Effect of Lifestyle Intervention on the Placebo Response in Anti-Obesity Trials: Considerations for Exposure-Response Modeling

Brenda Cirincione[1], Julie Passarell[1], Melannie Cummings[1], Elizabeth Ludwig[1], Ted Grasela[1], Amy Halseth[2], Mark Fineman[2]

[1]Cognigen Corporation, Buffalo, NY; [2]Amylin Pharmaceuticals, Inc., San Diego, CA

Abstract (revised)

BACKGROUND: Successful clinical development of anti-obesity agents is aided by characterization of the placebo effect, which can be significant when lifestyle intervention (LSI) accompanies pharmacologic therapy. Understanding the time course and dynamics of the placebo effect is important when developing pharmacodynamic (PD) models that isolate drug effects. Exploratory analyses were therefore performed to examine time-dependent changes in body weight during placebo treatment with and without LSI in obese subjects.

METHODS: Data were obtained from obese subjects (BMI ≥30 to ≤50 kg/m²) enrolled in the placebo arms of 2 obesity studies. In both, subjects injected placebo subcutaneously TID prior to meals for 16 weeks. Both studies included a 1-week placebo run-in prior to any LSI and a 16-week placebo treatment with or without LSI (LEARN® Program with lessons on eating, exercise, and behavioral modification) designed for weight loss. An extension to the LSI study was also performed, in which subjects received an additional 36 weeks of placebo treatment and participated in LSI designed for weight maintenance. The effect and time course of LSI on weight loss were evaluated using mean (SD) plots and boxplots of weight versus week since first dose (wsfd). The percentage of subjects achieving a 5% weight loss and the timing of subject withdrawals were also determined.

RESULTS: A total of 748 weight measurements from 94 subjects were evaluated. The mean (SD) baseline weight was 105 (20) kg (range 72 to 162 kg). The population was 77% female and 56% of the population participated in LSI. Mean (SD) weight loss was -0.23 (0.95) kg at the end of placebo run-in. No overall reduction in body weight was observed without LSI. For subjects in the LSI program, the mean weight nadir occurred at approximately 20 to 24 weeks, followed by a consistent mean weight gain throughout the extension period. At the end of placebo treatment (Week 16), 5% weight loss was achieved in 24% of subjects utilizing LSI, versus 5% of subjects without LSI. Among subjects in the LSI group, this target was achieved in 19% at Week 52. The mean (SD) time to subject withdrawal was 18.4 (16) weeks.

CONCLUSIONS: This analysis highlights the need to consider the duration and intensity of LSI when modeling the placebo effect in obesity trials. Knowledge of the time course of placebo response, the influence of LSI, and the pattern of subject withdrawal provide an informed basis for further PD model development.

Introduction

- Approximately 400 million adults worldwide are considered obese, a condition that substantially increases the risk of morbidity due to cardiovascular disease and type 2 diabetes mellitus[1]
- Strategies for weight loss include lifestyle interventions, such as diet, exercise, and behavioral programs, and pharmacologic therapy
- Placebo-controlled trials of anti-obesity agents have demonstrated weight loss in placebo-treated subjects; however, the contribution of the duration and intensity of other lifestyle modifications have not been considered in the interpretation of this weight loss. In addition, the rate of subject withdrawal has tended to be substantial
- Understanding the time course and dynamics of the placebo response is important when developing pharmacodynamic models that isolate drug effects

Objectives

- To perform an exploratory analysis that examines time-dependent changes in body weight in obese subjects receiving placebo with and without LSI
- To evaluate the characteristics and time-course of the achievement of 5% weight loss in obese subjects receiving placebo with and without LSI
- To evaluate the characteristics, rate, and reason for subject withdrawal in obese subjects receiving placebo with LSI

Methods

Figure 1. Study design

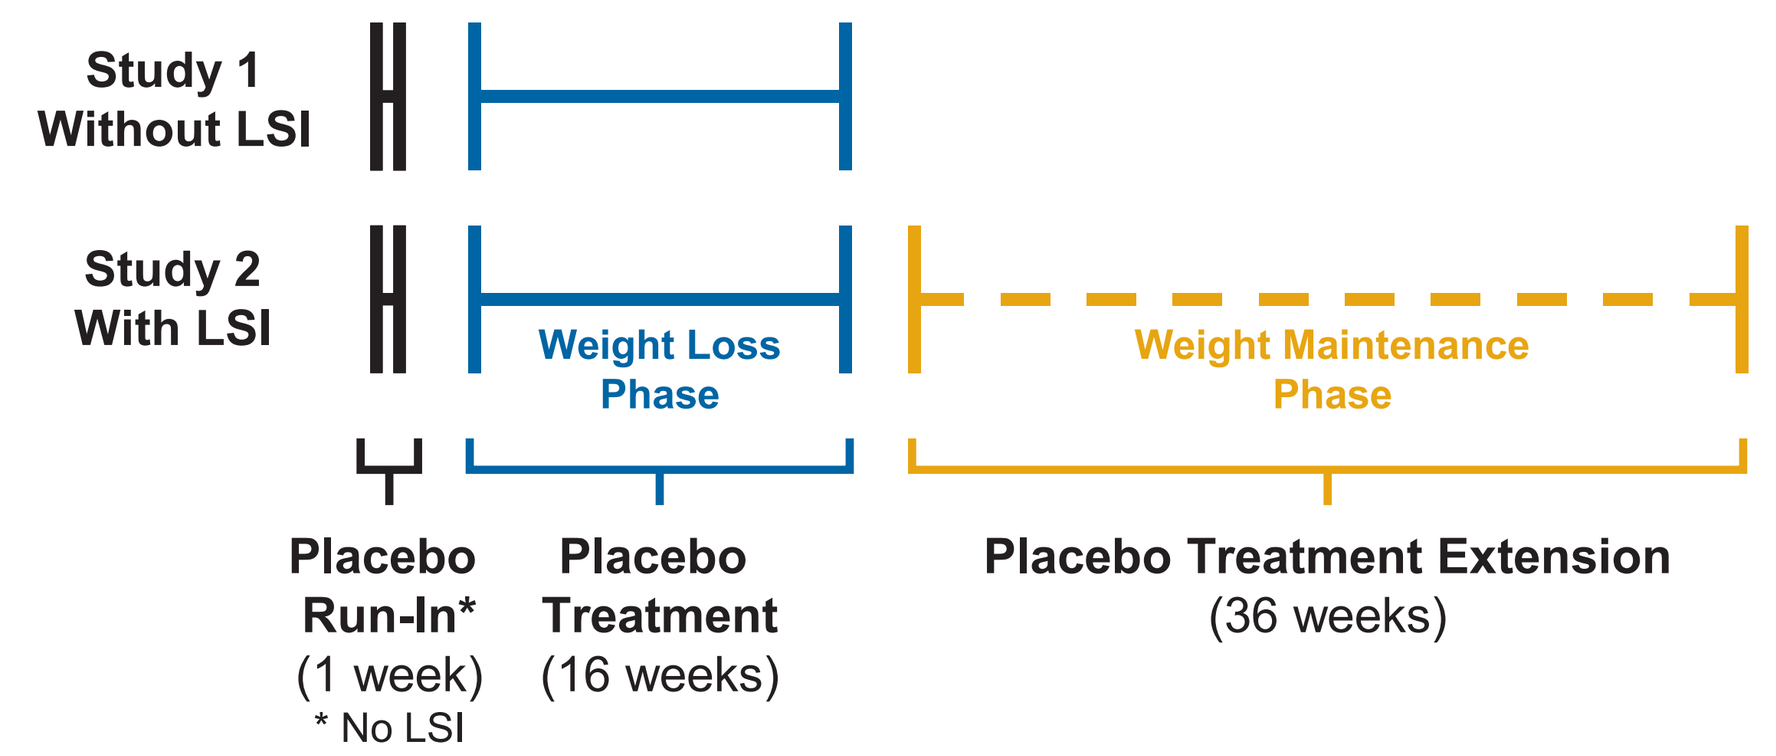
Study 1
Without LSI
Study 2
With LSI
Weight Loss Phase
Weight Maintenance Phase
Placebo Run-In* (1 week)
* No LSI
Placebo Treatment (16 weeks)
Placebo Treatment Extension (36 weeks)

