F-009

Presented at the
Society for Gynecologic Investigation 60th Annual Scientific Meeting
Orlando, FL • March 20-23, 2013

Pharmacokinetic Profile of an Ascending-Dose, Estrogen/Progestin Combination Oral Contraceptive

Mona Darwish, PhD[1]; Mary Bond, MS, MBA[1]; Nancy Ricciotti, MSN[2]; Jennifer Hsieh, MS[3]; Jill Fiedler-Kelly, MS[4]; Ted Grasela, PharmD, PhD[4]

[1]Clinical Pharmacology and Experimental Medicine, Teva Pharmaceuticals, Inc; [2]R&D, Teva Pharmaceuticals, Inc; [3]Global Biostatistics, Teva Pharmaceuticals, Inc; [4]Pharmacometric Services, Cognigen Corp.

ABSTRACT

Introduction: The estrogen component of oral contraceptives provides stability to the endometrium so that irregular shedding and unwanted breakthrough bleeding are minimized. An ascending-dose, extended-regimen ethinyl estradiol/levonorgestrel (EE/LNG) combination oral contraceptive (ADER) has been developed with the intent of providing better protection against unscheduled bleeding (UB) or spotting (US). It is hypothesized that gradual increases in EE may be more effective in preventing UB/US than sustained low levels of estrogen. The stepwise increase may also be better suited to preventing these events as compared to persistent high levels of EE which may desensitize the estrogen receptors. The current analysis characterizes the pharmacokinetics of EE following administration of ADER.

Methods: A population pharmacokinetic (PK) model was used to predict EE plasma concentrations over time following administration of ADER to non-pregnant women of childbearing age. ADER consists of the following: LNG, 150 mcg for 84 days with EE, 20 mcg Days 1-42; 25 mcg Days 43-63; 30 mcg Days 64-84; 10 mcg EE Days 85-91.

Results: The model-predicted plasma concentration-time profiles demonstrated that there is a step-wise increase in systemic exposure to EE with increase in dose over the first 84 days of the cycle as assessed by steady-state C_{max}, C_{min}, and AUC prior to each EE dose change. PK profiles of ADER on the final day prior to each EE dose change showed that C_{max} rose from 55.77 pg/mL at day 42 to 69.72 pg/mL at day 63 and 83.66 pg/mL at Day 84 while C_{min} increased from 9.67, to 12.08, and 14.5 pg/mL at Days 42, 63, and 84, respectively. These increases are followed by lower concentrations of EE during the 7-day period of low-dose estrogen (10 mcg) which replaces the typical hormone-free interval at the end of the cycle. Predicted C_{max} and C_{min} values during the final 7 days of the cycle were 27.92 pg/mL and 4.85 pg/mL, respectively.

Conclusions: The increasing dose of EE in ADER results in a step-wise increase in systemic exposure to EE over the 84-day active-treatment period followed by lower EE concentrations. This changing concentration of EE may provide better protection against UB/US than sustained EE doses.

INTRODUCTION

- Unscheduled bleeding with extended oral contraceptive (OC) regimens is a common occurrence, and although the incidence decreases over time with therapy,[1-3] it may contribute to treatment discontinuation.[4]
- Studies have found that the unscheduled spotting/bleeding associated with extended regimens tends to occur between days 43 and 56.[2,5]
- In addition, the hormone free interval (HFI) of OC has been associated with fluctuations in estradiol levels, which may destabilize the endometrial lining[4,6] and increase the risk of breakthrough bleeding.
- It has also been proposed that the higher consistent levels of EE in extended OC regimens may desensitize or downregulate estrogen receptors,[7,8] which could contribute to unscheduled bleeding.
- An investigational ascending-dose, extended-regimen (ADER) EE/levornorgestrel (LNG) combination oral contraceptive has been developed consisting of a gradually increasing estrogen dose combined with a continuous low progestin (LNG) dose for 84 days followed by a low dose of EE for 7 days.
- This design also provides a ratio of estrogen to progestin that becomes relatively more estrogenic across the extended cycle, with the estrogen increasing at specific times in the cycle prior to the time when breakthrough bleeding tends to be experienced by the greatest proportion of women using extended-regimen OCs.
- The use of a low dose of EE during the last 7 days of the cycle allows for a low circulating level of estrogen that replaces the traditional 7-day HFI.
 – Enhancing ovarian suppression by using estrogen during the usual HFI may improve efficacy and increase cycle control.[4,9]
- The pharmacokinetics of EE and LNG are well studied, and while the ADER uses comparable doses of LNG as other extended-regimen OCs, the pharmacokinetics of EE, on the other hand, have not been assessed in the context of the new 91-day ADER.
- The objectives of this analysis, which used a population model approach, were to characterize the pharmacokinetic profile of EE following administration of the ADER including absorption, distribution, metabolism, and elimination; assess dose proportionality, single, and multiple dose EE pharmacokinetics; and compare the EE pharmacokinetic profile of ADER with that of the same EE from extended-regimen products.

