEs. Parameters such as baseline concentration of TGF-β, αV integrins, number of TGF-β binding sites in the ECM, drug inhibition constants are taken from or estimated based on available literature data (Balestro 2019, King 2011, Hynes 2009). Drug concentrations are represented, including specified C_{max} and C_{trough} values, based on the dosing regimen of interest.

Results: *An optimal αV integrin inhibitor dosing regimen for IPF patients would persistently decrease the activated TGF-β concentrations. We have simulated effects of αV integrin inhibitor PK profile (C_{max} and C_{trough}) on lowering activated TGF-β concentration as well as uncertainty in parameters defining ECM turnover and αV integrins distribution in the tissue. We have identified that 90% suppression of activated TGF-β will require maintaining C_{trough} values close to the concentration of available αV integrin molecules in the tissue. Also we found that αV integrin inhibition drives an increase in the latent ECM-bound TGF-β pool. The elevated levels can compete with the drug for αV integrin binding sites. Moreover, therapy cessation will cause a temporary spike in activated TGF-β that could lead to undesired side effects.*

Conclusions: *We have developed a computer model describing TGF-β dynamics in the lung tissue in response to drugs blocking αV integrins. It was identified that effective suppression of TGF-β activation can be achieved by maintaining trough concentrations close to concentrations of αV integrins in the tissue.*

INTRODUCTION

- TGF-β plays a substantial role in pulmonary fibrosis, driving pathophysiologic levels of myofibroblasts and extracellular matrix (ECM)
- Integrins within the lungs can liberate ECM-bound TGF-β, and integrin inhibitors may prove an effective treatment option to reduce active TGF-β
- A small QSP model has been developed to predict optimal integrin inhibition, accounting for factors that could improve or worsen fibrosis

RESULTS

QSP Model

Model of integrins, ECM, TGF-β dynamics

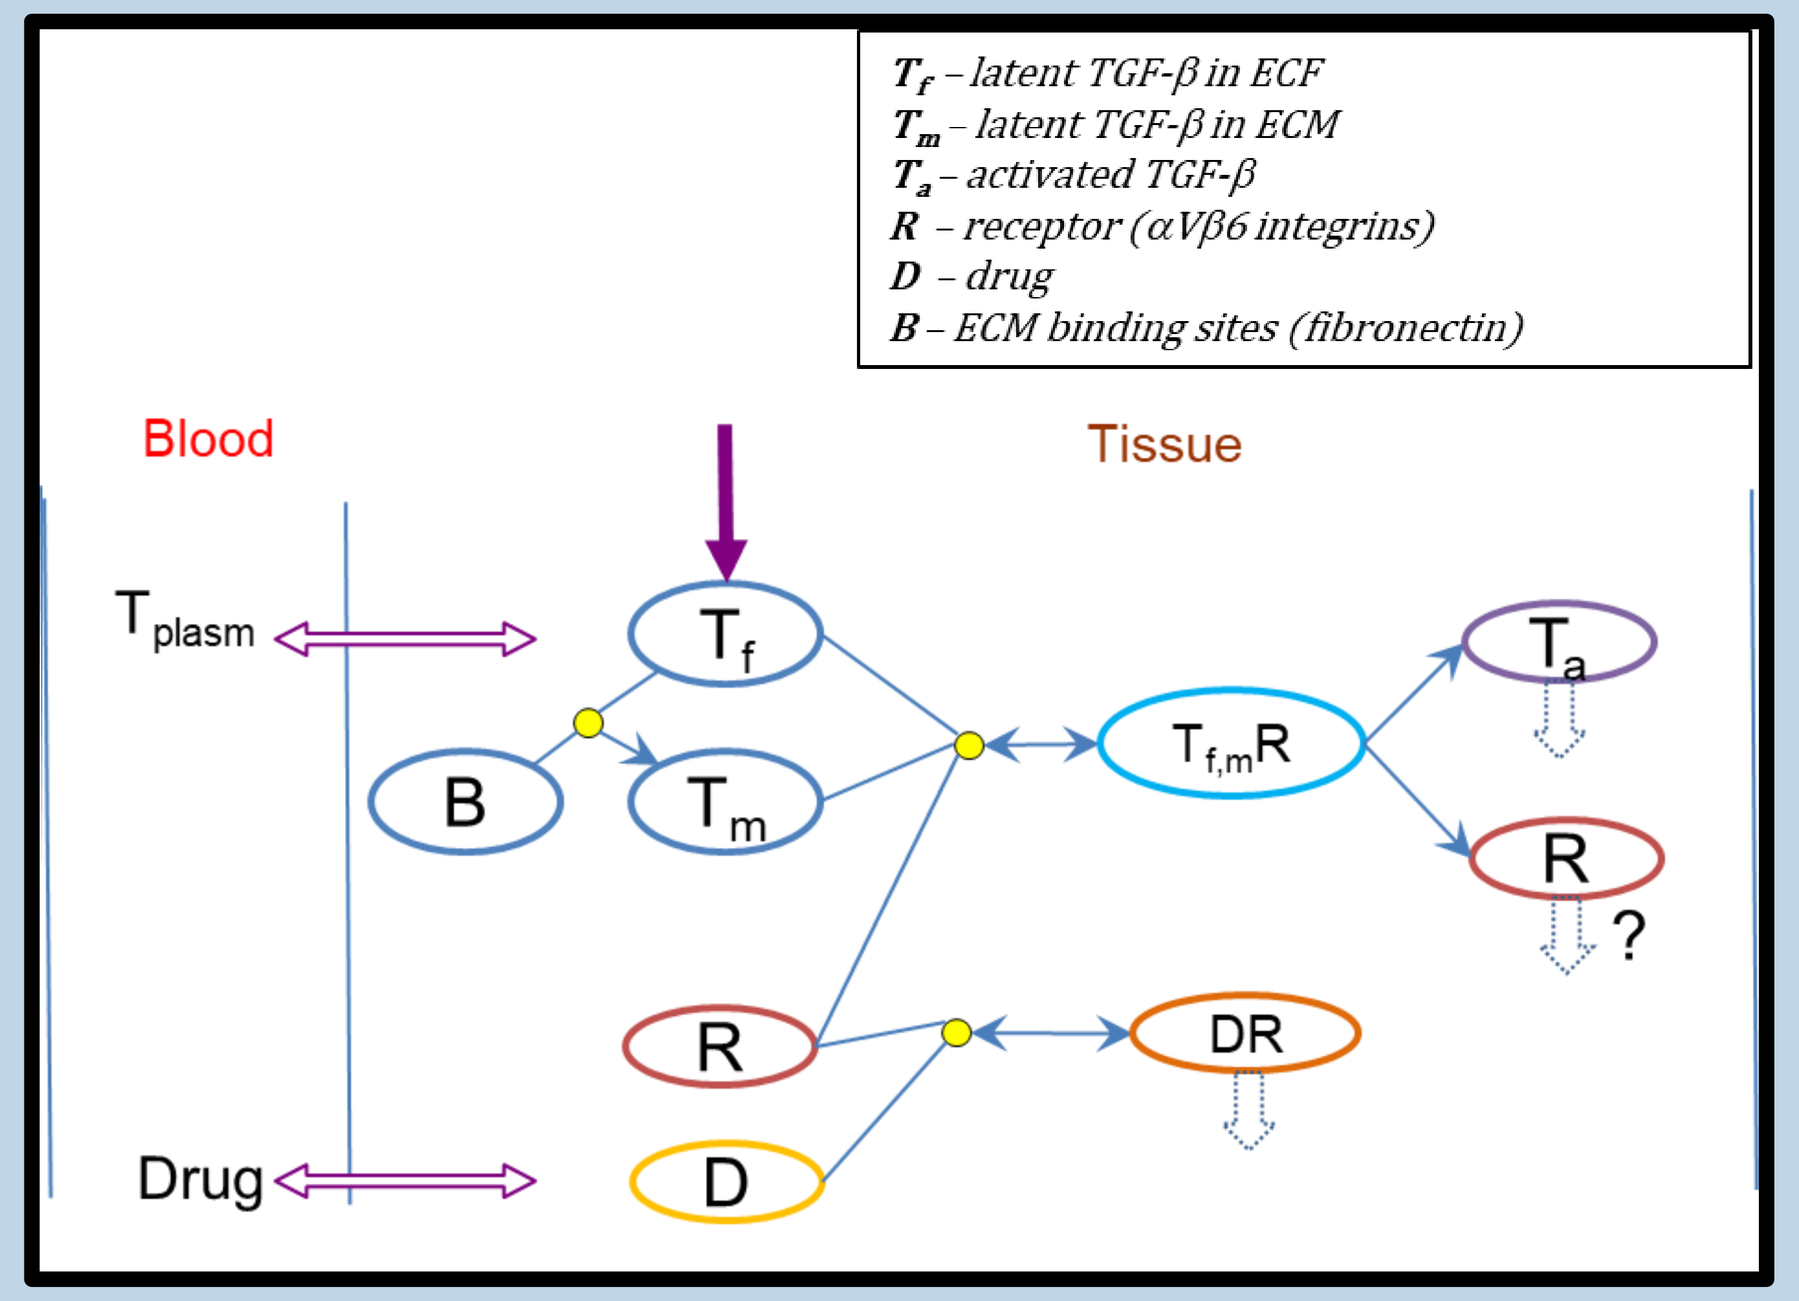
T_f – latent TGF-β in ECF
T_m – latent TGF-β in ECM
T_a – activated TGF-β
R – receptor (αVβ6 integrins)
D – drug
B – ECM binding sites (fibronectin)
Blood
Tissue
T_{plasm}
T_f
B
T_m
$T_{f,m}R$
T_a
R
?
R
DR
Drug
D