- Data were obtained from obese subjects (BMI: ≥30 to ≤50 kg/m²) enrolled in the placebo arms of two randomized, double-blind, Phase 2 obesity studies
- Both studies consisted of a 1-week run-in period and a 16-week placebo treatment period
 - In Study 1, no lifestyle intervention was performed
 - In Study 2, subjects received the workbook from the LEARN (Lifestyle, Exercise, Attitudes, Relationships, Nutrition) Program and also had monthly individual sessions of ~15-20 min with lifestyle counselors
- The LEARN Program is a behavioral modification program to influence thoughts and habits concerning nutrition and exercise[2]
 - The program consists of a weight-loss phase followed by a weight-maintenance phase
 - A typical caloric recommendation is 1200 kcal/day in women and 1500 kcal/day in men with a recommended increase in exercise to 10,000 steps/day
- An extension of the LSI study was performed, in which subjects received an additional 36 weeks of placebo treatment and participated in the weight-maintenance phase of the LEARN Program

Methods Cont.

Data Analyses

- The analysis population included all placebo subjects who entered the placebo run-in period
- Exploratory analyses were performed to evaluate the change in body weight over time, the number of subjects who achieved a 5% weight loss, and the withdrawal rate over time in the LSI group only
- Withdrawal subjects were classified according to reason:
 - Withdrawal of consent
 - Adverse event
 - Protocol violation
 - Lost to follow-up
- Kaplan-Meier curves were generated to further characterize subjects who withdrew
- All data processing and statistical analyses were performed using SAS software, Version 8.0

Table 1. Baseline subject characteristics

Characteristic		Overall (N = 94)	Study 1 (Without LSI) (N = 41)	Study 2 (With LSI) (N = 53)
Age (years)	Mean (SD)	48.4 (11.4)	50.9 (9.8)	46.5 (12.2)
Weight (kg)	Mean (SD)	105.1 (19.6)	107.2 (21.5)	103.6 (18.0)
BMI (kg/m²)	Mean (SD)	37.8 (5.1)	38.5 (5.9)	37.3 (4.4)
BMI category, n (%)	<35 kg/m²	34 (36.2)	15 (36.6)	19 (35.9)
	≥35 kg/m² to <40 kg/m²	28 (29.8)	8 (19.5)	20 (37.7)
	≥40 kg/m²	32 (34.0)	18 (43.9)	14 (26.4)
Waist circumference (cm)	Mean (SD)	112.2 (14.6)	110.9 (16.6)	113.2 (12.9)
Ideal body weight (kg)[3]	Mean (SD)	59.4 (9.8)	59.4 (8.5)	59.5 (10.8)
Gender, n (%)	Male	22 (23.4)	9 (22.0)	13 (24.5)
	Female	72 (76.6)	32 (78.1)	40 (75.5)

Results

Figure 2. Time course of mean (SD) weight loss in obese subjects treated with placebo with and without LSI

