MECHANISTIC MODELING OF MITOCHONDRIAL BIOGENESIS WITHIN DILISYM COULD EXPLAIN CLINICALLY OBSERVED ADAPTATION OF SERUM ALANINE AMINOTRANSFERASE ELEVATIONS

Kyunghee Yang[1], Jahid Ferdous[1], Jeffrey L Woodhead[1], David Oldach[2], Chris MacLauchlin[2], Prabhavathi Fernandes[2], Paul B Watkins[3], Brett A Howell[1], and Scott Q Siler[1]

[1]DILIsym Services Inc. Research Triangle Park, NC 27709; [2]Cempra, Chapel Hill, NC 27517 (affiliation at time of work);
[3]UNC School of Medicine & UNC Eshelman School of Pharmacy, The University of North Carolina at Chapel Hill, Chapel Hill, NC 27599

INTRODUCTION

- Resolution of serum ALT elevations despite continued drug dosing, termed "adaptation", is commonly observed in clinical trials, but the underlying mechanisms behind this phenomenon remain unclear.
- Mitochondrial dysfunction is one of the major mechanisms underlying drug-induced liver injury (DILI).[1] When mitochondrial function is insufficient for energy demand, mitochondrial biogenesis is often activated and contributes to adaptation.[2-4]
- Solithromycin, a 4th generation macrolide developed for the treatment of community acquired pneumonia, caused serum ALT elevations in a minority of patients in clinical studies, with improvement often observed during continued dosing (or with rapid recovery thereafter).[5]
- DILIsym® is a quantitative systems toxicology (QST) model which integrates *in vitro* mechanistic toxicity data, *in vivo* dynamic drug disposition, known biochemistry, and patient characteristics. DILIsym predicts the hepatotoxic potential of new drug candidates and also provides an enhanced understanding of the mechanisms underlying compounds that generate liver signals in the clinic. [6]
- QST modeling of five macrolide antibiotics using DILIsym showed that mechanisms underlying ALT elevations were significantly different within the same class of antibiotics. ALT elevations mediated by solithromycin and clarithromycin were predominantly due to mitochondrial electron transport chain (ETC) inhibition, whereas erythromycin effects were mainly due to bile acid (BA) transporter inhibition. Telithromycin and azithromycin hepatotoxicity was not explained by mechanisms represented in DILIsym (i.e., mitochondrial dysfunction, BA transporter inhibition, oxidative stress). [7]
- Mechanism analysis using QST modeling suggest that mitochondrial biogenesis might have contributed to the observed adaptation. In the current study, mitochondrial biogenesis was mechanistically represented within DILIsym, and its impact on time dependent ALT elevations resulting from solithromycin treatment was assessed.

METHODS

- The mitochondria sub-model within DILIsym [8] was updated to represent mitochondrial biogenesis. Relevant parameters were obtained from literature or optimized to ALT profiles in patients treated with solithromycin.[9,10]
- Three exemplars with different hepatotoxicity mechanisms were simulated in the simulated human population that includes variability in hepatotoxicity pathways in the absence and presence of mitochondrial biogenesis: solithromycin and phenformin (mitochondrial ETC inhibition), and AMG 009 (bile acid transporter inhibition)