IPFsym

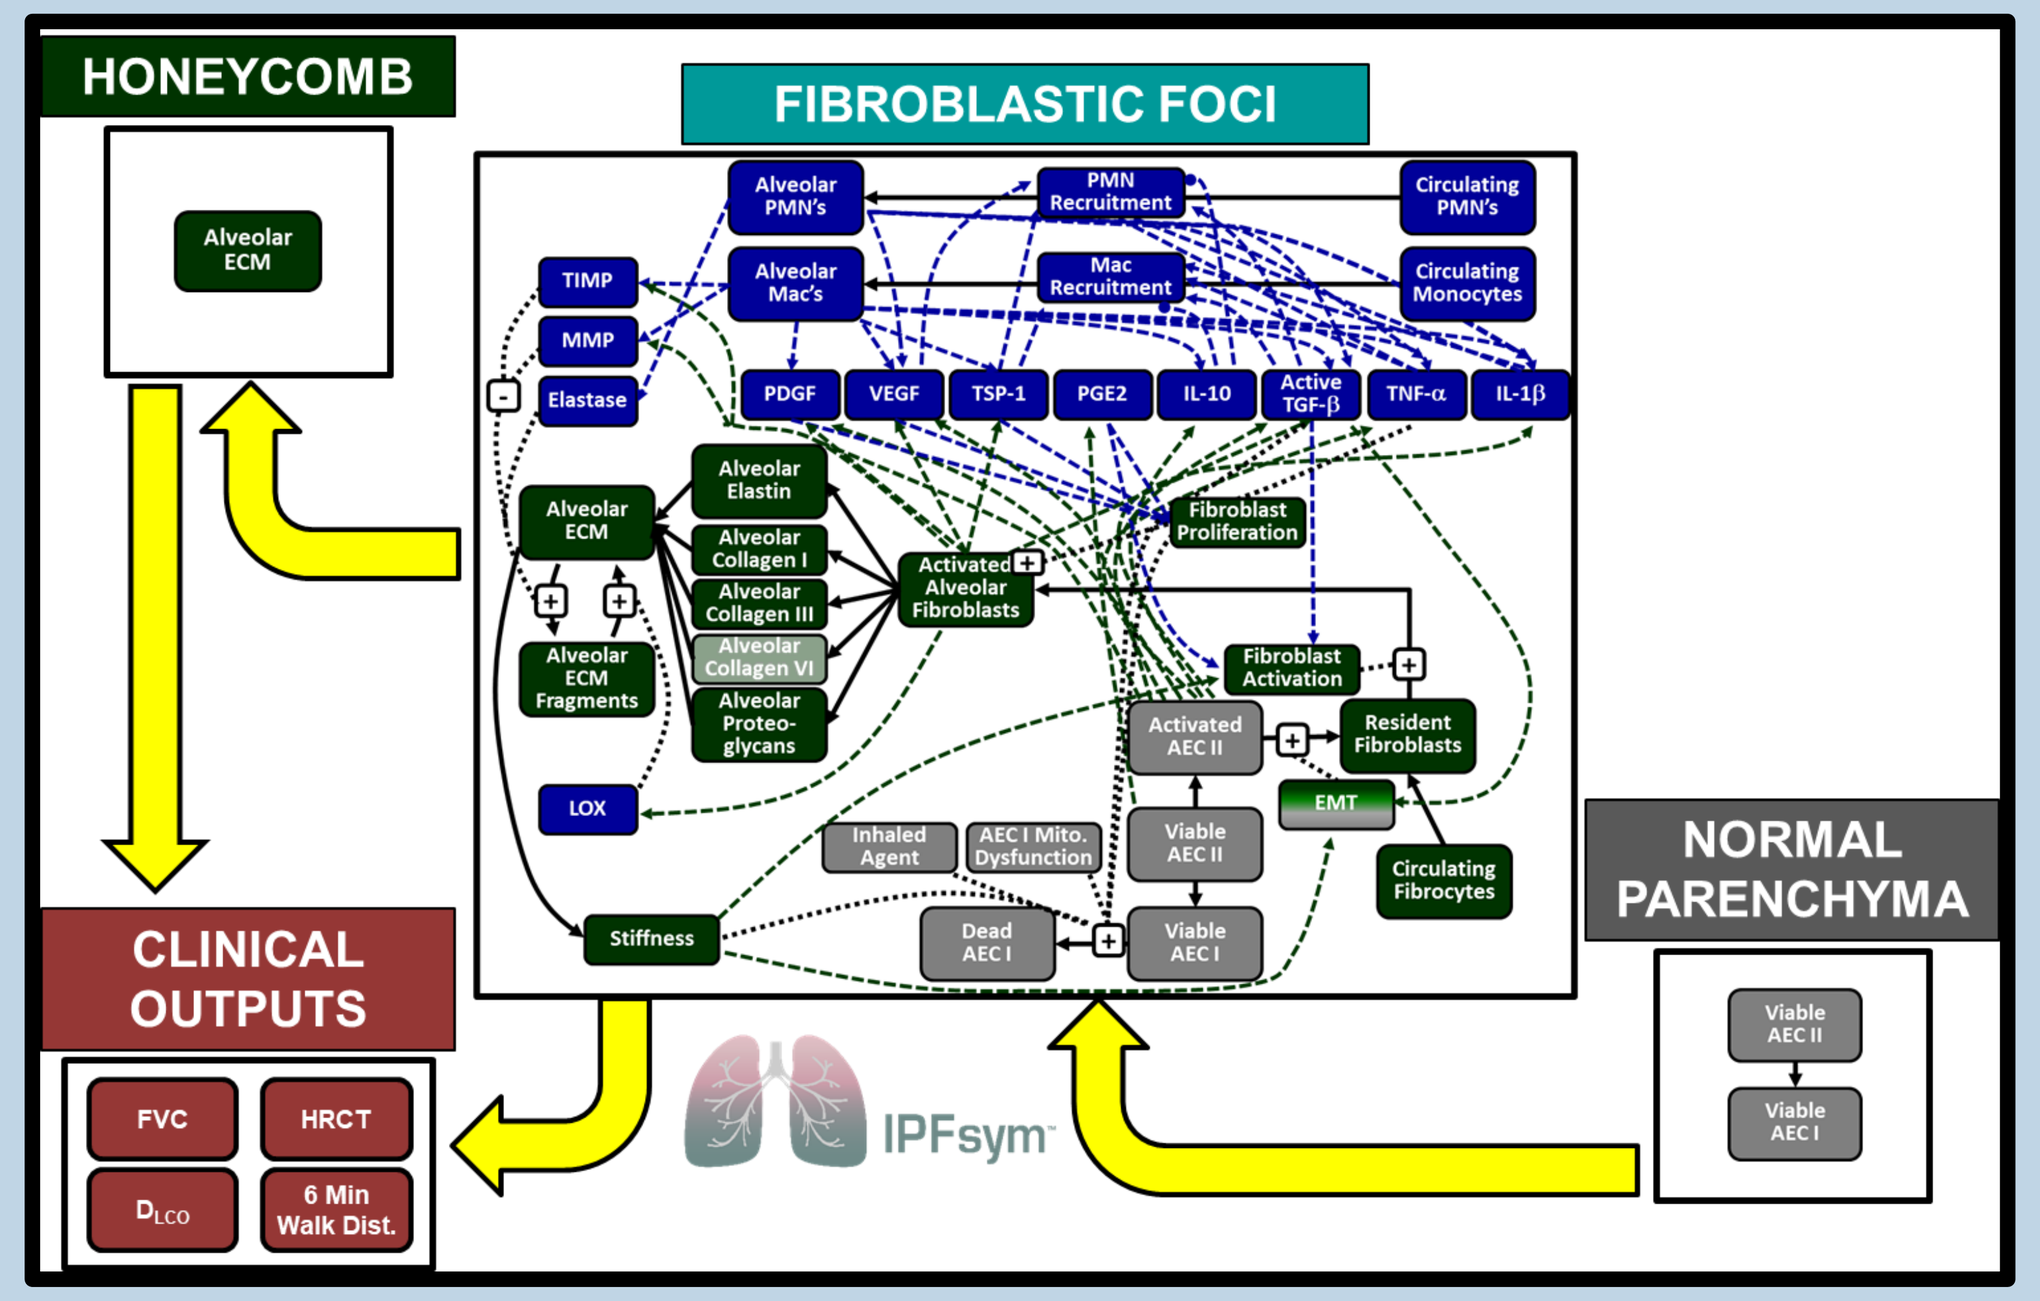
HONEYCOMB
Alveolar ECM
FIBROBLASTIC FOCI
Alveolar PMN's
PMN Recruitment
Circulating PMN's
TIMP
Alveolar Mac's
Mac Recruitment
Circulating Monocytes
MMP
Elastase
PDGF
VEGF
TSP-1
PGE2
IL-10
Active TGF-β
TNF-α
IL-1β
Alveolar Elastin
Alveolar ECM
Alveolar Collagen I
Alveolar Collagen III
Alveolar Collagen VI
Activated Alveolar Fibroblasts
Fibroblast Proliferation
Alveolar ECM Fragments
Alveolar Proteo-glycans
Fibroblast Activation
Activated AEC II
Resident Fibroblasts
EMT
LOX
Inhaled Agent
AEC I Mito. Dysfunction
Viable AEC II
Circulating Fibrocytes
Stiffness
Dead AEC I
Viable AEC I
NORMAL PARENCHYMA
Viable AEC II
Viable AEC I
CLINICAL OUTPUTS
FVC
HRCT
DLCO
6 Min Walk Dist.
IPFsym