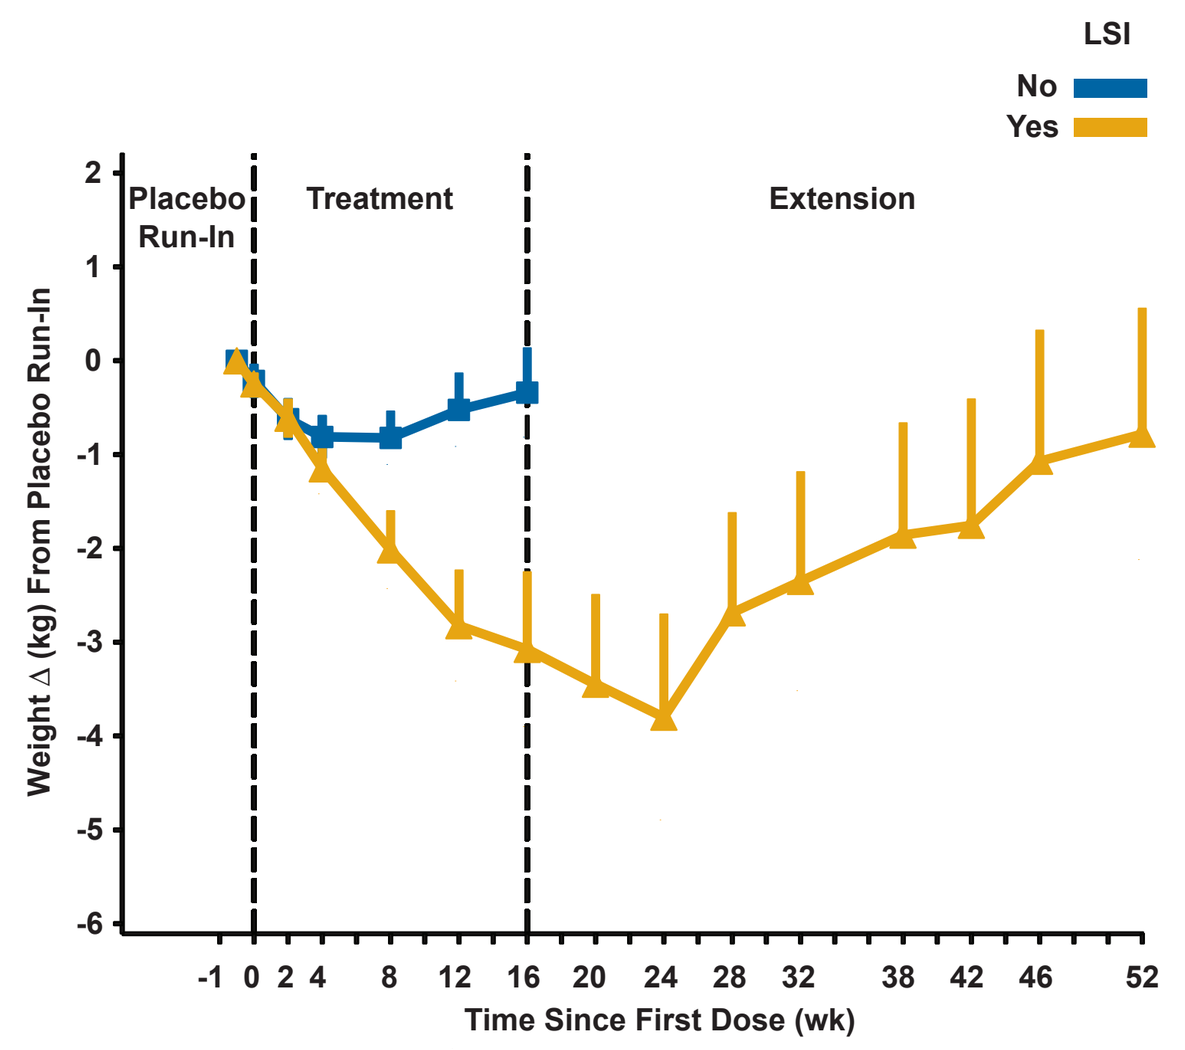
LSI
No
Yes
Placebo Run-In
Treatment
Extension
Weight Δ (kg) From Placebo Run-In
Time Since First Dose (wk)

- A total of 748 weight measurements from 94 placebo subjects were collected
- Mean (SD) weight loss was -0.23 (0.95) kg at the end of the placebo run-in period
- For subjects in the LSI program, maximum weight loss occurred between 20 and 24 weeks, followed by a consistent mean weight gain throughout the extension period. Without LSI, the maximum weight loss occurred at Week 4; however, no overall reduction in body weight was observed by the end of treatment
- Mean (SD) weight loss was -2.67 (4.61) kg at the end of the 16-week treatment period in the LSI arm versus -0.11 (3.09) kg without LSI
- At 52 weeks, a -0.57 (5.27) kg weight change was observed from baseline in the LSI arm

Results Cont.

Evaluation of Subjects Achieving 5% Weight Loss

Figure 3. Time course of individual weight loss considering LSI participation and achievement of 5% weight loss at any time during the study

