P15

Integration of Pharmaceutical Product Development in Asia/Japan Into the Global Program

Tamie Bergstrom, PhD[1], Ellick Wong, PhD[2], Ed Antal, PhD[1] and Ted Grasela, PharmD, PhD[3]
[1]Pharmacia Corporation, USA; [2]Pharmacia Pte Ltd, Singapore; [3]Cognigen Corporation, USA

ABSTRACT

With increasing global acceptance of the provisions in various guideline documents resulting from the International Conferences on Harmonization (ICH) and the advent of regulatory changes in Japan/Asia Pacific region, it has become possible to integrate the requirements for pharmaceutical product registration in this region into global development programs. Multinational research-based pharmaceutical companies have begun global drug development programs more efficiently providing patients throughout the world with safe and efficacious new treatments in the shortest possible time. ICH E5 (Ethnic Factors in the Acceptability of Foreign Clinical Data) formalizes the opportunity to use “bridging” for registration of global pharmaceutical products, with clinical science as the driver.

The bridging package should be determined by what is needed to show similarity between patients in Asia/Japan and other regions. Similarity of clinical response or surrogate variable, supported by clinically meaningful argument is more relevant to treating patients than proving pharmaceutical bioequivalence. New definitions of similarity by context of drug and indication require novel approaches like population pharmacokinetics and pharmacodynamics analyses and/or inclusion of native and expatriate Japan/Asia Pacific patients in pivotal studies.

Global clinical studies have been successfully conducted in Asia, Europe and USA. The results obtained from these trials have been utilized for global registrations. Consequently, as bridging is bi-directional, the scientific basis and content of the strategy should be equally applicable to a mainly Asian/Japanese package being used for registration in the US and EU.

INTRODUCTION

The ICH E5 guidelines (Ethnic Factors in the Acceptability of Foreign Clinical Data) formalize the opportunity to use bridging for registration in Japan/Asia Pacific. The purpose of this guidance is to facilitate the registration of medicines among ICH regions by recommending a framework for evaluating the impact of ethnic factors upon medical effect (i.e., efficacy and safety at a particular development stage) that will permit adequate evaluation of the influence of ethnic factors while minimizing duplication of clinical studies and supplying medicines expeditiously to patients for their benefit.

“Similar” is the term used in the ICH E5 guideline to indicate that the difference between groups of different ethnicity is small enough to allow a valid basis for bridging. No formal definition or criteria for similarity has been given, but is defined here as follows:

> Two ethnic populations can be described as similar for a specific parameter if the difference in the parameter between the populations (including the confidence interval that covers an appropriately large proportion of the population) is such that it is likely to cause no relevant difference in clinical efficacy or safety.

The definition of similarity may be drug- and indication-specific, and is a key issue in the development of a bridging strategy.

ASSESSMENT OF THE LIKELY EASE OF BRIDGING FOR LINEZOLID

The profile shown in table 1 suggests that linezolid is a good candidate for use of a bridging strategy for global drug development.

TABLE 1. Linezolid Properties as Related to Properties Identified by ICH E5 ICH E5 Bridging Characteristics

Properties Suggesting Minimal Ethnic Sensitivity*	Linezolid Properties
Linear pharmacokinetics	At the recommended dosage, deviation from linearity is about 20%, which is not considered clinically relevant.
Flat effect-concentration curve	Bridging analysis demonstrates lack of effect of exposure on safety laboratory parameters
Minimal metabolism or metabolism distributed among multiple pathways	Neither a substrate nor an inhibitor of the major human cytochrome P-450 isomers Non-enzymatic chemical oxidation
High bioavailability	100% bioavailability No first-pass effect No significant food effect
Low potential for protein binding	Protein binding 31%
Little potential for drug-drug, drug-diet and drug-disease interactions	No cytochrome P-450-mediated drug-drug interactions Mild, reversible inhibitor of MAO-A and MAO-B; this is well-defined in terms of interacting foods and drugs.
Non-systemic mode of action	Yes--inhibits bacterial protein synthesis
Little potential for inappropriate use	Antibiotics generally considered to have low abuse potential

* From ICH E5, Appendix D

RATIONALE FOR BRIDGING STRATEGY FOR LINEZOLID

The in-depth analysis of microbiology, pharmacokinetics, and pharmacokinetic-pharmacodynamic (PK-PD) relationships both for safety and efficacy provides a strong basis for extrapolation of safety and efficacy data.

The bridging package for linezolid demonstrates the following (see Figure 1):

- Susceptibility patterns of clinical isolates from Japan/Asia are similar to overseas (US/EU) test results, including MRSA, MRSE, PRSP, and VREF.
- Pharmacokinetics and safety in healthy Japan/Asia Pacific volunteers (Phase I) are similar to those seen in healthy US/EU volunteers (Step 1).
- Pharmacokinetics and safety in healthy volunteers (Phase I) are similar to those in the patient populations (Phase II and III) (Step 2).
- Established models of pharmacokinetic-pharmacodynamic (safety and efficacy) relationships based on the global database (Phase II and III), including body weight effect analysis, simulate, predict, and extrapolate safety and efficacy outcomes (Step 3). These simulations demonstrate the ability to extrapolate clinical safety and efficacy outcomes in Japan/Asia Pacific patients.

FIGURE 1. Diagram of bridging strategy.