METHODS

Subjects

- Participants in the pooled studies included healthy women of childbearing potential between the ages of 18 to 51 years (**Table 1**).

Table 1. Description of included studies

Study	Dosing regimen	Pharmacokinetic Sampling	Subjects*	Patients
ADER	Days 1-42: EE 20 mcg/ LNG 150 mcg Days 43-63: EE 25 mcg/ LNG 150 mcg Days 64-84: EE 30 mcg/ LNG 150 mcg Days 85-91: EE 10 mcg	Predose (0) and at 0.5, 1, 1.25, 1.5, 1.75, 2, 2.5, 3, 4, 6, 8, 10, 12, 16, 24, 36, 48, 72, and 96 hours after dose	n=18	• Healthy, non-tobacco using adult female patients with normal menstrual cycle • 18-45 years old • BMI 18-30 kg/m²
Seasonale	Days 1-84: EE 30 mcg/ LNG 150 mcg Days 85-91: placebo	Predose (0) and at 0.5, 1.0, 1.25, 1.5, 1.75, 2, 2.5, 3, 4, 6, 8, 10, 12, 16, 24, 36, 48, 72, and 96 hours after dose	n=29	• Healthy female adult patients • 18-35 years old • Within 15% of their ideal body weight
Seasonique	Days 1-84: EE 30 mcg/ LNG 150 mcg Days 85-91: EE 10 mcg	Days 1, 21: predose (0) and at 0.5, 1, 1.33, 1.67, 2, 3, 4, 6, 8, 11, 15, and 24 hours after dose Day 84: predose (0) and at 0.5, 1, 1.33, 1.67, 2, 3, 4, 6, 8, 11, 15, 24, 36, 48, 72, 96, 120, and 144 hours after dose Day 91: predose (0) and at 0.5, 1, 1.33, 1.67, 2, 3, 4, 6, 8, 11, 15, 24, 36, 48, 72, and 96 hours after dose	n=30†	• Healthy female adult patients with normal menstrual cycle • 19-51 years old • 119-191 lb
Seasonique	Days 1-84: EE 30 mcg/ LNG 150 mcg Days 85-91: EE 10 mcg after dose (LNG only)	Predose (0) and at 0.33, 0.67, 1, 1.33, 1.67, 2, 2.5, 3, 4, 6, 8, 12, 16, 24, 36, 48, and 72 hours after dose (EE and LNG) 96 and 120 hours after dose (LNG only)	n=29	• Healthy, non-tobacco using, female adult patients with normal menstrual cycle • 19-51 years old • BMI 18-30 kg/m²
Seasonique	Days 1-84: EE 30 mcg/ LNG 150 mcg Days 85-91: EE 10 mcg	Predose (0) and at 0.33, 0.67, 1, 1.33, 1.67, 2, 2.5, 3, 4, 6, 8, 10, 12, 16, 24, 36, 48, 72, and 96 hours after dose	n=17	• Healthy female patients • 18-47 years old • 53-87 kg
LoSeasonique	Days 1-84: EE 20 mcg/ LNG 100 mcg Days 85-91: EE 10 mcg	Predose (0) and at 0.5, 1.0, 1.25, 1.5, 1.75, 2, 2.5, 3, 4, 6, 8, 10, 12, 16, 24, 36, 48, 72, and 96 hours after dose	n=30	• Healthy, adult female patients with regular menstrual cycle • 18-35 years old • 48-75 kg

*All studies assessed single-dose pharmacokinetics with the exception of 1 Seasonique study†, which included both multidose and single-dose assessments.

