Modeling and Simulation of Acetaminophen Pharmacokinetics and Hepatic Biomarkers After Overdoses of Extended-Release and Immediate-Release Formulations with DILIsym, a Quantitative Systems Toxicology (QST) Software Platform

B. A. Howell[1], K. Yang[1], J. J. Beaudoin[1], Z. Kenz[1], V. V. Lakhani[1], J. L. Woodhead[1], J. C. Lai[2], C. K. Gelotte[3], S. Sista[2], E. Atillasoy[2,*]

[1]QSP Solutions, Simulations Plus, Inc., RTP, NC; [2]Kenvue, Montgomery Township, NJ; [3]Independent Consultant, Westbrook, CT

[*]**Contact information:** eatillas@kenvue.com

BACKGROUND & PURPOSE

- The analgesic/antipyretic acetaminophen (APAP) has multiple formulations including immediate-, modified-, and extended-release preparations
- In 2017, the European Medicines Agency recommended suspending medicines containing a modified-release (MR) preparation from the market as there was concern that in large overdose, it could form a bezoar resulting in unexpected pharmacokinetics (PK), with markedly prolonged absorption and delayed plasma peak concentrations, compared to an immediate-release (IR) formulation
- There was also a concern that current treatment guidelines developed for APAP overdose were inappropriate for MR formulations
- In the US, the extended-release (ER) formulation of APAP (TYLENOL® 8 HR Arthritis Pain, Johnson & Johnson Consumer Inc., NJ) is designed with an IR layer and an erodible ER layer
- This project utilized a modeling and simulation approach to compare predicted PK and hepatotoxicity biomarkers following various acute overdose and repeated supratherapeutic ingestion (RSTI) scenarios to determine if there is a difference between the US APAP-IR and APAP-ER preparations

METHODS

- The existing APAP-IR representation within DILIsym® v8A, a QST model of drug-induced liver injury (DILI), was updated [*e.g.*, formation of the reactive metabolite N-acetyl-*p*-benzoquinone imine (NAPQI) and other APAP metabolites] using newly acquired *in vitro* and clinical datasets (**Fig. 1**)
- Exposure-dependent cytochrome P450 (CYP) 2E1- and CYP3A4-mediated contributions to NAPQI formation (>85% and <15%, respectively) were implemented
- An APAP-ER model was developed by modifying the representation of APAP absorption in the APAP-IR model to recapitulate the fast- and slow-release from IR and ER layers of the APAP-ER caplet and was verified with clinical data (**Fig. 2**)
- *In vitro* dissolution of APAP from the IR and ER formulations was experimentally evaluated using different systems (*e.g.*, Tiny-TNO Gastro-Intestinal Model smartificialgut, Tiny-TIM*sg*)
- These experimental data were used as input for the Z-factor (Takano) dissolution model within GastroPlus® to inform the *in vivo* dose-vs-fraction absorbed (Fa) relationship for APAP-IR and APAP-ER that was subsequently leveraged within the DILIsym model (**Fig. 3**)
- Simulated populations (SimPops®) representing healthy adults, moderate chronic alcohol users (MCAU), excessive chronic alcohol users (ECAU), and individuals with low glutathione (GSH) were developed and verified using clinical PK and hepatic biomarker data (**Fig. 4**)
- The DILIsym model was then used to simulate PK and three clinically useful hepatic biomarkers [plasma alanine aminotransferase (ALT), total bilirubin (TB), and international normalized ratio (INR)] after single acute overdoses (3.9, 9.75, 19.5, 32.5, 65, 78 and 100.1 g; **Fig. 5**; **Table 1**) and RSTI (3.9, 5.2 and 7.8 g/day; **Table 2**) of APAP-ER and APAP-IR
- Simulations were carried out in the absence of clinical interventions (*e.g.*, decontamination, N-acetylcysteine treatment), such that they represent worst-case scenarios

RESULTS

Fig. 1: Updated APAP metabolism simulations reasonably recapitulated clinically observed percent urinary recovery of metabolites over wide dose ranges[1-32]. Simulations averaged within 9.55% of the dose-response lines generated from clinical data.

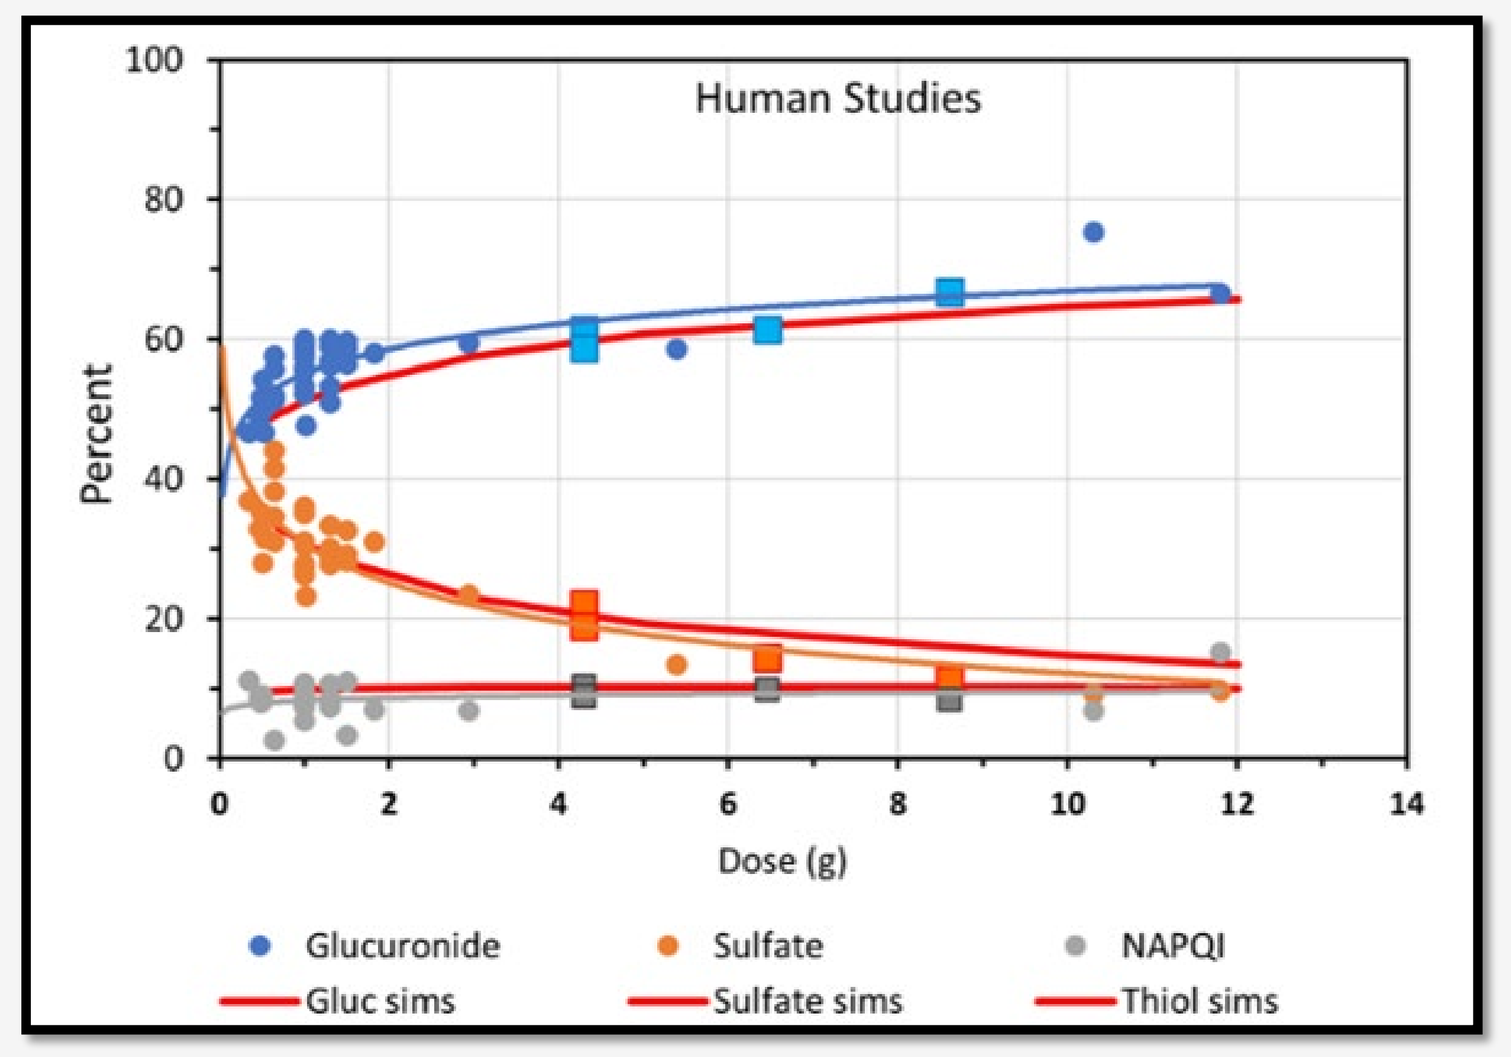
Human Studies
Percent
Dose (g)
Glucuronide
Sulfate
NAPQI
Gluc sims
Sulfate sims
Thiol sims

