Model-Based Evaluations to Select and Confirm Doses in the Clinical Development of Exenatide

Mark Fineman[1], Luann Phillips[2], David J Jaworowicz[2], Brenda Cirincione[2], Elizabeth Ludwig[2], Kristin Taylor[1], Prajakti A Kothare[3], Alain D Baron[1], Thaddeus Grasela[2]

[1]Amylin Pharmaceuticals, Inc., San Diego, CA; [2]Cognigen Corporation, Buffalo, NY; [3]Eli Lilly and Company, Indianapolis, IN

Abstract

BACKGROUND: Population pharmacokinetic (PK) and pharmacodynamic (PK/PD) models based on early Phases 1 and 2 data for the new antidiabetic medication exenatide (ex) supported transition from weight-based to fixed 5 and 10 µg subcutaneous BID dosing for Phase 3 trials (CPT 2002;71:P29). The model-based dose selection process was designed to target exenatide exposure that would maximize glucose reduction and mitigate gastric AEs. The models were iteratively refined with the inclusion of additional Phases 2 and 3 data to further describe the PK and exposure-response (ER) relationships. These models confirmed the appropriateness of the selected doses and allowed for the quantitative assessment of the influence of intrinsic variables such as age, weight, and gender.

METHODS: 4870 ex concentrations from 242 patients with type 2 diabetes (subj) across 10 clinical trials were applied to the existing PK model (1-CMT with nonlinear plus linear absorption and linear elimination). The relationship between ex AUC_{0-3h} and glucose (gluc) AUC_{0-3h} (540 pairs from 183 subj) was assessed in an inhibitory sigmoid-E_{max} model. Covariate evaluation included the influence of selected subject descriptors, clinical chemistries, concomitant oral antidiabetic agent use, and anti-ex antibody (Ab) status on PK and PK/PD. Model validation was performed by bootstrapping.

RESULTS: For the PK model, weight and anti-ex Ab status were significant covariates for CL/F and V/F, and gender for absorption rate. In the PK/PD model, significant factors for E_{max} were baseline gluc AUC and anti-ex Ab status. The magnitude of these effects did not support dose adjustment. The population mean estimated decrease in gluc was ~27% and 33% for the 5 and 10 µg BID doses, respectively. Both doses approach the asymptotic portion of the exposure-response curve. With ex doses of 5 µg, 55% of subj achieved 75% of their predicted maximum glucose reduction. With 10 µg doses, 90% achieved that same target.

CONCLUSION: This model-based drug development approach demonstrated the value of the learn-and-confirm paradigm. Dose selection was supported by prelim PK and PK/PD models in a small number of subj, and confirmed with larger numbers of subj from Phases 2 and 3 trials. These results verified that ex doses of 10 µg BID optimize clinical benefit in terms of glycemic control.

Introduction

- In preclinical and clinical studies, exenatide has been shown to have multiple mechanisms of action that improve metabolic control:[1-4]
 - Enhancement of glucose-dependent insulin secretion[1,4]
 - Suppression of inappropriately elevated postprandial glucagon secretion[1,4]
 - Slowing of gastric emptying[1,4]
 - Reduction of food intake and weight[1,4]
- In patients with type 2 diabetes, exenatide is administered as an immediate release formulation twice daily before main meals (BID) by subcutaneous injection[5]
- Exenatide has been approved for the treatment of type 2 diabetes in patients failing to achieve adequate glycemic control with metformin (MET), a sulfonylurea (SFU), a thiazolidinedione (TZD), a combination of MET and an SFU, or a combination of MET and a TZD
- PK/PD models were used throughout the development program to select Phase 3 dosing and to evaluate the need for dose adjustment for special populations[5]

Objectives

- To utilize PK and PK/PD models from early Phases 1 and 2 data to justify a shift from weight-based to fixed dosing for late-stage drug development
- To evaluate effects of various demographic characteristics and anti-exenatide antibodies on exenatide PK and PK/PD
- To confirm the appropriateness of the fixed (non-weight based) doses from simulations based on population analysis of early Phases 1 and 2 data[6]

Methods

Study Design/Data

- Data were pooled from 10 clinical trials
- A total of 242 subjects with type 2 diabetes, controlled by either diet or oral anti-diabetic agents, were utilized for the PK model; 183 of these subjects (enrolled in 8 of the clinical trials) were available for the PK/PD analysis
- Dosage regimens (single and multiple subcutaneous doses)
 - Weight based: QD: 0.02, 0.05, 0.1, 0.2, and 0.4 µg/kg
 BID: 0.08 and 0.1 µg/kg
 TID: 0.08 µg/kg
 QID: 0.05 µg/kg
 - Fixed: QD: 1 and 10 µg
 BID: 5 and 10 µg
- PK Sampling Design:
 - Full profiles (≥9 samples per subject) from 7 studies ranging from 5 to 23 h
 - Three Phase 3 studies: pre-dose, 30 min, and 1, 1.5, 2, and 3 h
- PD Sampling Design: only samples collected out to 3 h post dose following specified meals (≥6 samples per subject) were used to calculate glucose AUC_{0-3h} ($gAUC_{0-3h}$) via the trapezoidal rule; corresponding $exAUC_{0-3h}$ was calculated using exenatide concentrations obtained at simultaneous time points