Study Design

- Because the oral bioavailability of EE when administered as various extended-regimen OCs were comparable to that of ADER, which contains the same EE in a different dosing regimen, pooled data across studies were used to characterize the pharmacokinetics of EE when administered within ADER.
- Extensive pharmacokinetic sampling was performed in all studies prior to and up to 144 hours after drug administration (**Table 1**).
- The population pharmacokinetic analysis was completed using nonlinear mixed effects models (NONMEM) software, version 6, level 2.0 (ICON Development Solutions 2006).

RESULTS

- The population PK analysis included data from 152 female patients of childbearing age (**Table 1**).
- Systemic exposure at steady-state was approximately 20% greater than single-dose assessment (**Figure 1**).

Figure 1. Mean predicted single-dose and steady-state plasma concentration by time profiles of EE following administration of a 20 mcg dose

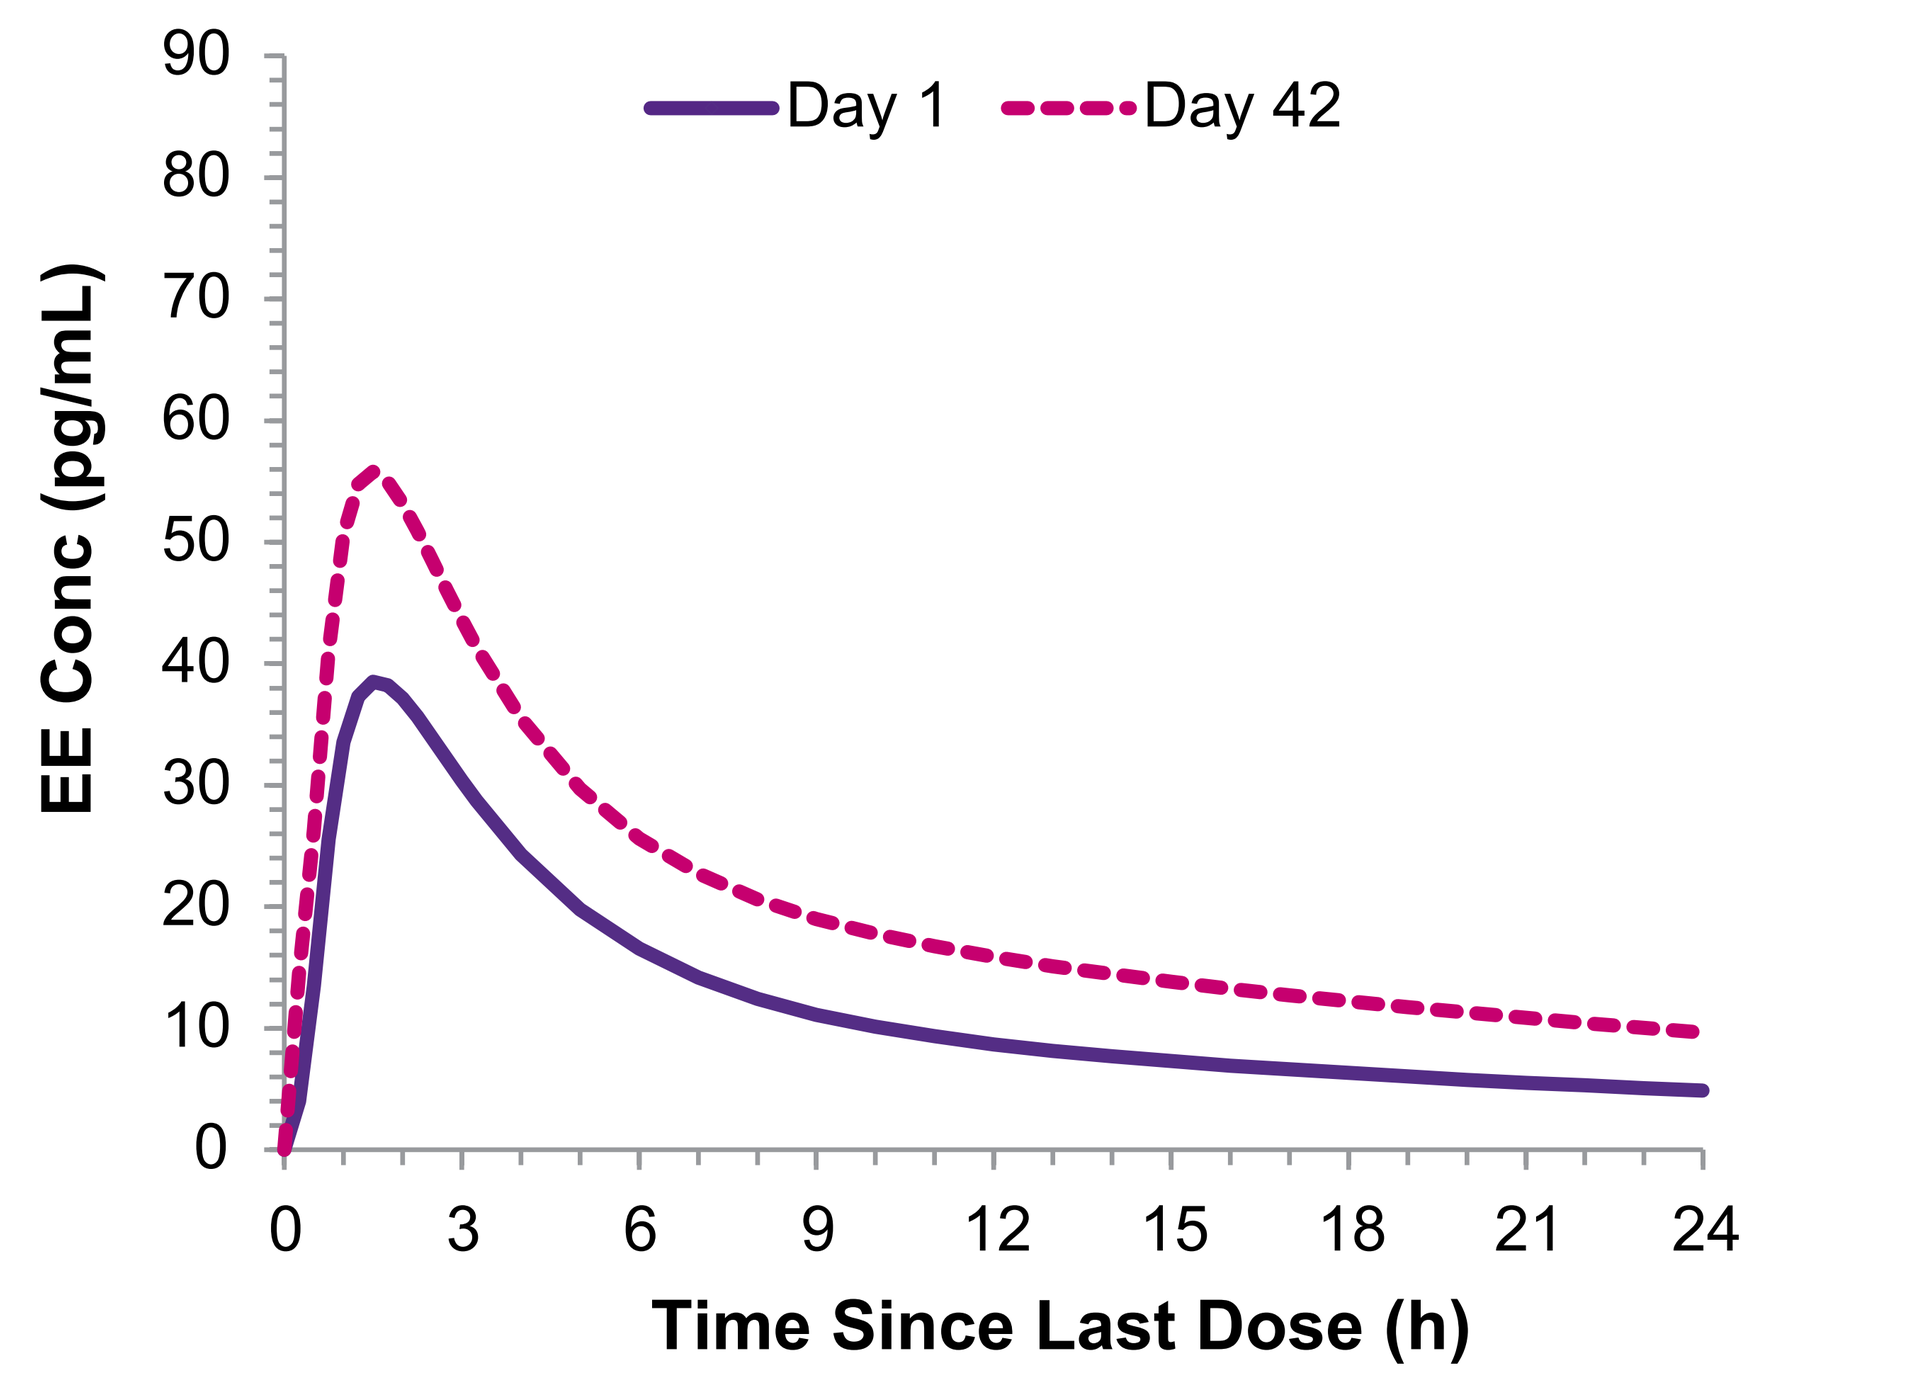
Day 1
Day 42
EE Conc (pg/mL)
90
80
70
60
50
40
30
20
10
0
0
3
6
9
12
15
18
21
24
Time Since Last Dose (h)