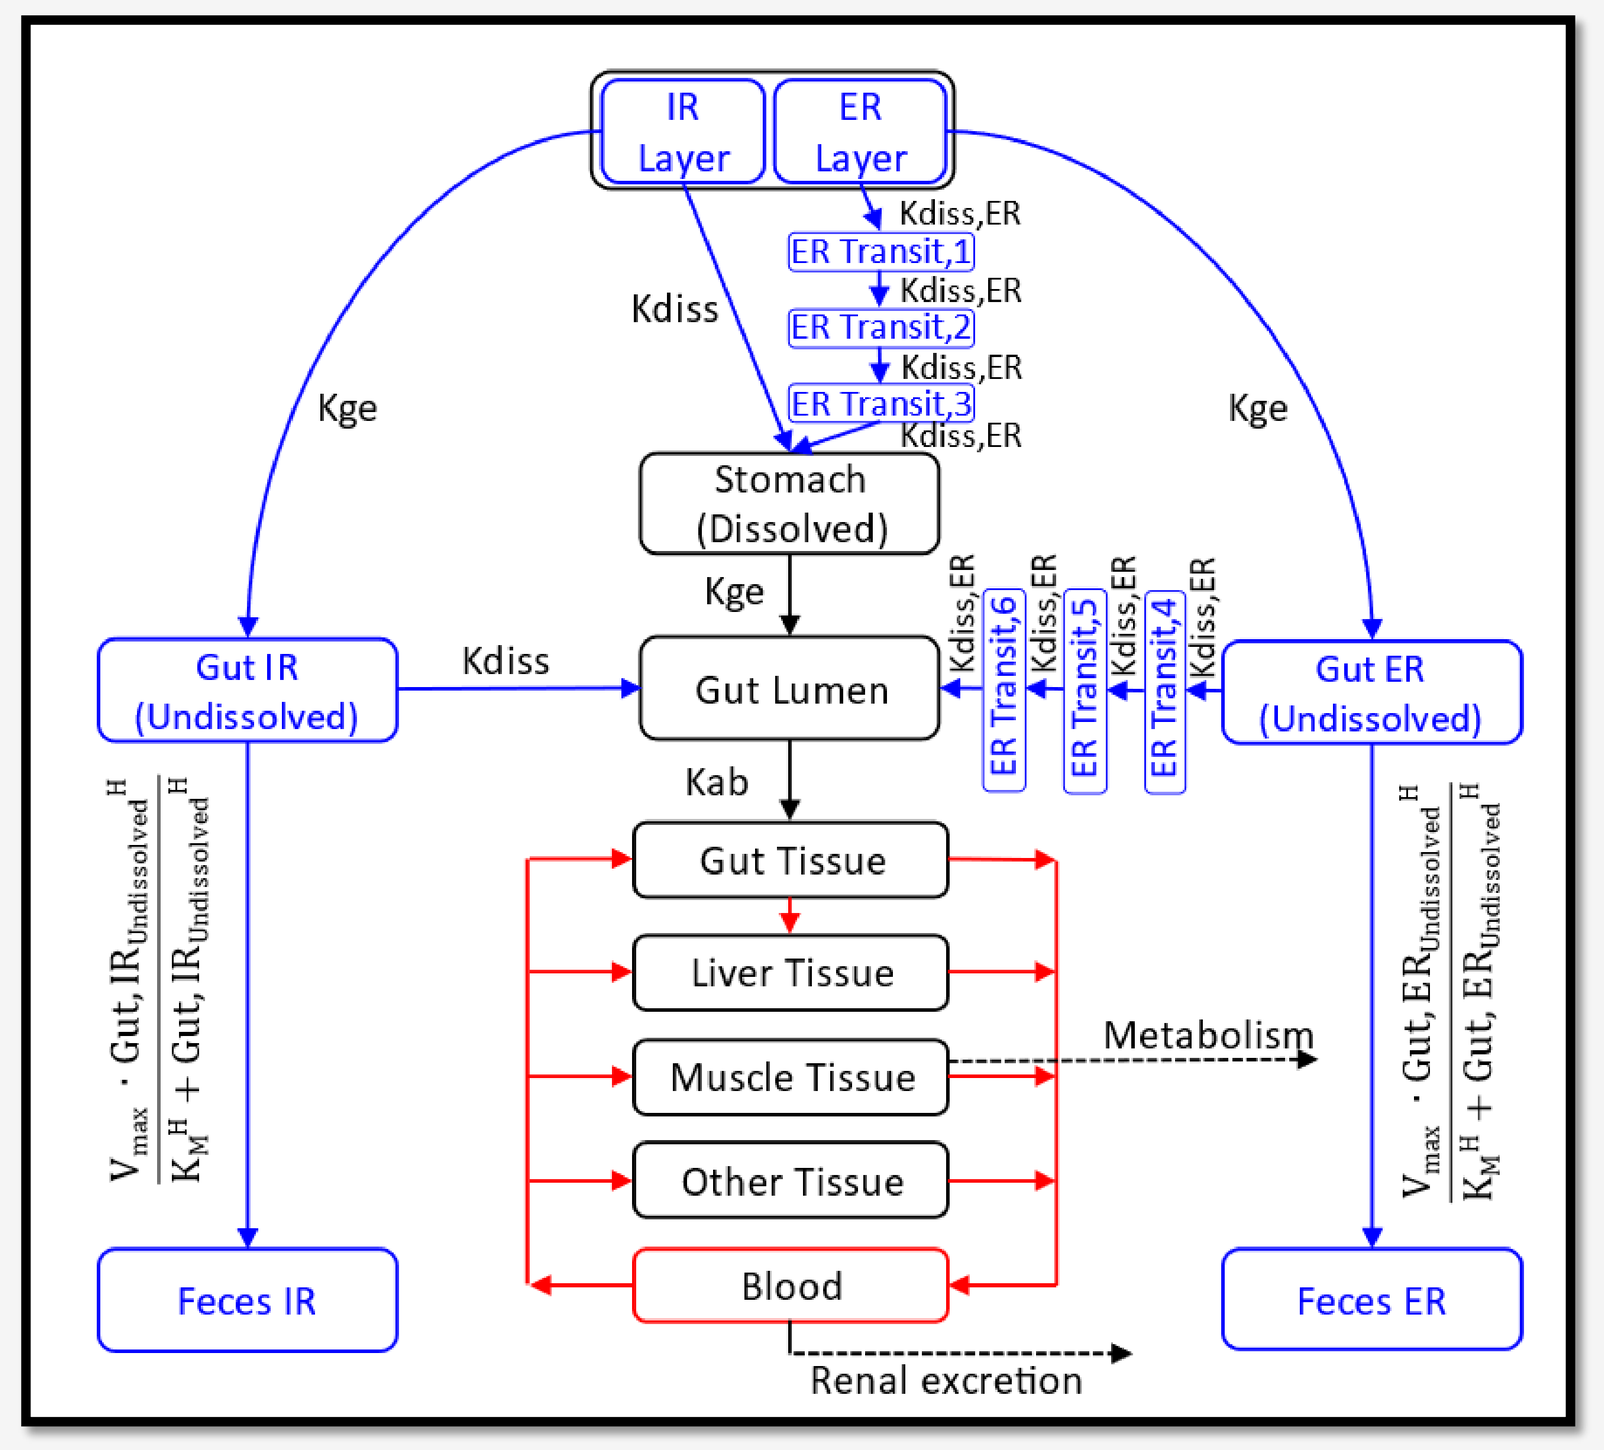
IR Layer
ER Layer
ER Transit,1
ER Transit,2
ER Transit,3
ER Transit,4
ER Transit,5
ER Transit,6
Kdiss,ER
Kdiss
Kge
Stomach (Dissolved)
Gut IR (Undissolved)
Gut Lumen
Gut ER (Undissolved)
Kab
Gut Tissue
Liver Tissue
Muscle Tissue
Other Tissue
Blood
Metabolism
Renal excretion
Feces IR
Feces ER