Subject Covariates

- PK and PK/PD:
 - Anti-exenatide antibody: status and titer ratio
 - Demographics: gender, race, height, weight, age, ideal body weight, percent of ideal body weight, body mass index, and body surface area
 - Clinical chemistries: serum creatinine, albumin, alkaline phosphatase, alanine aminotransferase, aspartate aminotransferase, gamma-glutamyltransferase, total bilirubin, and creatinine clearance
 - Concomitant medications: SFUs, MET, and TZDs
 - Other descriptors: dosing regimen, average hourly dose rate, breakfast type, baseline HbA_{1c}, method of diabetes control prior to treatment, and concomitant medications (SFUs, MET, and TZDs)

Methods Cont.

Pharmacostatistical Model

- NONMEM® V, Level 1.1, using first-order estimation
- PK: previously developed one-compartment model with first-order elimination; absorption modeled as a combination of a Michaelis-Menten process plus a linear absorption rate
- PK/PD: inhibitory sigmoid-E_{max} model for $gAUC_{0-3h}$ vs. $exAUC_{0-3h}$ relationship
- Interindividual variability models: exponential error for PK, and exponential and or additive error for PK/PD
- Residual variability models: log error for PK and additive error for PK/PD

Statistical Analyses

- Covariates were evaluated for inclusion into the final PK and PK/PD models by univariate stepwise forward selection followed by backward elimination procedures
 - Continuous covariates modeled using linear, exponential, and/or power models
 - Dichotomous and categorical covariates modeled using proportional or additive shift models
- Statistical significance for retention of covariates was determined by a change in the minimum value of the objective function of ≥3.84 (α = 0.05, 1 d.f.) during forward selection and ≥10.83 (α = 0.001, 1 d.f.) for backward elimination
- Goodness-of-fit for each NONMEM analysis was assessed by:
 - Scatterplots of population and individual predicted vs. measured concentrations or $gAUC_{0-3h}$
 - Scatterplots of weighted residuals vs. predicted concentrations (or $gAUC_{0-3h}$) and time since last dose
 - %SEMs of PK or PK/PD parameter estimates
 - Changes in the estimates of interindividual and residual variability

Population PK Model Development Process Overview

1. Initial Model Development and Simulation: Phases 1 and 2 clinical trials provided PK, safety, and efficacy data for an initial population PK and PK/PD model that justified the transition from weight-based dosing to model based fixed 5 µg and 10 µg BID dosing in Phase 3 clinical trials[6]
2. Population PK and PK/PD model refinement:
 - Data from 6 additional Phases 2 and 3 trials: a total of 4870 exenatide plasma concentrations (n = 229 subjects) were utilized in the refined PK model; a total of 540 pairs of exenatide and $gAUC_{0-3h}$ values (n = 183 subjects) were used in the analysis
 - Evaluating the effect of various subject descriptors, organ function descriptors, and concomitant medications on PK and PK/PD parameters
 - Assessment of the effect of anti-exenatide antibodies on the disposition of exenatide and on glucose response
3. Estimation of Refined Final Population PK and PK/PD model
4. Model-based dose confirmation assessment evaluating Phase 3 clinical outcomes for therapeutically recommended fixed-dose regimens (5 µg and 10 µg BID)

Results

Table 1. Final parameter estimates and standard errors for the final PK model

Parameter	Final Parameter Estimate: Population Mean	Final Parameter Estimate: %SEM	Magnitude of Interindividual Variability: Estimate (%CV)	Magnitude of Interindividual Variability: %SEM
Maximum fractional reduction of Ka (µg/h)	2.67	59.2	—	—
Amount required for 50% reduction in Ka (µg)	3.56	49.4	—	—
Linear Ka component (1/h)	0.237	36.6	135.65	65.2
CL/F intercept (L/h)	4.51	14.9	55.50	19.5
V/F intercept (L)	168	20.6	64.5	22.0
Shift for no Ab titer on CL/F	1.03	33.9	—	—
Slope for Ab titer ≤125 on V/F	1.08	25.6	—	—
Shift for female gender on linear Ka	0.679	38.3	—	—
Power for WTKG on CL/F	1.12	16.5	—	—
Power for WTKG on V/F	0.205	24.6	—	—
RV (SD-log units)	0.32	7.9	—	—