- Following the 20 mcg EE dose of ADER, EE was rapidly absorbed, with a maximum plasma concentration attained within 2 hours and the decline from peak occurring in a biphasic manner (**Figure 1**).
- A cross-study comparison of systemic exposure following intravenous administration of EE and administration of ADER resulted in an EE oral bioavailability of 40% following ADER administration.
- EE was well distributed with a mean steady-state volume of distribution of approximately 873 L (**Table 2**).
- Metabolism of orally administered EE involves the formation of ethinyl estradiol-3-sulfate in the gut wall, as well as 2-hydroxylation via the cytochrome P450 3A4 enzyme (CYP3A4).
- EE is excreted in the urine and feces as glucuronide and sulfate conjugates.
- The terminal elimination half-life was 16.5 and 17.8 hours following single and multiple doses of ADER, respectively (**Table 2**).

Table 2. Mean predicted single-dose and steady-state pharmacokinetic parameters of EE following administration of 20 mcg EE dose within ADER

Parameter (unit)	Single-dose	Steady-state
AUC_{inf} (pg•h/mL)	464	NA
AUC_{0-24} (pg•h/mL)	NA	500
C_{max} (pg/mL)	40	55.8
t_{max} (h)	1.66	1.59
V_{ss} (L)	NA	873
$t_{½}$ (h)	16.5	17.8

AUC_{inf}=area under the plasma concentration by time curve from time 0 to infinity; AUC_{0-24}= area under the plasma concentration time curve from 0 to 24 hours; C_{max}=maximum observed plasma drug concentration; t_{max}=time to maximum observed plasma drug concentration; V_{ss}=volume of distribution at steady state; $t_{½}$ =elimination half-life; NA=not applicable.

- As determined from the population analysis, systemic exposure (AUC) increased in a dose-proportional manner over the dose range of 10 to 60 mcg indicating there was no relationship between EE dose and clearance.
- The model-predicted steady-state plasma concentration by time profiles of EE depicted in **Figure 2** are representative of the daily profiles for each dose of EE within ADER.

Figure 2. Model-predicted steady-state plasma concentration by time profiles of EE prior to each change in EE dose during administration of ADER (20 mcg [day 42], 25 mcg [day 63], 30 mcg [day 84], and 10 mcg [day 91])