Fig. 2: The APAP model within DILIsym was revised to accommodate administration of APAP-IR and APAP-ER formulations. For APAP-IR simulations, 100% of APAP follows the IR layer pathway, while for APAP-ER simulations, 50% of APAP uses the IR layer pathway, and another 50% uses the ER layer pathway. Once absorbed in the gut, APAP derived from the APAP-IR or APAP-ER layers are handled in a similar fashion.

Fig. 3: Mechanistic modeling of *in vitro* dissolution data predicted that the dose-dependent human *in vivo* Fa (%) of APAP-ER was generally lower than for APAP-IR and declined from ~100% to ~50% when increasing the APAP-IR and APAP-ER dose from therapeutic doses to 100 g overdoses. APAP Fa for both formulations as predicted in GastroPlus (solid lines) was reasonably recapitulated by the DILIsym model (dashed lines) of APAP-IR and APAP-ER. USP, United States Pharmacopeia.

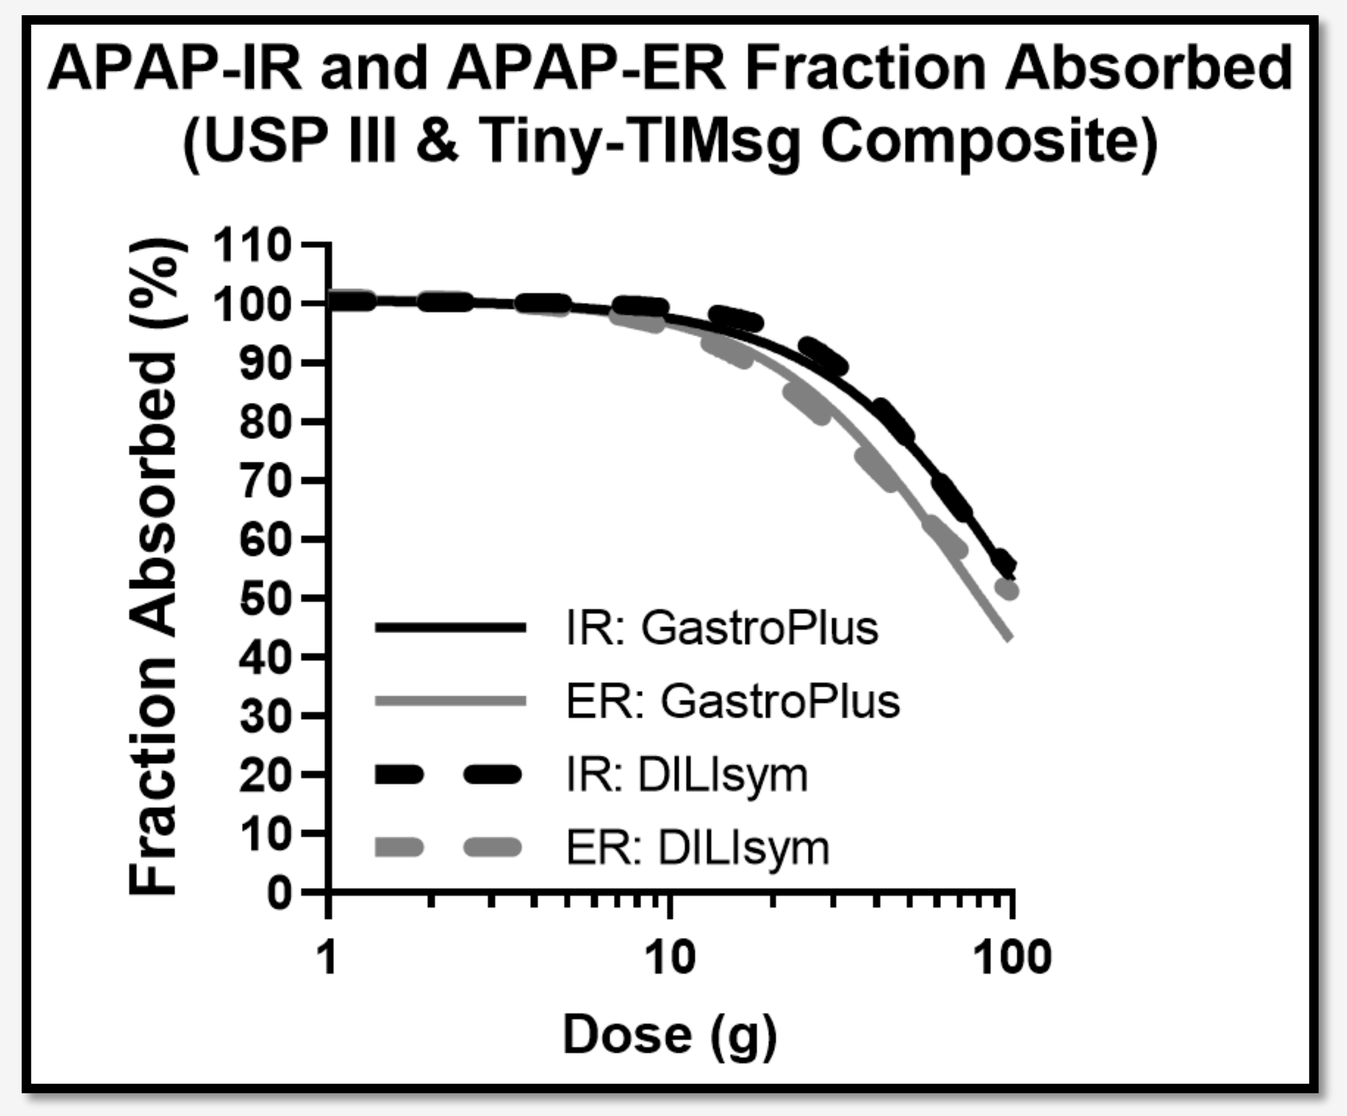
APAP-IR and APAP-ER Fraction Absorbed
(USP III & Tiny-TIMsg Composite)
Fraction Absorbed (%)
Dose (g)
IR: GastroPlus
ER: GastroPlus
IR: DILIsym
ER: DILIsym