MVOF=-4455.110

$$Ka_{ij} = 2.67^{*}\, A(1)_{ij} / (3.56 + A(1)_{ij}) + 0.237^{*}\,(1 + 0.679^{*}\, SEXF_j)$$

$$CL/F_{ij} = (4.51^{*}\,(WTKG_j/87)^{1.12} {}^{*}\,(1 + 1.03^{*}\, ANTIneg_{ij}))$$

$$V/F_{ij} = 168^{*}\,(WTKG_j/87)^{0.205} + 1.08^{*}\,(TITER_{ij} - 125) \cdot (TTRG_{ij})$$

- The refined model identified (1) body weight and antibody status as predictors of CL/F, (2) body weight and antibody titer ratio as predictors of V/F, and (3) absorption rate was a combination of linear and non-linear processes, with gender as a significant predictor
- None of the other covariates tested could explain the interindividual variability in any of the parameters to a statistically significant extent. Thus, no dosing adjustments would be recommended for these variables based upon these analyses

Table 2. Population mean values of CL/F and V/F based upon significant covariates identified in the final PK model

	Antibody Status	Titer Ratio	Body Weight: Minimum (50 kg)	Body Weight: 25th Percentile (80.9 kg)	Body Weight: Median (93.3 kg)	Body Weight: 75th Percentile (105.9 kg)	Body Weight: Maximum (161.9 kg)
CL/F (L/h)	+	N/A	2.43	4.16	4.88	5.62	9.04
	–	0	4.93	8.44	9.91	11.41	18.35
V/F (L)	+	5	20.37	35.91	40.83	45.31	61.2
		25	41.97	57.51	62.43	66.91	82.81
		125	149.97	165.51	170.43	174.91	190.81
	–	0	14.97	30.51	35.43	39.91	55.81

Results Cont.

Figure 1. Goodness-of-fit plot for PK model

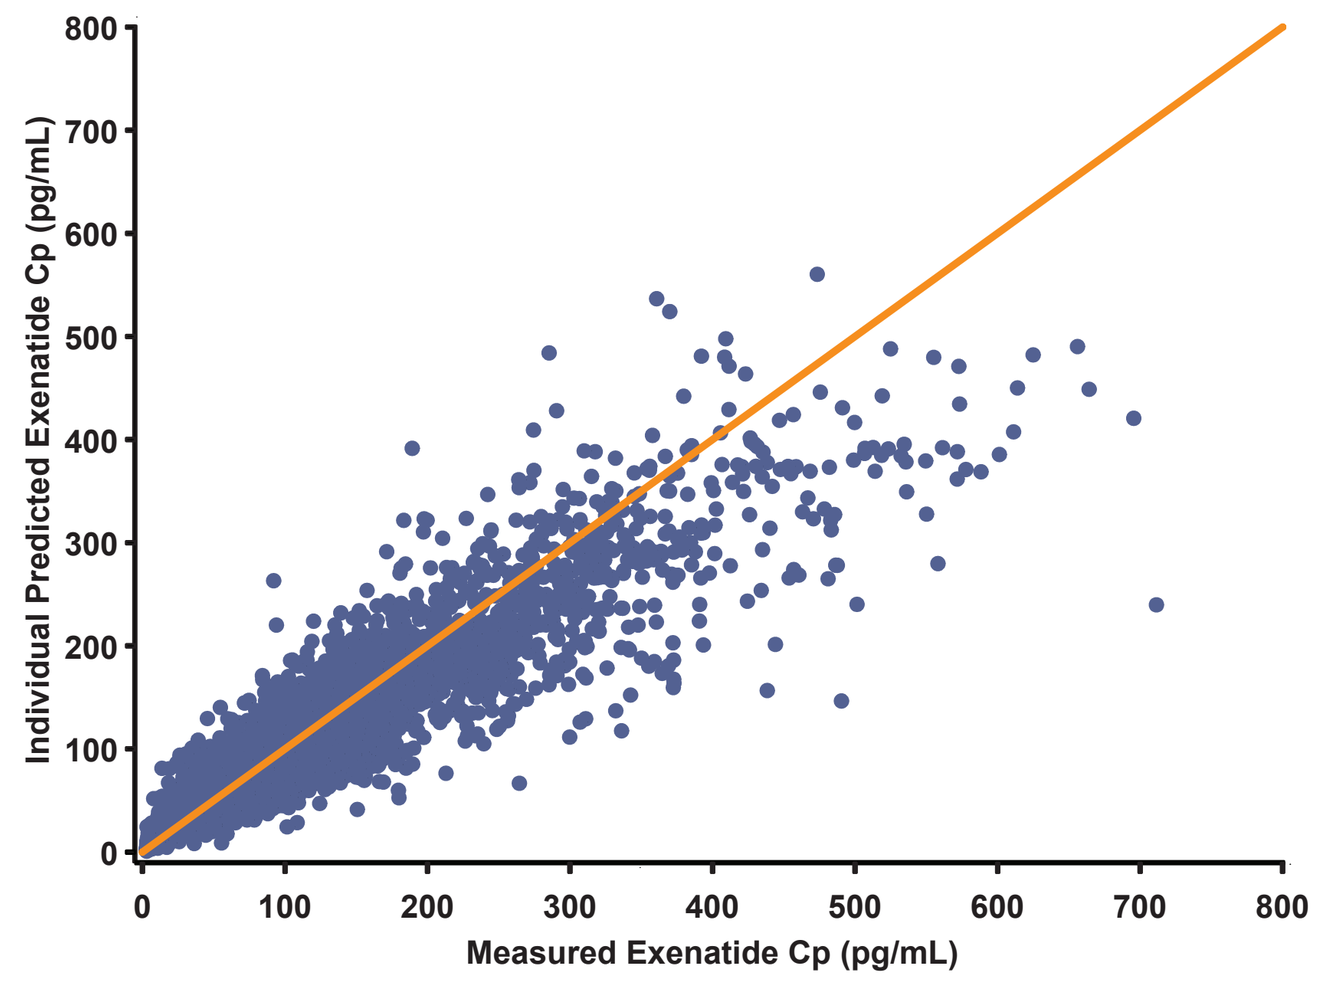
Individual Predicted Exenatide Cp (pg/mL)
Measured Exenatide Cp (pg/mL)