DILI-sim Initiative

RESULTS

Mechanistic Modeling of Mitochondrial Biogenesis

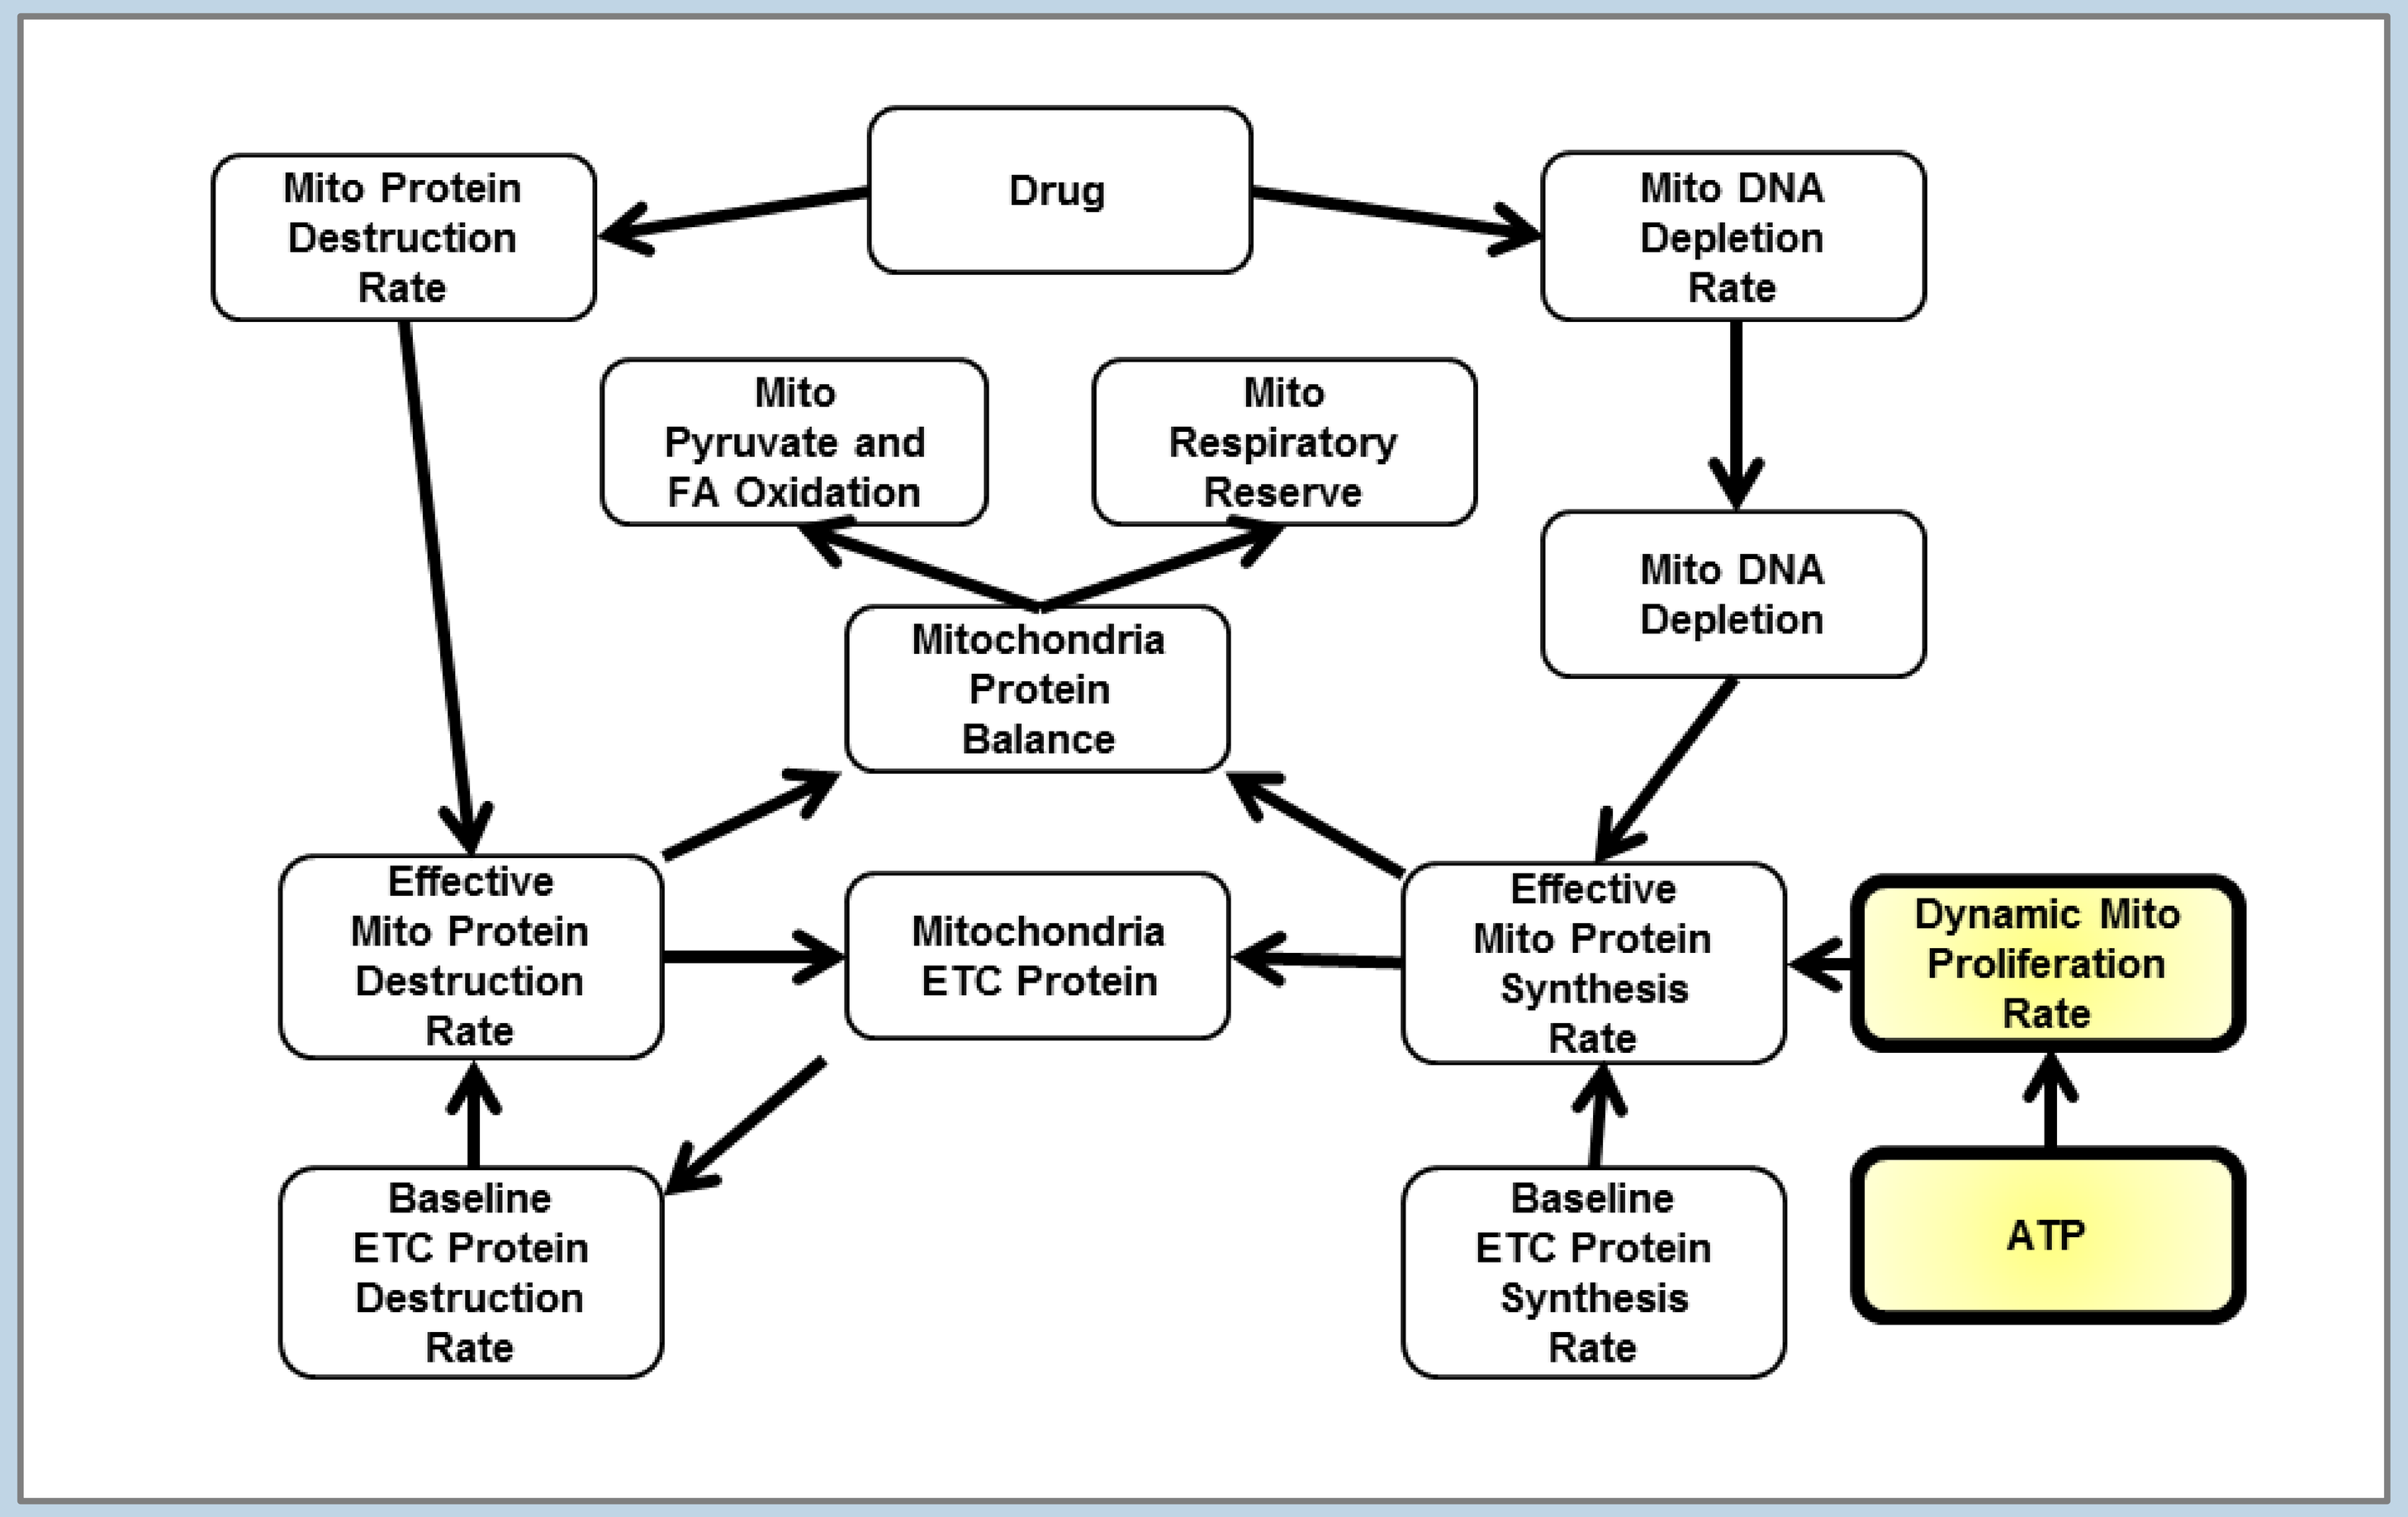

Mito Protein Destruction Rate
Drug
Mito DNA Depletion Rate
Mito Pyruvate and FA Oxidation
Mito Respiratory Reserve
Mito DNA Depletion
Mitochondria Protein Balance
Effective Mito Protein Destruction Rate
Mitochondria ETC Protein
Effective Mito Protein Synthesis Rate
Dynamic Mito Proliferation Rate
Baseline ETC Protein Destruction Rate
Baseline ETC Protein Synthesis Rate
ATP

DILIsym Biogenesis Parameters

Parameter	Unit	Value
Basal mitochondria ETC protein content[a]	mmol	$9.56e^{-14}$
Rate constant for baseline mitochondria ETC protein synthesis[a]	1/hr	0.0069
Mitochondria protein proliferation V_{max}[b]	mmol/hour	$4e^{-14}$
Mitochondria protein proliferation K_m[b]	dimensionless	0.8
Mitochondria protein proliferation Hill[b]	dimensionless	1.5
ATP decrement delay constant for mitochondria[b]	hr	96