Ingested Dose (g)	AUC_∞ (µg·h/mL)		C_{max} (µg/mL)		Fa		ALT_{max} (U/L)		TB_{max} (mg/dL)		INR_{max}	
	APAP-ER	APAP-IR	APAP-ER	APAP-IR	APAP-ER	APAP-IR	APAP-ER	APAP-IR	APAP-ER	APAP-IR	APAP-ER	APAP-IR
3.9	195.7 (24.10%)	200.0 (23.97%)	32.8 (19.73%)	46.1 (18.67%)	1.00 (1.00-1.00)	1.00 (1.00-1.00)	30.0 (30.00-30.00)	30.0 (30.00-30.00)	0.55 (0.55-0.55)	0.55 (0.55-0.55)	1.0 (1.00-1.00)	1.0 (1.00-1.00)
9.75	542.7 (25.35%)	553.9 (25.44%)	84.5 (19.78%)	103.4 (19.08%)	0.97 (0.95-0.99)	1.00 (0.99-1.00)	36.8 (30.00-201.65)	38.3 (30.00-225.90)	0.55 (0.55-0.59)	0.55 (0.55-0.59)	1.0 (1.00-1.00)	1.0 (1.00-1.00)
19.5	1046.6 (26.79%)	1174.2 (27.02%)	155.7 (20.27%)	201.2 (19.07%)	0.87 (0.81-0.94)	0.97 (0.89-0.99)	272.5 (30.00-4491.62)	421.0 (30.00-6998.37)	0.62 (0.55-2.29)	0.68 (0.55-6.60)	1.0 (1.00-1.86)	1.0 (1.00-4.93)
32.5	1634.6 (27.99%)	1903.2 (28.28%)	234.2 (20.53%)	311.4 (20.27%)	0.77 (0.69-0.87)	0.89 (0.74-0.97)	1268.8 (30.01-8640.56)	1984.7 (32.37-8885.89)	1.12 (0.55-13.86)	1.64 (0.56-12.05)	1.2 (1.00-7.91)	1.5 (1.00-7.92)
65	2841.9 (29.96%)	3255.1 (31.66%)	392.5 (20.83%)	495.6 (22.89%)	0.61 (0.51-0.74)	0.70 (0.50-0.88)	4104.9 (116.74-8894.11)	4756.5 (197.77-8816.65)	3.44 (0.58-15.08)	4.10 (0.59-14.43)	2.5 (1.00-7.90)	2.7 (1.00-7.87)
78	3237.5 (30.59%)	3646.7 (32.59%)	443.7 (21.04%)	545.9 (23.75%)	0.57 (0.46-0.70)	0.64 (0.44-0.84)	4688.0 (176.50-8688.58)	5119.3 (263.87-8741.29)	3.95 (0.59-14.22)	4.53 (0.60-14.88)	2.6 (1.00-7.79)	2.9 (1.00-7.91)
100.1	3822.7 (31.53%)	4190.9 (33.88%)	518.6 (21.54%)	612.9 (24.97%)	0.51 (0.39-0.65)	0.56 (0.36-0.79)	5249.2 (273.63-8800.01)	5398.2 (366.74-8690.06)	4.66 (0.62-14.86)	4.97 (0.61-15.23)	2.9 (1.00-7.92)	3.0 (1.00-7.94)

Parameter	Scenario A (3.9 g/day)		Scenario B (5.2 g/day)		Scenario C (7.8 g/day)	
	APAP-ER	APAP-IR	APAP-ER	APAP-IR	APAP-ER	APAP-IR
AUC_τ (µg·h/mL)	55.2 (25.69%)	54.9[b] (25.73%)	56.6 (26.60%)	57.5 (26.30%)	59.5 (27.97%)	60.2 (27.70%)
$C_{max,ss}$ (µg/mL)	11.9 (21.52%)	10.9 (21.45%)	13.7 (22.94%)	18.6 (20.49%)	18.0 (25.42%)	23.2 (22.63%)
AccR	0.97 (0.85-1.18)	1.01 (0.85-1.29)	0.99 (0.85-1.26)	0.97 (0.84-1.21)	1.04 (0.85-1.43)	1.01 (0.84-1.36)
Fu APAP (%)	3.2 (23.22%)	3.2 (23.34%)	3.3 (24.20%)	3.4 (23.90%)	3.5 (25.72%)	3.5 (25.45%)
Fu APAP-G (%)	60.3 (13.36%)	60.1 (13.41%)	61.7 (12.58%)	62.5 (12.43%)	64.2 (11.26%)	64.8 (11.19%)
Fu APAP-S (%)	21.7 (28.49%)	22.1 (28.26%)	20.1 (28.73%)	19.2 (29.14%)	17.2 (29.22%)	16.6 (29.49%)
Fu Thiols (%)	9.8 (22.81%)	9.7 (22.88%)	9.8 (22.39%)	9.7 (22.50%)	9.4 (21.61%)	9.3 (21.73%)
Fu Catechols (%)	4.9 (35.64%)	4.9 (35.71%)	5.0 (35.99%)	5.0 (35.93%)	5.2 (36.43%)	5.2 (36.39%)
ALT_{max} (U/L)	31.5 (30.00-137.79)	31.5 (30.00-136.25)	36.0 (30.00-263.33)	35.9 (30.00-262.47)	56.7 (30.00-386.29)	56.1 (30.00-385.04)
TB_{max} (mg/dL)	0.55 (0.55-0.57)	0.55 (0.55-0.57)	0.55 (0.55-0.59)	0.55 (0.55-0.59)	0.56 (0.55-0.62)	0.56 (0.55-0.62)
INR_{max}	1.0 (1.00-1.00)	1.0 (1.00-1.00)	1.0 (1.00-1.00)	1.0 (1.00-1.00)	1.0 (1.00-1.00)	1.0 (1.00-1.00)

Table 1 (left) and **Table 2** (right): Comparison of simulated plasma APAP PK parameters and hepatic biomarkers after acute overdoses (Table 1) or RSTI (Table 2) of APAP-ER and APAP-IR in healthy adults. On average, APAP exposure after acute overdose or RSTI in healthy adults was predicted to be lower for APAP-ER compared to APAP-IR. Similar ALT, TB and INR levels were predicted for APAP-ER and APAP-IR after overdoses in healthy adults. These findings were similar when making comparisons between APAP-ER and APAP-IR in the compromised adult SimPops (*i.e.*, MCAU, ECAU, low GSH).