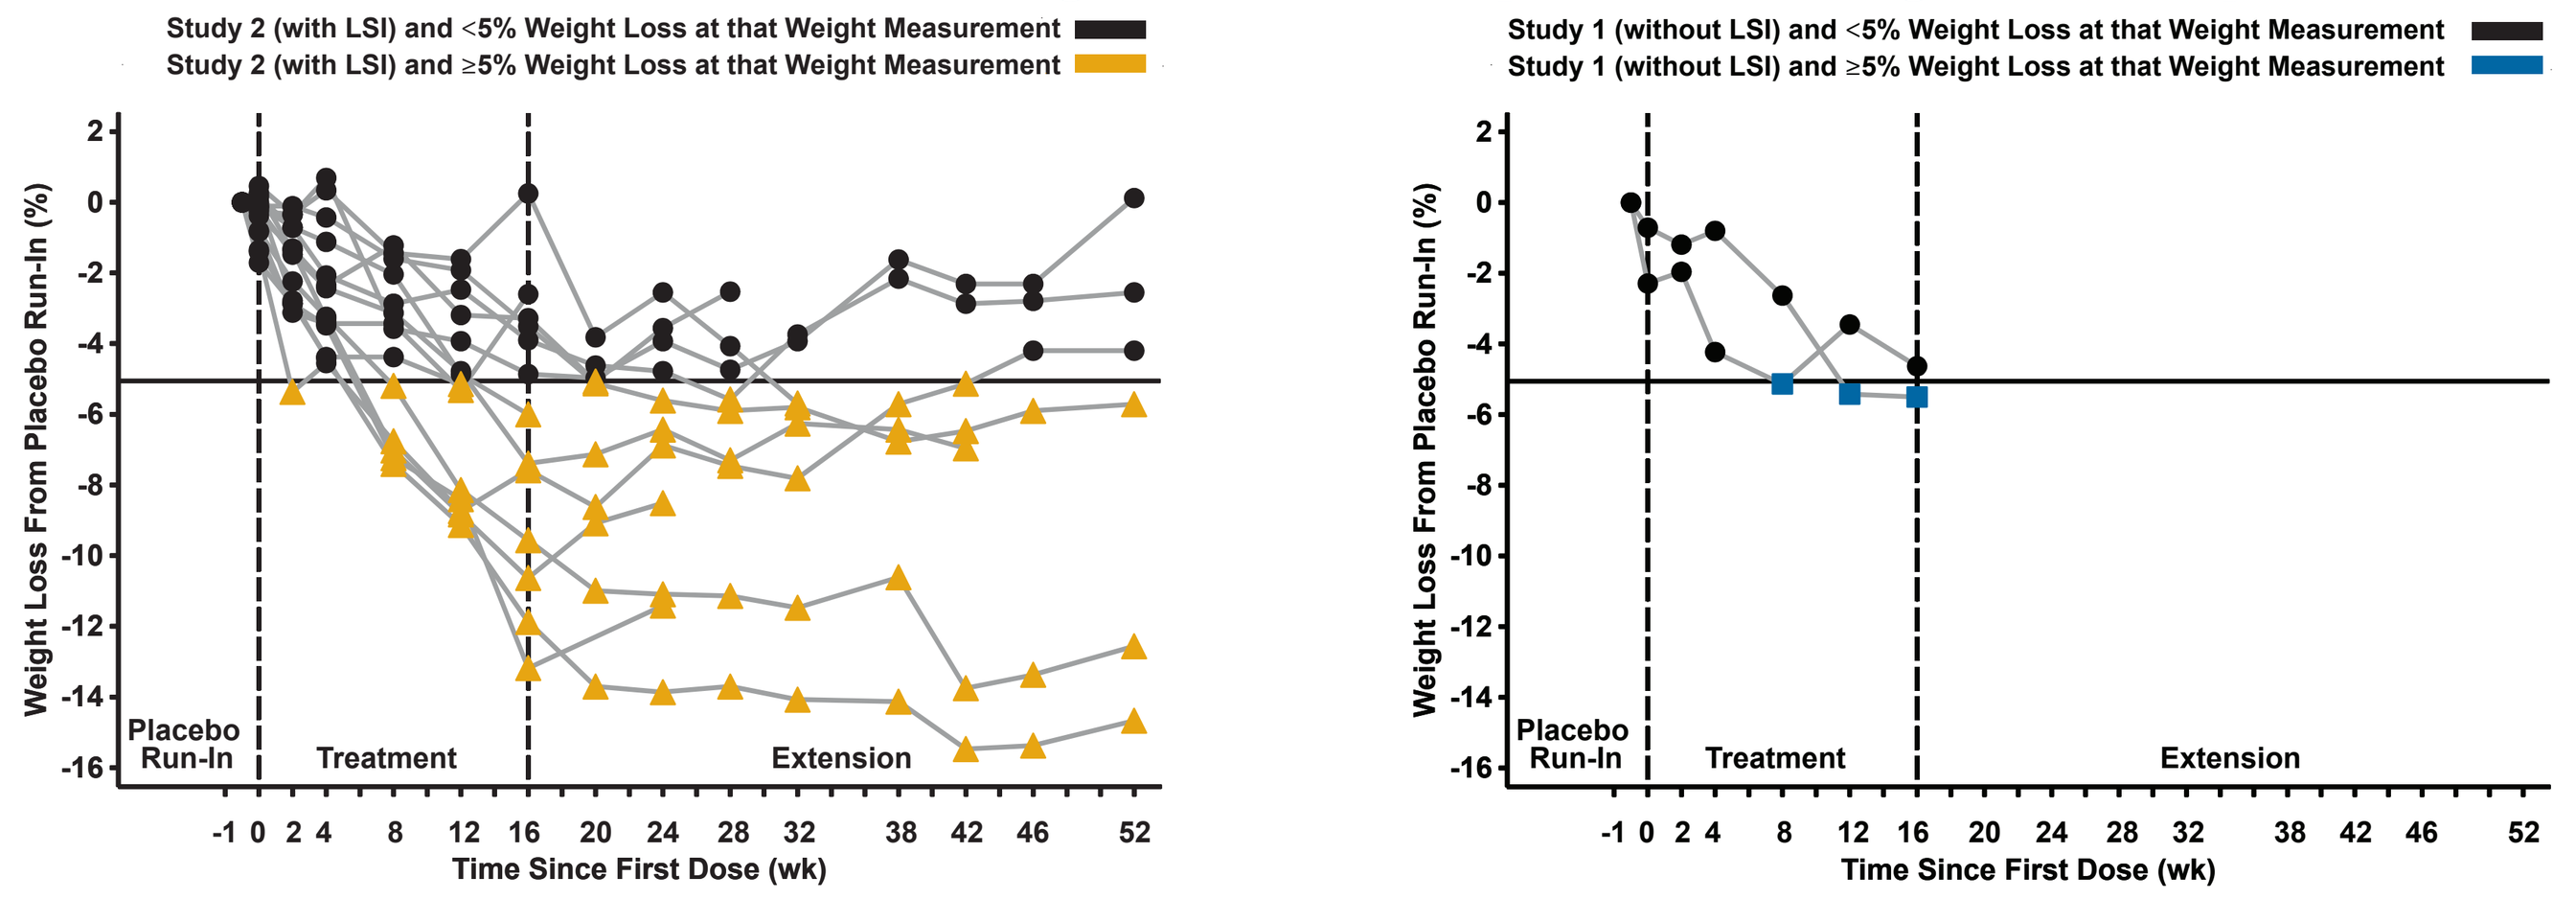
Study 2 (with LSI) and <5% Weight Loss at that Weight Measurement
Study 2 (with LSI) and ≥5% Weight Loss at that Weight Measurement
Study 1 (without LSI) and <5% Weight Loss at that Weight Measurement
Study 1 (without LSI) and ≥5% Weight Loss at that Weight Measurement
Weight Loss From Placebo Run-In (%)
Placebo Run-In
Treatment
Extension
Time Since First Dose (wk)