Table 3. Final parameter estimates and standard errors of the final PK/PD model

Parameter	Final Parameter Estimate: Population Mean	Final Parameter Estimate: %SEM	Magnitude of Interindividual Variability: Estimate[a]	Magnitude of Interindividual Variability: %SEM
AUC_{50} DREG=1, 2, 5, 6[b]	169	12.5	62.45	20.4
S	2.42	19.1	—	—
Slope for placebo AUC_{0-5h} Effect on E_{max}	0.258	15.2	114.02	19.7
Shift for No Ab on E_{max}	0.329	66.3	—	—
AUC_{50} shift DREG=3, 4[2]	1.25	52.4	—	—
RV (SD)	10.25	16.6	—	—

MVOF = 5421.127

a The IIV of AUC_{50} is expressed as %CV and the IIV of E_{max} is expressed as a standard deviation.
b DREG = Dosage regimen
DREG = 1 – Single doses with a 48-h washout,
DREG = 2 – Single doses with a 24-h washout,
DREG = 3 – Multiple doses for 2 d,
DREG = 4 – Multiple doses for 5 d,
DREG = 5, 6 – Multiple doses ≥4 wk.

$$\text{Glucose}AUC_{0-3\,h\,ij} = \text{Placebo}_j - \left(\frac{E_{max\,j} \cdot (\text{Exenatide}AUC_{0-3hr\,ij})^{s}}{(AUC_{50\,j})^{s} + (\text{Exenatide}AUC_{0-3hr\,ij})^{s}}\right)$$