1. $\frac{dR}{dt} = r_{syn}\frac{EC_{50}}{EC_{50}+R_{tot}} - k_{T,on}(T_f+T_m)R - k_{D,on}\cdot D\cdot R + k_{T,off}([T_fR]+[T_mR]) + k_{D,off}\cdot[DR] - k_{R,degr}\cdot R + k_{act}([T_mR]+[T_mR])$
2. $\frac{dT_f}{dt} = q_{syn} + \propto\cdot(T_{plasm}-T_f) - k_{T_f,clear}\cdot T_f - k_{att}\cdot T_f\cdot B + k_{T_m,degr}\cdot T_m$
3. $\frac{d[T_fR]}{dt} = k_{T,on}\cdot T_f\cdot R - k_{T,off}\cdot[T_fR] - k_{act}\cdot[T_fR]$
4. $\frac{dT_a}{dt} = k_{act}\cdot([T_fR]+[T_mR]) - k_{Ta,degr}\cdot T_a$
5. $\frac{dD}{dt} = \beta\cdot(D_{plasm}-D) - k_{D,on}\cdot D\cdot R + k_{D,off}\cdot[DR] - k_{D,clear}\cdot D$
6. $\frac{d[DR]}{dt} = k_{D,on}\cdot D\cdot R - k_{D,off}\cdot[DR] - k_{DR,int}\cdot[DR]$
7. $\frac{dT_m}{dt} = -k_{T,on}\cdot T_m\cdot R + k_{T,off}\cdot[T_mR] - k_{T_m,degr}\cdot T_m + k_{att}\cdot T_f\cdot B$
8. $\frac{d[T_mR]}{dt} = k_{T,on}\cdot T_m\cdot R - k_{T,off}\cdot[T_mR] - k_{act}\cdot[T_mR]$
9. $\frac{dB}{dt} = b_{syn} - k_{B,degr}\cdot B - k_{att}\cdot T_f\cdot B$

$T_m = [T_fB]$
$R_{tot} = R + [T_fR] + [T_mR] + [DR];$

T_f – latent TGF-β in ECF
T_m – latent TGF-β in ECM
T_a – activated TGF-β
R – receptor (αVβ6 integrins)
D – drug
B – ECM binding sites (fibronectin)

- The small QSP model of integrins, ECM, and TGF-β dynamics includes binding and catalysis for the relevant processes
- Integrin inhibition being pursued as a possible treatment for idiopathic pulmonary fibrosis (IPF)
- Predictions of TGF-β levels due to integrin inhibition helps inform potential influence on IPF disease progression
- A larger QSP model of IPF, IPFsym, can be informed with these simulation results
- The small QSP model of integrins, ECM, and TGF-β dynamics includes equations and parameters that allow simulation of:
 - The liberation and activation of ECM-TGF (T_a) by integrin (R)
 - Integrin (R) subtype parameterized to represent αVβ6 (John 2020)
 - Binding of inhibitor (D) to integrin
 - Inhibition of integrin-mediated liberation and activation of ECM-TGF by D
 - Internalization and degradation of R after binding D
 - Physiologic turnover of ECM, including B and T_m; T_m re-enters the T_f pool
- Parameter values taken from or derived from literature (see references)