[a] calculated using experimental data [9,10]
[b] optimized to ALT profiles in patients administered solithromycin

- Mitochondrial electron transport chain (ETC) protein content is determined by the balance of synthesis and destruction rates
- At steady-state, baseline synthesis and destruction rates are assumed equal
- Changes in mitochondria protein balance subsequently affect ETC function, pyruvate and fatty acid oxidation, and the respiratory reserve
- The effective synthesis rate is regulated by changes in liver ATP and mtDNA content

Adaptation in Patients Treated with Solithromycin: Clinical Data

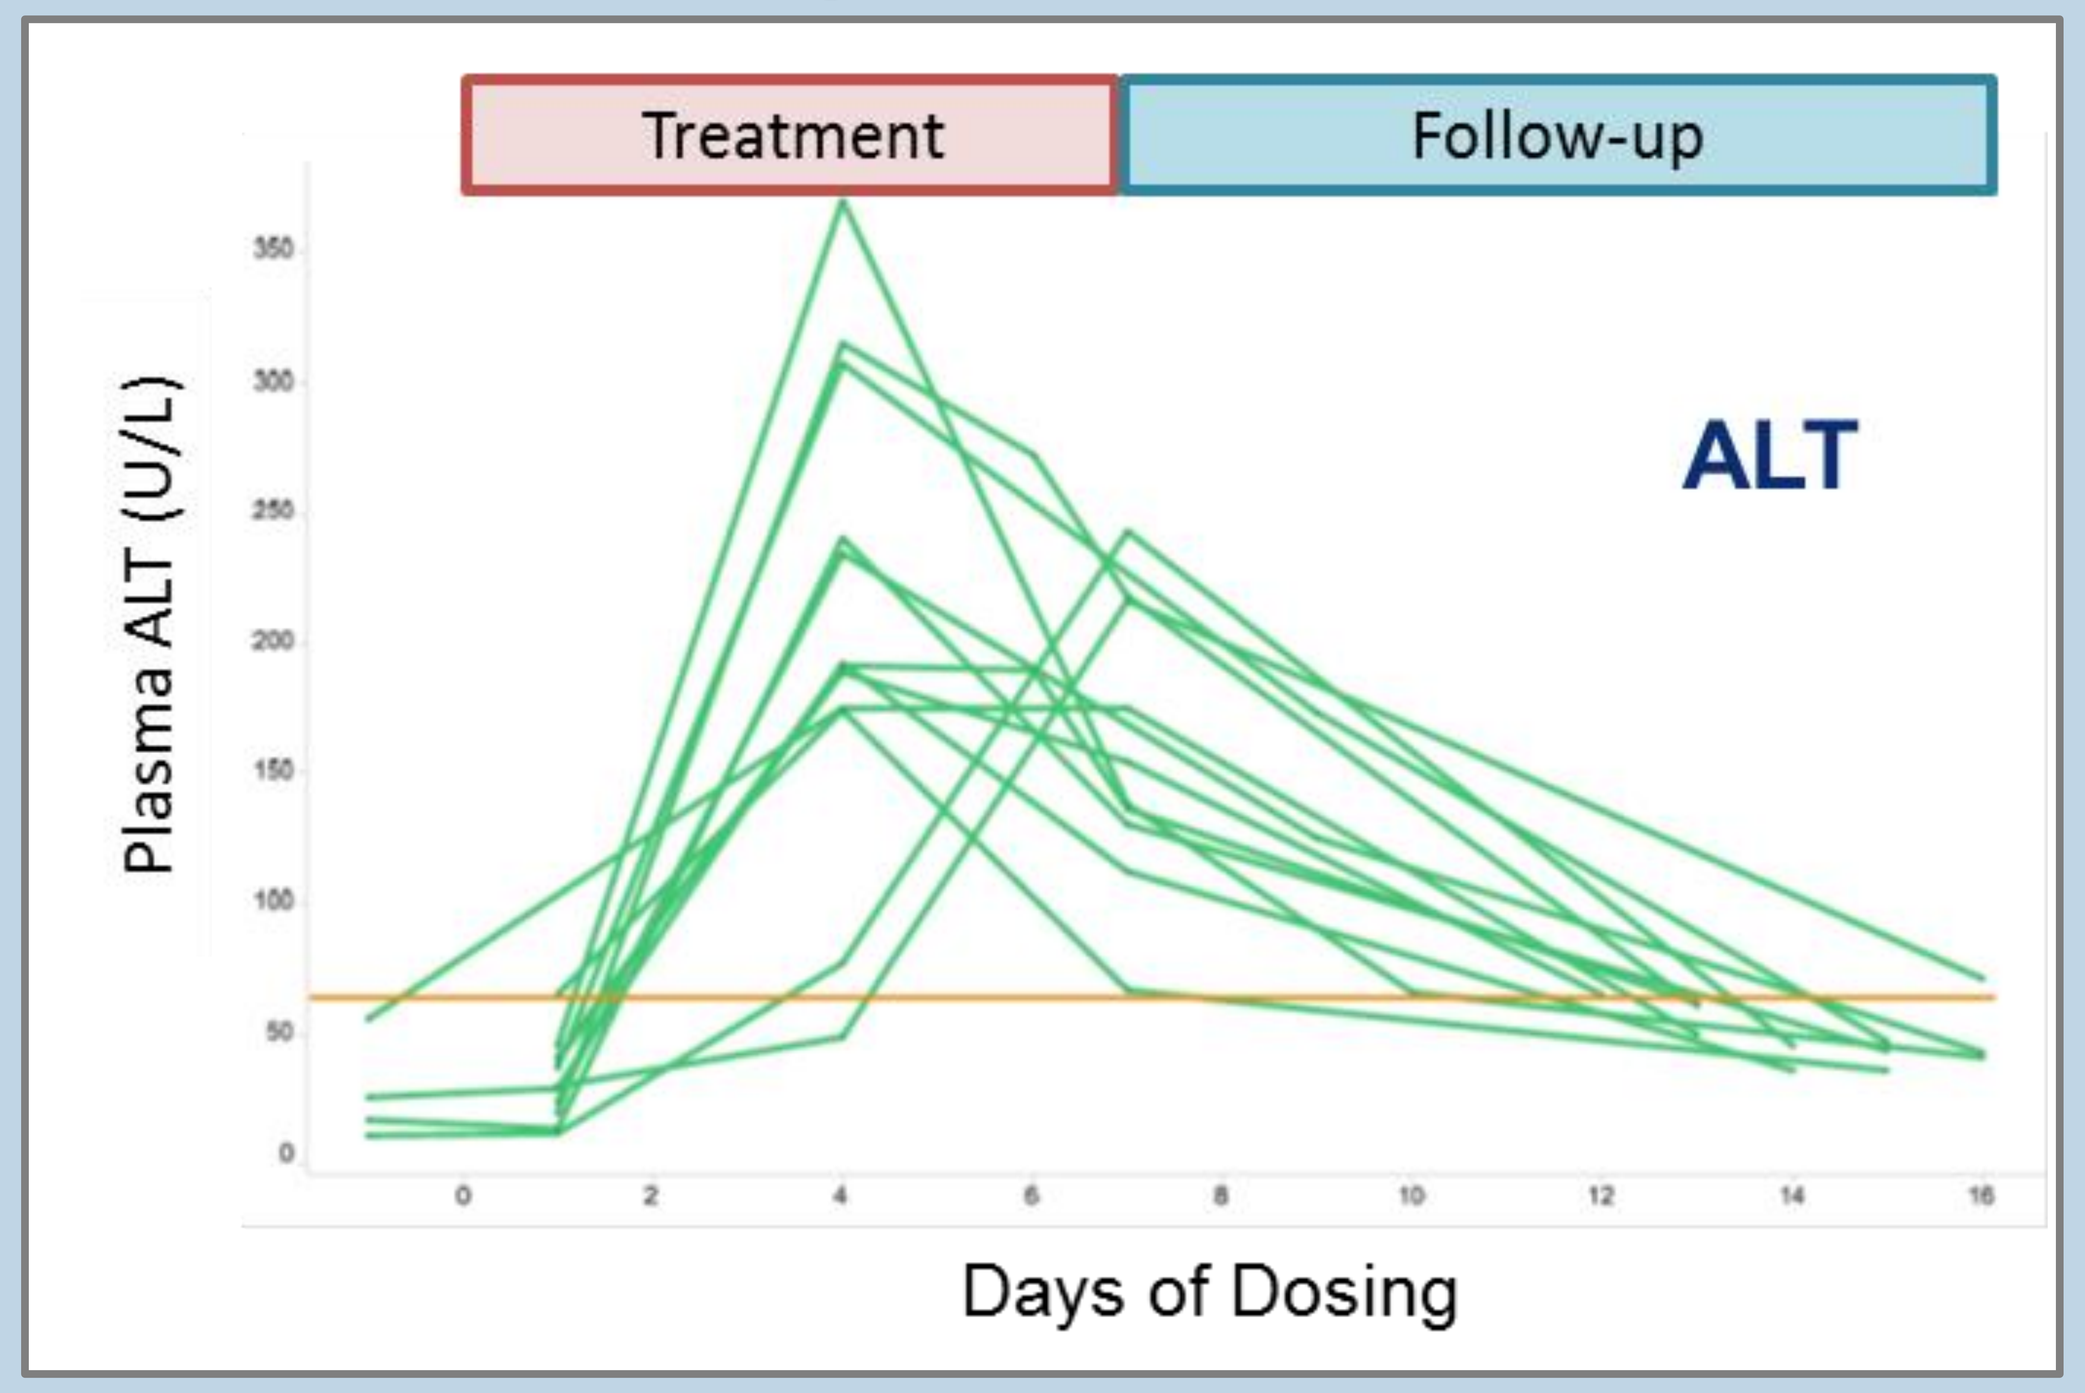

Treatment
Follow-up
ALT
Plasma ALT (U/L)
Days of Dosing

- Patients were treated with solithromycin for 7 days
- ALT was measured at baseline, on days 4 and 7, and then 5-10 days after therapy
- ALT at day 7 was less than ALT at day 4 for most patients, despite continued dosing through day 7
- ALT elevations were asymptomatic
- No bilirubin elevations were observed