Simulated exposure parameters (AUC_∞, C_{max}, AUC_τ and $C_{max,ss}$) as well as fraction excreted in urine (Fu) for 300 virtual adults in the APAP-ER and APAP-IR SimPops are reported as mean (% coefficient of variation), whereas Fa, and simulated maximum concentrations of hepatic biomarkers (ALT_{max}, TB_{max}, and INR_{max}) are reported as median and range. Baseline biomarker values in the DILIsym model are 30, 0.55 and 1 for ALT (U/L), TB (mg/dL) and INR, respectively.

Fig. 4: Calibration and validation of APAP-IR and APAP-ER models[32-34]. A large variety of clinical PK datasets (examples shown) and hepatic biomarker datasets (not shown) at therapeutic and overdose levels for both APAP-IR and APAP-ER were reasonably recapitulated by the baseline (top panels) and SimPops (bottom panels) models. Healthy SimPops for both APAP-IR and APAP-ER covered >80% of individual clinical PK data (C_{max} and AUC values).

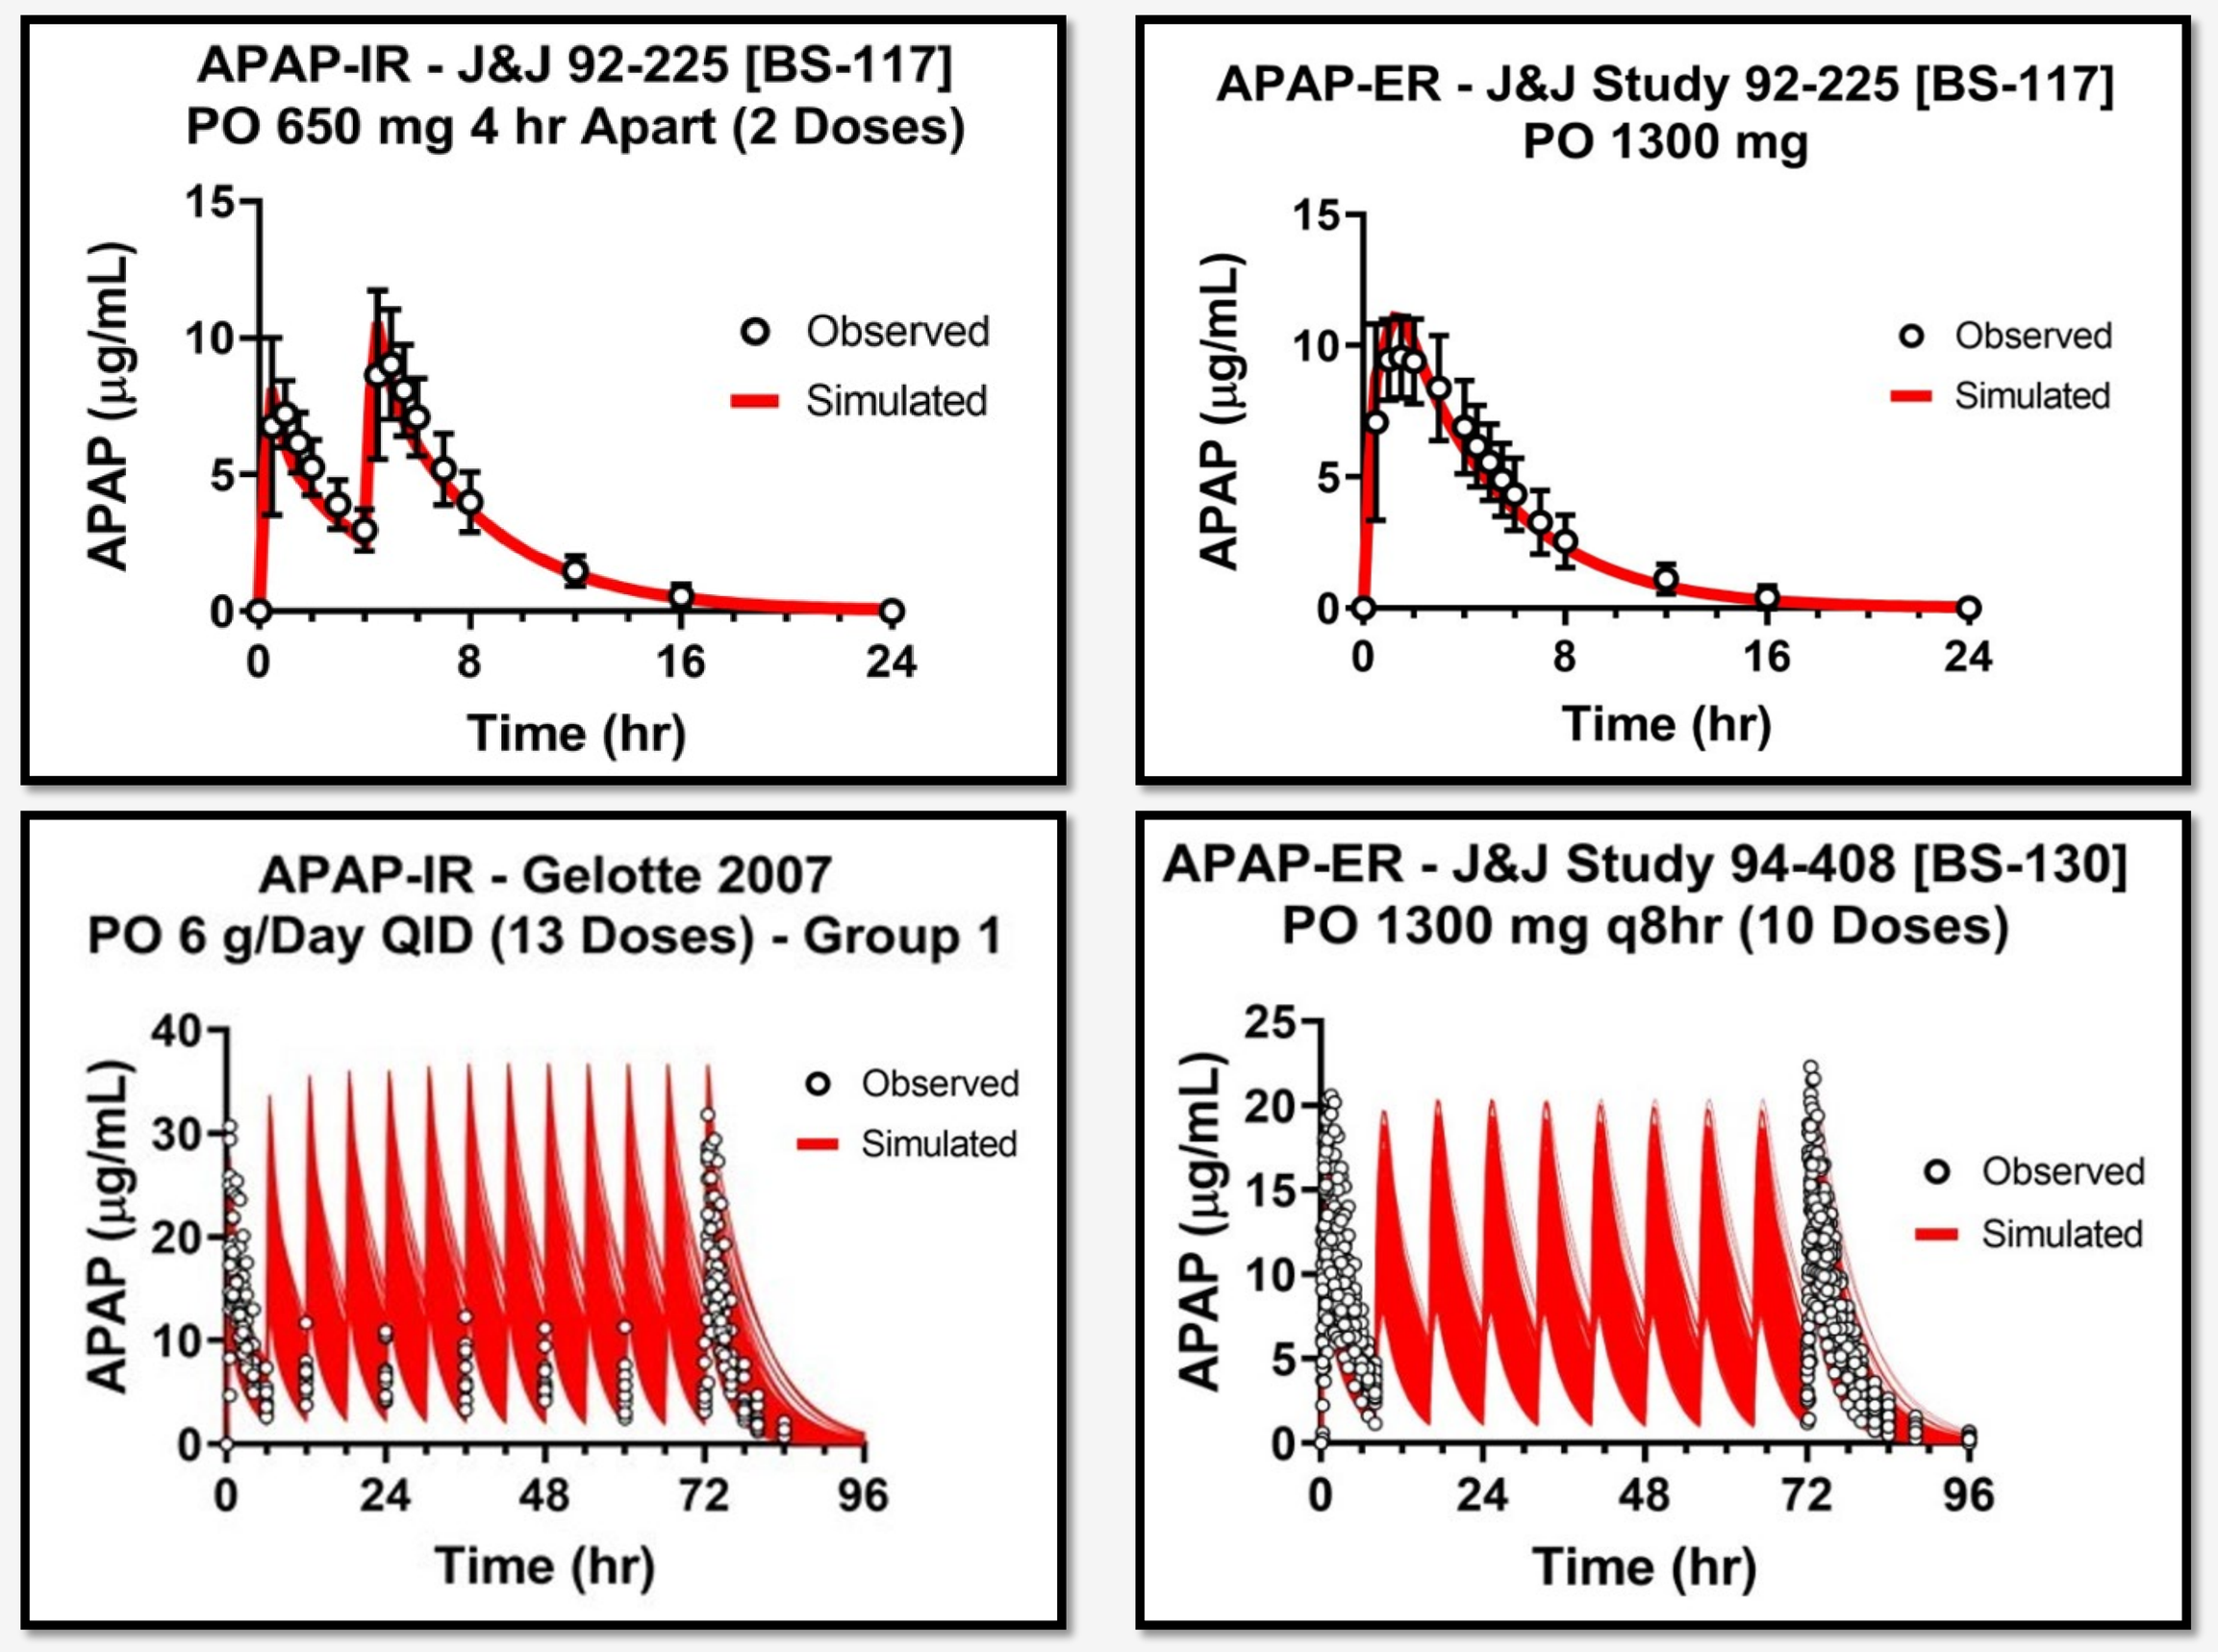
APAP-IR - J&J 92-225 [BS-117]
PO 650 mg 4 hr Apart (2 Doses)
APAP-ER - J&J Study 92-225 [BS-117]
PO 1300 mg
APAP-IR - Gelotte 2007
PO 6 g/Day QID (13 Doses) - Group 1
APAP-ER - J&J Study 94-408 [BS-130]
PO 1300 mg q8hr (10 Doses)
APAP (µg/mL)
Time (hr)
Observed
Simulated