Simulation Results: Treatment

Periodic High Integrin Inhibition

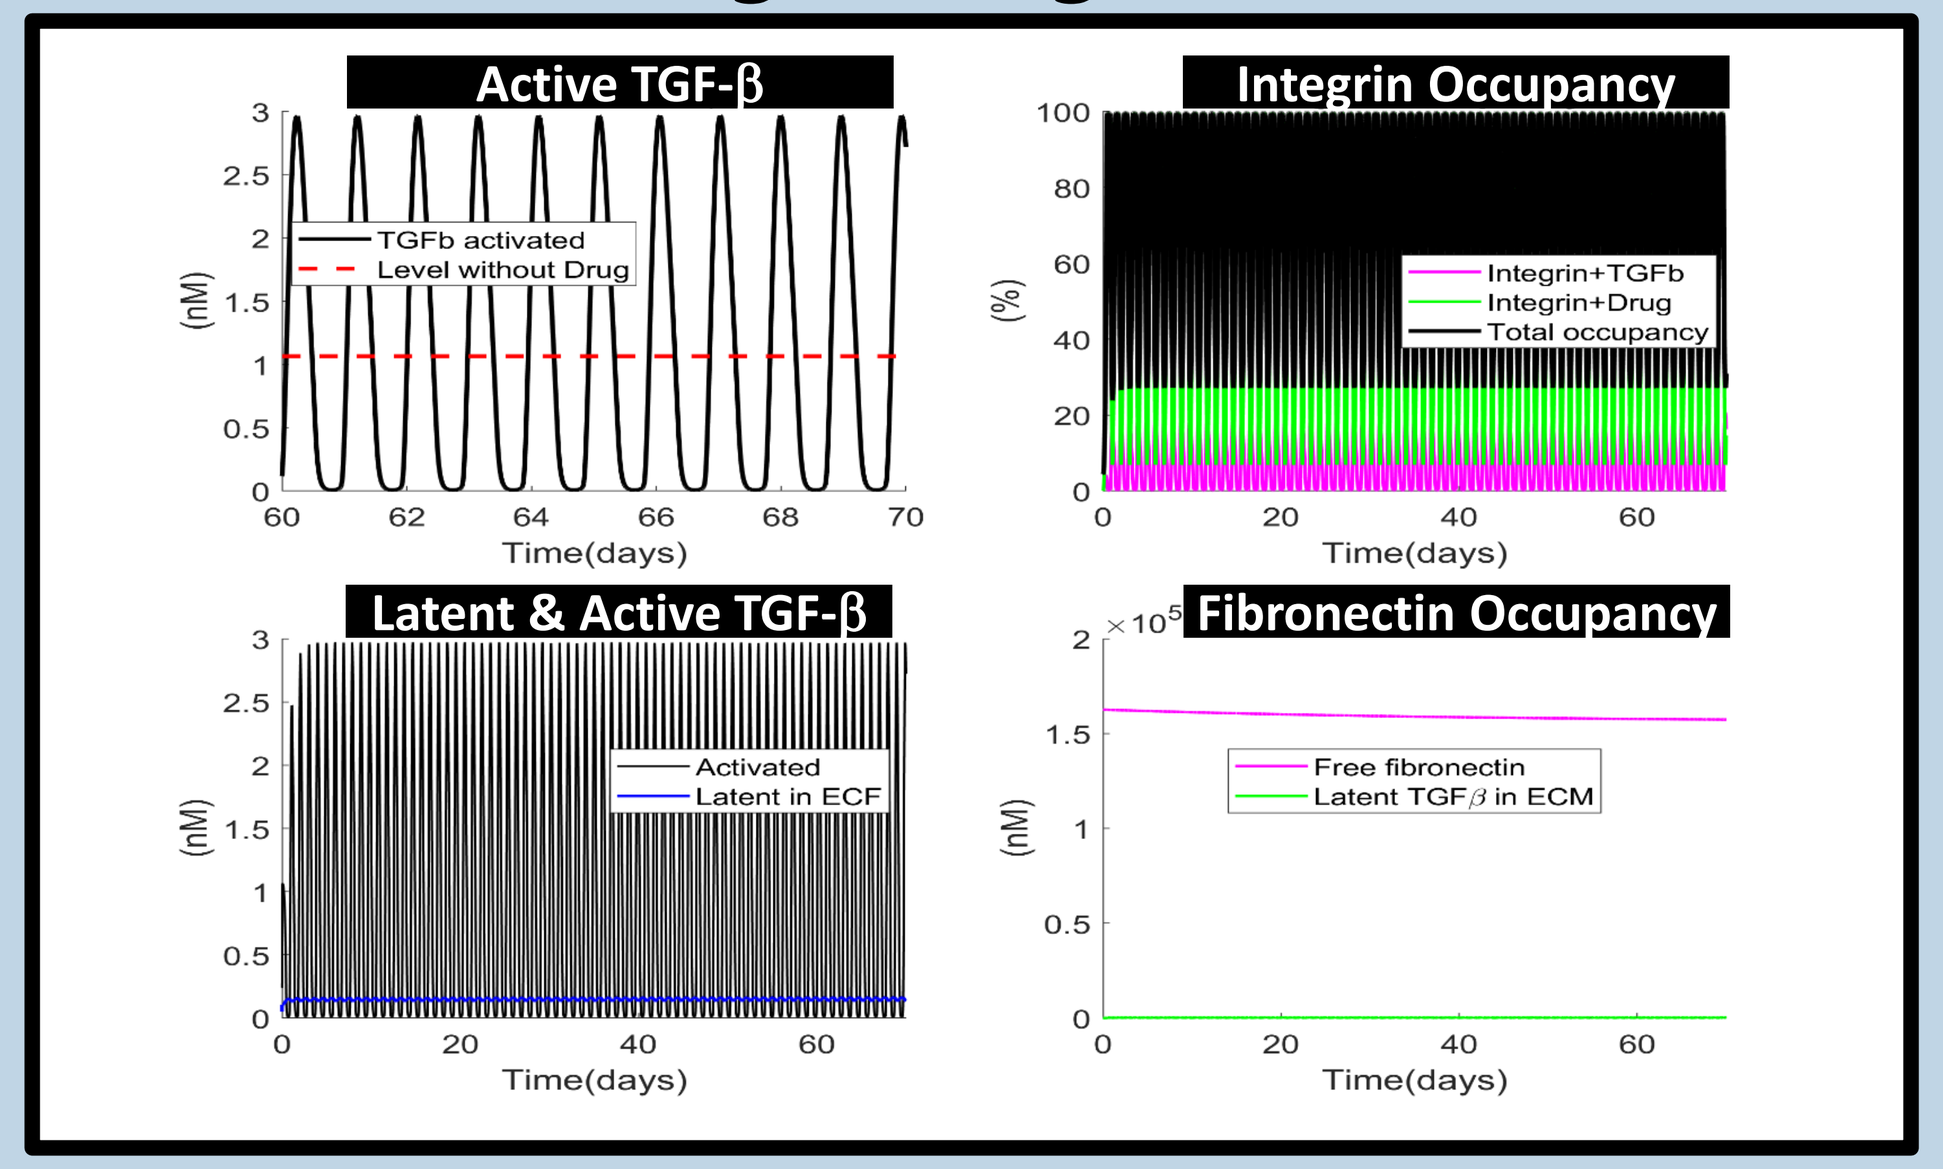
Active TGF-β
TGFb activated
Level without Drug
Time(days)
(nM)
Integrin Occupancy
Integrin+TGFb
Integrin+Drug
Total occupancy
(%)
Latent & Active TGF-β
Activated
Latent in ECF
Fibronectin Occupancy
Free fibronectin
Latent TGFβ in ECM