Simulated Liver Enzyme Profiles in the Absence of Mitochondrial Biogenesis

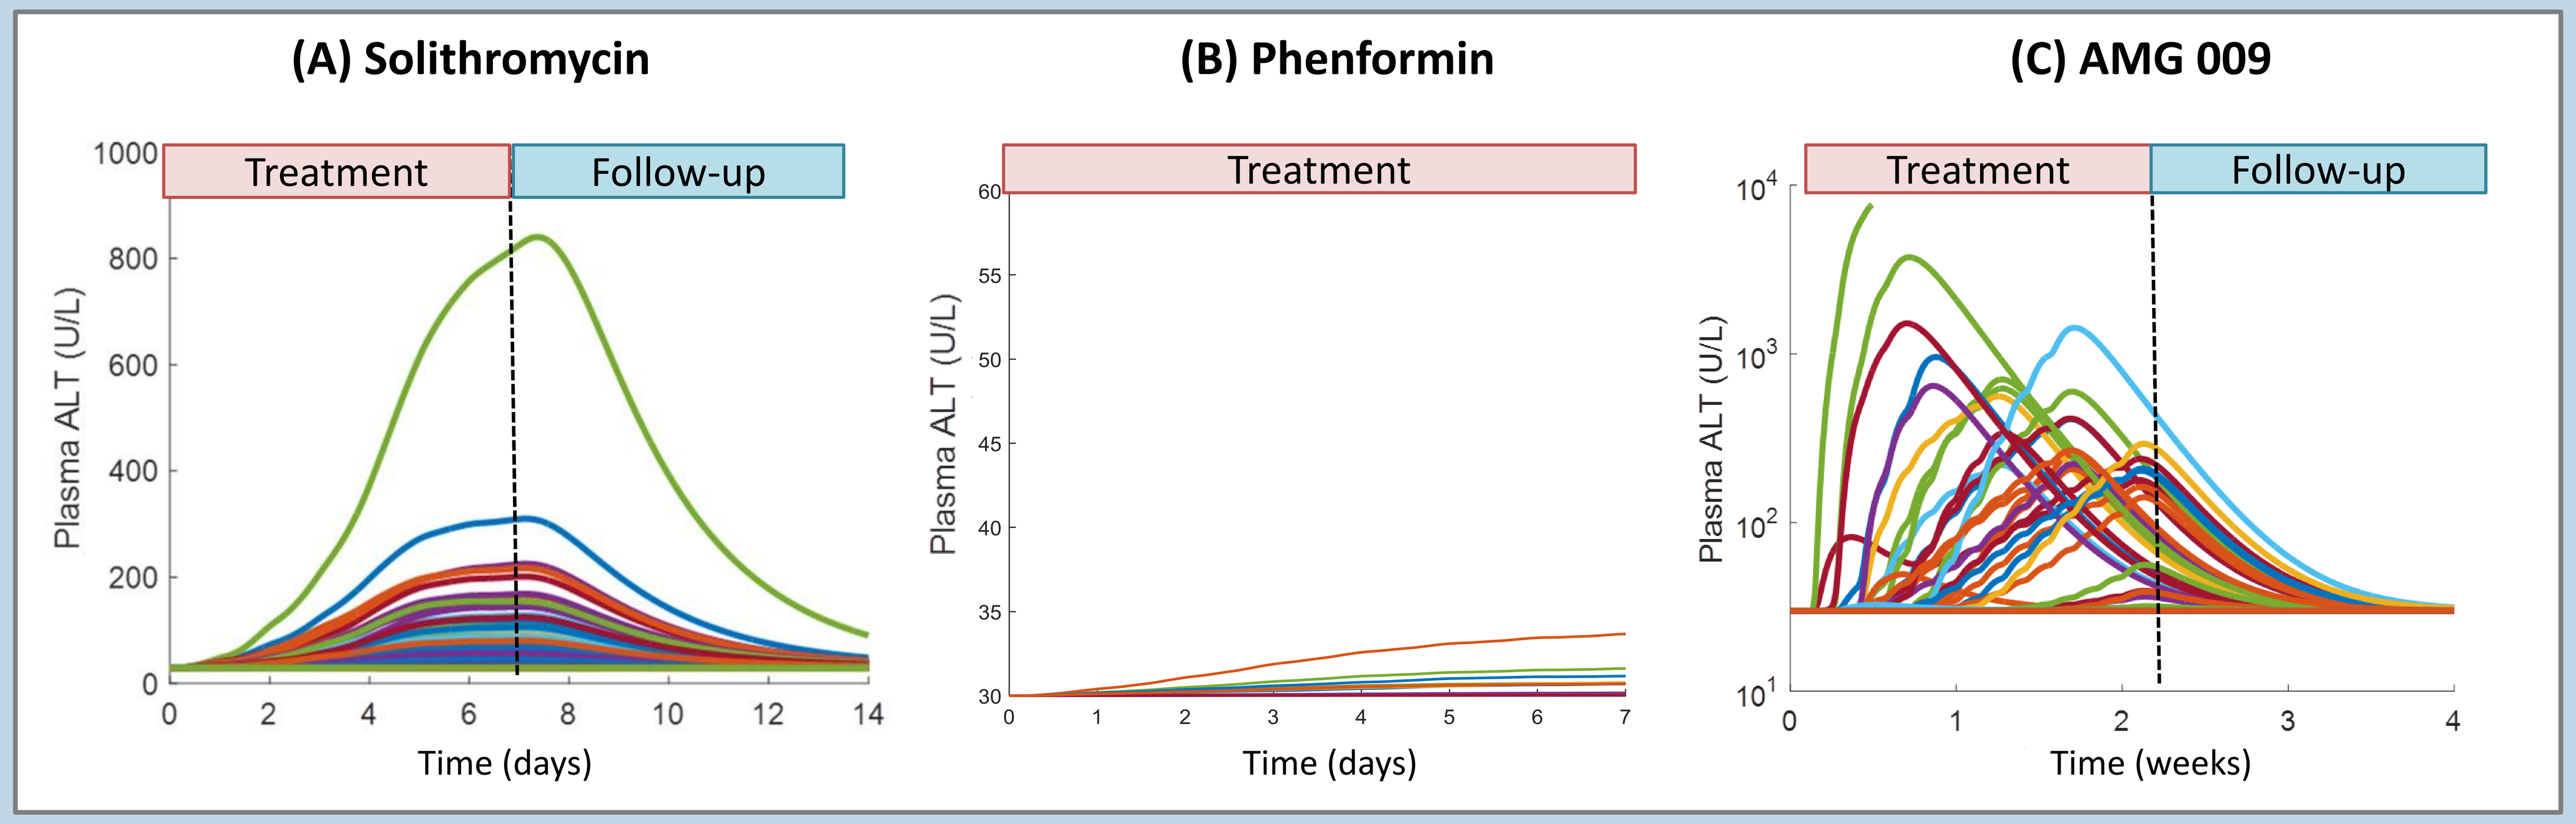

(A) Solithromycin
(B) Phenformin
(C) AMG 009
Treatment
Follow-up
Plasma ALT (U/L)
Time (days)
Time (weeks)