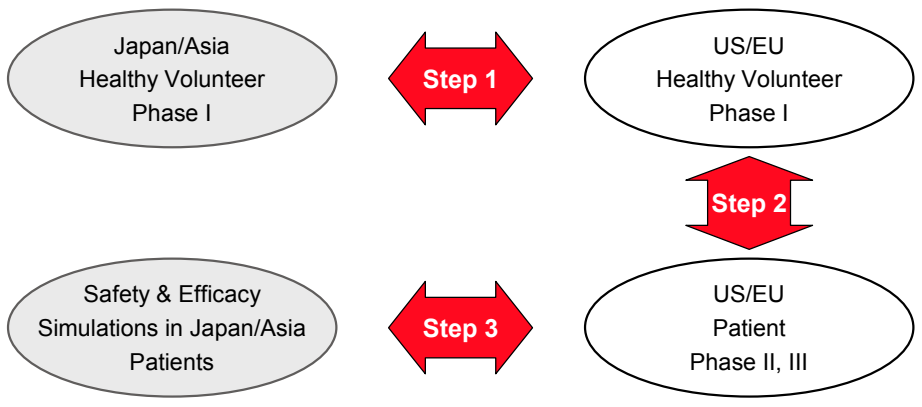
Japan/Asia
Healthy Volunteer
Phase I
Step 1
US/EU
Healthy Volunteer
Phase I
Step 2
Safety & Efficacy
Simulations in Japan/Asia
Patients
Step 3
US/EU
Patient
Phase II, III

SUMMARY OF RESULTS

- **Susceptibility patterns of clinical isolates from Japan/Asia Pacific were similar to overseas (US/EU) test results, including results for MRSA, MRSE, PRSP, and VREF.**
- **Pharmacokinetics and safety in Japan/Asia Pacific healthy volunteers (Phase I) were similar to those seen in US/EU healthy volunteers (Step 1).**
- **Pharmacokinetics and safety in healthy volunteers (Phase I) are similar to those in the patient populations (Phase II and III) (Step 2).**
- **Established models of pharmacokinetic-pharmacodynamic (safety and efficacy) relationships based on the global database (Phase II and III), including body weight effect analysis, simulate/predict/extrapolate safety and efficacy outcomes in Japan/Asia Pacific patients (Step 3).**
 - We demonstrated 1) the similarity of safety experience in Japan/Asia Pacific versus US/EU volunteers given the same linezolid exposure and 2) the similarity of safety experience with linezolid in volunteers versus patients.
 - We conclude that Japan/Asia Pacific patients with similar exposure to linezolid will have the same safety experience as that observed for patients in the global database.
 - The analysis of body-weight effect allows us to conclude that the hematological and hepatic safety profile of linezolid 600 mg twice daily is the same for low-weight patients and the database as a whole.
 - In order to compare the expected efficacy of linezolid at the 500-mg and 600-mg twice-daily dose regimens in Japan/Asia Pacific patients, population pharmacokinetic and pharmacodynamic simulations were performed.
 - For the 60-kg patients, thought to be typical of the body size observed in Japan/Asia Pacific patients, the efficacy rate for linezolid with 500 mg administered twice daily is expected to be 72.1%. By increasing the dose to 600 mg twice daily, the expected cure rate increases to 76.7%. Based on the 95% confidence intervals about the cure rate, this difference is statistically significant ($p<0.05$).
- **The linezolid clinical program has encompassed a wide range of environmental factors, which strengthens the validity of a bridging strategy.**
 - Environmental factors also need to be considered when evaluating the potential for use of a bridging strategy. Environment effects include differences in medical practices, diet, disease state, disease definition, and co-administered drug.
 - This bridging analysis is based on a large, high-quality, international and multicultural experience with linezolid. The Phase II program was conducted primarily in the United States and included linezolid administration to over 600 White and over 150 Black patients. The Phase III program included linezolid administration to over 1400 White patients, over 200 Black patients, and approximately 125 Asian patients.
 - The program was multi-national, with patients enrolled in centers in North America (>900 linezolid-treated patients), Europe (>600 linezolid-treated patients), Latin America (>300 linezolid-treated patients), and Asia (>100 linezolid-treated patients). The Phase II and III experience includes linezolid administration to more than 800 patients over age 65 years and over 1100 women. As such, the linezolid NDA represents a wide-ranging experience in several races, both sexes, a wide age range, and many social and medical cultures throughout the world.
 - All of the studies were 1) pre-submitted to the US IND for FDA review, 2) conducted under the laws and regulations set forth by the US FDA and GCPs for clinical trials at the time they were conducted and 3) submitted to and accepted by the US FDA in the US NDA. As such, this analysis represents a high-quality experience over a wide range of social and medical environments.

CONCLUSIONS

- **The in-depth analysis of microbiology, pharmacokinetics, and pharmacokinetics-pharmacodynamic (PK-PD) relationships both for safety and efficacy provides a strong basis for extrapolation of safety and efficacy data.**
- **The linezolid NDA represents a wide-ranging experience in several races, both sexes, a wide age range, and many social and medical cultures throughout the world.**
- **These bridging strategy analyses and simulations, based on established PK-PD models and using parameters representative of Japan/Asia Pacific patients, strongly demonstrate the similarities among multiethnic populations and the ability to extrapolate safety and efficacy clinical outcomes.**
- **Thus, Pharmacia believes the complete data package meets ICH E5 guidance for “Ethnic Factors in the Acceptability of Foreign Clinical Data”.**