Persistent Mid Integrin Inhibition

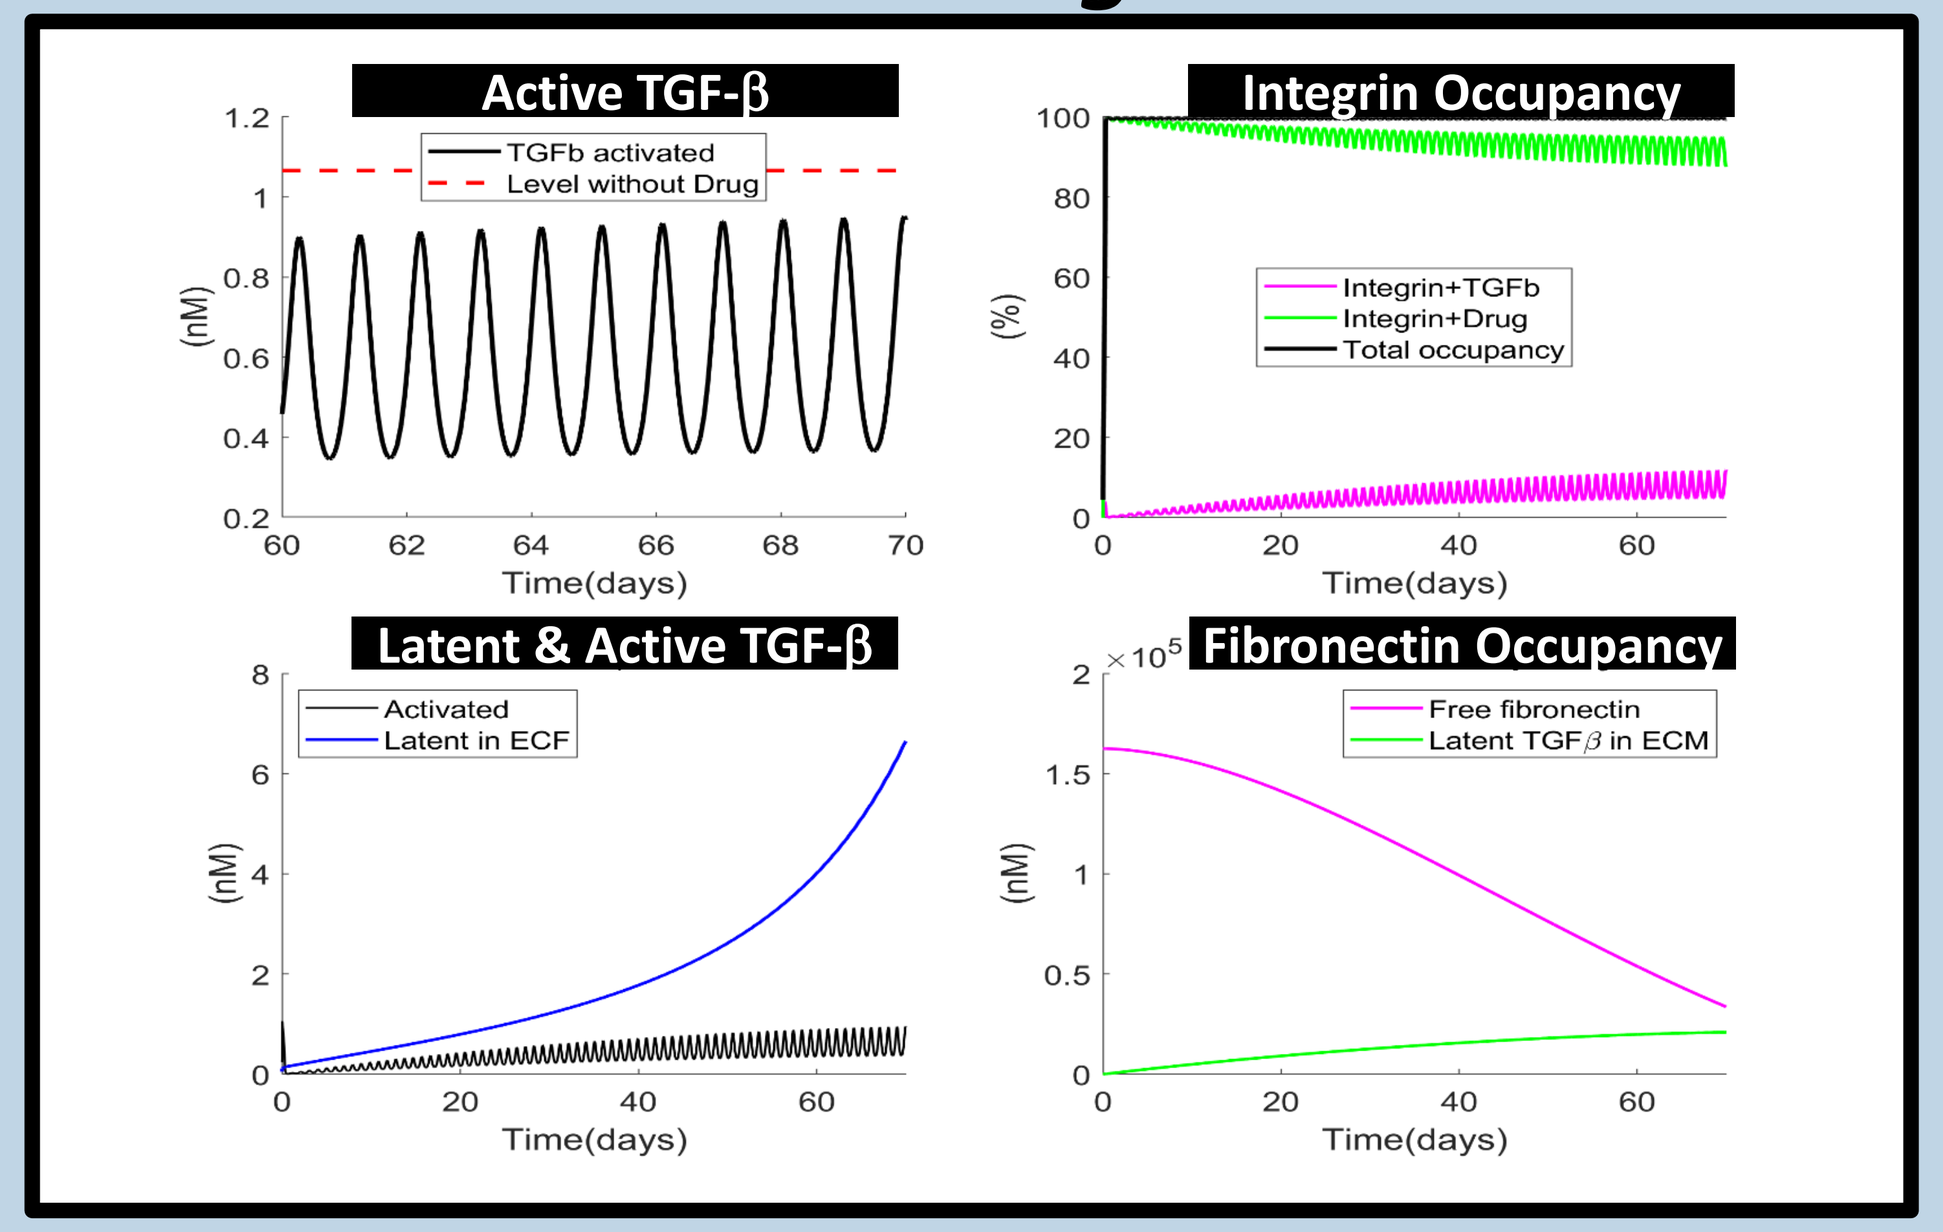
Active TGF-β
TGFb activated
Level without Drug
Time(days)
(nM)
Integrin Occupancy
Integrin+TGFb
Integrin+Drug
Total occupancy
(%)
Latent & Active TGF-β
Activated
Latent in ECF
Fibronectin Occupancy
Free fibronectin
Latent TGFβ in ECM