$$AUC_{50\,j} = 169 \cdot (1 + 1.25 \cdot DR1_{ij})$$

$$E_{max\,j} = (0.258 \cdot BLGAUC_j) \cdot (1 + 0.329 \cdot NANTI_{ij})$$

Figure 2. Goodness-of-fit plot for PK/PD model

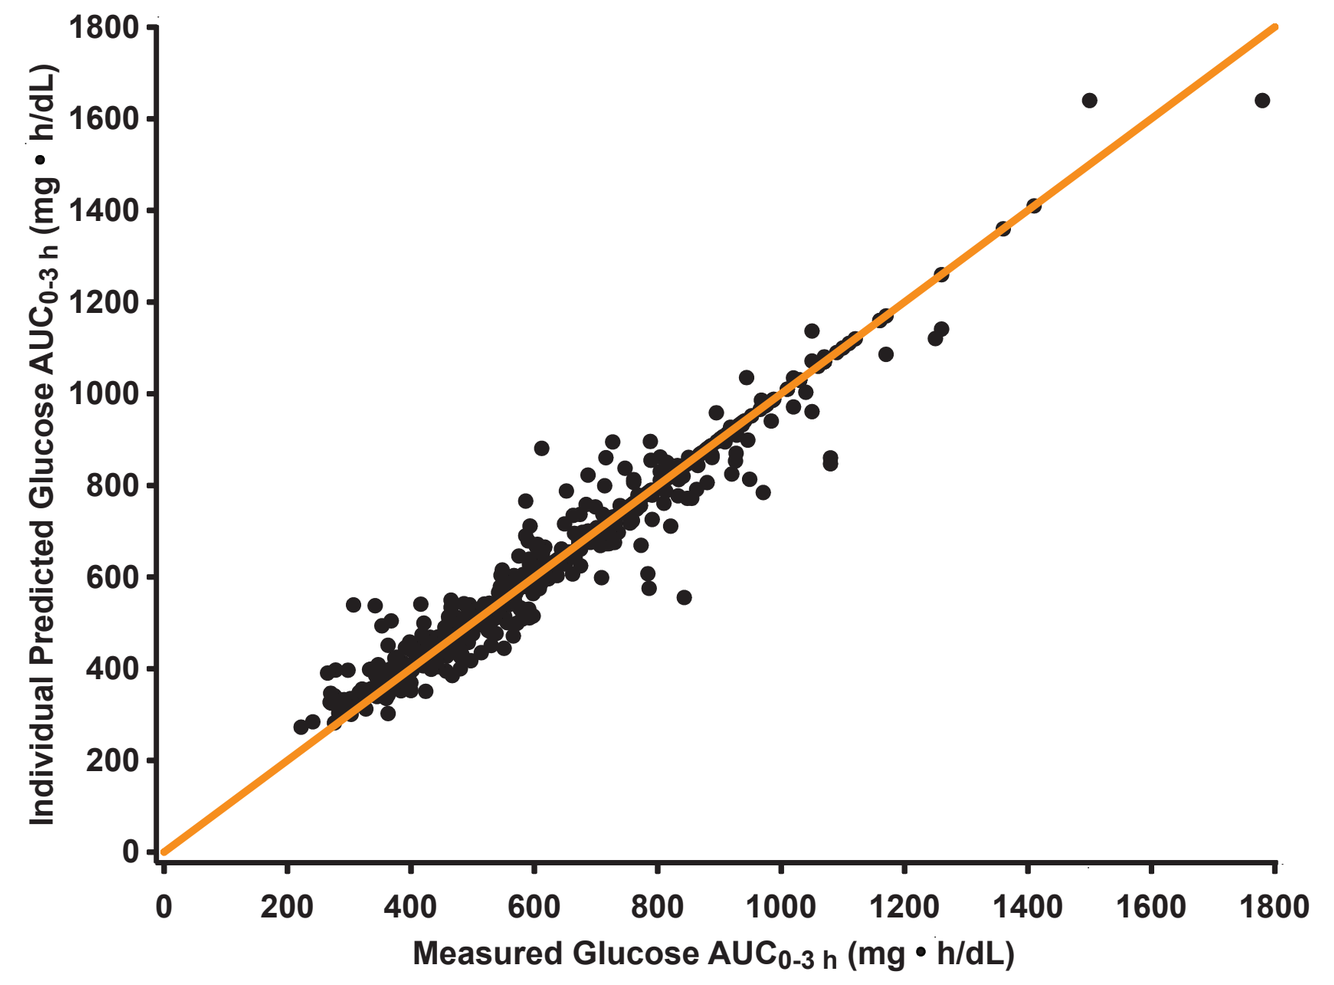
Individual Predicted Glucose $AUC_{0-3\,h}$ (mg • h/dL)
Measured Glucose $AUC_{0-3\,h}$ (mg • h/dL)

Figure 3. Model predicted $gAUC_{0-3h}$ response, stratified by dosage regimen and antibody status, overlaid on observed data