- Of those subjects who had a 5% weight loss, 10 subjects (62.5%) achieved this weight loss between 8 and 12 weeks
- By Week 16, 10 (18.9%) subjects in Study 2 (with LSI) and 2 (4.9%) subjects in Study 1 (without LSI) had achieved a 5% weight loss. By Week 52, 4 more subjects had achieved a 5% weight loss in Study 2 (with LSI)
- Ten (62.5%) subjects who achieved a 5% weight loss maintained it until their last weight measurement across both Study 1 (without LSI) and Study 2 (with LSI)

Table 2. Baseline characteristics of subjects achieving 5% weight loss at any time during the study

Characteristic		≥5% Weight Loss (N = 16)	<5% Weight Loss (N = 78)
Age (years)	Mean (SD)	49.4 (12.5)	48.2 (11.2)
Weight (kg)	Mean (SD)	104.7 (18.7)	105.2 (19.9)
BMI (kg/m²)	Mean (SD)	37.4 (4.9)	37.9 (5.2)
BMI category, n (%)	<35 kg/m²	6 (37.5)	28 (35.9)
	≥35 kg/m² to <40 kg/m²	5 (31.3)	23 (29.5)
	≥40 kg/m²	5 (31.3)	27 (34.6)
Waist circumference (cm)	Mean (SD)	113.2 (13.9)	112.0 (14.8)
Ideal body weight (kg)[3]	Mean (SD)	59.6 (8.2)	59.4 (10.2)
Gender, n (%)	Male	3 (18.8)	19 (24.4)

Evaluation of Withdrawal Subjects in Study 2 (with LSI)

- Withdrawal rates were only evaluated in Study 2 (with LSI) based on National Institutes of Health recommendations that medications for obesity be used only in conjunction with lifestyle intervention[4]

Table 3. Baseline characteristics of subjects who withdrew vs. completed Study 2 (with LSI)

Characteristic		Withdrew (N = 30) (56.6%)	Completed (N = 23) (43.4%)
Age (years)	Mean (SD)	43.4 (12.1)	50.4 (11.5)
Weight (kg)	Mean (SD)	105.5 (19.0)	101.0 (16.7)
BMI (kg/m²)	Mean (SD)	38.5 (4.1)	35.8 (4.4)
BMI category, n (%)	<35 kg/m²	6 (20.0)	13 (56.5)
	≥35 kg/m² to <40 kg/m²	15 (50.0)	5 (21.7)
	≥40 kg/m²	9 (30.0)	5 (21.7)
Waist circumference (cm)	Mean (SD)	114.7 (11.7)	111.4 (14.3)
Ideal body weight (kg)[3]	Mean (SD)	58.0 (10.9)	61.3 (10.6)
Gender n (%)	Male	6 (20.0)	7 (30.4)
	Female	24 (80.0)	16 (69.6)

Figure 4. Cumulative withdrawal rate in Study 2 (with LSI)

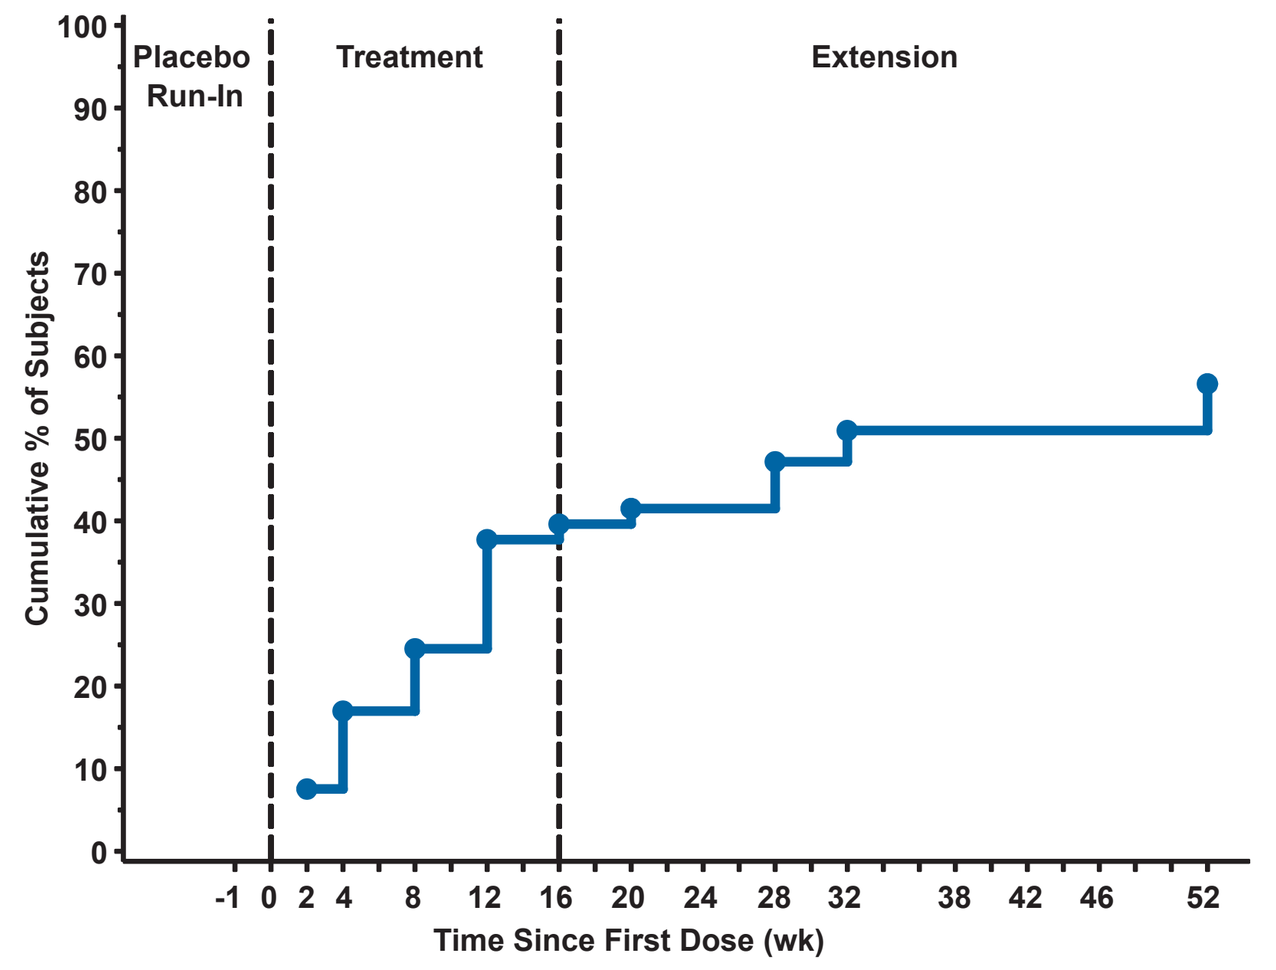
Placebo Run-In
Treatment
Extension
Cumulative % of Subjects
Time Since First Dose (wk)