Persistent High Integrin Inhibition

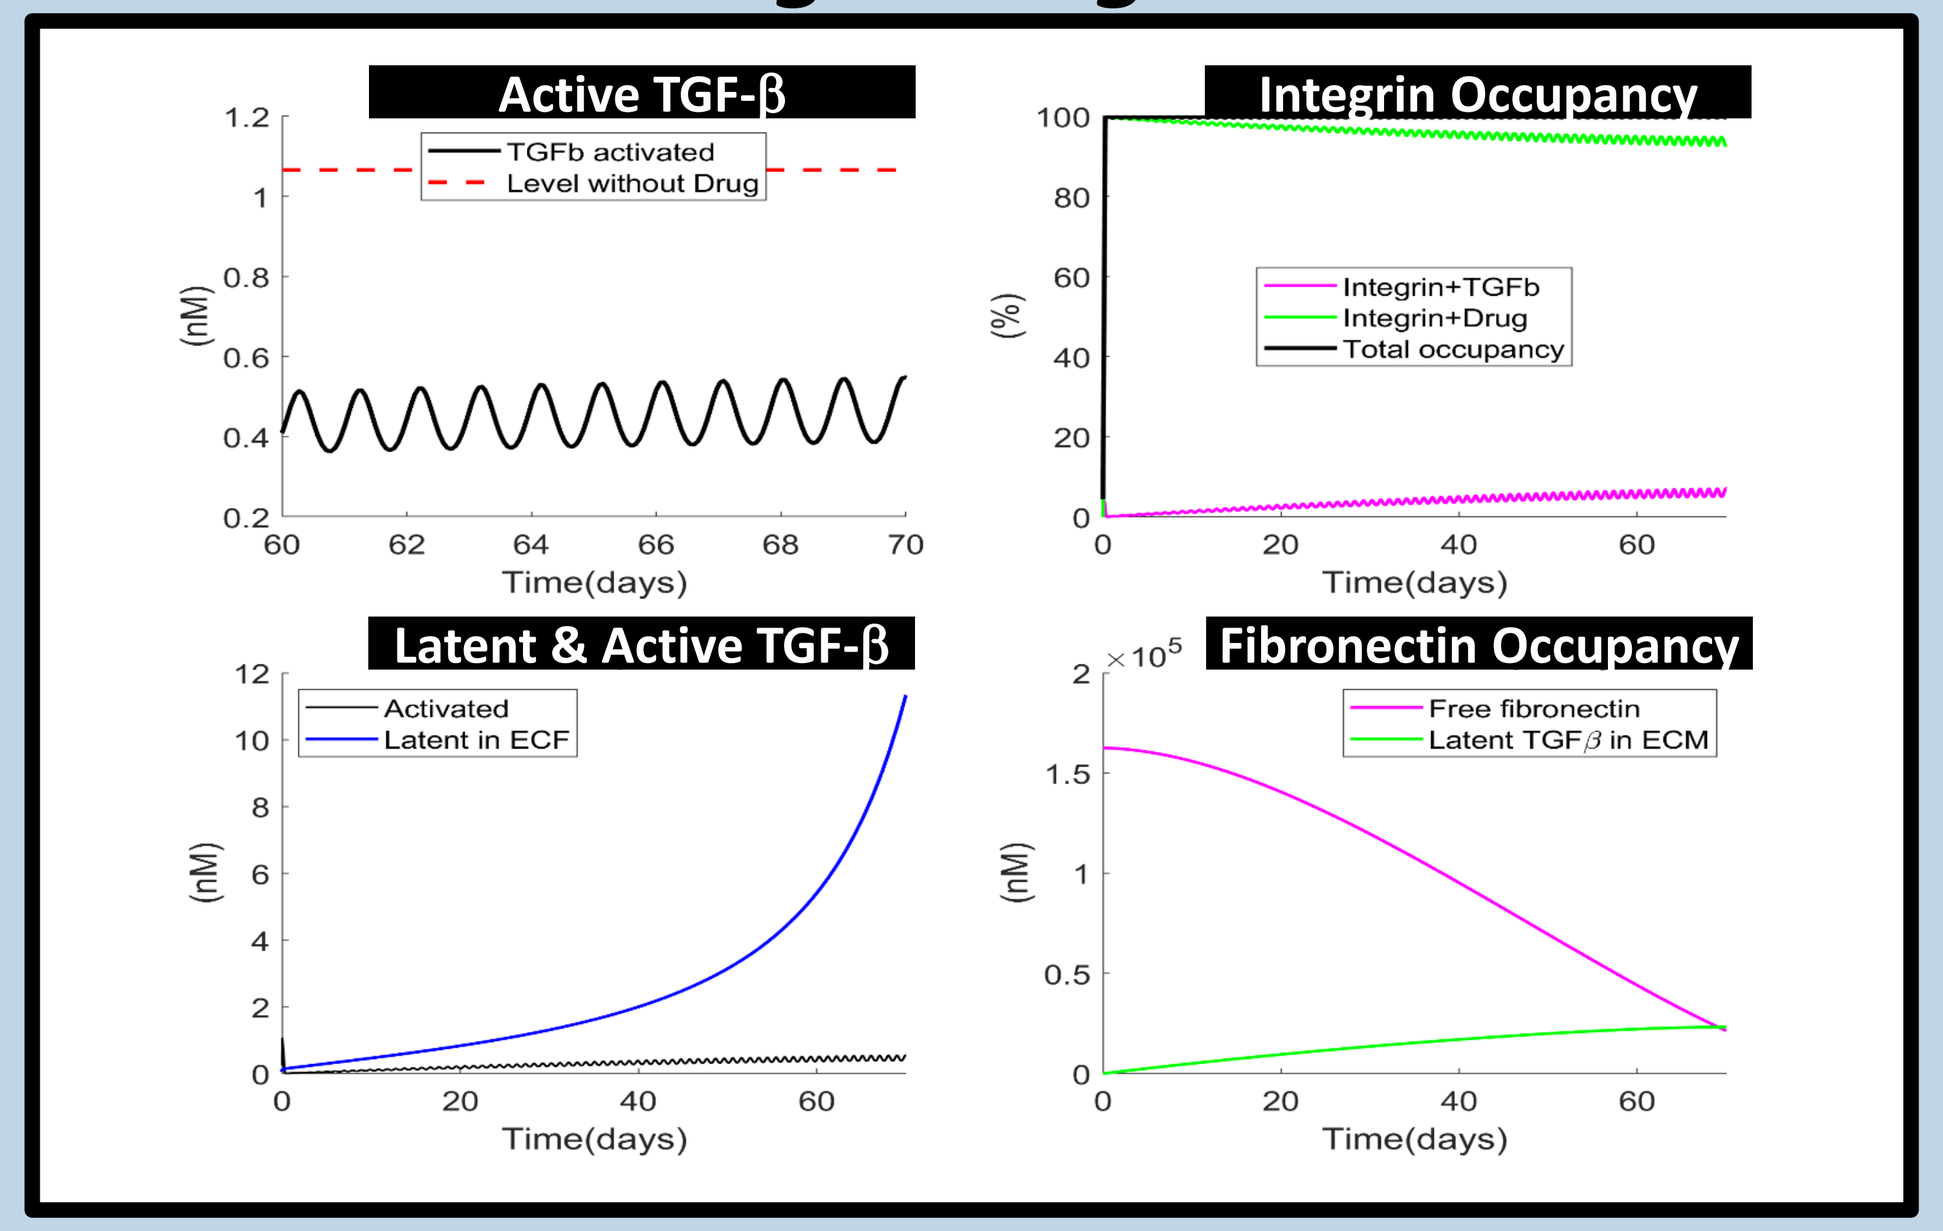
Active TGF-β
TGFb activated
Level without Drug
Time(days)
(nM)
Integrin Occupancy
Integrin+TGFb
Integrin+Drug
Total occupancy
(%)
Latent & Active TGF-β
Activated
Latent in ECF
Fibronectin Occupancy
Free fibronectin
Latent TGFβ in ECM