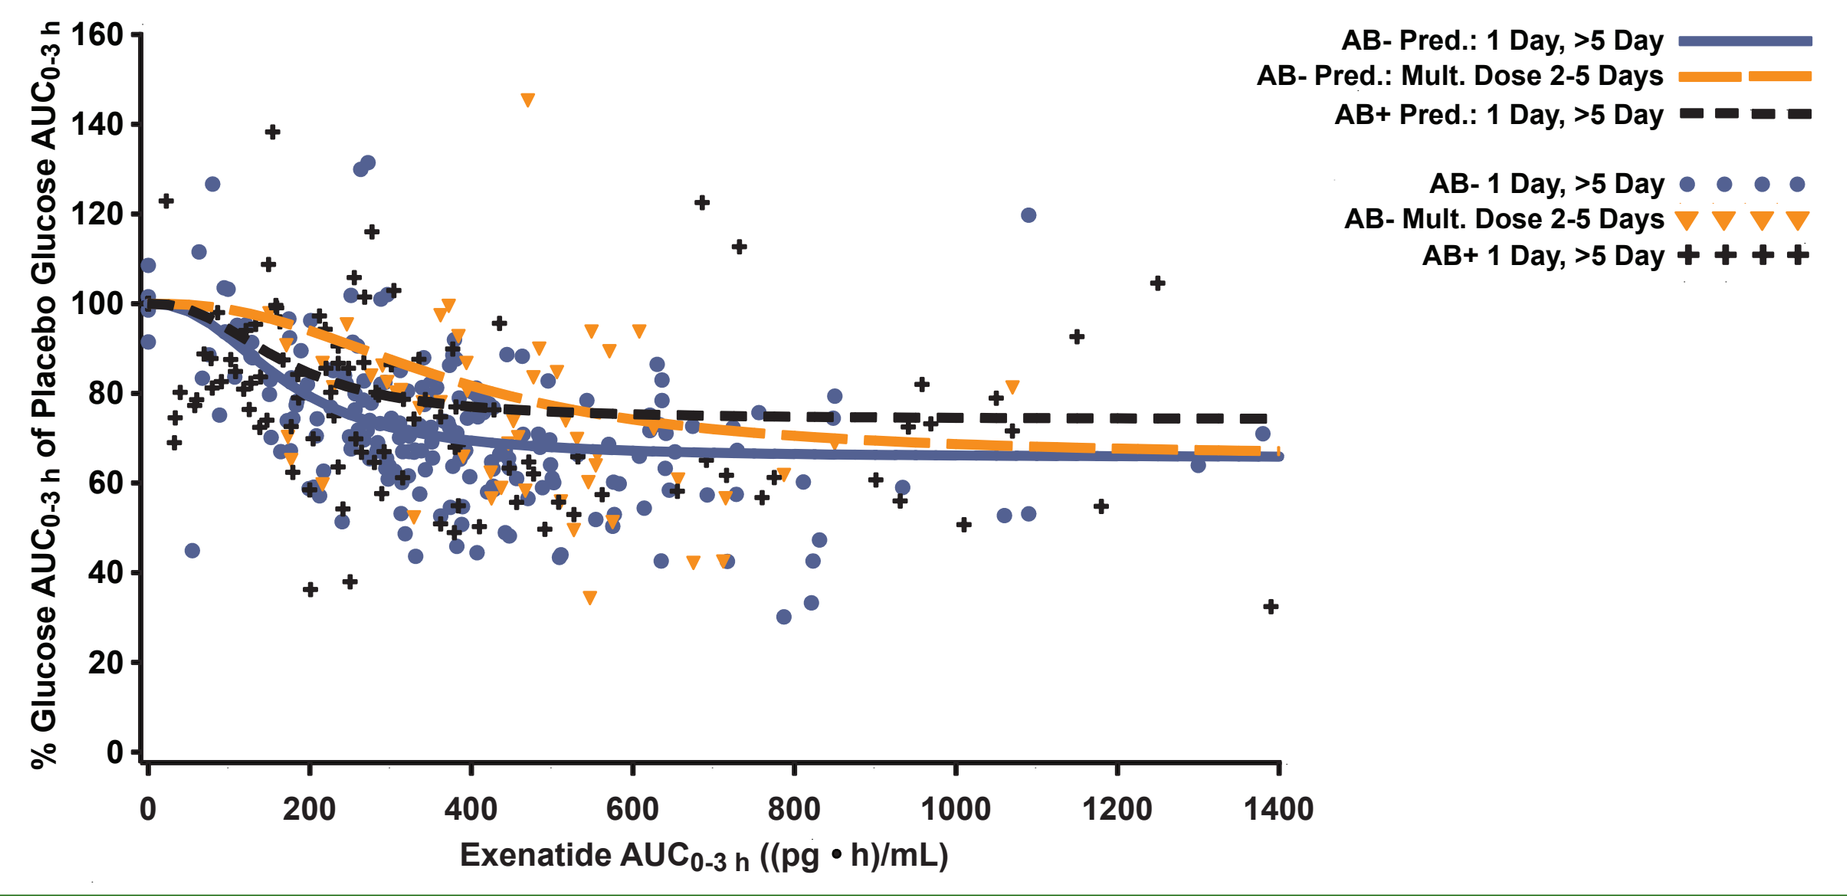
AB- Pred.: 1 Day, >5 Day
AB- Pred.: Mult. Dose 2-5 Days
AB+ Pred.: 1 Day, >5 Day
AB- 1 Day, >5 Day
AB- Mult. Dose 2-5 Days
AB+ 1 Day, >5 Day
% Glucose $AUC_{0-3\,h}$ of Placebo Glucose $AUC_{0-3\,h}$
Exenatide $AUC_{0-3\,h}$ ((pg • h)/mL)

- The population mean estimated decrease in glucose was predicted for all subjects following chronic doses of 5 µg and 10 µg BID
- The results are shown in Figure 4