Simulated Liver Enzyme Profiles in the Presence of Mitochondrial Biogenesis

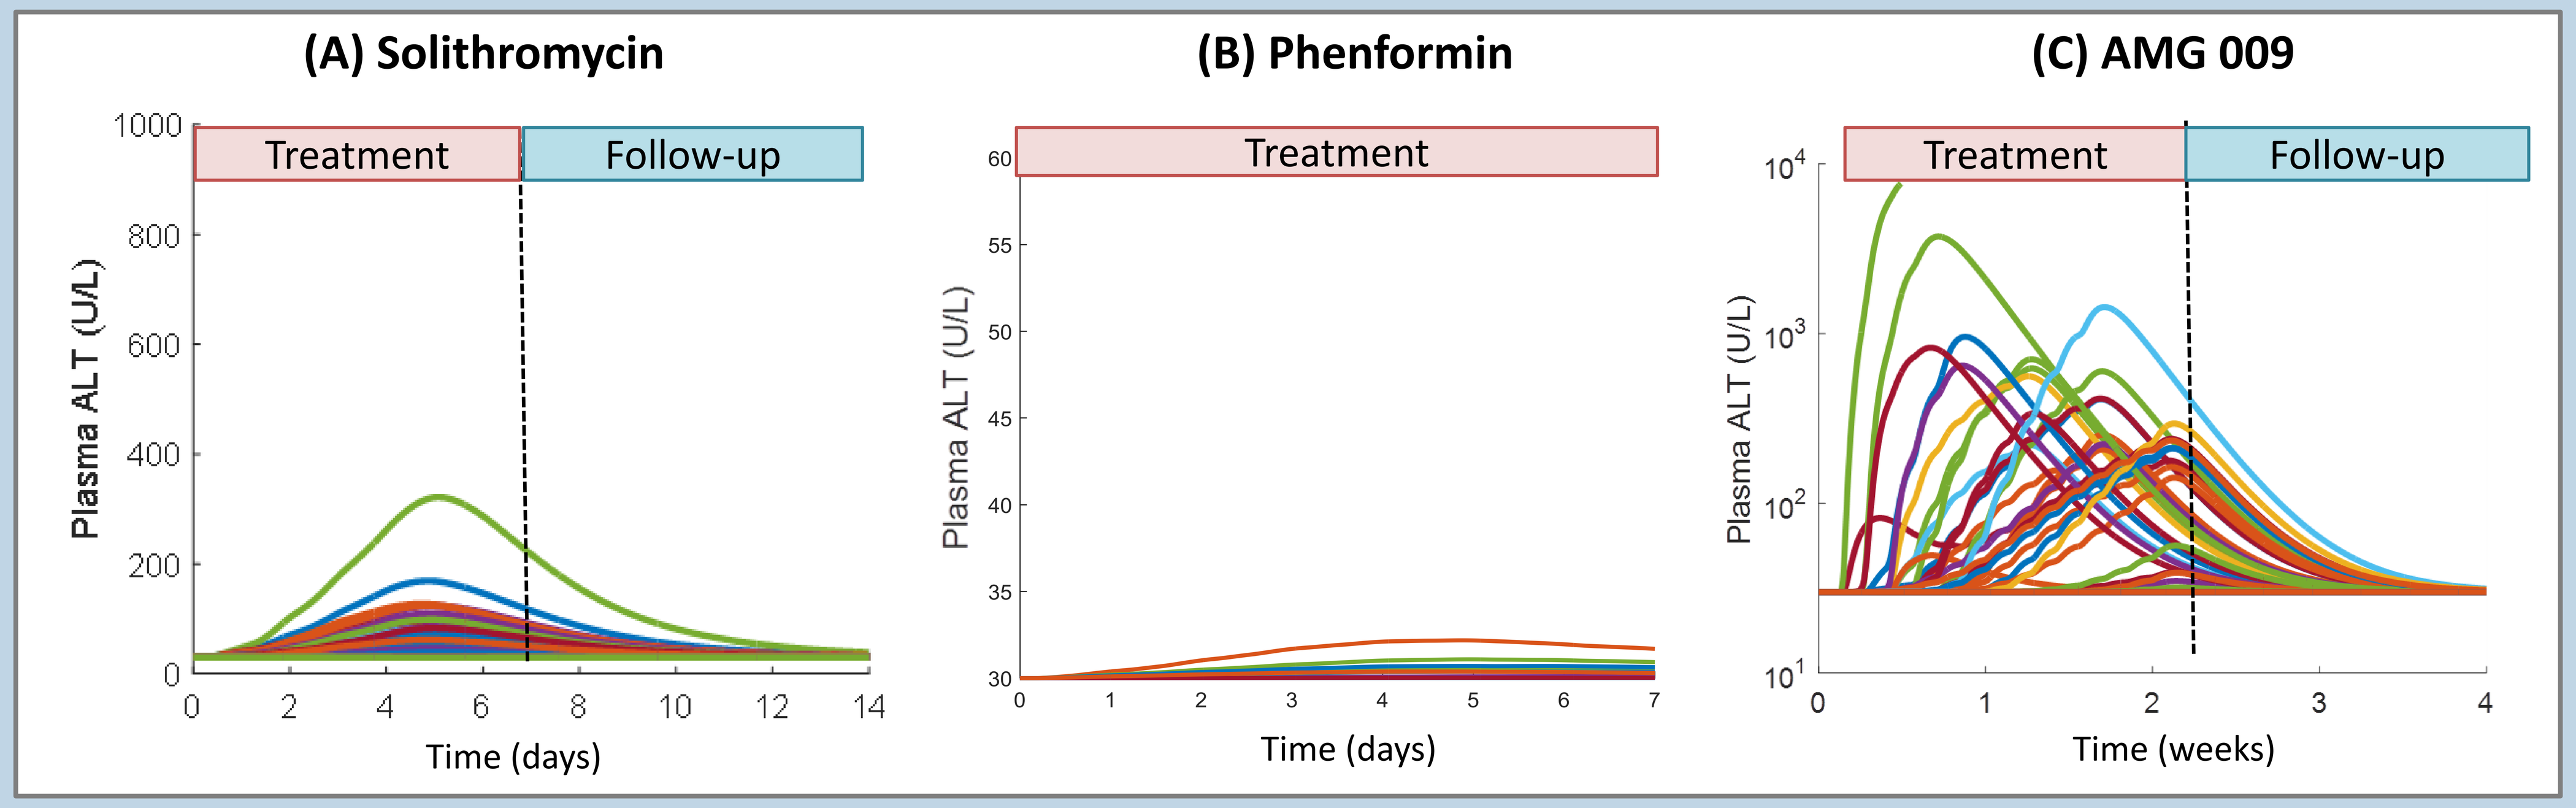

(A) Solithromycin
(B) Phenformin
(C) AMG 009
Treatment
Follow-up
Plasma ALT (U/L)
Time (days)
Time (weeks)

Simulated ALT in the human SimPops treated with (a) solithromycin, (b) phenformin, and (c) AMG 009 in the absence (top) and presence (bottom) of mitochondrial biogenesis. Liver enzyme elevations were predicted in a simulated human population (n=285) administered three compounds by combining PBPK-predicted exposure and mechanistic toxicity data. Each line represent a simulated individual. AMG 009 simulations employed a monitoring protocol to recapitulate the clinical protocol; serum ALT levels were monitored before AM dosing on days 2, 3, 4, 5, 8, 11, and 14. If ALT > 3X ULN, dosing was discontinued after 24 hr.

Solithromycin

- ALT elevations in excess of those observed clinically (see above) were predicted in a subset of individuals which was normalized after discontinuation of treatment
- With biogenesis, ALT peaks occur earlier and resolve with continuing treatment, enhancing consistency with the clinical data (see above)

Phenformin

- Early, subclinical increase in ALT is predicted in some patients without biogenesis
- Simulations of phenformin including mitochondrial biogenesis show no risk of DILI, consistent with clinical observations