- Active TGF-β levels predicted to be reduced compared to pre-treatment levels when integrins persistently inhibited (mid or high)
- High fraction of drug occupancy of integrins associated with reduced active TGF-β
- Periodic high (100%) integrin inhibition does reduce nadir active TGF-β levels, but not average concentrations
- Persistent integrin inhibition is predicted to increase size of ECM-TGF pool (reduced exit from pool) and to increase latent TGF-β levels (reduced entrance into pool)
- Note that this small QSP model of integrins, ECM, and TGF-β dynamics does not include changes to ECM or TGF-β production rates due to disease progression or treatment; each could influence results and could be evaluated by implementing this small QSP model into the larger QSP model, IPFsym

METHODS

- Simulations performed in the presence of a theoretical integrin inhibitor
 - Full inhibition of catalytic activity when drug is bound to integrin; magnitude of overall inhibition dictated by integrin occupancy
 - Theoretical drug concentrations are simulated, including C_{max} and C_{trough} values; 24 hour dosing period simulated
 - Periodic High integrin inhibition includes 100% max/0% nadir occupancy
 - Persistent Mid integrin inhibition includes 100% max/60% nadir occupancy
 - Persistent High integrin inhibition includes 100% max/90% nadir occupancy
 - ECM levels and latent TGF-β production rates do not change (i.e., no influence of disease progression nor response to treatment)
- Simulations were performed using Matlab software

CONCLUSIONS

- Effective, persistent suppression of active TGF-β levels can be achieved by maintaining trough drug concentrations close to the concentrations of αV integrins in the tissue.
- αV integrin inhibition drives an increase in the latent ECM-bound TGF-β pool; the elevated levels can compete with the drug for αV integrin binding sites
- Size of latent ECM-bound TGF-β pool has not been measured in IPF patients; the size of this pool (and occupancy of ECM binding sites) could influence the potential efficacy of integrin inhibition
- The small QSP model of integrins, ECM, and TGF-β dynamics enables exploration of αV integrin inhibitor dosing paradigms on TGF-β levels

REFERENCES

Balestro, E. et al. J. Clin. Med. 8, (2019)
Hynes, R. O., Science 326, 1216–1219 (2009)
John, A.E., et al. Nat Commun. 11, 4659-4673 (2020)
King, T. E. J., et al. Lancet Lond. Engl. 378, 1949–1961 (2011)
Laurent, G.J., Am. J. Physiol. 252:C1-C9 1987
Westergren-Thorsson, G., et al., Int. J. Biochem. Cell Biol. 83, 27-38 (2017)

ACKNOWLEDGEMENTS

- This work was supported by Bristol Meyers Squibb

SimulationsPlus
Cognigen | DILIsym Services | Lixoft