Figure 4. Population mean predicted % reduction in $gAUC_{0-3h}$

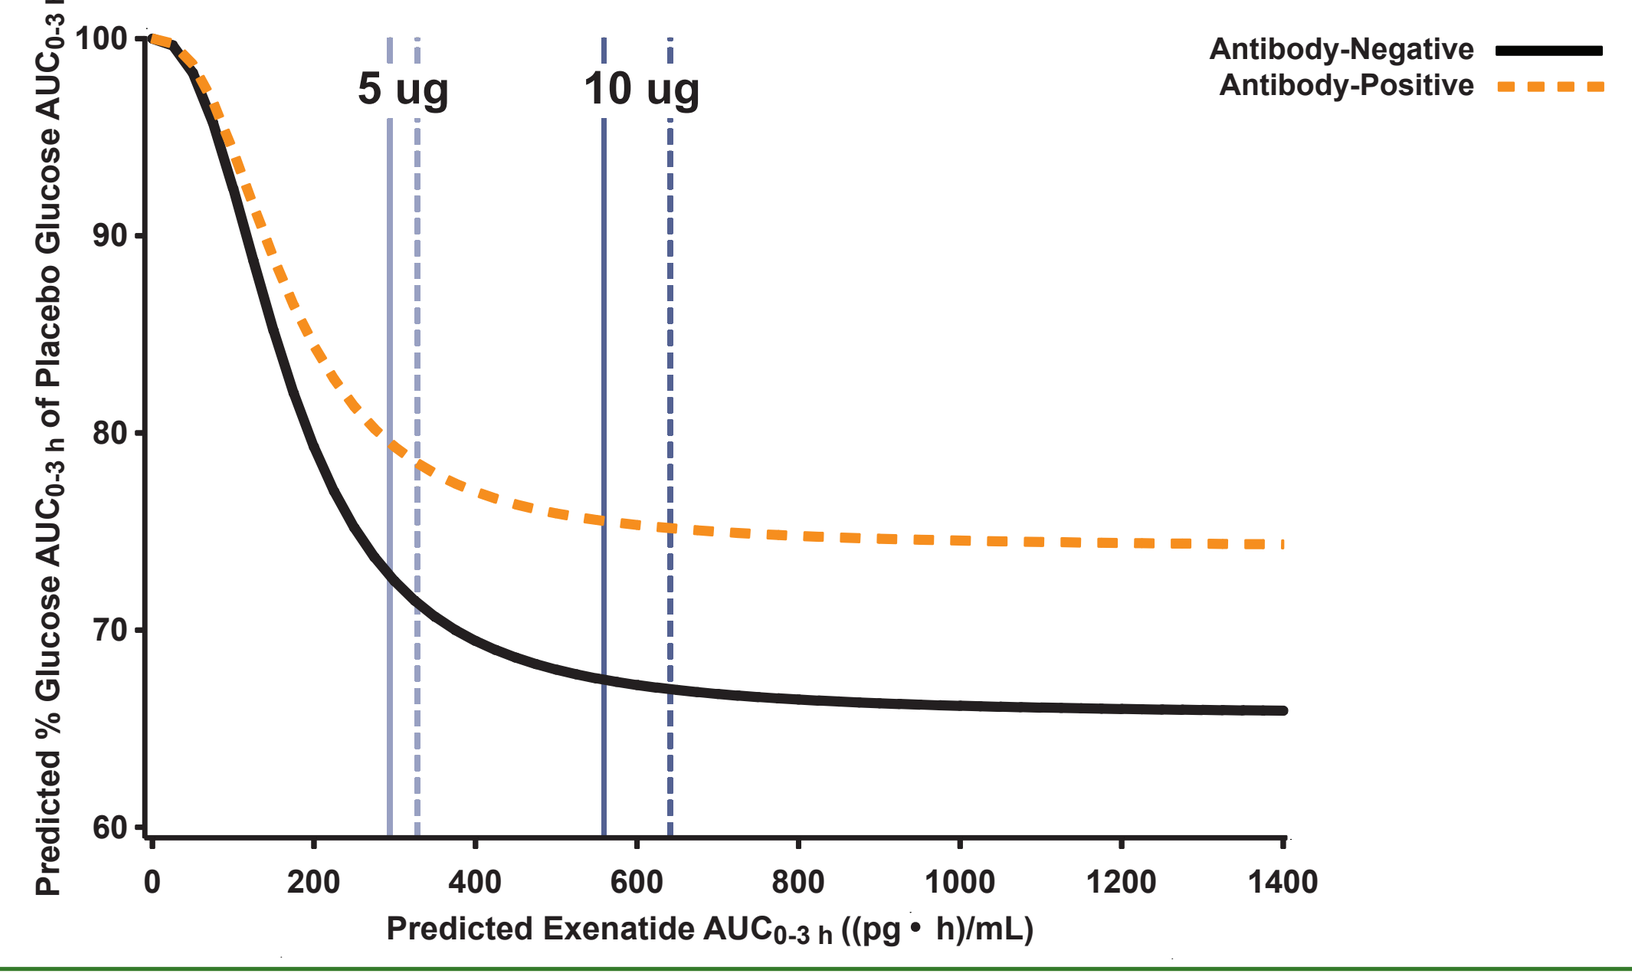
Antibody-Negative
Antibody-Positive
5 ug
10 ug
Predicted % Glucose $AUC_{0-3\,h}$ of Placebo Glucose $AUC_{0-3\,h}$
Predicted Exenatide $AUC_{0-3\,h}$ ((pg • h)/mL)