AMG 009

- Significant ALT elevations are predicted in a subset of patients due to accumulating bile acids
- Mitochondrial biogenesis has minimal impact of serum ALT levels

CONCLUSIONS

- Exploratory simulations using DILIsym suggest that mitochondrial biogenesis can mitigate DILI caused by mitochondrial dysfunction
- Mitochondrial biogenesis may be one of the mechanisms underlying frequent observation of adaptation during DILI
- More *in vitro* and clinical data are needed to refine mitochondrial biogenesis parameters and relevant variability to enhance the accuracy of prospective predictions for new chemical entities

REFERENCES

[1] Aleo et al. (2014) Hepatology 60(3):1015-22.
[2] Than et al. (2011) J Biol Chem 286(25):22047-54.
[3] Hayasaka et al. (1986) Neurology 36:351-356.
[4] Sitarz et al.(2014) Mol Genet Metab 112:57-63.
[5] File et al.(2016) Clin Infect Dis 63(8):1007-1016.
[6] Shoda et al. (2014) Biopharm Drug Dispos 35(1):33-49.
[7] Yang et al. (2017) ASCPT Annual Meeting. Abstract LB-028.
[8] Yang et al. (2015) Pharm Res 32(6):1975-92.
[9] Schwerzmann et al. (1987) J Cell Biol 102:97-103.
[10] Price et al. (2012) Mole Cell Proteomics 11(12):1801-14.

ACKNOWLEDGEMENTS

- This research was supported by Cempra.

DILIsym Services
A SIMULATIONS PLUS COMPANY