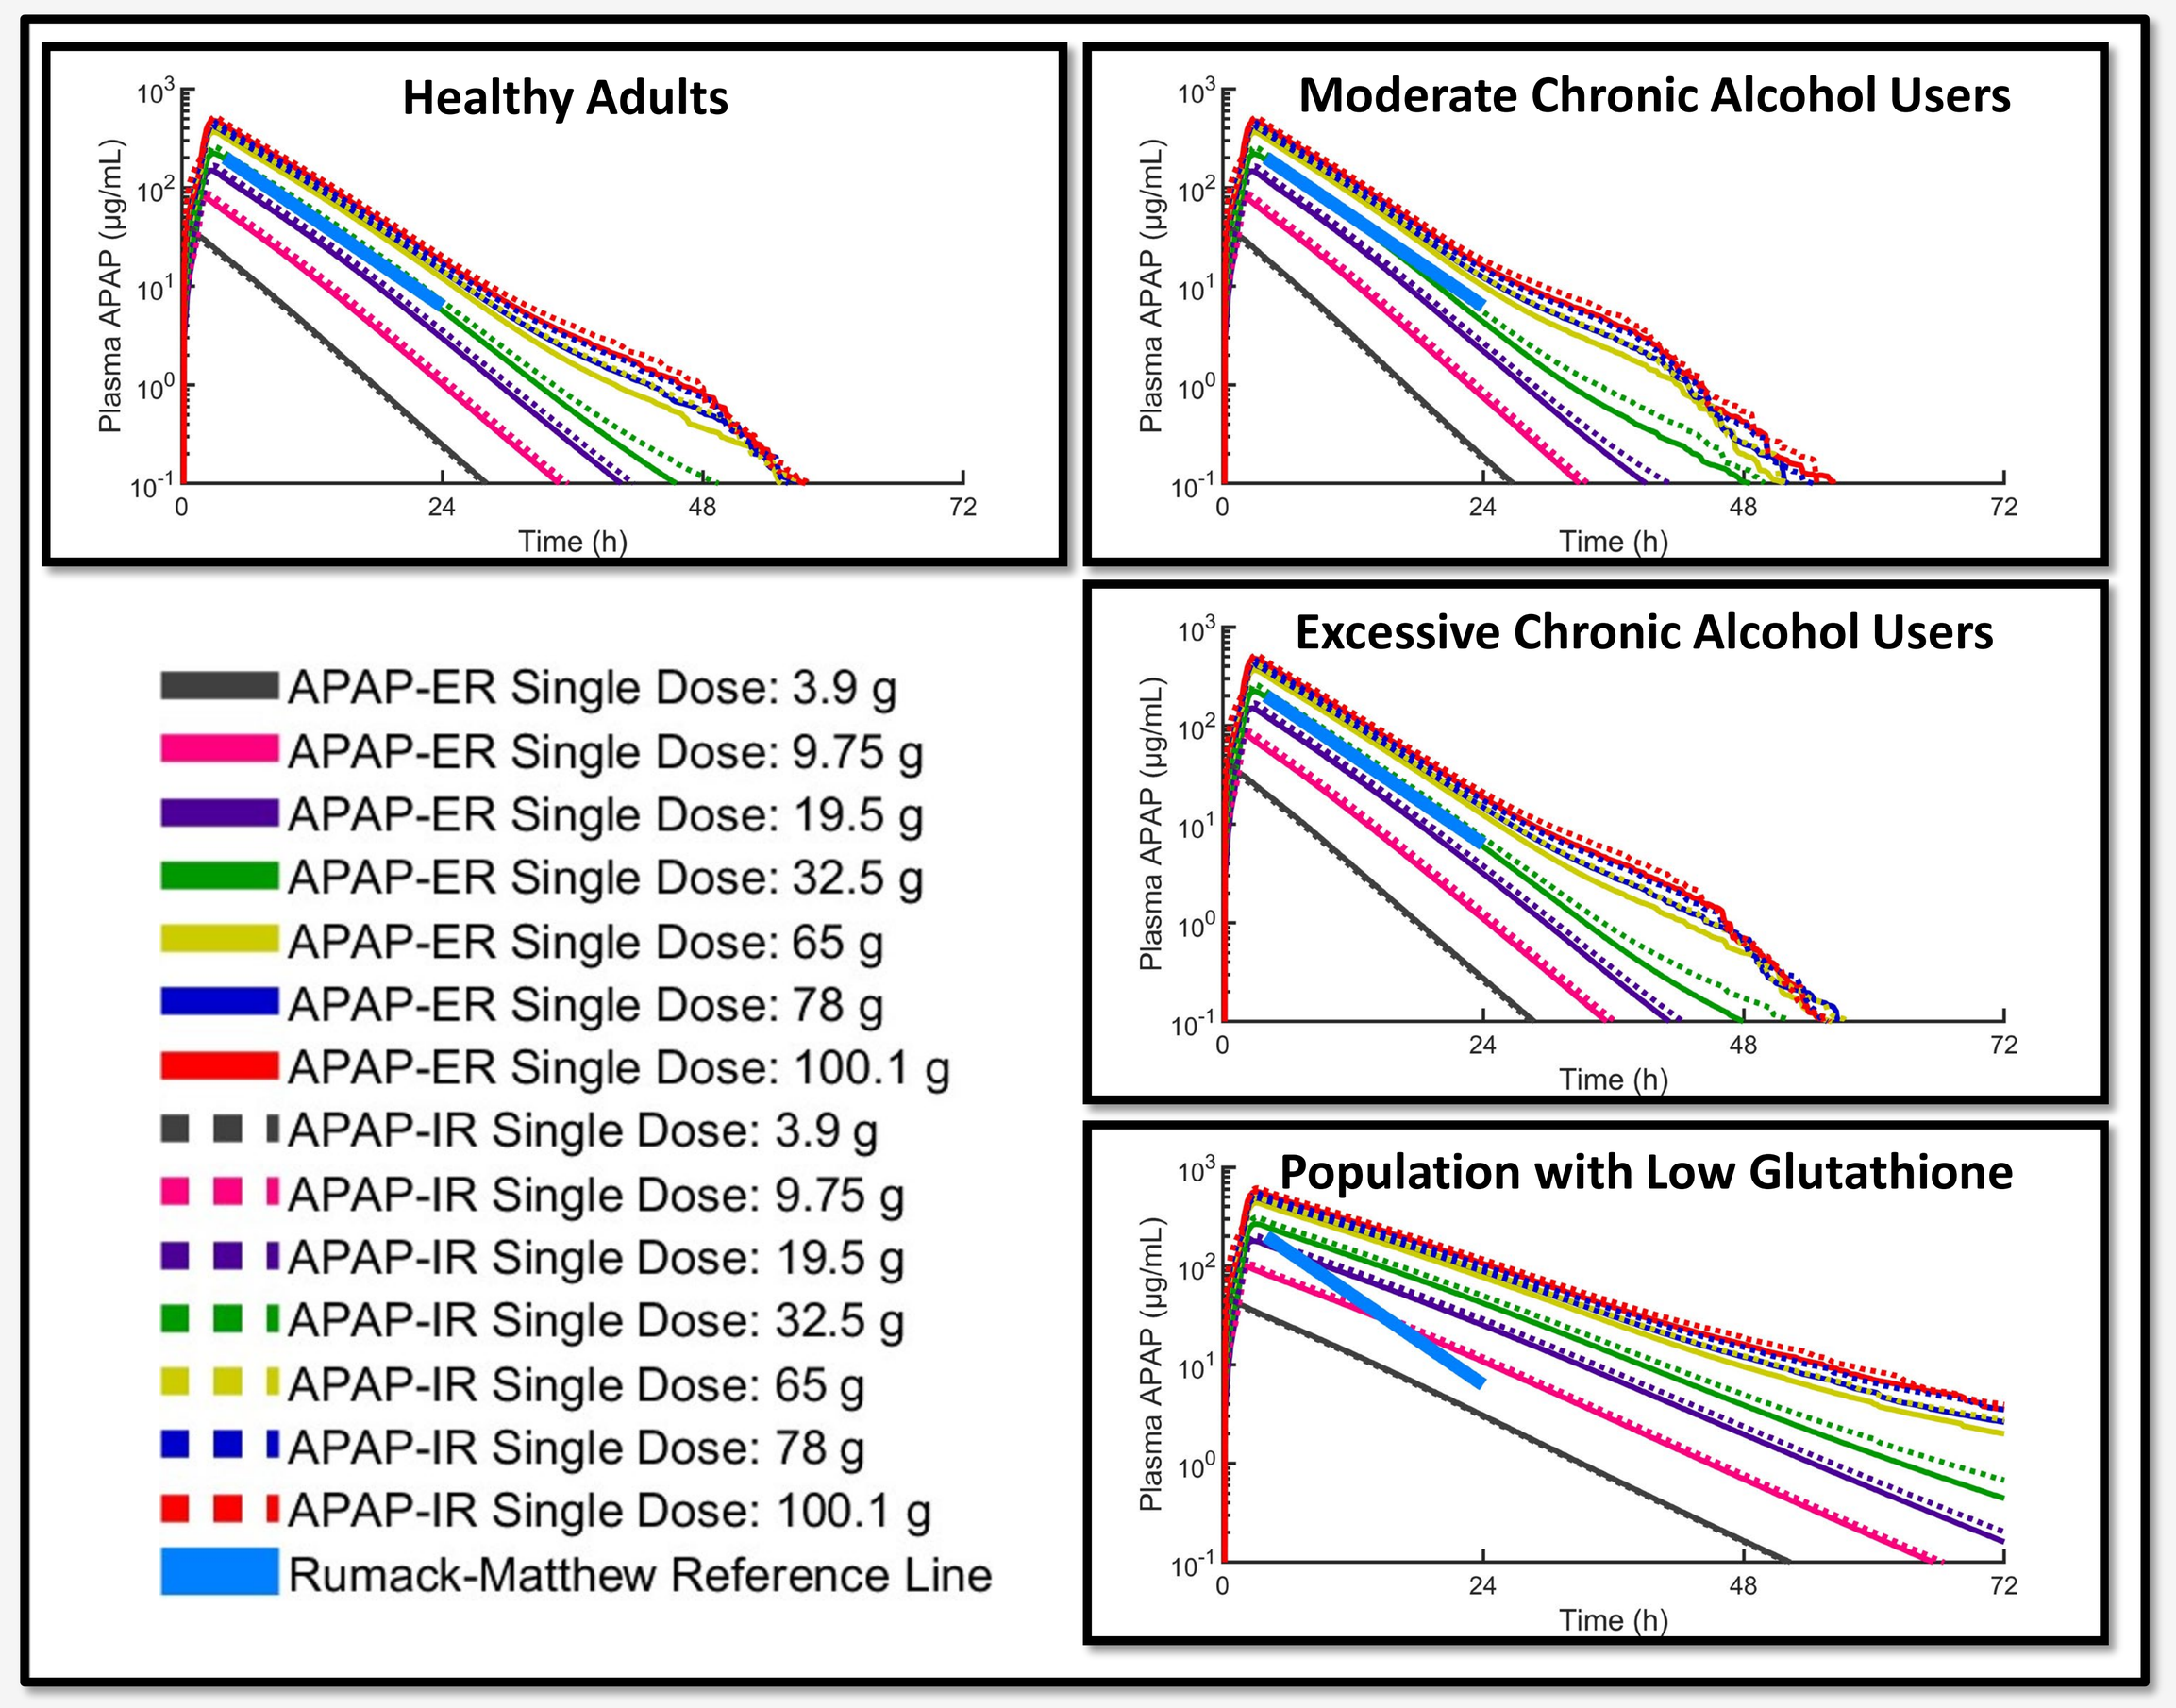
Healthy Adults
Moderate Chronic Alcohol Users
Excessive Chronic Alcohol Users
Population with Low Glutathione
Plasma APAP (µg/mL)
Time (h)
APAP-ER Single Dose: 3.9 g
APAP-ER Single Dose: 9.75 g
APAP-ER Single Dose: 19.5 g
APAP-ER Single Dose: 32.5 g
APAP-ER Single Dose: 65 g
APAP-ER Single Dose: 78 g
APAP-ER Single Dose: 100.1 g
APAP-IR Single Dose: 3.9 g
APAP-IR Single Dose: 9.75 g
APAP-IR Single Dose: 19.5 g
APAP-IR Single Dose: 32.5 g
APAP-IR Single Dose: 65 g
APAP-IR Single Dose: 78 g
APAP-IR Single Dose: 100.1 g
Rumack-Matthew Reference Line