Figure 5. Change in weight at withdrawal vs. time of withdrawal in Study 2 (with LSI)

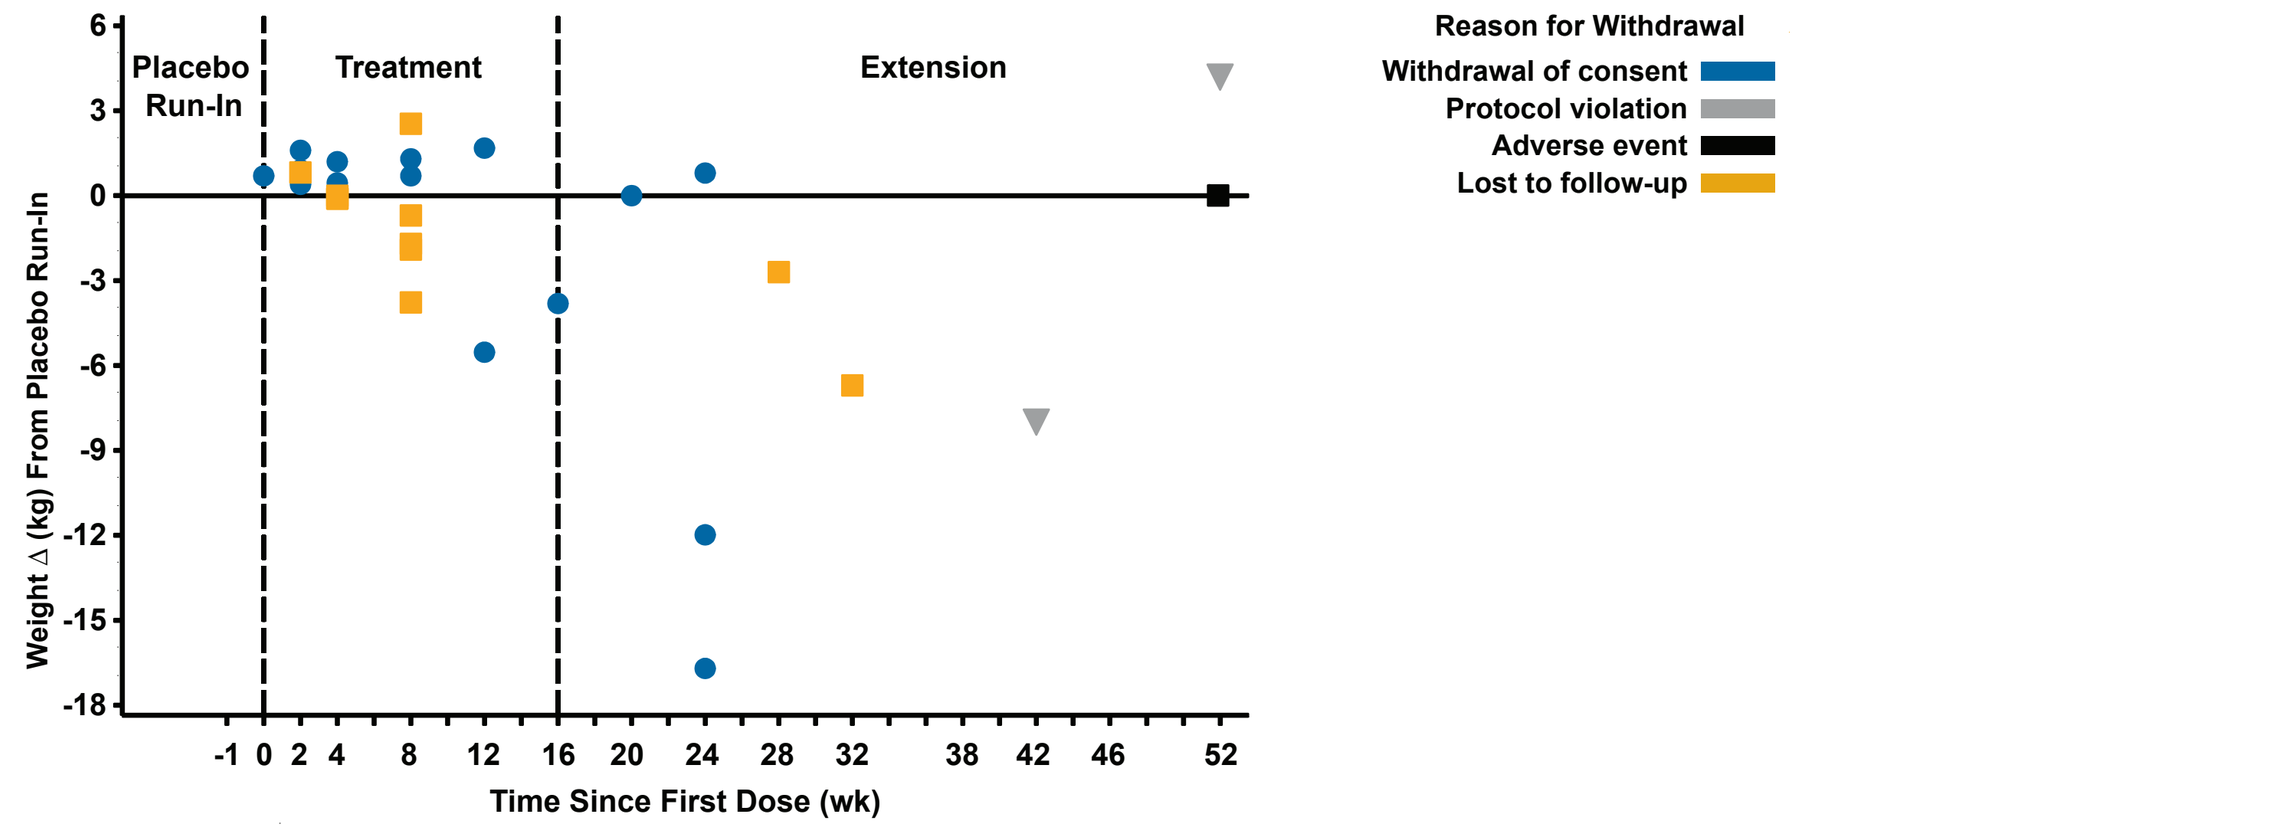
Reason for Withdrawal
Withdrawal of consent
Protocol violation
Adverse event
Lost to follow-up
Placebo Run-In
Treatment
Extension
Weight Δ (kg) From Placebo Run-In
Time Since First Dose (wk)