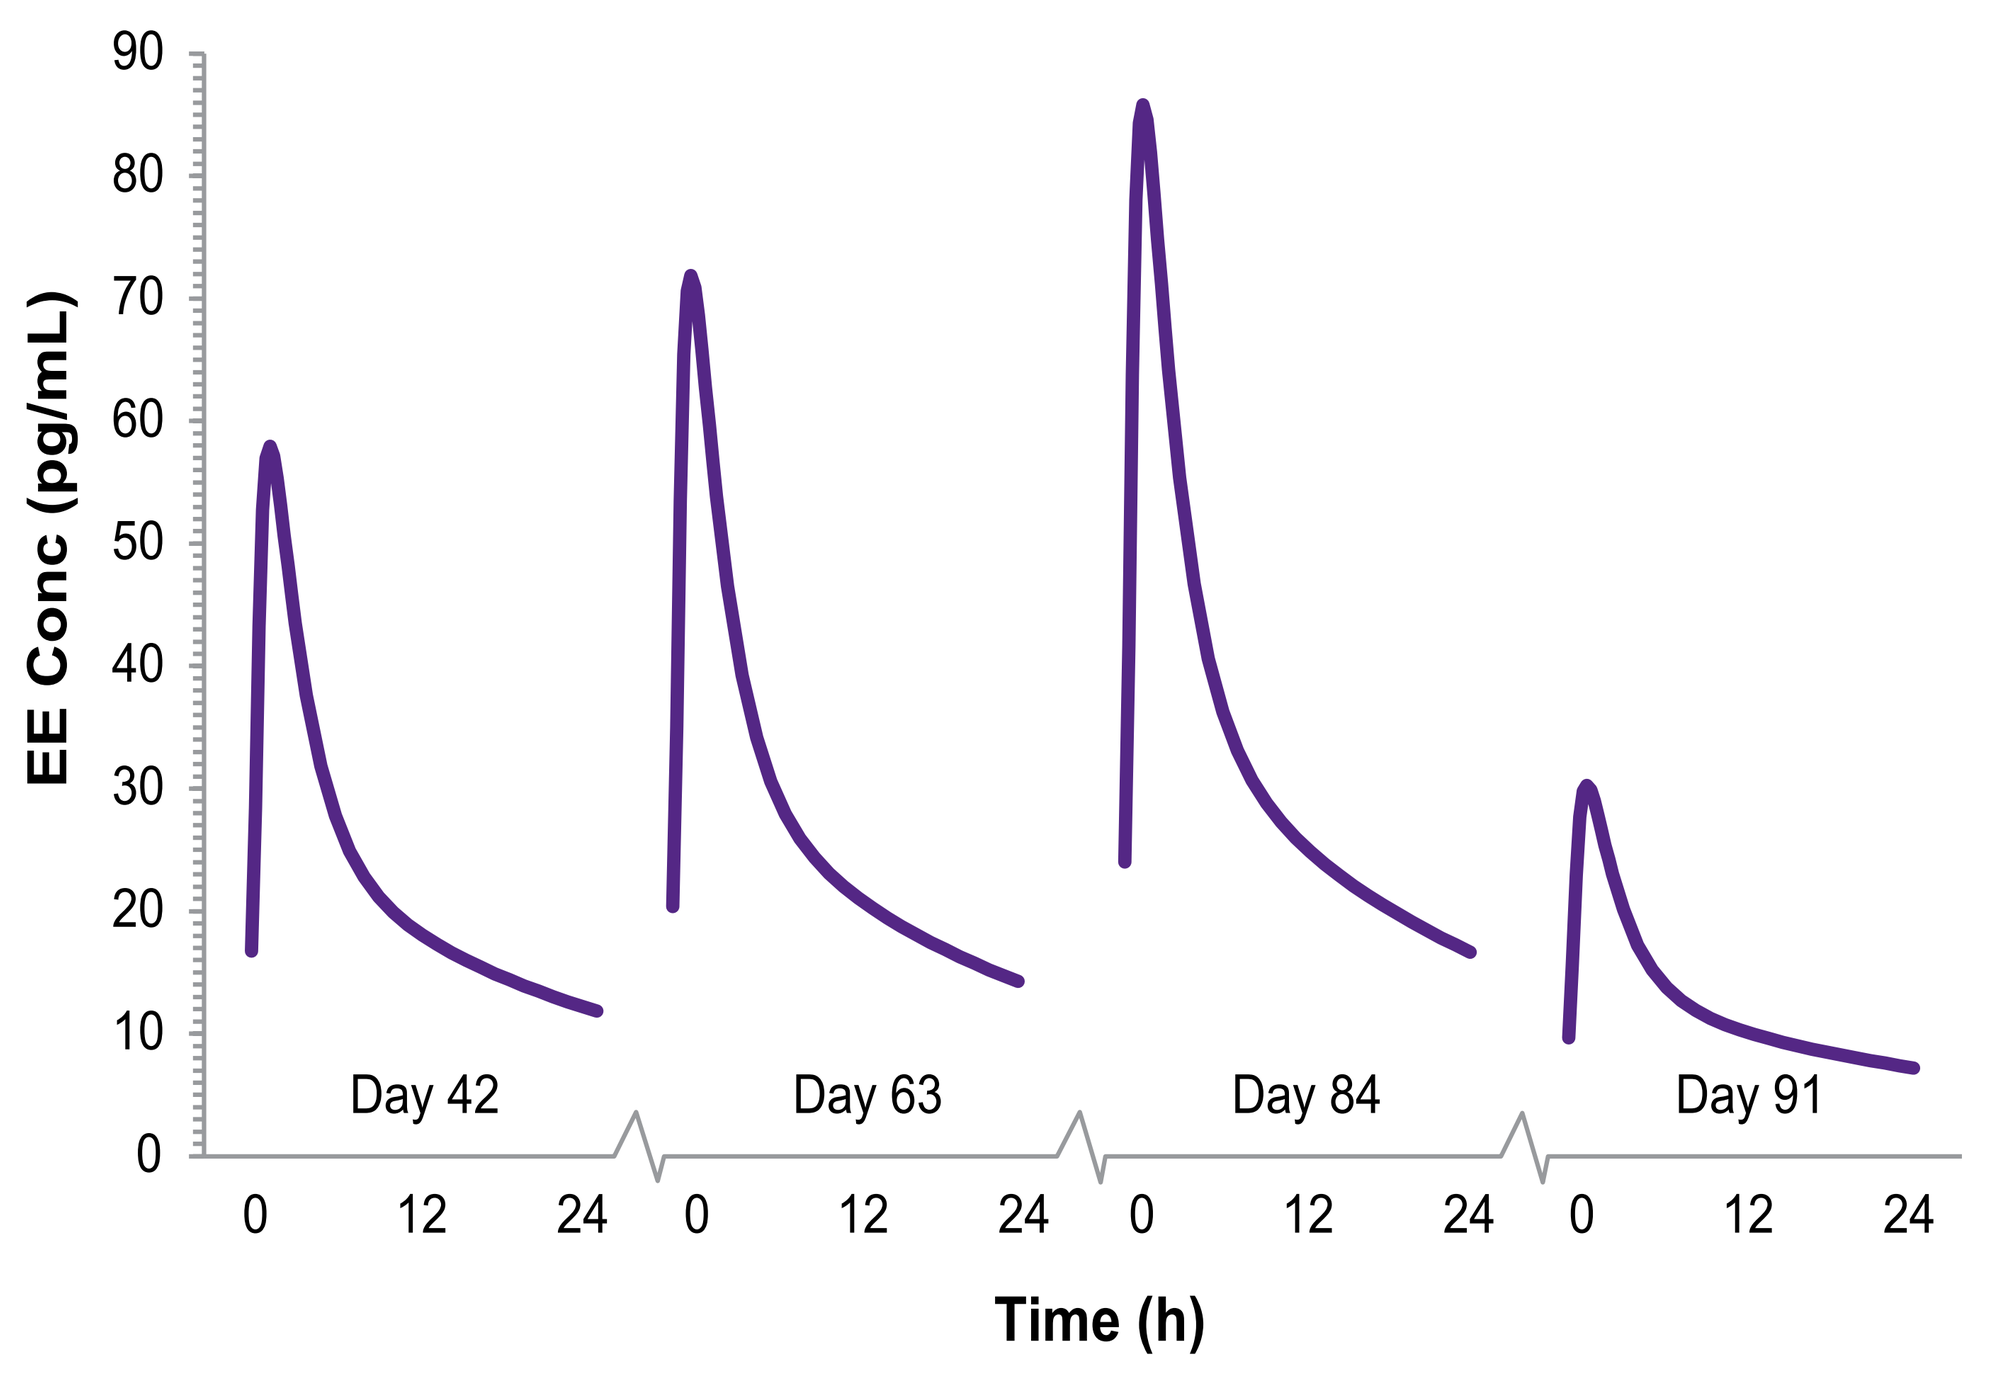
EE Conc (pg/mL)
90
80
70
60
50
40
30
20
10
0
Day 42
Day 63
Day 84
Day 91
0
12
24
Time (h)