Fig. 5: Simulated mean APAP PK profiles after acute overdoses of APAP-ER (solid lines) and APAP-IR (dotted lines) in healthy adults, moderate chronic alcohol users, excessive chronic alcohol users, and a population with low glutathione. Simulated APAP PK profiles showed no apparent Rumack-Matthew reference line crossing differences between APAP-ER and APAP-IR within each population.

CONCLUSION

- Modeling and simulation consistently demonstrate that the difference in PK between the two APAP formulations in the US:
 - ➢ has a relatively small, if any, effect on APAP metabolism and biomarkers of hepatotoxicity following overdose
 - ➢ does not result in marked differences in the expected time course of APAP plasma concentrations
- Based on these results, the updated APAP overdose treatment guidelines, published in 2023, are not further impacted by this report

REFERENCES

1:Slattery et al. CPT 41, 413–418 (1987); 2:Prescott. Arch. Intern. Med. 141, 386–389 (1981); 3:Manyike et al. CPT 67, 275–282 (2000); 4:Robertson et al. Xenobiotica 2, 339–347 (1972); 5:Amsel and Davison. J Pharm Sci 61, 1474–1475 (1972); 6:Mrochek et al. Clin. Chem. 20, 1086–1096 (1974); 7:Miller et al. CPT 19, 284–294 (1976); 8:Prescott. BJCP 10 Suppl 2, 291S-298S (1980); 9:Reiter and Weinshilboum. CPT. 32, 612–621 (1982); 10:Critchley et al. Lancet 1, 1375–1376 (1983); 11:Miners et al. BJCP 16, 503–509 (1983); 12:Mitchell et al. CPT. 34, 48–53 (1983); 13:Clements et al. BJCP 18, 481–485 (1984); 14:Miners et al. CPT. 35, 480–486 (1984); 15:Mitchell et al. JCI 73, 383–91 (1984); 16:Jayasinghe et al. BJCP 22, 363–366 (1986); 17:Miners et al. BJCP 22, 359–362 (1986); 18:Amouyal et al. Biochem. Pharmacol. 36, 2349–2352 (1987); 19:Bock et al. Eur. J. Clin. Pharmacol. 31, 677–683 (1987); 20:Notarianni et al. BJCP 24, 63–67 (1987); 21:Ullrich et al. Eur. J. Clin. Invest. 17, 237–240 (1987); 22:Miners et al. Eur. J. Clin. Pharmacol. 35, 157–160 (1988); 23:Epstein et al. BJCP 31, 139–142 (1991); 24:Hindmarsh et al. Vet Hum Toxicol 33, 441–445 (1991); 25:Leung et al. Cancer Lett. 57, 45–48 (1991); 26:Rose et al. Ann Emerg Med 20, 1064–1068 (1991); 27:Patel et al. Pharmacogenetics 2, 38–45 (1992); 28:Esteban and Pérez-Mateo. J. Hepatol. 18, 257–258 (1993); 29:Roos et al. Neurology 43, 1373–1376 (1993); 30:Zapater et al. Aliment. Pharmacol. Ther. 20, 29–36 (2004); 31:Critchley et al. J Clin Pharm Ther 30, 179–184 (2005); 32:Gelotte et al. CPT 81, 840–848 (2007); 33:McNeil Consumer Products Company. Report on Biostudy No. 117 (1992); 34:McNeil Consumer Products Company. Report on Study 94-408 (BS-130) (1995).

ACKNOWLEDGMENTS

We thank scientists from the TIM company and Johnson & Johnson for performing the Tiny-TIMsg and USP II/III experiments, respectively.

CONFLICTS OF INTEREST

B.A.H., K.Y., J.J.B, Z.K., V.V.L. and J.L.W are employees of Simulations Plus Inc. J.C.L., S.S., and E.A. are employees of Kenvue Inc. C.K.G. served as a paid consultant for Kenvue Inc. as part of this work.

www.simulations-plus.com