- Both doses are approaching the asymptotic portion of the exposure-response curve

Table 4. Percentage of subjects with a Bayesian-predicted glucose response (reduction in glucose AUC_{0-3h}) greater than 50%, 75%, 85%, and 90% of their individual Bayesian-predicted E_{max}

Dose	Frequency of Subjects (n) Above % of Individual Bayesian E_{max}: >50%	>75%	>85%	>90%
5 µg	82.5% (151)	54.6% (100)	38.3% (70)	26.8% (49)
10 µg	94.5% (173)	89.6% (164)	79.2% (145)	65.6% (120)

- With exenatide doses of 5 µg, 55% of subjects achieved 75% of their predicted maximum glucose reduction; with 10-µg doses, 90% achieved that same target

Table 5. Relationship between fixed dose of exenatide and predicted percentage decrease in gAUC, stratified by model refinement

Dose	Initial Model (Phases 1 and 2): 5 µg	Initial Model (Phases 1 and 2): 10 µg	Refined Final Model (Phases 1, 2, and 3): 5 µg	Refined Final Model (Phases 1, 2, and 3): 10 µg
% decrease in gAUC (mg • h/dL)				
<20%	39	23.4	31.1	15.8
20-40%	47.5	55.9	56.8	61.2
>40%	13.5	19.8	12.0	23.0
Predicted % of subjects with ≥20% decrease in gAUC	61	77	69	81

- Simulated outcomes were similar between initial model development efforts and refined model

Confirmation of Fixed-Dose Regimen with Clinical Data

- Final Population PK model predicts an approximate 25% and 33% decrease in gAUC for 5-µg and 10-µg fixed doses, respectively
- This translates into a gAUC ratio (treatment to placebo) of 0.75 and 0.67 for 5 µg and 10 µg, respectively
- Phase 3 Clinical Trial Experience (3 clinical trials combined)

Table 6. Geometric mean (SE) ratio of postprandial $gAUC_{0-3h}$ (mg·min/mL)

Ratio*	Dose: 5 µg (n = 42)	Dose: 10 µg (n = 52)
Week 4: Day 1	0.68 (0.032)	0.70 (0.026)
Week 30: Day 1	0.76 (0.030)	0.72 (0.033)

*Day 1 measurement taken prior to the date of the first dose.

Conclusions

- These results corroborate the 5 µg and 10 µg fixed doses that were selected for later Phases 2 and 3 clinical trials based upon population modeling and simulation analyses implemented early in clinical drug development
- The value and effectiveness of the learn-and-confirm paradigm was successfully demonstrated from these analyses
- Preliminary population exposure-response modeling of data from early clinical studies and simulations suggested that 10 µg BID dosing would best balance effectiveness and tolerability (with ~77% of subjects expected to achieve ≥20% decrease in glucose exposure)
- Model predictions remained comparable as additional data became available from Phases 2 and 3 trials

References

1. Kolterman OG, Buse JB, Fineman MS, Gaines E, Heintz S, Bicsak TA, et al. Synthetic exendin-4 (exenatide) significantly reduces postprandial and fasting plasma glucose in subjects with type 2 diabetes. J Clin Endocrinol Metab 2003; 88:3082-3089.
2. Nielsen LL, Baron AD. Pharmacology of exenatide (synthetic exendin-4) for the treatment of type 2 diabetes. Curr Opin Investig Drugs 2003; 4:401-405.
3. Fineman MS, Bicsak TA, Shen LZ, Taylor K, Gaines E, Varns A, et al. Effect on glycemic control of exenatide (synthetic exendin-4) additive to existing metformin and/or sulfonylurea treatment in patients with type 2 diabetes. Diabetes Care 2003; 26:2370-2377.
4. Nielsen LL, Young AA, Parkes DG. Pharmacology of exenatide (synthetic exendin-4): A potential therapeutic for improved glycemic control of type 2 diabetes. Regul Pept 2004; 117:77-88.
5. BYETTA® (exenatide) injection prescribing information.
6. Phillips L, Fineman M, Taylor K, Baron A, Ludwig E, Grasela T. Population modeling to guide phase 3 dose selection for AC2993 (synthetic exendin-4). Clin Pharmacol Ther 2002; 71:P29.