- The increases in exposure occur in a stepwise fashion with increasing EE dose over the first 84 days of the extended cycle followed by lower concentrations of EE with the 10-mcg daily dose of estrogen that replaces the traditional 7-day HFI at the end of the cycle.
- The population PK model was also used to compare and contrast the pharmacokinetics of ADER with Seasonique and LoSeasonique over the entire 91-day cycle length.
- Predicted measures of EE exposure during ADER administration changed over the initial 84 day cycle period, in contrast the same measures of EE exposure remained constant for the other extended-regimen OCs.

CONCLUSIONS

- Single- and multiple-dose pharmacokinetic parameters of EE following administration of 20 mcg ADER were consistent with the known profile of EE.
- Systemic exposure to EE following ADER administration increases proportionally with an increase in dose with steady-state plasma concentrations attained prior to day 21.
- Increases in systemic exposure occur in a stepwise fashion, with increasing EE dose over the first 84 days of the extended cycle followed by lower concentrations of EE with the 10-mcg daily dose of estrogen, which replaces the traditional 7-day HFI at the end of the cycle.
- ADER contains approximately 20% lower total hormonal content compared to the currently approved extended-dose regimen, Seasonique.
- The gradually increasing dose of EE within ADER may provide the benefit of reduced breakthrough bleeding or spotting compared with sustained levels of EE found in traditional extended regimens with constant dose EE, which may desensitize or downregulate the estrogen receptors of the endometrium.[7,8]
- Further studies are needed to investigate the incidence of unscheduled bleeding with ADER and to substantiate the pharmacokinetic association between ascending dose and monophasic extended OC regimens and the incidence of unscheduled bleeding.

Acknowledgements

This study was sponsored by Teva Women's Health, a division of Teva Branded Pharmaceutical Products R&D, Inc. Funding for development, editorial, design, and production support was provided by Teva Women's Health to MedVal Scientific Information Services, LLC, Skillman, NJ.

REFERENCE LIST

1. Sillem M, Schneidereit R, Heithecker R, Mueck AO. Use of an oral contraceptive containing drospirenone in an extended regimen. *Eur J Contracept Reprod Health Care*. 2003;8:162-169.
2. Anderson FD, Hait H. A multicenter, randomized study of an extended cycle oral contraceptive. *Contraception*. 2003;68:89-96.
3. Miller L, Notter KM. Menstrual reduction with extended use of combination oral contraceptive pills: randomized controlled trial. *Obstet Gynecol*. 2001;98:771-778.
4. Kaunitz AM, Portman DJ, Hait H, Reape KZ. Adding low-dose estrogen to the hormone-free interval: impact on bleeding patterns in users of a 91-day extended regimen oral contraceptive. *Contraception*. 2009;79:350-355.
5. Stewart FH, Kaunitz AM, LaGuardia KD, Karvois DL, Fisher AC, Friedman AJ. Extended use of transdermal norelgestromin/ethinyl estradiol: a randomized trial. *Obstet Gynecol*. 2005;105:1389-1396.
6. Endrikat J, Gerlinger C, Plettig K et al. A meta-analysis on the correlation between ovarian activity and the incidence of intermenstrual bleeding during low-dose oral contraceptive use. *Gynecol Endocrinol*. 2003;17:107-114.
7. Brown TJ, Scherz B, Hochberg RB, MacLusky NJ. Regulation of estrogen receptor concentrations in the rat brain: effects of sustained androgen and estrogen exposure. *Neuroendocrinology*. 1996;63:53-60.
8. Klinge, C. M. and Rao, V. The steroid hormone receptors. In: Hiscock T, et al, eds. Gynecology & Obstetrics. Volume 5: Reproductive endocrinology, infertility, and genetics. Available at: http://www.glowm.com/resources/glowm/cd/pages/v5/v5c004.html.
9. Reape KZ, DiLiberti CE, Hendy CH, Volpe EJ. Effects on serum hormone levels of low-dose estrogen in place of placebo during the hormone-free interval of an oral contraceptive. *Contraception*. 2008;77:34-39.