- Mean (SD) time to subject withdrawal was 18.4 (16.0) weeks
- Seventy percent of withdrawal subjects withdrew by Week 16
- All except three of the subject withdrawals were classified as withdrawal of consent or lost to follow-up

Table 4. Mean body weight changes in subjects who withdrew vs. completed Study 2 (with LSI) at ≤16 weeks and >16 weeks

	≤16 weeks	>16 weeks
Subjects in study (n)	53	23
Subject withdrawal n (%)	21 (39.6)	9 (39.1)
Mean body weight change (%) from baseline to withdrawal in subjects who withdrew	-0.21	-4.55
Mean body weight change (%) from baseline to end of study in subjects who completed	-2.08	-1.20

- Nine subjects completed the weight-loss phase but chose not to enter into the weight-maintenance phase; these subjects were classified as completers
- During the first 16 weeks, subjects who withdrew had less change in body weight than those who completed Study 2 (with LSI); this trend was not apparent after 16 weeks
- Seventeen (56.7%) subjects who withdrew during Study 2 (with LSI) had gained weight at the time of withdrawal

Figure 6. Kaplan-Meier plot of estimated probability of withdrawal vs. time since first dose grouped by BMI classification

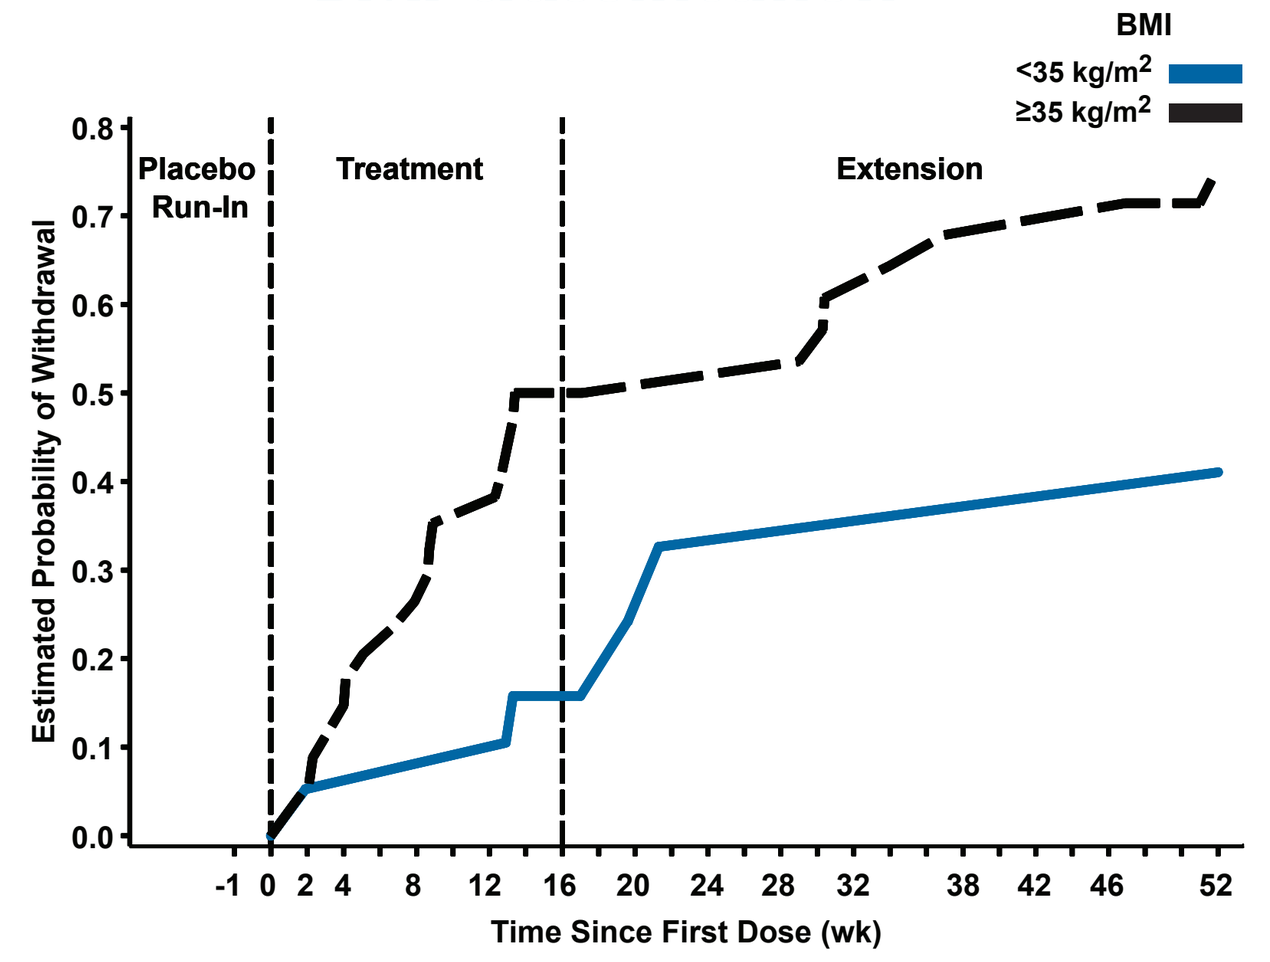
BMI
<35 kg/m²
≥35 kg/m²
Placebo Run-In
Treatment
Extension
Estimated Probability of Withdrawal
Time Since First Dose (wk)

- Subjects with a BMI ≥35 kg/m² were more likely to withdraw

Figure 7. Kaplan-Meier plot of estimated probability of withdrawal vs. time since first dose grouped by age

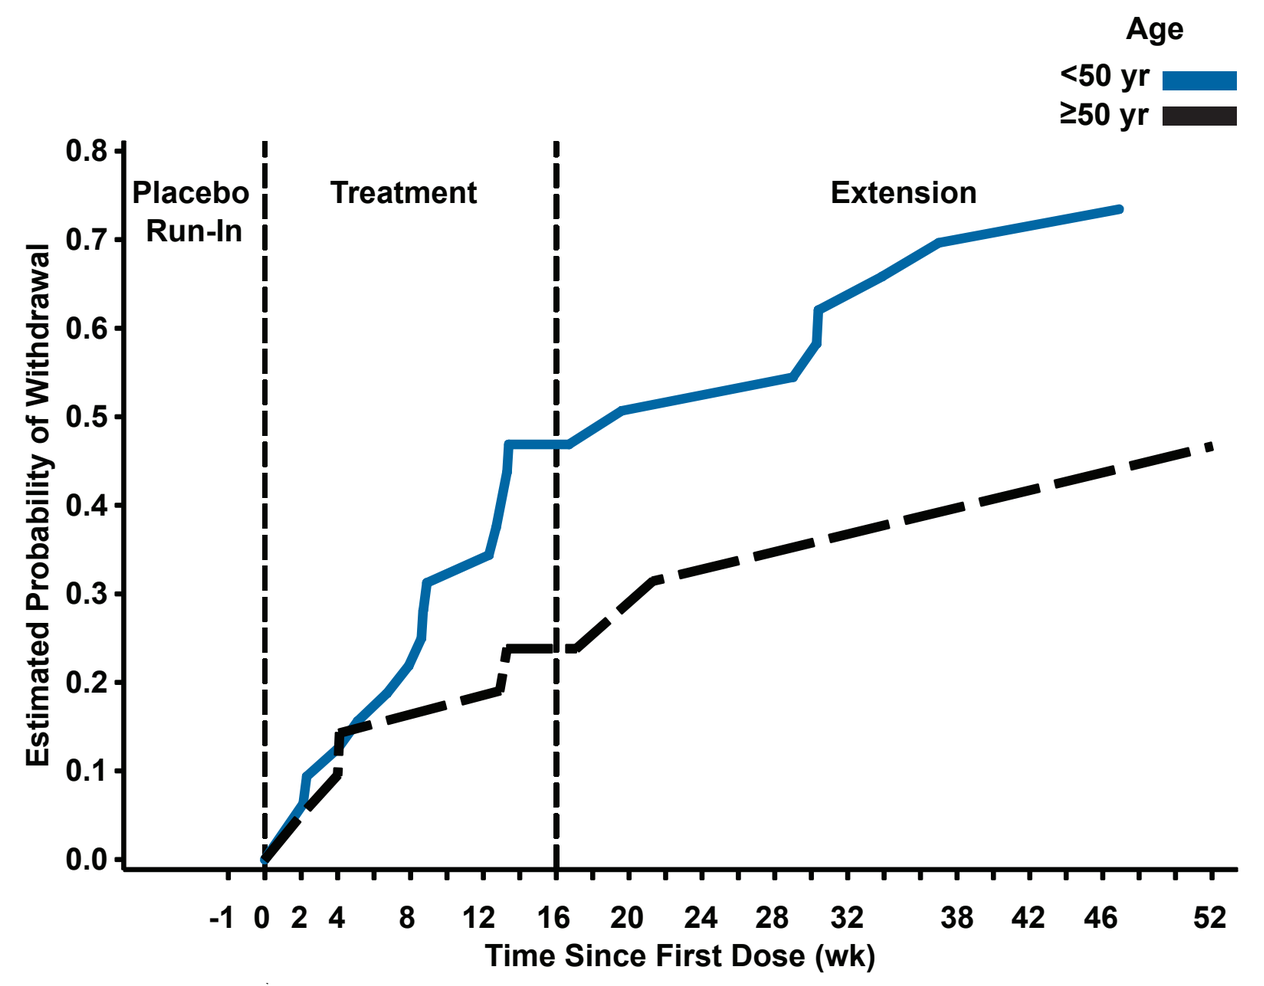
Age
<50 yr
≥50 yr
Placebo Run-In
Treatment
Extension
Estimated Probability of Withdrawal
Time Since First Dose (wk)

- A trend toward an increased withdrawal rate was observed for subjects less than 50 years old

Conclusions

- Overall, no weight loss was observed without LSI. During the LSI treatment period, there was initial weight loss followed by weight gain throughout the LSI maintenance period
- This analysis highlights the need to consider the duration and the intensity of LSI when modeling the placebo effect in obesity trials
- Knowledge of the time course of the placebo response, the influence of LSI, and the pattern of subject withdrawal provide an informative basis for further pharmacodynamic model development

References

1. Obesity and overweight. World Health Organization. Available at: http://www.who.int/mediacentre/factsheets/fs311/en/. Accessed January 12, 2006.
2. Brownell KD. The LEARN Program for Weight Management, 10th ed. Dallas, TX: American Health Publishing Company; 2004.
3. Peck CC, Conner DP, Murphy MG. Bedside Clinical Pharmacokinetics: Simple Techniques for Individualizing Drug Therapy. Vancouver, WA: Lippincott Williams & Wilkins; 1989.
4. Clinical Guidelines on the Identification, Evaluation, and Treatment of Overweight and Obesity in Adults. National Institutes of Health. National Heart, Lung, and Blood Institute. NIH Publication No. 98-4083. Available at: http://www.nhlbi.nih.gov/guidelines/obesity/ob_gdlns.pdf. Accessed February 7, 2007.

ACKNOWLEDGEMENT: We thank Monika Sokolowski, BA, for her contribution to the statistical and graphical programming for these analyses.